A Phase 3 study of gedatolisib plus fulvestrant with and without palbociclib in patients with HR+/HER2- advanced breast cancer previously treated with a CDK4/6 inhibitor plus a non-steroidal aromatase inhibitor (VIKTORIA-1)

Sara Hurvitz[1], Fabrice Andre[2], Massimo Cristofanilli[3], Giuseppe Curigliano[4], Antonio Giordano[5], Hyo Han[6], Miguel Martin[7], Barbara Pistilli[2], Hope Rugo[8], Robert Wesolowski[9], Samuel Suzuki[10], Sarah C. Mutka[10], Igor Gorbatchevsky[10], Sibylle Loibl[11]

[1] David Geffen School of Medicine, University of California Los Angeles, Los Angeles, CA; [2] Gustave Roussy Cancer Center, Villejuif, France; [3] Weill Department of Medicine, Cornell University, New York, NY; [4] Istituto Europeo di Oncologia, IRCCS, Milano, Italy and University of Milano, Milan, Italy; [5] Dana Farber Cancer Institute, Harvard University, Boston, MA; [6] Moffit Cancer Center Department of Breast Oncology, Florida; [7] Hospital General Universitario Gregorio Marañón, Universidad Complutense, Madrid, Spain; [8] University of California San Francisco Comprehensive Cancer Center, San Francisco, CA; [9] James Cancer Hospital and the Ohio State University Comprehensive Cancer Center, Columbus, OH; [10] Celcuity, Minneapolis, MN; [11] German Breast Group (GBG), Neu-Isenburg, Germany

San Antonio
Breast Cancer Symposium®
December 6–10, 2022

BACKGROUND

Gedatolisib is a Potential First-in-Class PI3K/mTOR Inhibitor

Mechanism of Action

- **First** small molecule dual inhibitor of the PI3K/mTOR pathway administered intravenously
- Inhibits all isoforms of PI3K and mTOR at **low or sub-nanomolar** concentrations

Efficacy

- Compelling efficacy relative to 1st & 2nd line SOC with HR+/HER2- ABC with gedatolisib + ET + CDK4/6i
 - Phase 1b trial (N=103) reported **62% ORR** in 94 response evaluable patients across four expansion arms
 - Median PFS not reached in 1L arm, and 12.9 months in 2L arm with Phase 3 dosing schedule
 - NCT02684032 ; SABCS 2022 Spotlight Discussion PD13-05

Tolerability

- Addition of gedatolisib to palbociclib and fulvestrant in the Phase 1b trial was shown to be well-tolerated with manageable TEAEs
- **Few patients discontinued treatment due to an AE, with only one (4%) discontinuation in cohort with Phase 3 dosing**
- Low incidence of the Grade 3/4 adverse events that are generally associated with the PI3K/mTOR class of inhibitors: hyperglycemia (7%), diarrhea (6%), AST/ALT increase (4%), and no Grade 3/4 colitis

TREATMENT STRATEGY

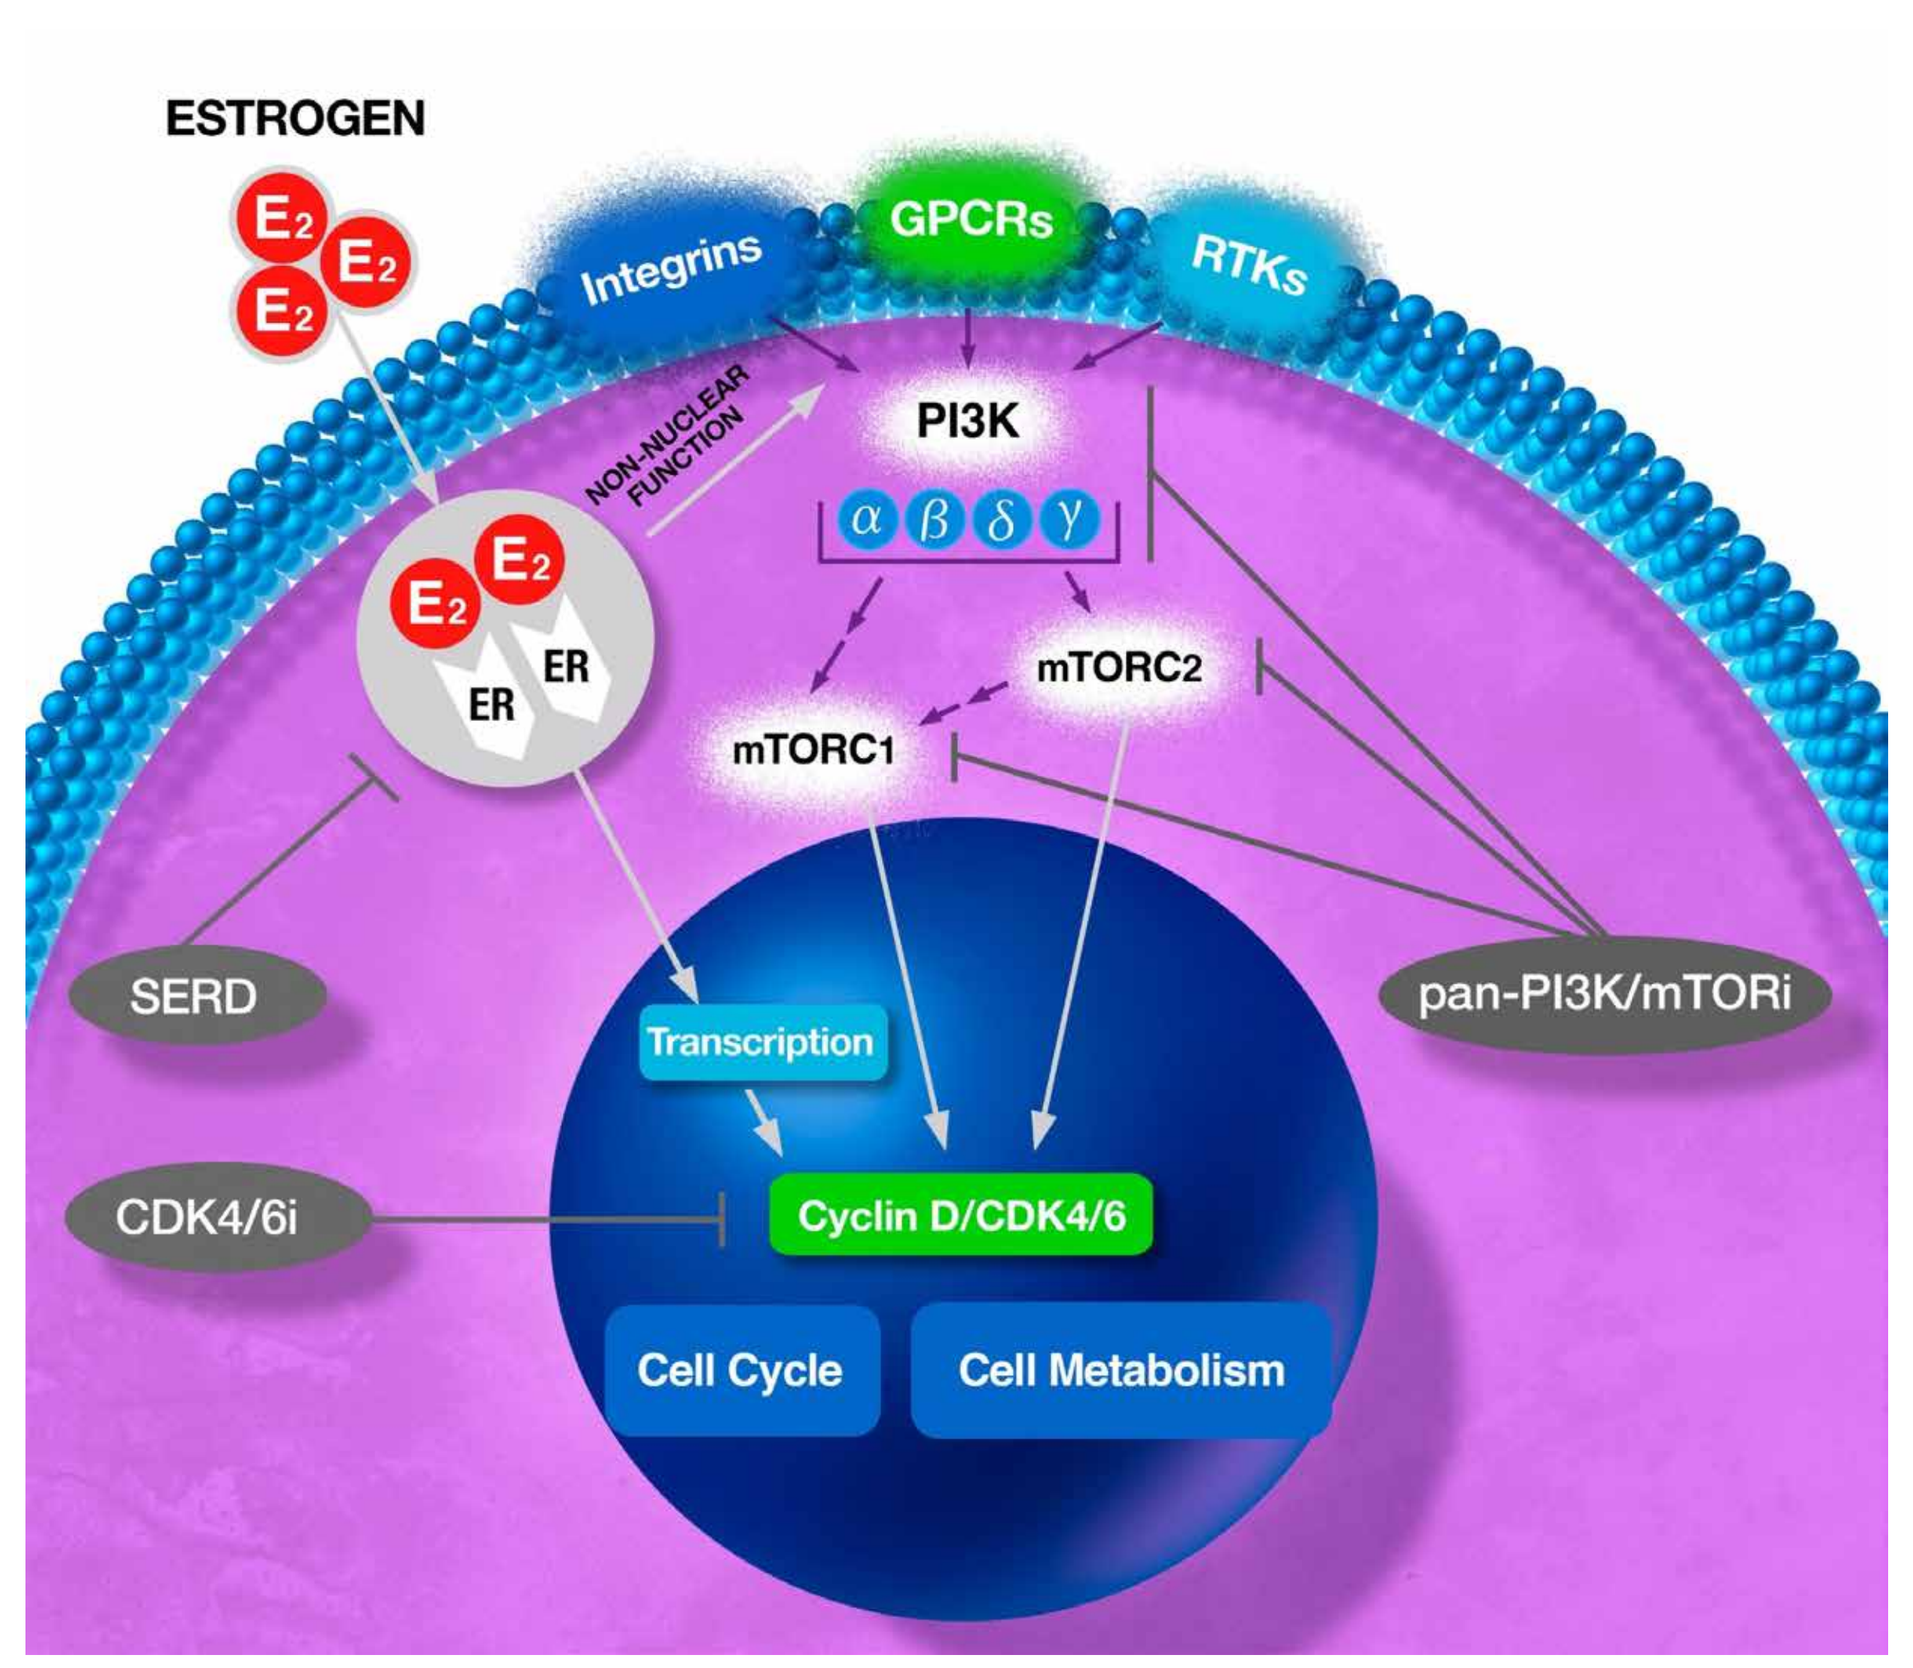
ESTROGEN
E2
Integrins
GPCRs
RTKs
NON-NUCLEAR FUNCTION
PI3K
α β δ γ
ER
mTORC2
mTORC1
SERD
pan-PI3K/mTORi
Transcription
CDK4/6i
Cyclin D/CDK4/6
Cell Cycle
Cell Metabolism

Simultaneously blockading PI3K/mTOR, CDK4/6, and ER signaling pathways disrupts complex cooperation between these pathways to inhibit tumor growth

- The upregulation of the PI3K/AKT/mTOR pathway promotes hormone dependent and independent estrogen receptor (ER) transcriptional activity.
- This contributes to endocrine resistance, leading to tumor cell growth, survival, motility, and metabolism.
- Available evidence indicates that resistance to CDK4/6 inhibition is a transient adaptive mechanism, most likely involving the PI3K/mTOR pathway.
- These data indicate that continuing CDK4/6 inhibitor treatment in combination with a PI3K/mTOR inhibitor in patients who progressed on their prior CDK4/6 inhibitor, would both block the reactivated CDK4/6 pathway and prevent adaptive activation of the PI3K/mTOR pathway.
- This suggests that patients whose disease progressed on a CDK4/6 inhibitor may benefit from continued treatment with a CDK4/6 inhibitor when it is combined with a PI3K/mTOR inhibitor as their next line of therapy.

VIKTORIA-1 is a Phase 3, open-label, randomized, two-part clinical trial to evaluate the efficacy and safety of gedatolisib in combination with fulvestrant with and without palbociclib. Two studies based on *PIK3CA* mutation status are included in the trial. *PIK3CA* mutation status will be assessed centrally using an FDA approved *PIK3CA* test. According to confirmed *PIK3CA* mutation status, subjects will be manually assigned to Study 1 (*PIK3CA* WT) or Study 2 (*PIK3CA* MT). The two studies will be randomized separately

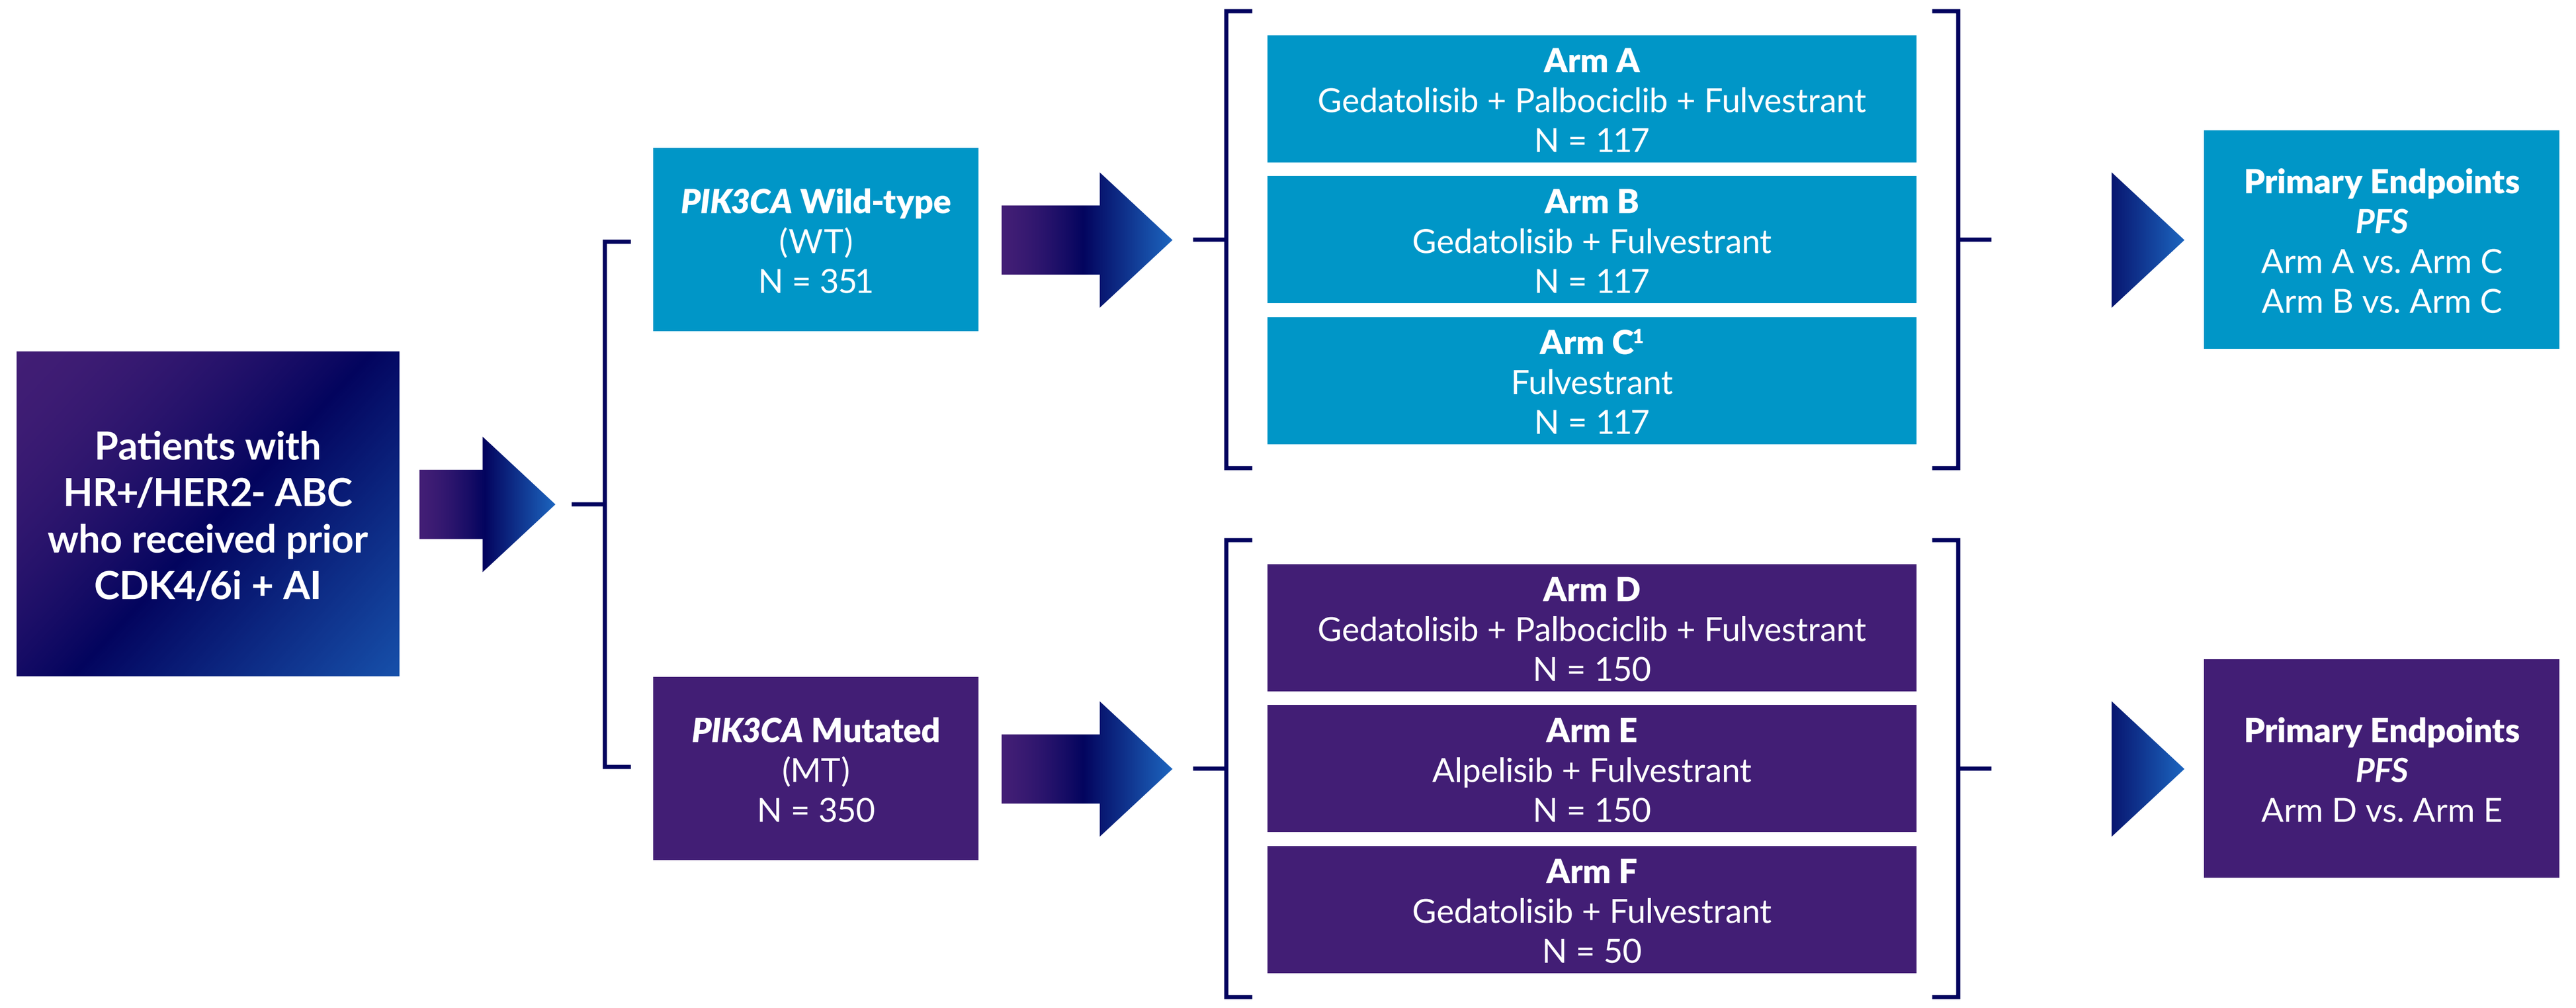
Patients with HR+/HER2- ABC who received prior CDK4/6i + AI
PIK3CA Wild-type (WT) N = 351
PIK3CA Mutated (MT) N = 350
Arm A
Gedatolisib + Palbociclib + Fulvestrant
N = 117
Arm B
Gedatolisib + Fulvestrant
N = 117
Arm C¹
Fulvestrant
N = 117
Arm D
Gedatolisib + Palbociclib + Fulvestrant
N = 150
Arm E
Alpelisib + Fulvestrant
N = 150
Arm F
Gedatolisib + Fulvestrant
N = 50
Primary Endpoints
PFS
Arm A vs. Arm C
Arm B vs. Arm C
Primary Endpoints
PFS
Arm D vs. Arm E

1) Optional Crossover from Arm C to Arm A or Arm B upon progressive disease

OBJECTIVES AND ENDPOINTS

VIKTORIA-1 will evaluate the efficacy and safety of gedatolisib and fulvestrant with or without palbociclib in patients with HR+/HER2- ABC previously treated with any CDK4/6i in combination with non-steroidal AI therapy.

Study 1 (*PIK3CA WT*)	Study 2 (*PIK3CA MT*)
Primary Objectives	
• Compare efficacy, as measured by progression-free survival, of gedatolisib in combination with palbociclib and fulvestrant (Arm A) to fulvestrant (Arm C) • Compare efficacy as measured by PFS, of gedatolisib in combination with fulvestrant (Arm B) to Arm C	• Compare the efficacy, as measured by PFS, of gedatolisib in combination with palbociclib and fulvestrant (Arm D) to alpelisib with fulvestrant (Arm E)
Key Secondary Objectives	
• Compare efficacy as measured by PFS of Arm A to Arm B • Compare the efficacy as measured by OS of Arm A to Arm C, Arm B to Arm C, and Arm A to Arm B • Compare safety & tolerability between treatment arms	• Compare the efficacy, as measured by PFS, of Arm D to Arm F (gedatolisib with fulvestrant) • Compare the efficacy, as measured by OS, of Arm D to Arm E • Compare the efficacy, as measured by OS, of Arm D to Arm F • Compare safety & tolerability between treatment arms
Additional Secondary Objectives	
• Evaluate contributory treatment effect of gedatolisib, palbociclib, & combined treatment effect of gedatolisib and palbociclib in the stratified Cox proportional hazard model • Estimate and compare PFS and OS based on HER2 status (HER2-low, defined as an IHC score of 1+ or IHC 2+ with a negative ISH score, and HER-negative status, defined as an IHC score of 0) • Compare efficacy, as measured by ORR, DOR, TTR, & CBR of Arm A to Arm C, Arm B to Arm C, & Arm A to Arm B. • Compare change in health status/QOL of Arm A to Arm C, Arm B to Arm C, and Arm A to Arm B. • PK of gedatolisib	• Compare the efficacy, as measured by PFS, of Arm E to Arm F • Estimate and compare PFS and OS based on HER2 status (HER2-low, defined as an IHC score of 1+ or IHC 2+ with a negative ISH score, and HER2-negative status, defined as an IHC score of 0) • Compare the efficacy, as measured by ORR, DOR, TTR, and CBR, of Arm D to Arm E • Compare change in health status/QOL of Arm D to Arm E • PK of gedatolisib

Trial Status

- VIKTORIA-1 is recruiting
- Approximately 701 subjects are expected to be randomized at sites in the Americas, Europe, and Asia-Pacific
- The primary completion date is estimated to occur in the second half of 2024

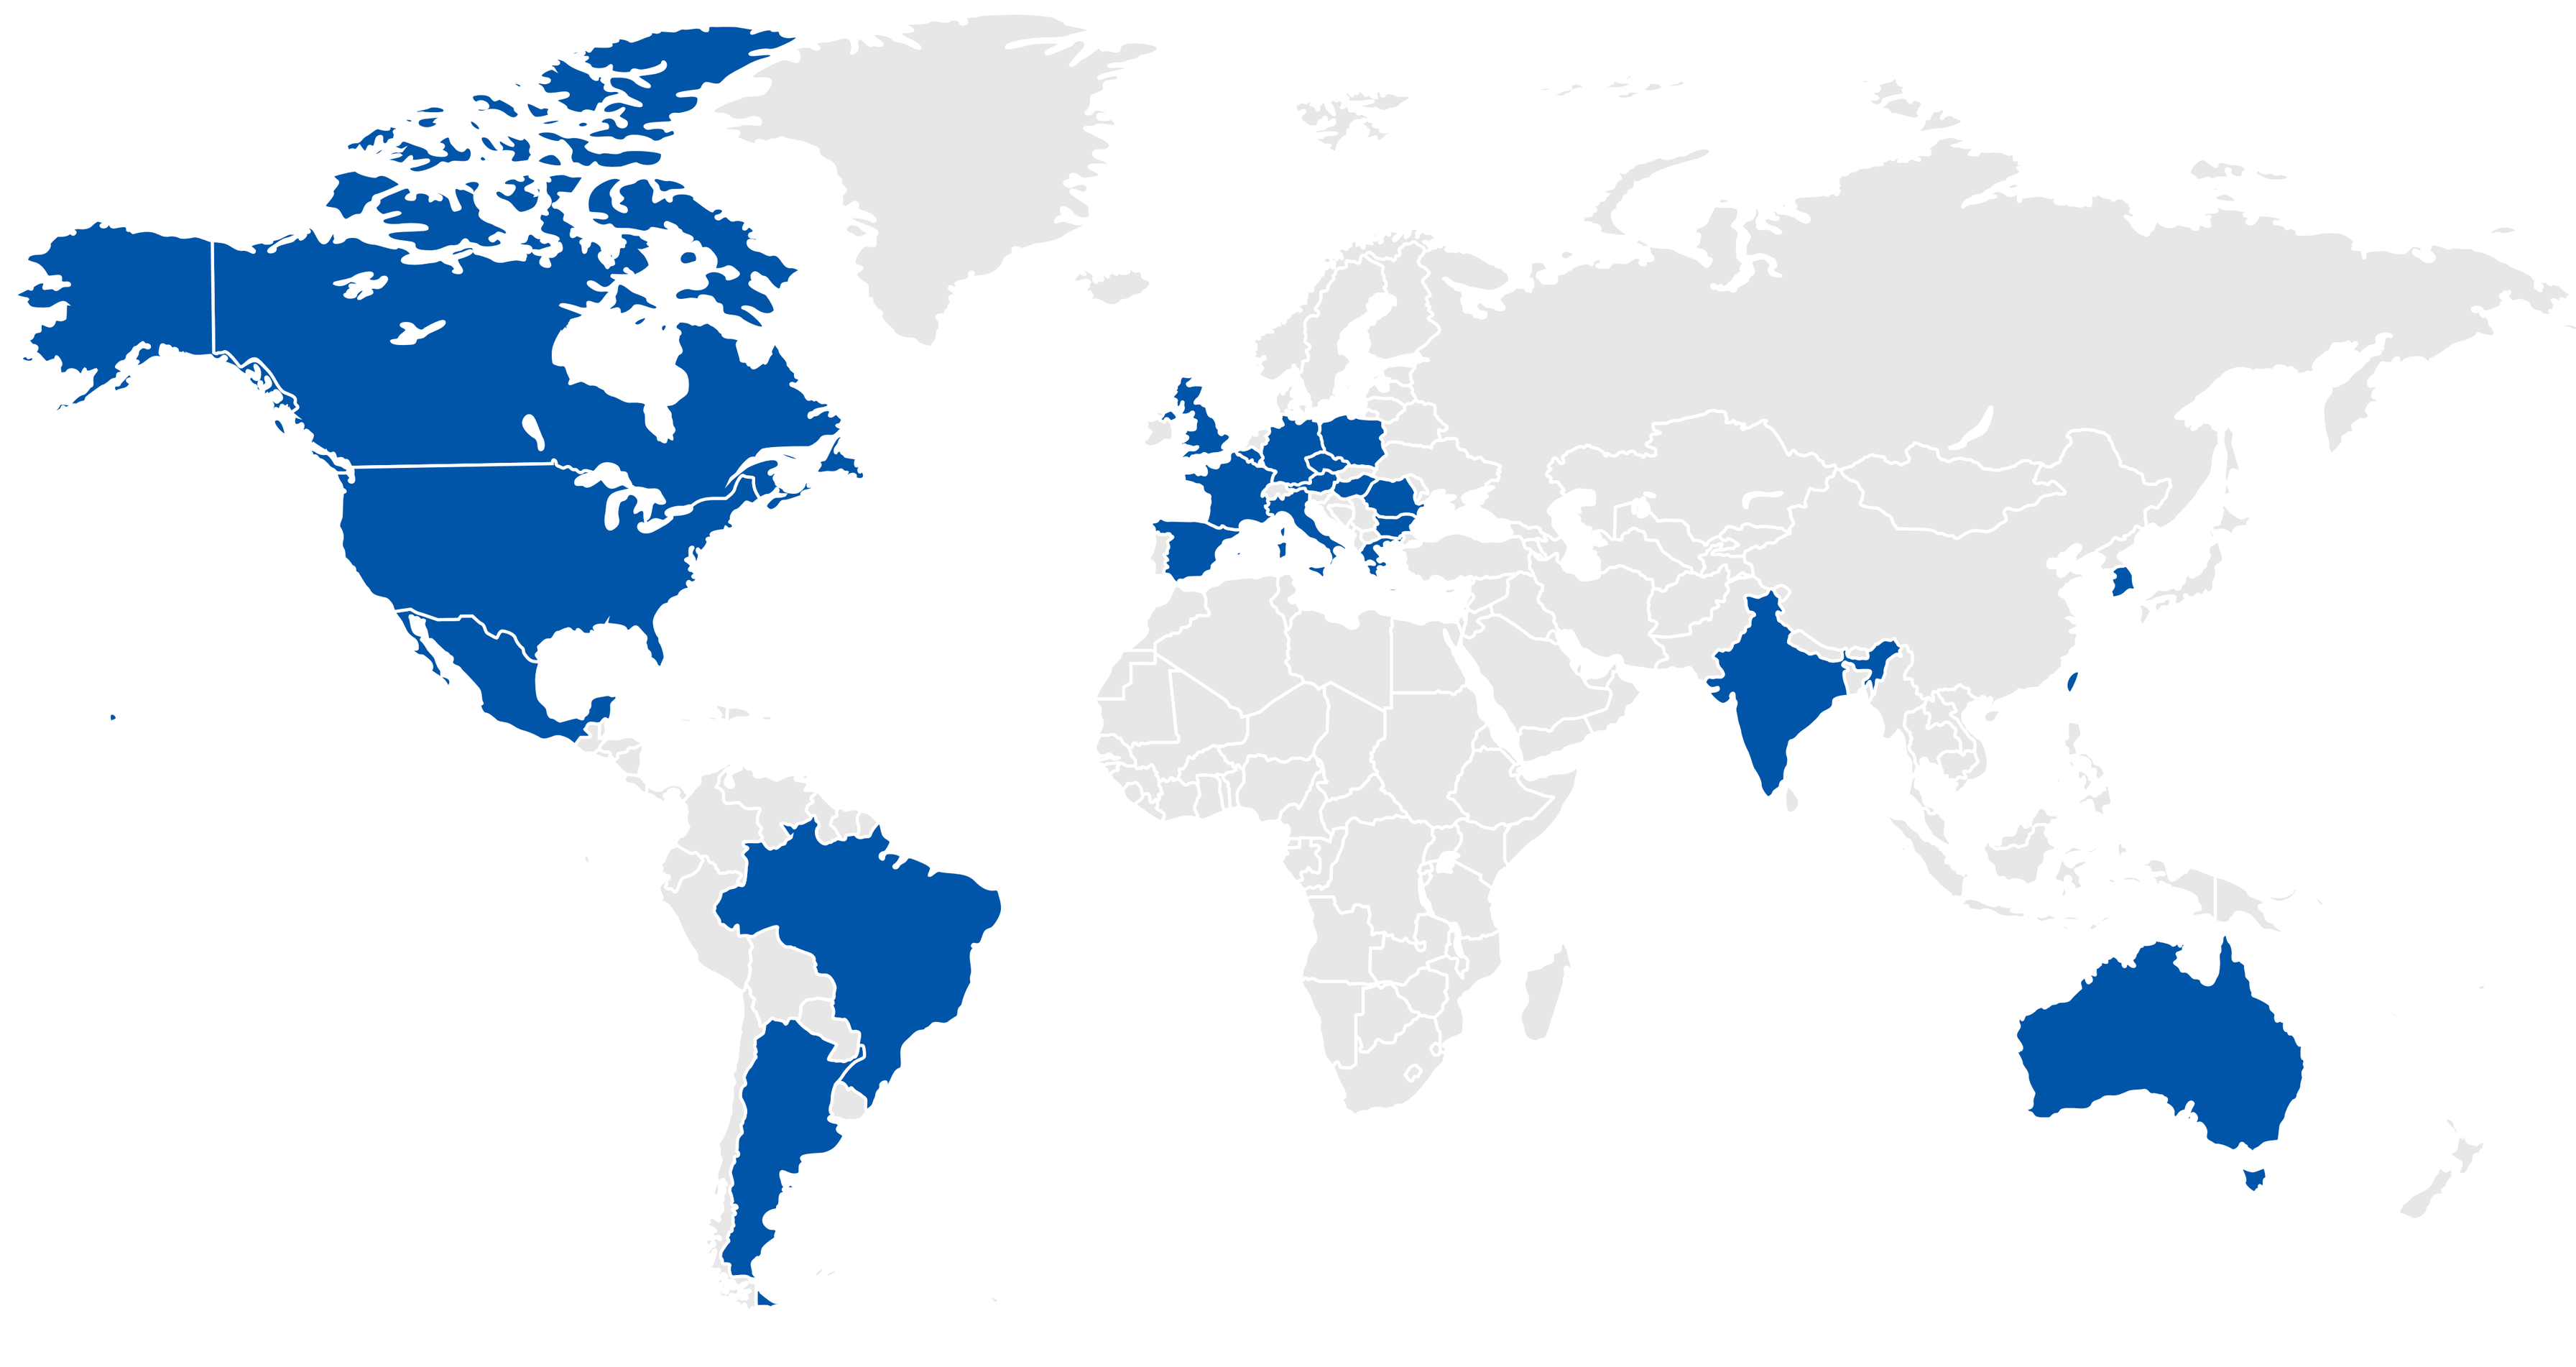

ELIGIBILITY CRITERIA

Key Inclusion Criteria	Key Exclusion Criteria
• Adults ≥ 18 years of age • Confirmed diagnosis of ER+ and/or PR+ as per ASCO-CAP 2020 guidelines • Documented HER2- as per ASCO-CAP 2018 guidelines • Adequate archival or fresh tumor tissue specimen for analysis of *PIK3CA* mutational status by a central lab using an FDA approved test • Radiologically evaluable disease according to RECIST v1.1 • Progressed during or after CDK4/6i combination treatment with non-steroidal AI • Adequate bone marrow, hepatic, renal and coagulation function as defined by acceptable laboratory parameters	• Prior treatment with PI3K, Akt, or mTOR inhibitors • Prior chemotherapy for advanced disease • More than 2 prior lines of endocrine therapy treatment • Bone only disease with no soft tissue component • Type 1 diabetes or uncontrolled type 2 diabetes • History of drug induced pneumonitis or interstitial lung disease • Pregnant or breast-feeding women

Acknowledgements

The authors would like to acknowledge the patients and their families/caregivers, investigators and their support staff for participating in this trial and Celcuity Inc. for sponsoring the study

Clinical Trial Registry Number

ClinicalTrials.gov Identifier: NCT05501886
EU CT 2022-502145-10-00

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

Poster

Study Site

Abstract: OT3-26-02

This presentation is the intellectual property of the author/presenter. Contact **igorbatchevsky@celcuity.com** for permission to reprint and/or distribute.