Updated results of a Phase 1b study of gedatolisib plus palbociclib and endocrine therapy in women with hormone receptor positive advanced breast cancer: Subgroup analysis by *PIK3CA* mutation status

Robert Wesolowski[1*], Hope S. Rugo[2*], Erica M. Stringer-Reasor[3], Hyo Han[4], Jennifer M. Specht[5], E. Claire Dees[6], Peter Kabos[7], Ulka Vaishampayan[8], Seth A. Wander[9], Janice Lu[10], Keerthi Gogineni[11], Alexander Spira[12], Anne F. Schott[13], Maysa Abu-Khalaf[14], Pratima Nayak[15], Brian F. Sullivan[15], Igor Gorbatchevsky[15], Rachel M. Layman[16*]

[1]The Ohio State University Comprehensive Cancer Center, Columbus, OH; [2]University of California, San Francisco Comprehensive Cancer Center, San Francisco, CA; [3]Division of Hematology Oncology, Department of Medicine, O'Neal Comprehensive Cancer Center, University of Alabama at Birmingham, Birmingham, AL; [4]Moffit Cancer Center, Tampa, FL; [5]Fred Hutch Cancer Center, University of Washington, Seattle, WA; [6]University of North Carolina Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [7]University of Colorado Hospital, Aurora, CO; [8]Karmanos Cancer Institute, Detroit, MI; [9]Massachusetts General Hospital, Harvard Medical School, Boston, MA; [10]University of Southern California Norris Comprehensive Cancer Center, Los Angeles, CA; [11]Emory University Hospital, Atlanta, GA; [12]Virginia Cancer Specialists Research Institute, Fairfax, VA; [13]University of Michigan, Ann Arbor, MI; [14]Sidney Kimmel Cancer Center at Jefferson Health, Philadelphia, PA; [15]Celcuity, Minneapolis, MN; [16]The University of Texas MD Anderson Cancer Center, Houston, TX

*These authors contributed equally to this work

Poster selected for Spotlight Discussion at the 2022 San Antonio Breast Cancer Symposium, December 6-10, 2022, San Antonio, Texas

BACKGROUND

- The addition of a PI3K/mTOR inhibitor after progression on CDK4/6 inhibitors (CDK4/6i) and endocrine therapy (ET) in estrogen receptor positive (ER+), HER2-negative advanced breast cancer (ABC) can
 - potentially restore sensitivity to CDK4/6i and
 - prevent adaptive activation of the PI3K/mTOR pathway[1-5].
- To evaluate this hypothesis, a Phase Ib study of gedatolisib (G), a dual inhibitor of PI3K/mTOR, palbociclib (P) a CDK4/6i, and ET (with letrozole [LET] or fulvestrant [FUL]) in women with estrogen receptor positive (ER+)/HER2- ABC was conducted.
- Manageable toxicity and preliminary antitumor activity were observed in 35 patients(pts) enrolled in the dose escalation portion of the study[6], and 103 pts enrolled in the expansion portion of the study[7].
- In this analysis, we report updated efficacy and safety data, and sub-group analysis by *PIK3CA* mutation status in the four expansion study arms with a June 29, 2022, database lock.

STUDY DESIGN

- Patients with ER+/HER2- MBC were treated in four-arms as shown in "Expansion" panel. Pre-/peri-menopausal women received ovarian suppression.
- Dosing information: Palbociclib: 125 mg/day in a 3 week on, 1 week off schedule; Letrozole: 2.5 mg/day; Fulvestrant: 500 mg intramuscular injection on cycle 1 days 1 and 15, and every 28 days ± 3 days thereafter; Gedatolisib: 180 mg IV weekly (Arms A, B, and C) or three weeks on/one week off (Arm D).
- Tumor assessment performed at baseline and every 12-16 weeks until disease progression or the start of a new anti-cancer therapy.
- Endpoints: Primary – objective response assessed by the investigator; Secondary – safety, duration of response (DOR), and PFS.

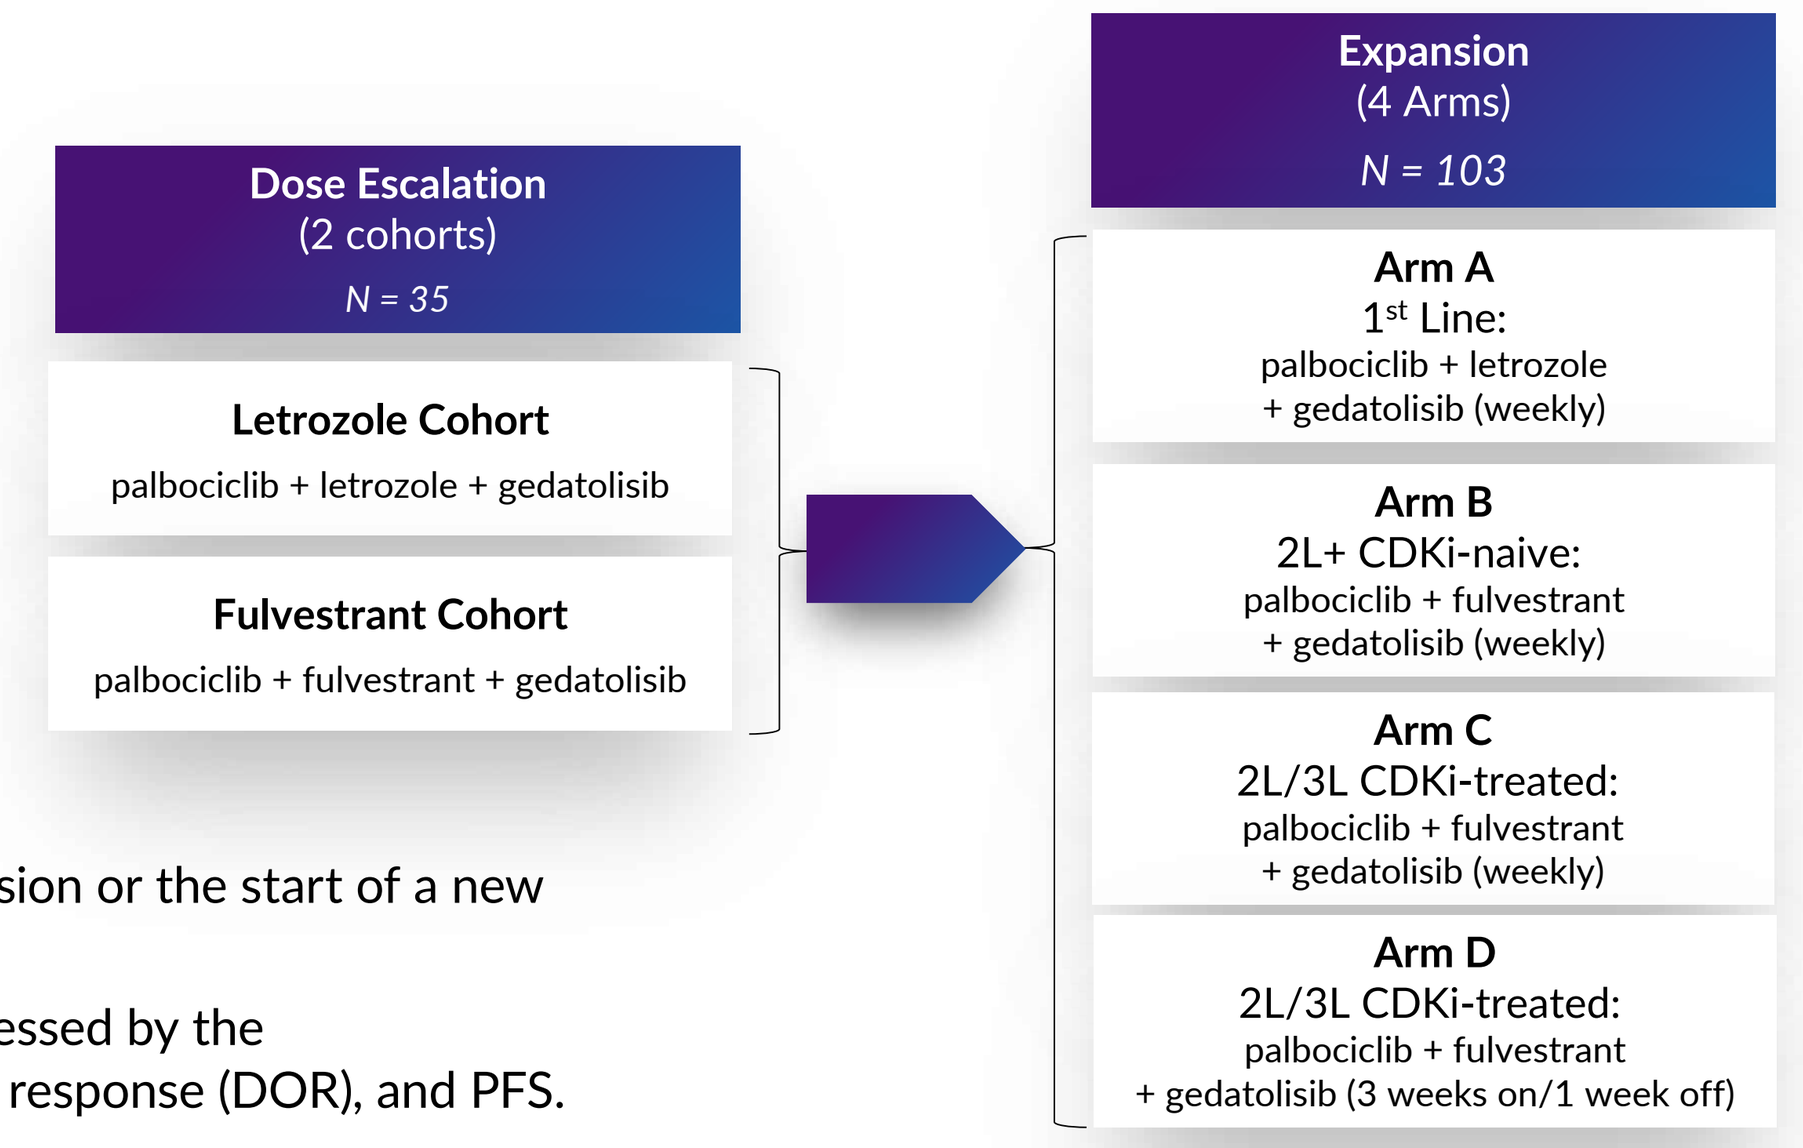
Dose Escalation
(2 cohorts)
N = 35
Letrozole Cohort
palbociclib + letrozole + gedatolisib
Fulvestrant Cohort
palbociclib + fulvestrant + gedatolisib
Expansion
(4 Arms)
N = 103
Arm A
1st Line:
palbociclib + letrozole + gedatolisib (weekly)
Arm B
2L+ CDKi-naive:
palbociclib + fulvestrant + gedatolisib (weekly)
Arm C
2L/3L CDKi-treated:
palbociclib + fulvestrant + gedatolisib (weekly)
Arm D
2L/3L CDKi-treated:
palbociclib + fulvestrant + gedatolisib (3 weeks on/1 week off)

RESULTS

Table 1: Baseline Characteristics

All data as of 29-June-2022 database lock date

Parameter	Expansion Arm A (N=31)	Expansion Arm B (N=13)	Expansion Arm C (N=32)	Expansion Arm D (N=27)
Age				
Median Years (range)	54 (28-78)	62 (41-71)	59 (41-74)	59 (34-79)
Prior Therapies – Advanced Breast Cancer, n (%)				
Prior Chemotherapy	1 (3.2)	4 (30.8)	15 (46.9)	5 (18.5)
Prior SERD or SERM Therapy[1]	0	5 (38.5)	14 (43.8)	10 (37.0)
Prior Aromatase Inhibitor Therapy	0	7 (53.8)	25 (78.1)	19 (70.4)
Prior CDK4/6 inhibitor	0	0	32 (100)	26 (96.3)
Number of Prior Therapies – Advanced Breast Cancer, n (%)				
0	30 (96.8)	2 (15.4)	0	0
1	1 (3.2)	9 (69.2)	15 (46.9)	18 (66.7)
2	0	2 (15.4)	11 (34.4)	8 (29.6)
3 or more	0	0	6 (18.8)	1 (3.7)
Measurable Baseline Disease, n (%)				
Yes	31 (100)	13 (100)	32 (100)	27 (100)
No	0	0	0	0
Disease Site Involved, n (%)				
Bone	18 (58.1)	11 (84.6)	25 (78.1)	18 (66.7)
Brain	0	0	1 (3.1)	0
Liver	14 (45.2)	10 (76.9)	20 (62.5)	17 (63)
Lung	7 (22.6)	3 (23.1)	7 (21.9)	6 (22.2)
Lymph Node	8 (25.8)	2 (15.4)	9 (28.1)	2 (7.4)
Pleural Effusion	4 (12.9)	0	3 (9.4)	2 (7.4)
Skin	1 (3.2)	0	1 (3.1)	0
Other	26 (83.9)	10 (76.9)	20 (62.5)	21 (77.8)
Number of Disease Sites Involved, n (%)				
1 – 3	26 (83.9)	11 (84.6)	26 (81.2)	24 (88.9)
≥4	5 (16.1)	2 (15.4)	6 (18.8)	3 (11.1)
***PIK3CA*, n (%)[2]**				
Wild Type	25 (80.6)	9 (69.2)	24 (75.0)	15 (55.6)
Mutation	5 (16.1)	4 (30.8)	8 (25.0)	11 (40.7)
Unknown/Missing	1 (3.2)	0	0	1 (3.7)

[1]Across 4 arms, 27 subjects received SERD and 2 subjects received SERM as a primary systemic therapy for advanced breast cancer. No subjects received both therapies

[2]*PIK3CA* status confirmed by liquid biopsy using a central lab

EFFICACY

Table 2: Efficacy Summary (All Expansion Arms)

	Total Expansion Arms (N=103, full analysis set)			
	Expansion			
Arm	A	B	C	D
Prior Therapy	1L: CDKi- naive	2L+: CDKi- naive	2L/3L: CDKi -pretreated	2L/3L: CDKi- pretreated
n (Full, response evaluable)	31, 27	13, 13	32, 28	27, 27
Study Treatment	P + L + G	P + F + G	P + F + G	P + F + G
Gedatolisib schedule	weekly	weekly	weekly	3 weeks on/1 week off
Median DOR, months (95% CI)[3]	NR (22.2, NR)	12.2 (3.7, 40.6)	16.6 (3.7, 30.3)	12.6 (7.3, 21.2)
ORR[1] (evaluable)	85%	77%	36%	63%
mPFS[2], mos (range)	NR (16.9, NR)	12.9 (7.6, 38.3)	5.1 (3.3, 7.5)	12.9 (7.4, 16.7)
Median Follow Up[2], mos (range)	33.1 (0.0+, 40.3+)	NE (2.1+, 42.5)	NE (0.0+, 32.1)	29.0 (1.7, 31.6+)
PFS % at 12 mos[2]	72.1%	54.5%	23.6%	53.2%

[1]Response evaluable analysis set per RECIST v1.1 including uPR; [2]full analysis set; [3]Kaplan Meier method and confidence intervals by the Brookmeyer and Crowley Method; Abbreviations: 1L= first line, 2L= second line; mos= months; NR = not reached; NE = could not be estimated per reverse KM method; DOR, duration of response; ORR, objective response rate; PFS, progression free survival; +=censored

SAFETY

Table 3: Treatment Related and Emergent Adverse Events (≥20% of subjects, by SOC and preferred term)

	All Expansion Arms (n=103)			
Adverse Event	Grade 1 %	Grade 2 %	Grade 3 %	Grade 4 %
Gastrointestinal disorders				
Stomatitis[1]	19.4	41.7	27.2	0
Nausea	42.7	34.0	0	0
Vomiting	32.0	12.6	1.0	0
Diarrhea	23.3	8.7	2.9	0
Dry mouth	25.2	1.9	0	0
Constipation	20.4	4.9	1.0	0
General disorders and administration site conditions				
Fatigue	21.4	35.9	10.7	0
Skin and subcutaneous tissue disorders				
Rash[2,3]	21.4	10.7	20.4	0
Pruritus	13.6	7.8	4.9	0
Investigations				
White blood cell count decreased	1.9	15.5	11.7	2.9
Lymphocyte count decreased	2.9	4.9	11.7	1.0
Blood and lymphatic system disorders				
Anemia	10.7	17.5	11.7	0
Neutropenia/Neutrophil count decreased[2,4]	1.0	11.7	53.4	9.7
Nervous system disorders				
Dysgeusia	42.7	2.9	0	0
Headache	18.4	5.8	0	0
Metabolism and nutrition disorders				
Decreased appetite	23.3	8.7	0	0
Hyperglycemia	12.6	5.8	3.9	1.9
Injury, poisoning and procedural complications				
Infusion related reaction	16.5	5.8	0	0

There were no Grade 5 treatment related TEAEs

[1]Prophylactic treatment for stomatitis was not implemented; [2]Number of patients with at least one of the terms. If a patient experienced multiple terms, it will be counted once for the highest grade; [3]Rash, Rash maculo-papular, Rash pruritic, Rash pustular, Rash papular, Rash erythematous, or Rash vesicular; [4]Neutropenia and neutrophil count decrease were reported interchangeably for many patients. In this table, neutropenia (SOC-blood and lymphatic system disorders) and neutrophil count decreased (SOC-investigations) were combined

Table 4: Patient Treatment Discontinuation

	Expansion Arm A (N=31)	Expansion Arm B (N=13)	Expansion Arm C (N=32)	Expansion Arm D (N=27)
Subjects who discontinued treatment	n (%)	n (%)	n (%)	n (%)
Discontinuation of Study Treatment				
Reasons other than AEs	28 (90.3)	11 (84)	29 (90.6)	26 (96.3)
Progression or relapse	12 (38.7)	10 (76.9)	24 (75.0)	18 (66.7)
Death[1]	0	0	0	1 (3.7)
Study terminated by sponsor[2]	8 (25.8)	1 (7.7)	0	2 (7.4)
Other[3]	8 (25.8)	0	5 (15.6)	5 (18.5)
Adverse Event[4]	3 (9.7)	2 (15.4)	3 (9.4)	1 (3.7)

[1]Death due to non treatment related septic shock; [2]After study termination, as of database lock date of June 29, 2022, 11 subjects rolled over to an expanded access protocol and continued treatment; [3]Other includes global deterioration, protocol violation, no longer willing to participate in study; [4]Treatment related AEs: stomatitis (n=4); mucosal inflammation, pneumonitis, psoriasis, pulmonary embolism (n=1 each); Non-treatment related AEs: edema peripheral (n=1)

Table 5: Efficacy Summary (*PIK3CA* Mutation Status)

	Total Expansion Arms (N=103, full analysis set)							
	Expansion							
Arm	A		B		C		D	
Prior Therapy	1L: CDKi- naive		2L+: CDKi- naive		2L/3L: CDKi -pretreated		2L/3L: CDKi- pretreated	
n (Full, response evaluable)	31, 27		13, 13		32, 28		27, 27	
Study Treatment	P + L + G		P + F + G		P + F + G		P + F + G	
Gedatolisib schedule	weekly		weekly		weekly		3 weeks on/1 week off	
	WT	MT	WT	MT	WT	MT	WT	MT
***PIK3CA* Status**	81%[2,3]	16%[2,3]	69%	31%	75%[2]	25%[2]	56%[2,3]	41%[2,3]
ORR[1] (evaluable)	81%	100%	78%	75%	25%	63%	60%	73%
PFS % at 12 mos[2]	74%	60%	50%	67%	22%	29%	49%	60%

[1]Response evaluable analysis set per RECIST v1.1 including uPR; [2]full analysis set; [3]Baseline *PIK3CA* mutation status missing for one patient; 1L= first line, 2L= second line; mos= months; WT=wild type; MT= *PIK3CA* mutation; ORR, objective response rate; PFS, progression free survival

Figure 1: Treatment-Naive (1L) Subjects Time to First Response, Duration of Response and Treatment

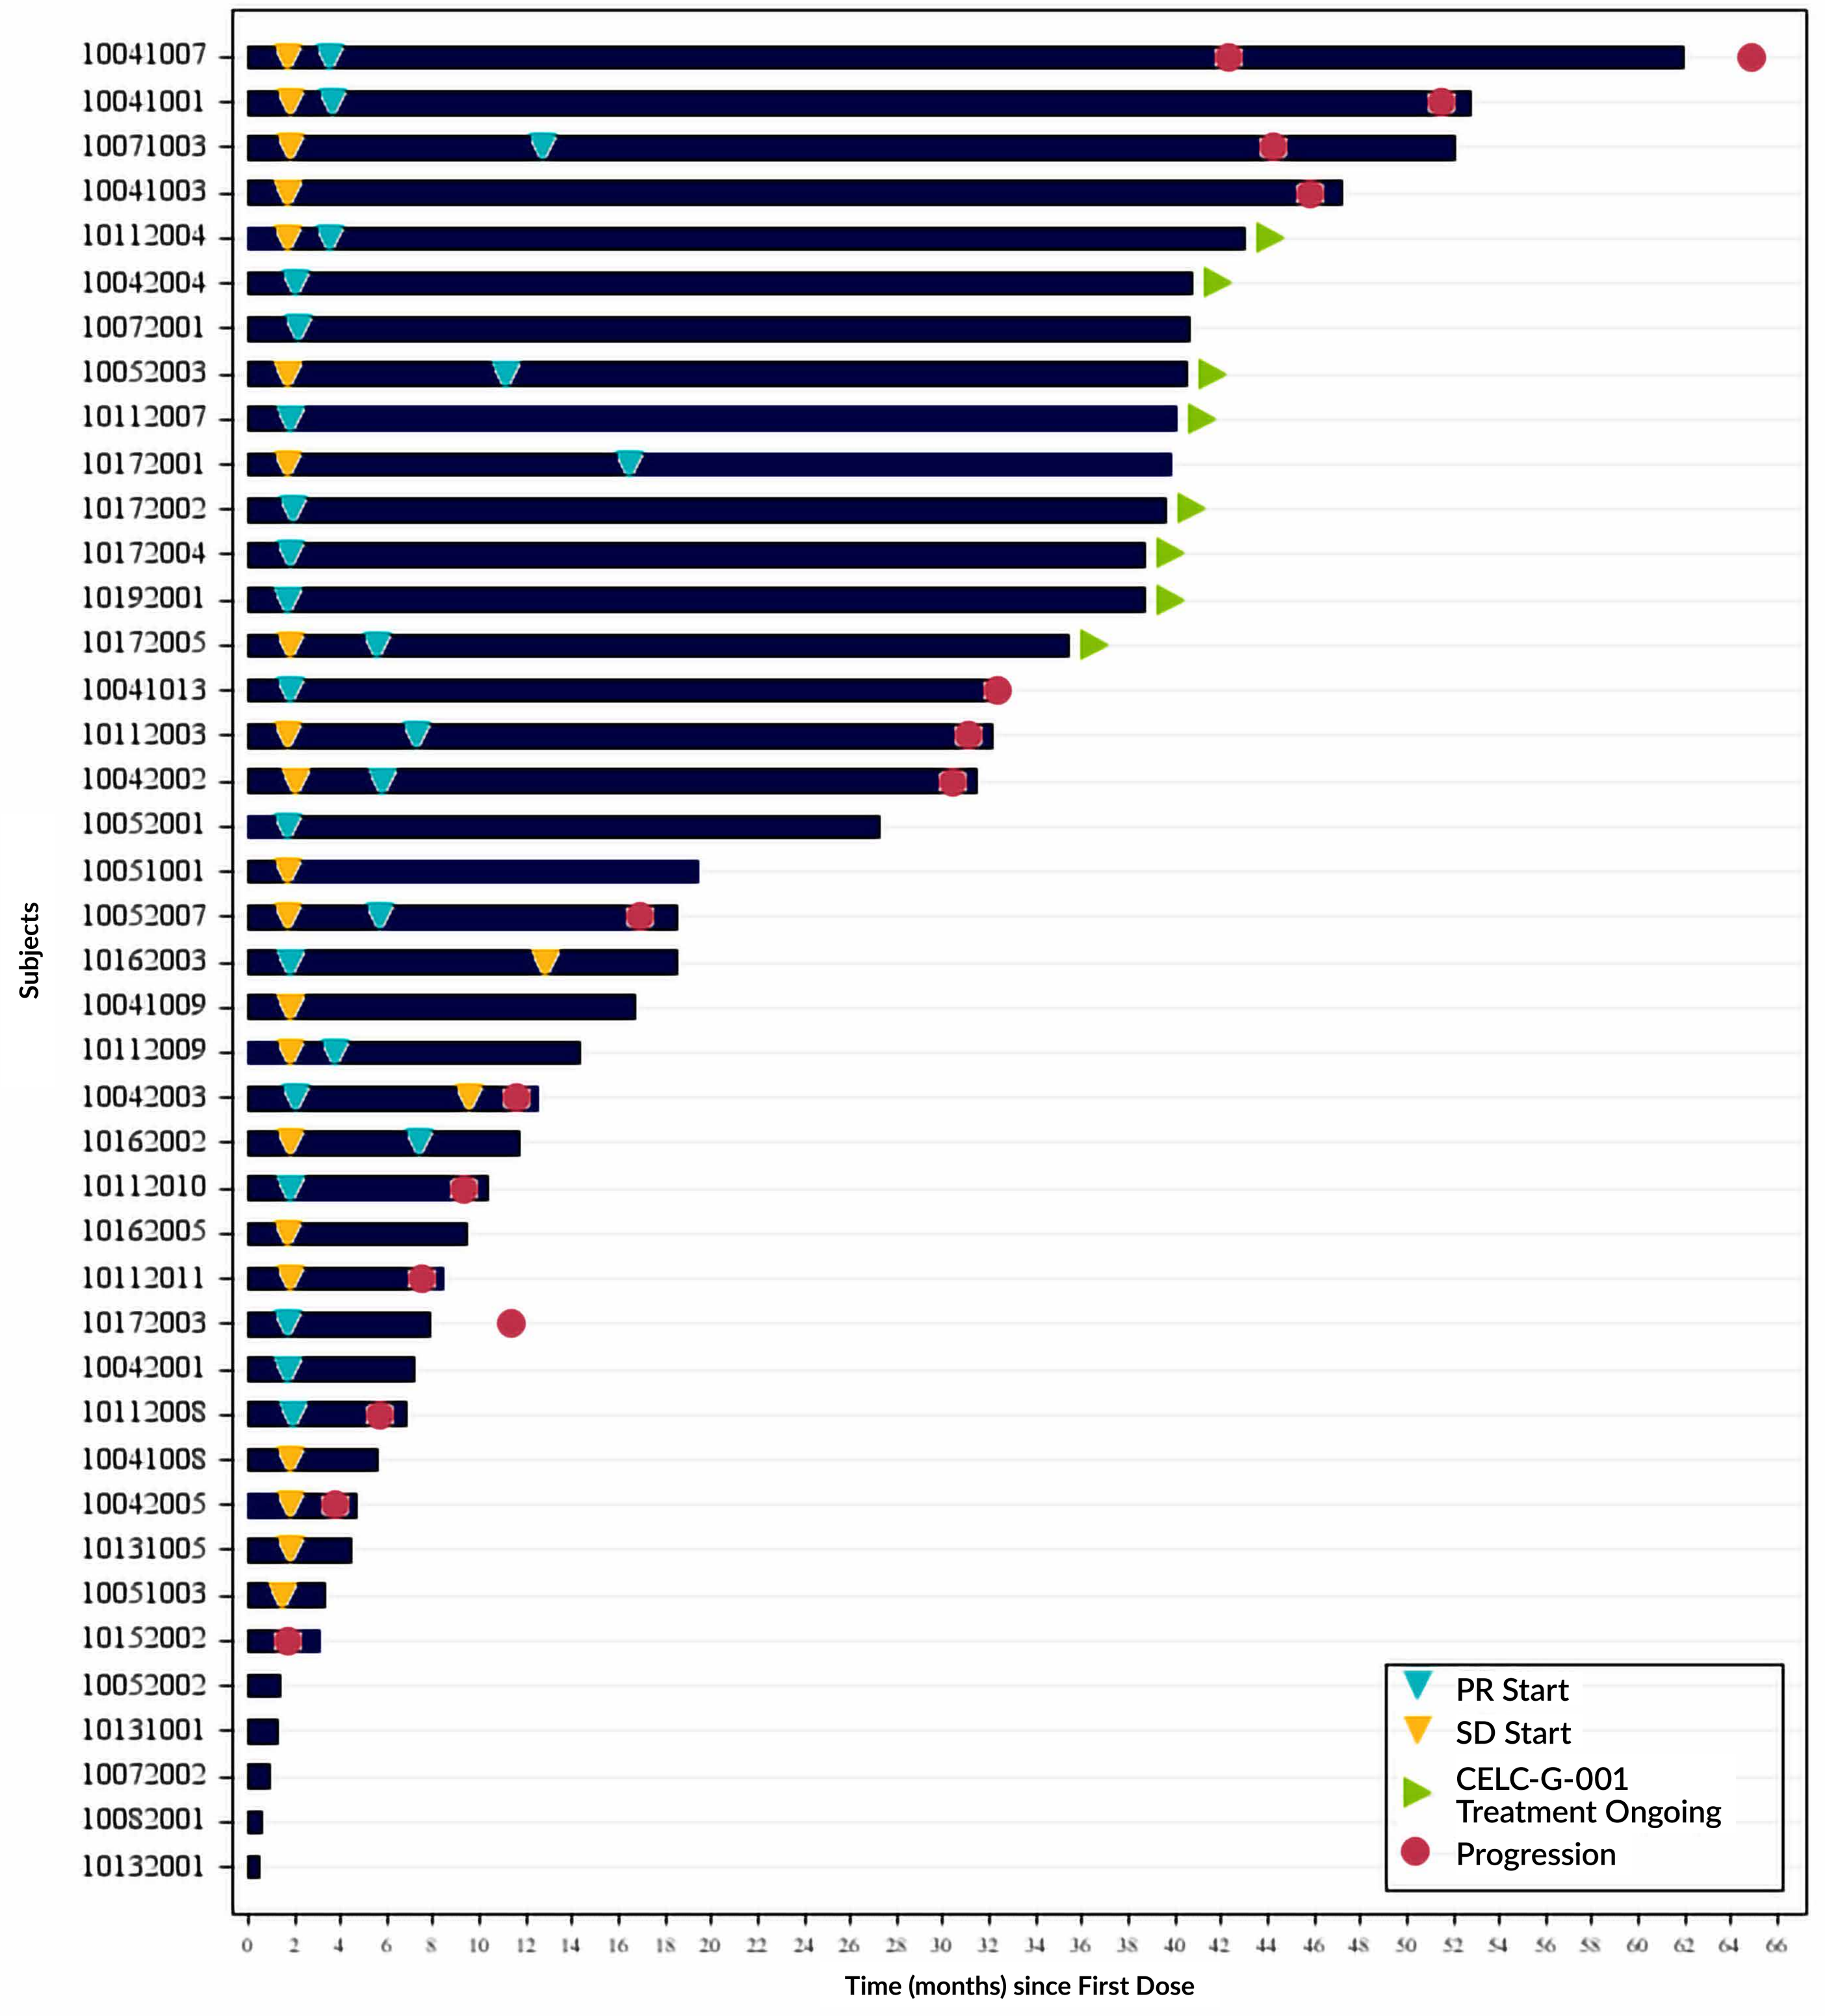
10041007
10041001
10071003
10041003
10112004
10042004
10072001
10052003
10112007
10172001
10172002
10172004
10192001
10172005
10041013
10112003
10042002
10052001
10051001
10052007
10162003
10041009
10112009
10042003
10162002
10112010
10162005
10112011
10172003
10042001
10112008
10041008
10042005
10131005
10051003
10152002
10052002
10131001
10072002
10082001
10132001
Subjects
PR Start
SD Start
CELC-G-001 Treatment Ongoing
Progression
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44 46 48 50 52 54 56 58 60 62 64 66
Time (months) since First Dose

Note: Each bar is presented for each subject starting from first dose date to last dose date, with time of disease progression. No subject died prior to experiencing disease progression. 8 subjects (green arrows) remained on treatment after transferring to an expanded access protocol (CELC-G-001) or individual IND as of June 29, 2022.

Table 6 : Efficacy Summary (Treatment-Naïve [1L] Population)

	Escalation Arm A	Expansion Arm A	Escalation Arm A + Expansion Arm A Total
n (full, response evaluable)	(11, 10)	(30, 26)[3]	(41, 36)
Responses (response evaluable)[1]	n (%)	n (%)	n (%)
CR	0	1 (3.8)	1 (2.8)
PR	4 (40.0)	21 (80.8)	25 (69.4)
SD	6 (60.0)	3 (11.5)	9 (25.0)
Unconfirmed PR	0	0	0
Durable SD (>=24 weeks)	3 (30.0)	2 (7.7)	5 (13.9)
PD	0	1 (3.8)	1 (2.8)
Not Evaluable	0	0	0
ORR[1]	4 (40.0)	22 (84.6)	26 (72.2)
Progression Free Survival (full analysis set)[2]			
Median PFS, mos (95%CI)[2]	44.2 (32.3, 51.4)	NR (11.6, NR)	42.3 (30.4, 45.8)[4]

Median time from the last prior therapy was 1 month for Escalation Arm A vs 26 months for Expansion Arm A.
These 2 arms were not randomized.

[1]Response evaluable analysis set per RECIST v1.1; [2]full analysis set; [3]one subject in expansion arm A was not treatment-naïve and has been excluded in this evaluation; [4]median PFS follow up time was 33.1 months; CR, complete response; mos, months; NR, Not Reached; PFS, progression free survival; PR, partial response; SD, stable disease; PD, progressive disease; ORR, objective response rate

Figure 2: Treatment-Naive (1L) Subjects Best Response (Maximum Improvement in Sum of Target Lesion Diameters from Baseline)

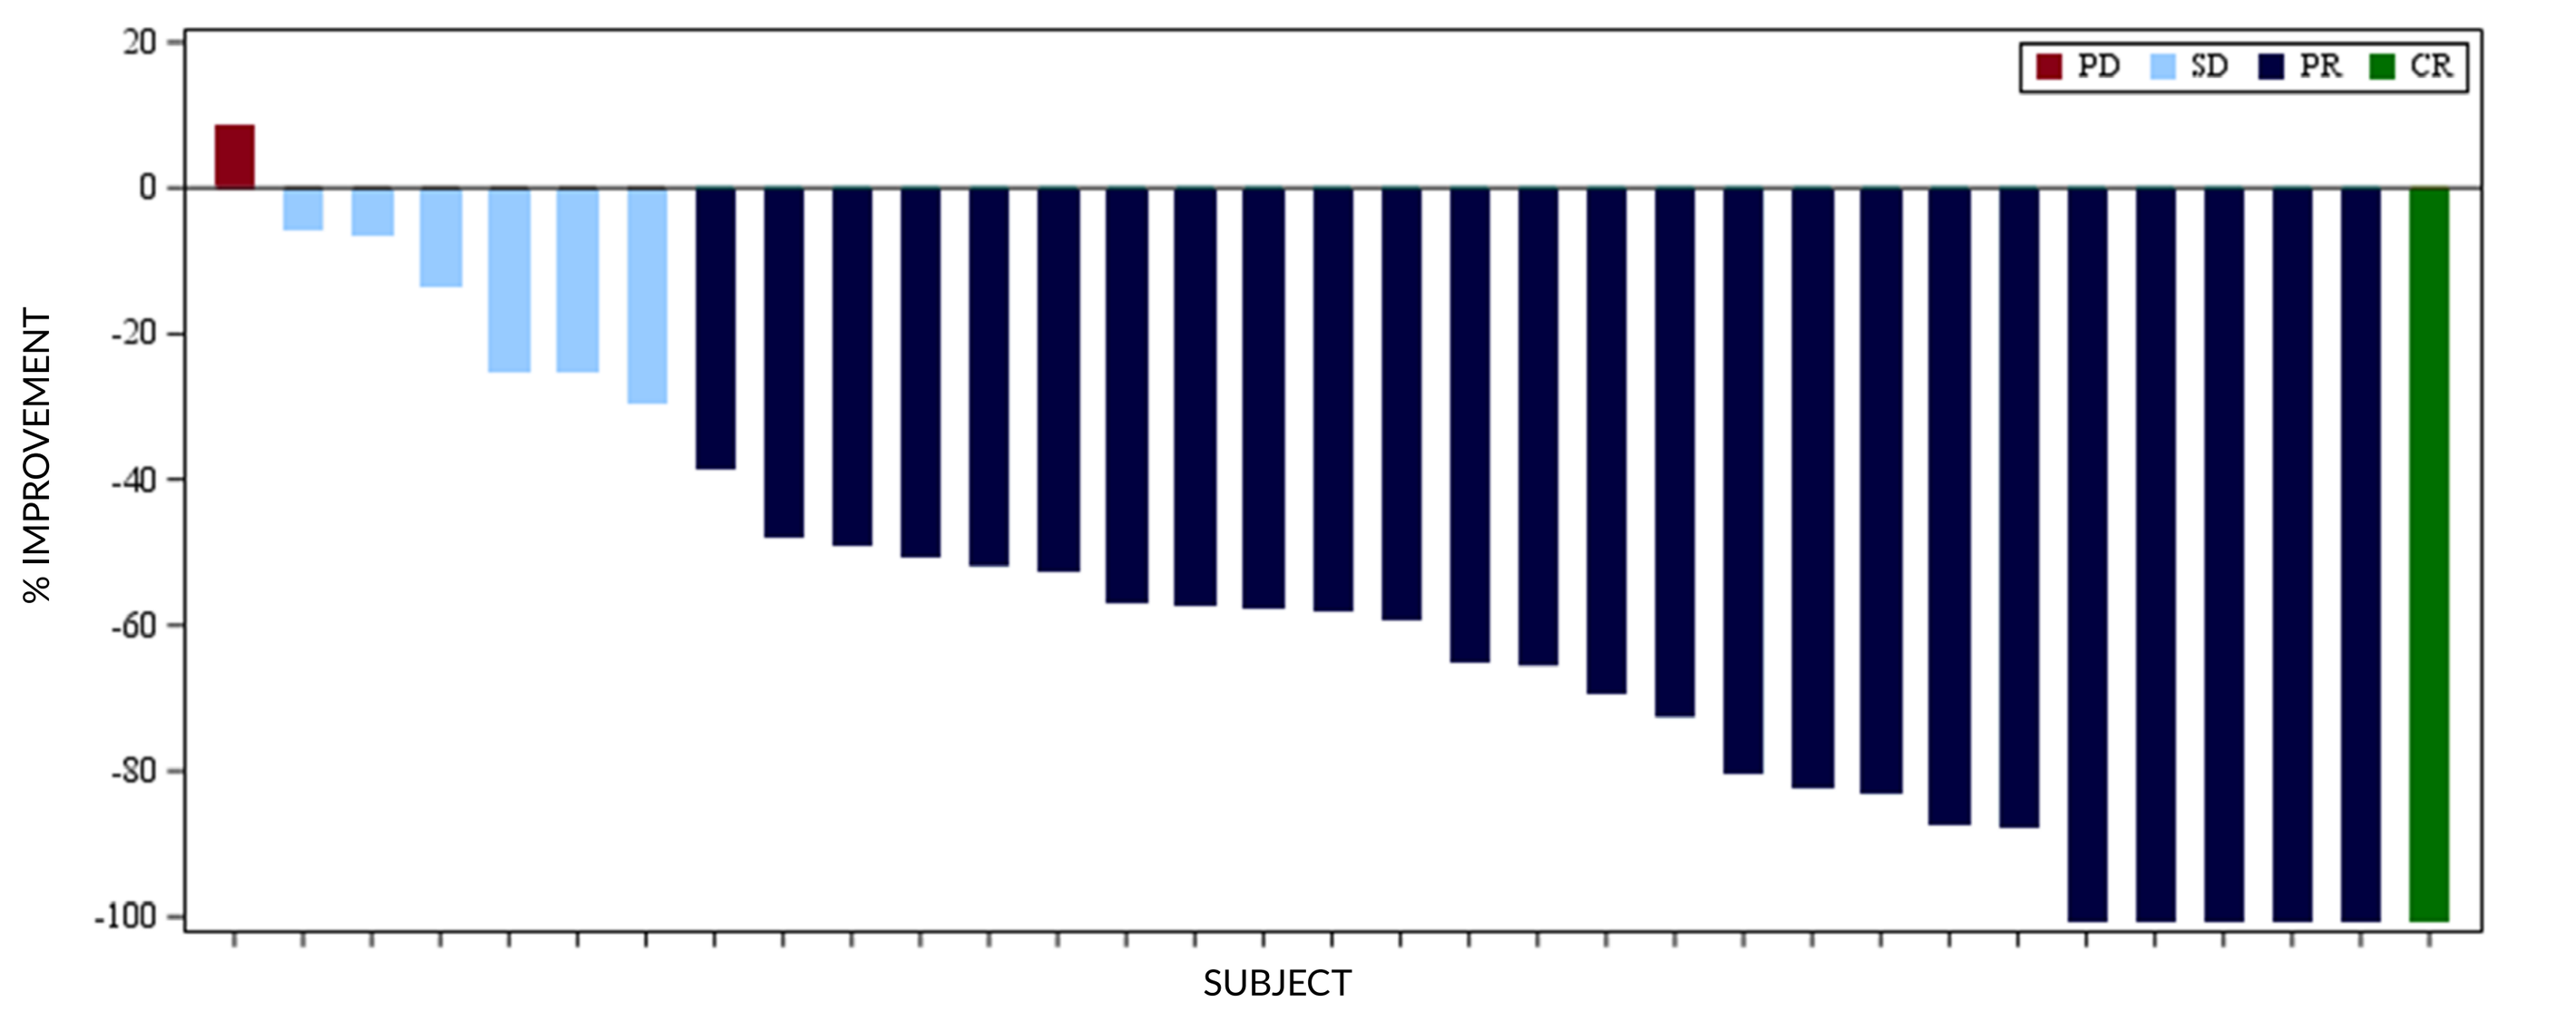
PD
SD
PR
CR
20
0
-20
-40
-60
-80
-100
% IMPROVEMENT
SUBJECT

Note: Three patients in Escalation Arm A did not have measurable target lesions but had non-target lesions. In this study, they were considered evaluable and included in all analyses of efficacy. Based on evaluation of their non-target lesions, each of the three patients had SD as BOR.

CONCLUSIONS

- Gedatolisib in combination with ET and palbociclib demonstrated encouraging efficacy and durable responses in both treatment naïve ER+/HER2- ABC as well as in later line settings.
- Promising efficacy results compared favorably to published data for current standard of care.
- High response rates were observed in subjects regardless of *PIK3CA* mutation status.
- Therapy was well tolerated with few subjects (<10%) discontinuing due to adverse events.
- A Phase 3 study evaluating gedatolisib in patients with HR+/HER2- ABC is ongoing (NCT05501886; Poster-ID-OT3-26-02)
- Further study of gedatolisib in treatment naïve HR+/HER2- ABC is warranted.

Acknowledgements

- Patients and their families/caregivers for participating in this trial
- Investigators and their support staff who participated in this work
- Celcuity Inc. for sponsoring the study

References

1. Turner NC, et al. (2015) doi: 10.1056/NEJMoa1505270
2. Andre F, et al. (2019) doi: 10.1056/NEJMoa1813904
3. Rugo HS, et al. (2020) doi: 10.1200/JCO.2020.38.15_suppl.1006
4. Baselga J, et al.(2012) doi: 10.1056/NEJMoa1109653
5. Dhakal A, et al. (2020). doi: 10.1177/1178223420944864
6. Forero-Torres, A., et al. (2018). *J Clin Oncol* **36**(15_suppl): 1040-1040
7. Layman, RM, et al. (2021) SABCS 2021 Dec 7-10; San Antonio, TX. Abstract 1017

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

Abstract: PD13-05

This presentation is the intellectual property of the author/presenter. Contact **igorbatchevsky@celcuity.com** for permission to reprint and/or distribute.