CELx™ | breast

HER2 Signaling Function Test

Quantification of HER2-driven signaling ($HER2_S$) inhibition of four different anti-HER2 drugs tested ex vivo in live primary HER2-negative breast cancer cell samples with abnormal HER2 signaling activity

Lance G Laing, David J Burns, Yao Huang, Ian A MacNeil, Benjamin E Rich, Sajjad M. Soltani, Samantha Myhre, Brian F Sullivan Celcuity LLC, 16305 36th Ave N, Suite 450, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®

December 6–10, 2016

celcuity™

www.celcuity.com

Background

HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15-20% of breast cancers and associated with more aggressive disease progression, metastasis, and a poorer prognosis [1-4]. The use of HER2-targeting agents, such as monoclonal antibodies (trastuzumab and pertuzumab) and small kinase inhibitors (lapatinib, afatinib, and neratinib) (Table 1), significantly improves clinical outcomes in HER2+ patients [4, 5]. Currently, a patient's eligibility for HER2-targeted therapies is determined by their IHC- or FISH-based HER2 testing scores [4]. However, clinical trials have indicated a weak correlation between HER2 expression levels and HER2 targeted therapy benefit [6, 7]. Other biological factors, such as HER2 signaling activity, may be important to measure, in addition to expression and amplification of HER2, when identifying patients eligible for HER2 therapies.

A new functional cellular analysis platform, the CELx HER2 Signaling Function (CELx HSF) Test, which uses a label-free impedance biosensor to measure HER2 signaling activity in live tumor cells, has been developed by Celcuity [8] (Figure 1). A recently completed study quantified HER2-driven signaling activity in epithelial cell samples extracted and cultured from fresh breast tissue specimens obtained from 34 patients with HER2-negative breast cancer (DAKO 0 or 1+). Of the cell samples tested, 7 of 34 HER2-negative breast tumor patients (20.5%; 95% CI = 10-37%) were found to have abnormal HER2 signaling activity ($HER2_S$+) [9] (Table 1).

The current study accomplishes the following:

1. Evaluate primary cell samples with abnormal HER2-driven signaling with four HER2 signal inhibitors—pertuzumab, lapatinib, neratinib, afatinib, and
2. Evaluate the same four HER2 signal inhibitors with 9 HER2-positive cell lines.
3. Quantify the percentage of HER2-driven signaling activity each drug could inhibit ex vivo in the primary cell samples and cell lines.
4. Compare the results between the HER2-negative primary cells and the HER2-positive cell lines.
5. Compare NRG1-driven HER2 signaling inhibition by trastuzumab and pertuzumab alone and in combination with HER2-positive cell lines and HER2-negatives cell lines and primary cells.

Methods

Specimens: A training set of de-identified fresh breast tissue specimens was obtained from 50 patients, 34 with HER2- breast cancer (IHC 0 or 1+) and 16 healthy patients.

Cell Culture: Methods for tissue extraction and primary cell culture are essentially as described previously [10, 11]. Modifications can be found in our recent publication [9]. All cell lines were maintained according to ATCC recommendations and authenticated by ATCC in March 2016.

Flow Cytometry: Flow cytometry was performed on a BD FACSCalibur using cells harvested at the time of CELx HSF test using FACS markers listed in Table 1. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2, ER, and PR that were provided for each specimen by the clinic.

CELx HSF Test: Real time live cell response to specific HER2 agonists (NRG1b or EGF) and with or without an antagonist (pertuzumab, lapatinib, afatinib, neratinib, trastuzumab) was measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, the net amount of HER2 participation in HER2 signaling initiated by the HER2 agonists ("$HER2_S$") was determined (Figure 1) [8, 9].

Table 1. Antibodies Used in Flow Cytometry

Description of Antibody	Vendor
Mouse anti-human HER2-phycoerythrin (PE), clone 24D2	Biolegend, San Diego, CA
Mouse anti-human HER1 conjugated with AlexaFluor 647, clone EGFR.1	BD Biosciences, San Jose, CA
Mouse anti-human HER3 conjugated with PerCP-sFluor710, clone SGP1	eBioscience, San Diego, CA
Mouse anti-human EPCAM conjugated with AlexaFluor 488, clone MH99	eBioscience, San Diego, CA
Mouse anti-human Claudin4 conjugated to PE, clone 382321	R&D Systems, Minneapolis, MN
Rat anti-human CD49f conjugated to PerCP/eFluor710, clone eBioGoH3	eBioscience, San Diego, CA
Mouse anti-human CD10 conjugated to Allophycocyanin (AP), cloneHL10a	BioLegend, San Diego, CA
Rabbit polyclonal anti-human estrogen receptor alpha (ERα) conjugated to AlexaFluor488	Bioss, Woburn, MA
Mouse anti-human progesterone receptor (PRG) conjugated to eFluor660, clone KMC912	eBioscience, San Diego, CA

All epitopes were extracellular with the exceptions of ER and PR. All antibodies were purchased from companies as listed who provided empirical demonstration of each of the antibodies for our applications.

Results

Figure 1. Platform Sensitivity Enables Quantification of HER2 Signaling

HER2 ligands and inhibitors used to turn on/off HER pathways

Very subtle, sub-nanometer, cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Create Pathway Dysfunction and Drug Response Scores

Typical Impedance vs. Time Data Set

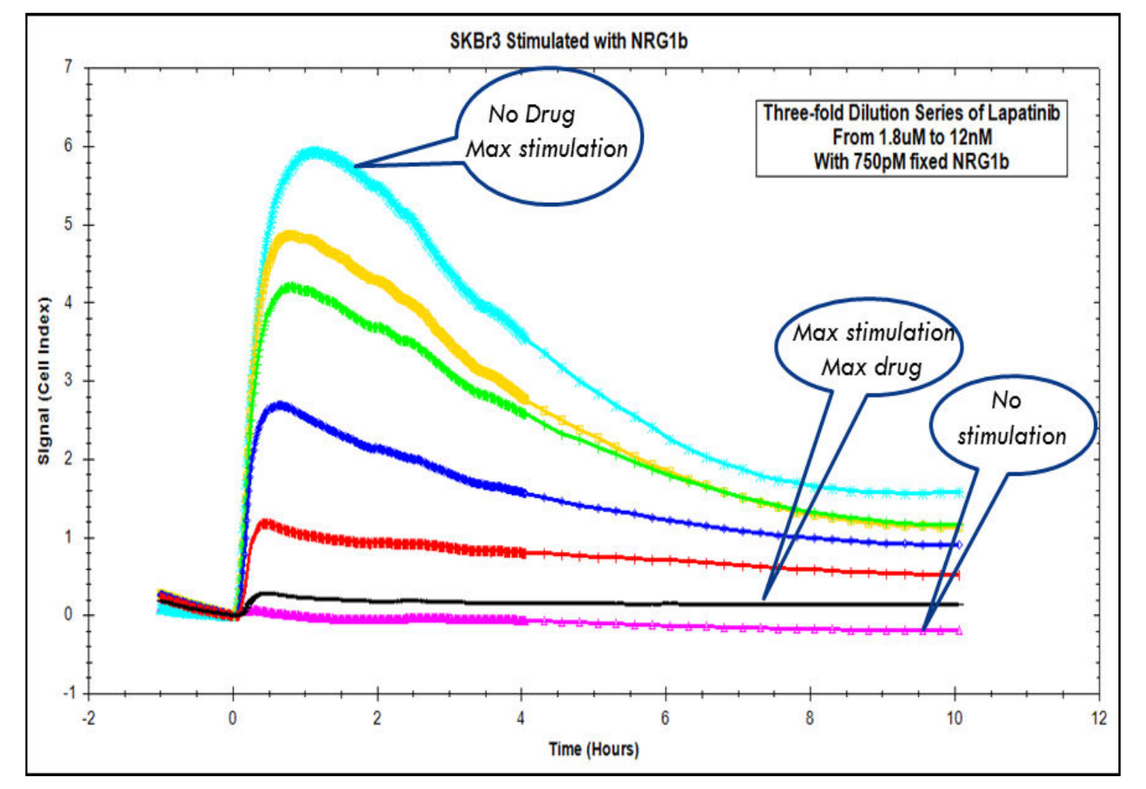
SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib
From 1.8uM to 12nM
With 750pM fixed NRG1b
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Table 2. CELx HSF Test Identified 7 HER2-negative (HER2-) Patient Samples With Abnormally High HER2-dependent Signaling ($HER2_S$+)

HER2- Patient ID	NRG1-driven HER2-dependent CELx Signal Units	EGF-driven HER2-dependent CELx Signal Units	Total HER2-dependent CELx Signal Units	CELx Test Result
R39 (tumor)	475	88	563	Abnormal
R20 (tumor)	409	120	529	Abnormal
R160 (tumor)	332	99	430	Abnormal
R82 (tumor)	272	40	312	Abnormal
R95 (tumor)	227	44	271	Abnormal
R25 (tumor)	238	29	267	Abnormal
R71 (tumor)	228	23	252	Abnormal
R22 (tumor)	1	78	79	Normal
R62 (healthy)	31	7	38	Normal
SKBr3 (HER2+ cell line)	401	143	544	Abnormal

CELx HSF tests were performed on 34 tumor cell samples from patients with breast cancer classified as HER2- to measure HER2 pathway stimulation and signal specificity. For comparative purposes, 16 primary breast epithelial cell samples from healthy patients, and DAKO IHC test standard breast cancer cell lines SKBr3 and MDA-231 were also analyzed with the CELx HSF Test. NRG1b-induced and EGF-induced CELx signals for tumor and healthy primary cells and a HER2+ reference cell line (SKBr3) (DAKO test score 3+) are summarized in the table.

The CELx HSF test identified seven HER2- patient samples having abnormally high HER2 signaling activities.

Table 3. HER2-targeting Drugs Used in This Study

HER2 Drugs	Structure	Mechanism of Action
Pertuzumab (Perjeta)	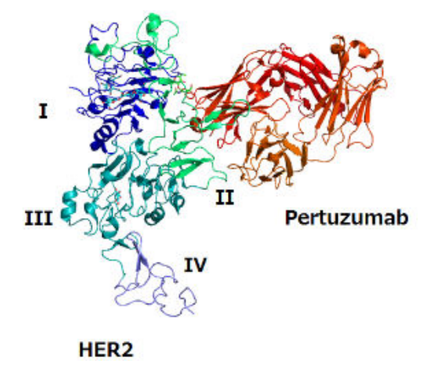	An inhibitor of HER2 hetereodimerization with other HER receptors, including EGFR, HER3, and HER4
Trastuzumab (Herceptin)	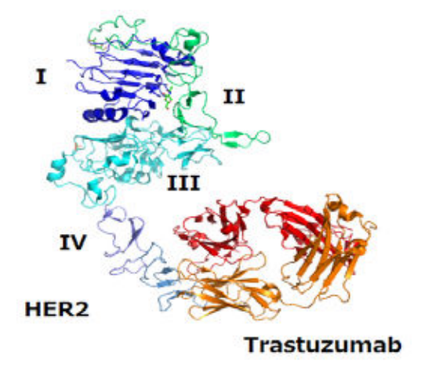	A mediator of ADCC and an indirect inhibitor of HER2-driven signaling activity
Lapatinib (Tykerb, Tyverb)	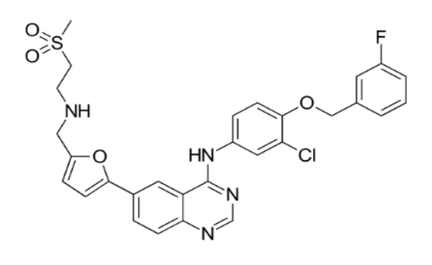	A reversible dual inhibitor of EGFR and HER2
Afatinib (Gilotrif)	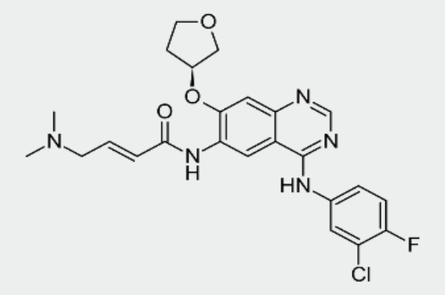	An irreversible covalent dual inhibitor of EGFR and HER2
Neratinib	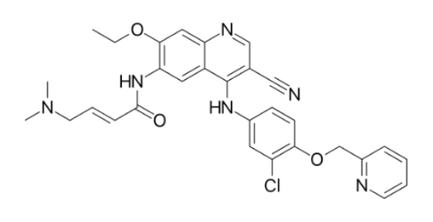	An irreversible covalent dual inhibitor of EGFR and HER2

Figure 2. CELx Test Identifies Four Different Subgroups for HER2 Signaling

Examples of CELx HSF test results (HER2+/$HER2_S$+): use of NRG1 (ligand) and pertuzumab (antagonist) to ID net HER2-driven signaling activity. Within HSF+ subgroups differential response to drug is demonstrated.

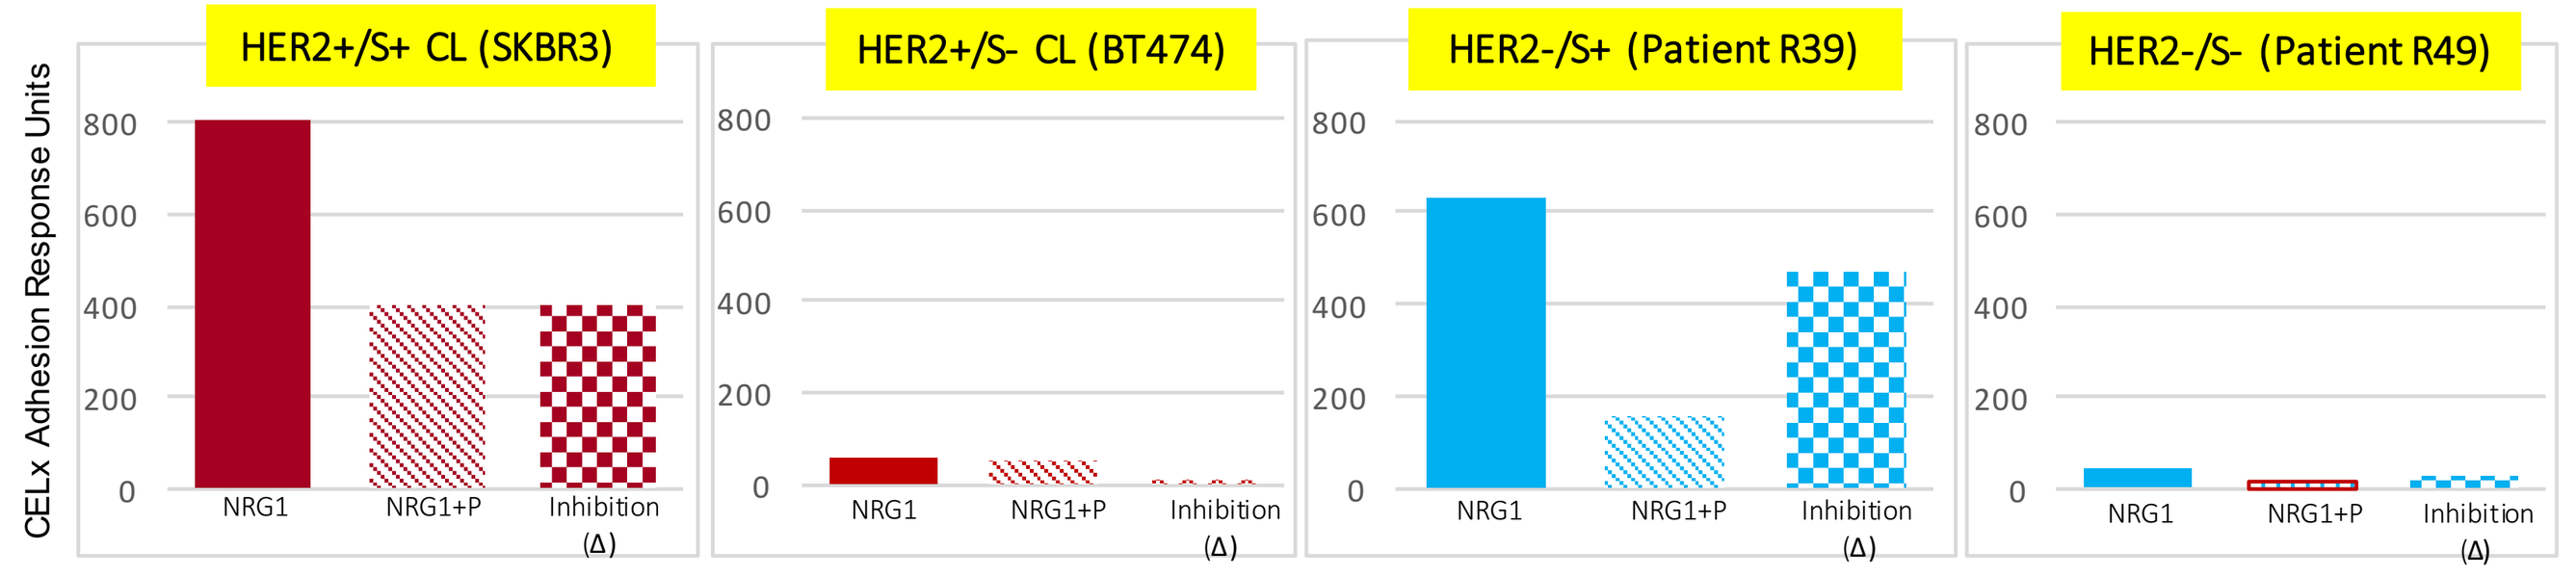
HER2+/S+ CL (SKBR3)
HER2+/S- CL (BT474)
HER2-/S+ (Patient R39)
HER2-/S- (Patient R49)
CELx Adhesion Response Units
NRG1
NRG1+P
Inhibition (Δ)

- NRG1 initiated signaling varied widely in the HER2+ samples
- SKBR3's signaling was one of highest HER2+ cell lines tested
- Difference between NRG1 and NRG1 + P above is amount of signaling involving HER2 in HER2/HER3 dimerization
- Pertuzumab inhibits 50% of the SKBR3 signal
- None of BT474's limited activity is inhibited

- HER2- primary tumor samples, R39 and R49, show a similar range of NRG1 initiated activity as the HER2+ cell lines
- The HER2-driven activity (Δ) for R39 is nearly identical to SKBR3's
- High Pz inhibition is similar to other HER2/$HER2_S$+ tumors
- R49's low signaling is typical of 80% of HER2- patient tumors

Table 4. Comparison of Inhibition of NRG1-driven HER2 Signaling Activity by Four Different HER2-targeting Drugs

Average % Inhibition of NRGI-driven HER2 Signaling Activity			
HER2 Drugs	Mechanism of Action	Cell Lines (HER2+)	Primaries (HER2-/$HER2_S$+)
Pertuzumab	HER2 dimerization inhibitor	46%	78%
Lapatinib	Reversible Dual RTKi	15%	69%
Afatinib	Irreversible Covalent Dual RTKi	47%	93%
Neratinib	Irreversible Covalent Dual RTKi	95%	100%

Figure 3. Comparison of NRG1-driven HER2 Signaling Inhibition by Trastuzumab (T) and Pertuzumab (P) Alone and in Combination

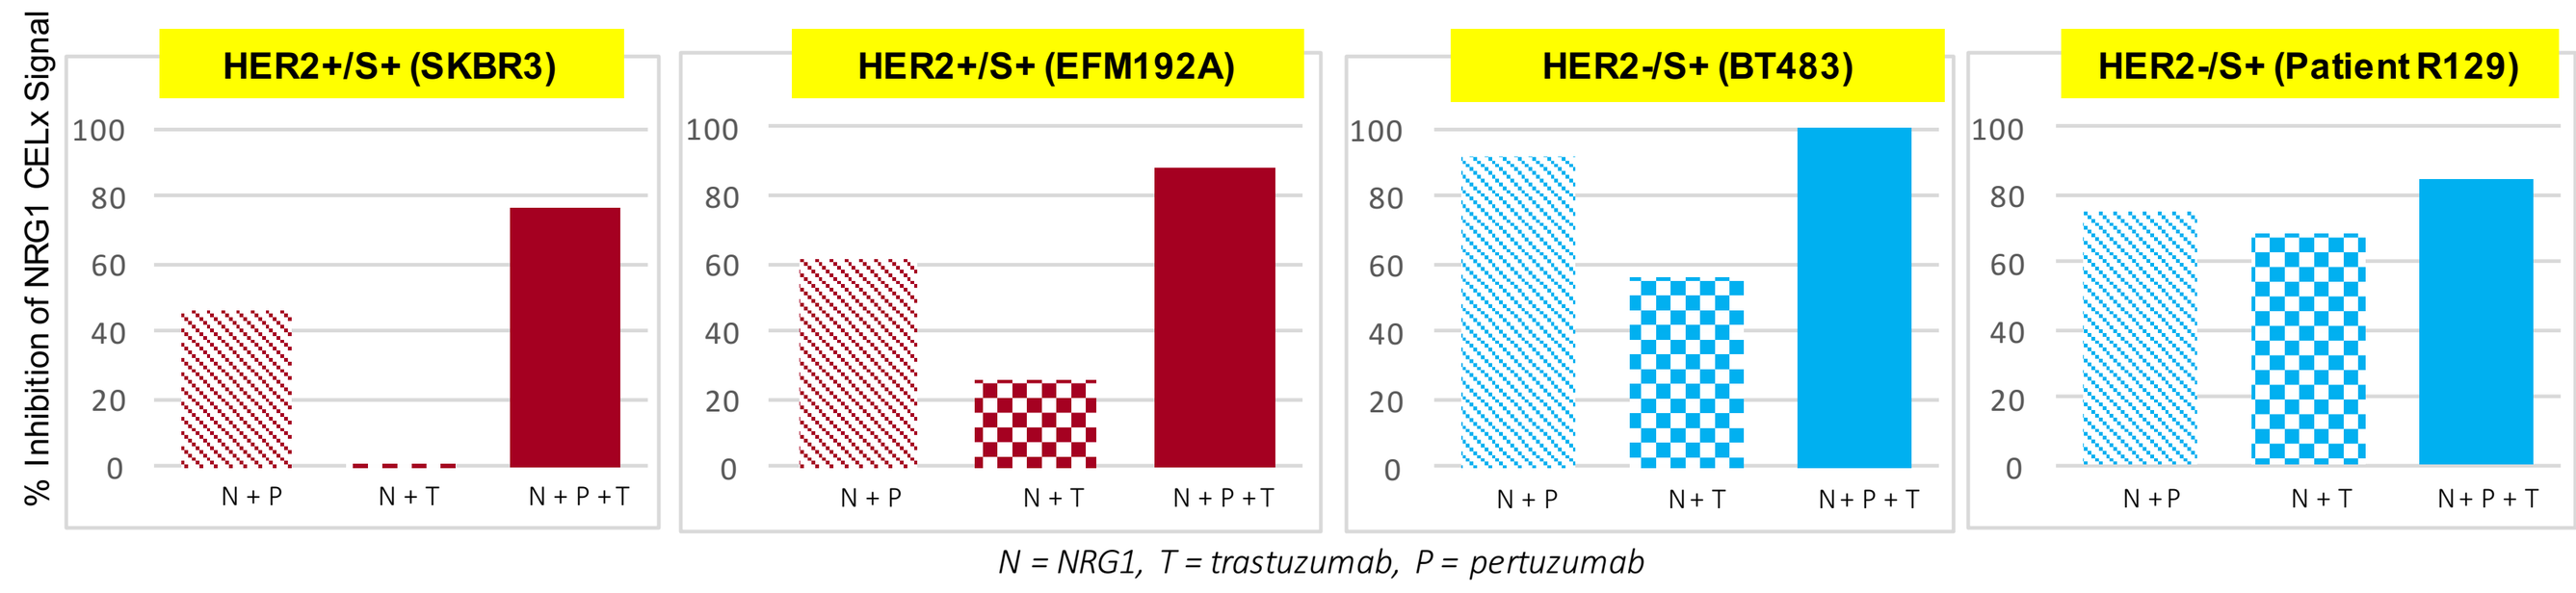
HER2+/S+ (SKBR3)
HER2+/S+ (EFM192A)
HER2-/S+ (BT483)
HER2-/S+ (Patient R129)
% Inhibition of NRG1 CELx Signal
N + P
N + T
N + P + T

N = NRG1, T = trastuzumab, P = pertuzumab

- Trastuzumab had little effect on signaling in either HER2+ cell line, particularly SKBR3
- Pertuzumab inhibited 45% and 75% in the two samples
- The combined IgG agents showed unexpected amplified response effect on SKBR3

- Trastuzumab had significant effect on both HER2-/$HER2_S$+ samples
- Pertuzumab inhibition was high in both samples
- T & P combination inhibited greater signal in both HER2- samples than either T or P alone

More signaling inhibited as combined agents in both HER2+ and HER2- cells is noted.

Summary of Results

- Four HER2 signal inhibitors each inhibited an average of at least 69% of the HER2-driven signaling activated by NRG1 stimulation in the HER2-negative primary cell samples.
- The highest level of inhibition was found with the two irreversible covalent RTK inhibitors—afatinib and neratinib.
- The two mAb's tested, trastuzumab and pertuzumab, inhibited greater signaling in both HER2- samples than either alone.
- The HER2 drugs all inhibited a higher percentage of HER2-driven signaling in the HER2- primary cells than the HER2+ cell lines.

Conclusions

- These findings provide strong evidence that HER2 signal inhibitors are effective in blocking abnormal levels of HER2-driven signaling ($HER2_S$+) ex vivo in live primary cells from breast cancer patients with normal expression levels of HER2.
- The data suggest a new group of breast cancer patients, HER2-negative with abnormal HER2 signaling (HER2-/$HER2_S$+), may benefit from the addition of HER2 signal inhibitors to current combination therapeutic regimens.
- Additional studies to confirm these findings are underway.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. Cancer Res. 2004;64(23):8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. J Clin Oncol. 2010;28(1):92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. Oncologist. 2009;14(4):320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. Science. 1987;235(4785):177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. Cancer Treat Rev. 2014;40(2):276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. N Engl J Med. 2008;358(13):1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. J Clin Oncol. 2010;28(28):4307-4315.
8. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. BMC Cancer. 2016; in revision.
9. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. Oncotarget. 2016; in press.
10. Stampfer MR, et al. Culture of human mammary epithelial cells. In: Freshney RI and Freshney MG, eds. Culture of Epithelial Cells.
11. Proia DA and Kuperwasser C. Reconstruction of human mammary tissues in a mouse model. Nat Protoc. 2006;1(1):206-214.

P4-12-06

This presentation is the intellectual property of Celcuity LLC. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.