CELx™ | breast
HER2 Signaling Function Test

New method to measure functional HER2-driven signaling activity in primary tumor cells identifies HER2-negative breast cancers with abnormal HER2 signaling activity: new group of patients may benefit from anti-HER2 therapy

Lance G Laing, David J Burns, Yao Huang, Ian A MacNeil, Benjamin E Rich, Sajjad M. Soltani, Samantha Myhre, Brian F Sullivan Celcuity LLC, 16305 36th Ave N, Suite 450, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 6–10, 2016

celcuity™
www.celcuity.com

Background

HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15-20% of breast cancers and is associated with more aggressive disease progression, metastasis, and a poorer prognosis [1-4]. Agents targeting HER2, such as trastuzumab, lapatinib, and pertuzumab, significantly improve clinical outcomes in HER2+ patients [4, 5]. Currently, a patient's eligibility for HER2-targeted therapies is determined by their IHC- or FISH-based HER2 testing scores [4]. However, clinical trials have indicated a weak correlation between HER2 expression levels and HER2 targeted therapy benefit [6, 7]. Other biological factors, such as HER2 signaling activity, may be important to measure, in addition to expression and amplification of HER2, when identifying patients eligible for HER2 therapies.

It has been well established that in breast tumors, the catalytically inactive HER family member HER3 is an obligate partner for HER2 and couples active HER2 to the PI3K/AKT pathway to drive tumor cell growth and survival [8-11]. Numerous other mechanisms have been proposed to examine biomarker correlation with drug efficacy that to date have not been clinically adopted, e.g., PI3K activating mutation [12] and HER2 mutants [13].

To measure the HER2-driven signaling activity of a patient's tumor cells, a new assay using an impedance biosensor, the CELx HER2 Signaling Function (CELx HSF) Test, was developed [14, 15].

This study provides an initial assessment of the CELx test, specifically to accomplish the following:

1. Quantify HER2-driven signaling activity ($HER2_S$) in cell lines and primary epithelial cells.
2. Assess the correlation between HER2 expression levels and HER2 signaling function.
3. Define a preliminary cut-point between normal and abnormal HER2 signaling function.
4. Estimate the proportion of HER2- primary breast cancer tumors with abnormal HER2 signaling.

Methods

Reference Breast Cancer Cell Lines: 9 HER2+, 10 HER2- were tested, including two cell lines used as 0 and +3 controls in IHC HER2 clinical tests.

Specimens: A training set of de-identified fresh breast tissue specimens was obtained from 50 patients, 34 with HER2- breast cancer (IHC 0 or 1+) and 16 healthy patients. Summary of 34 HER2- tumor patient characteristics is in Table 1.

Cell Culture: Methods for tissue extraction and primary cell culture are essentially as described previously [16, 17]. All cell lines were maintained according to ATCC recommendations and authenticated by ATCC in March 2016.

Flow Cytometry: Flow cytometry of all cells was performed on a BD FACSCalibur using cells harvested at the time of the CELx HSF test using FACS markers listed in Table 2. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2, ER, and PR that were provided for each specimen by the clinic.

CELx HSF Test: Real time live cell response to specific HER2 agonists (NRG1b or EGF) with or without an antagonist (pertuzumab, HER2 dimer blocker) was measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, the net amount of HER2 participation in HER2 signaling initiated by the HER2 agonists ("$HER2_S$") was determined [15, 16].

Table 1. Summary of Patient Characteristics

Characteristic	No.	Percentage (%)
No. of breast cancer patients	34	
Age, years		
Mean	57.5	
36-60 years old	18	53
61-79 years old	16	47
Clinical Stage		
I	5	15
II	22	65
III	5	15
N/A*	2	6
Histology		
Invasive only	13	38
Invasive Ductal/DCIS mixed	11	32
Lobular/other	8	24
N/A*	2	6
Lymph Status		
Metastatic	12	35
Not Metastatic	20	59
N/A*	2	6
Estrogen Receptor Status		
ER+	26	76
ER-	1	3
N/A*	7	21
HER2 IHC score/FACS		
0/1+	34	100
2+/3+	0	0

Information not available due to nature of some de-identified surplus tissue used in this study.

Table 2. Antibodies Used in Flow Cytometry

Description of Antibody	Vendor
Mouse anti-human HER2-phycoerythrin (PE), clone 24D2	Biolegend, San Diego, CA
Mouse anti-human HER1 conjugated with AlexaFluor 647, clone EGFR.1	BD Biosciences, San Jose, CA
Mouse anti-human HER3 conjugated with PerCP-sFluor710, clone SGP1	eBioscience, San Diego, CA
Mouse anti-human EPCAM conjugated with AlexaFluor 488, clone MH99	eBioscience, San Diego, CA
Mouse anti-human Claudin4 conjugated to PE, clone 382321	R&D Systems Minneapolis, MN
Rat anti-human CD49f conjugated to PerCP/eFluor710, clone eBioGoH3	eBioscience, San Diego, CA
Mouse anti-human CD10 conjugated to Allophycocyanin (AP), cloneHL10a	BioLegend, San Diego, CA
Rabbit polyclonal anti-human estrogen receptor alpha (ERα) conjugated to AlexaFluor488	Bioss, Woburn, MA
Mouse anti-human progesterone receptor (PRG) conjugated to eFluor660, clone KMC912	eBioscience, San Diego, CA

All epitopes were extracellular with the exceptions of ER and PR. All antibodies were purchased from companies as listed who provided empirical demonstration of each of the antibodies for our applications.

As shown in Table 1, we collected a random training set of tumors from UMN cancer patients with a distribution of stage, histology, and age to study a new test for identifying pathway dysfunction in HER2- breast cancer.

Results

Figure 1. Characterization of Primary Epithelial Cells Derived From Patient Tissue

A
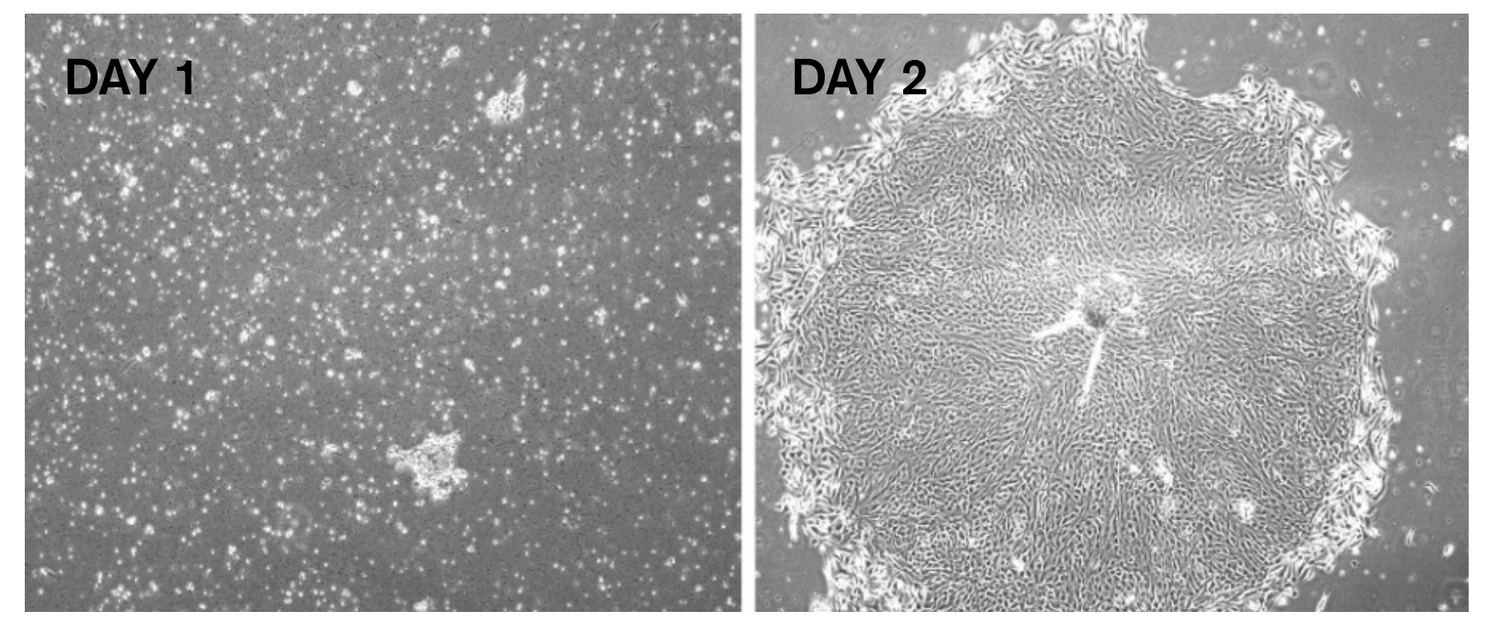
DAY 1
DAY 2

(A) A representative culture of primary cells from a digested tumor biopsy.
The results show that colonies appear typically epithelial, marked by a tight cobblestone structure.

B C
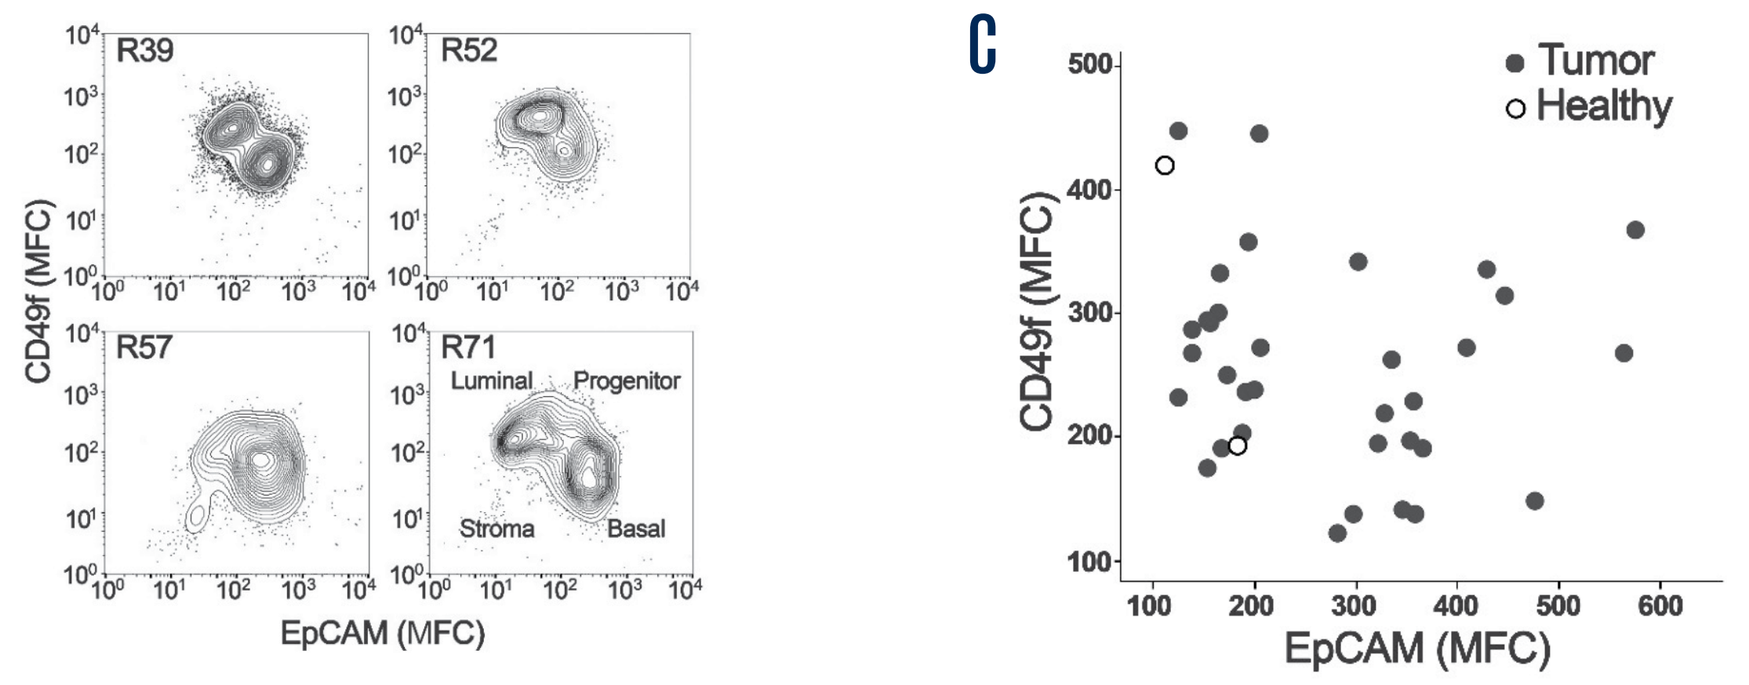
R39
R52
R57
R71
Luminal
Progenitor
Stroma
Basal
CD49f (MFC)
EpCAM (MFC)
Tumor
Healthy
CD49f (MFC)
EpCAM (MFC)

(B) Flow cytometric analysis of luminal (EpCAM+, Claudin4+) and basal (CD49f+, CD10+) markers on representative tumor primary cells harvested at the time of CELx HSF test.
(C) Plot showing the Mean Fluorescence Channel (MFC) of the luminal marker EpCAM (x-axis) and the basal/progenitor biomarker CD49f (y-axis) for all 34 tumor samples tested.
The results shown in B and C indicate populations containing both luminal-like and basal-like characteristics.

D

	Fluorescence (MFC)	
	Cell Lines	Primary Cells
HER2+	1000-4000	
HER2-	30-500	30-230
ERα	25-100	50-240
PR	40-1300*	60-240

** The majority of the cell lines expressed PR between 40 and 280*

(D) Comparison of expression levels of HER2, ERα, and PR, determined by flow cytometry, between primary cells and cell lines (HER2+ and HER2-).
The results show that primary cells grown from tumor samples of the ER+/PR+ patients continue to express ER and PR.

E
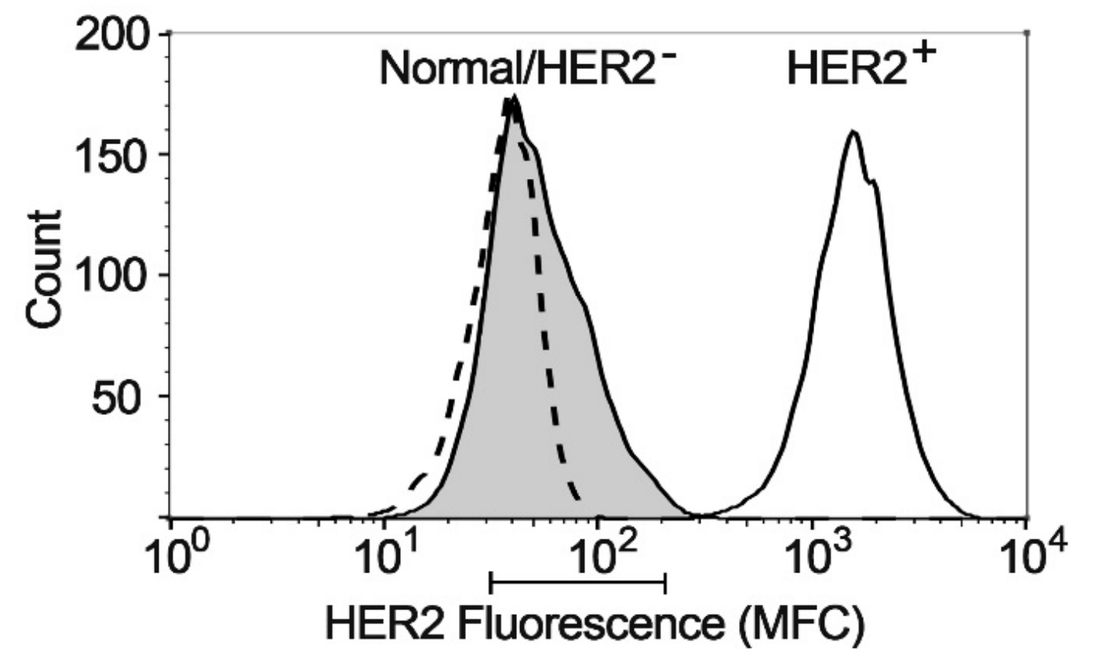
Normal/HER2-
HER2+
Count
HER2 Fluorescence (MFC)

(E) Histogram plot of HER2 expression of a representative tumor (shaded peak) compared to HER2+ cell line SKBR3 (DAKO 3+) (solid line) and HER2- cell line MDA231 (DAKO 0) (dashed line) that is coincidentally in the same range as the healthy samples. The bar below the graph represents the range of means from all the primary tumors and healthy tissue tested (MFC range 31-210).

The results shown in D and E indicate that all 34 tumor and 16 healthy samples had normal/low HER2 expression levels in the range of HER2- cell lines. This confirms the clinical pathology test results reported to us for each specimen, namely that the 34 primary tumor samples were true HER2-.

Table 3. HER1-3 Expression in Primary Tumor Cells

Sample #	Genotype	HER1	HER2	HER3
R131	ER+, HER2-, PR+	104	105	140
R160	ER+, HER2-, PR-	65.8	96.2	151
R20	ER+, HER2-, PR+	93.3	99.8	173
R22	ER+, HER2-, PR+	229	101	158
R23	ER+, HER2-, PR+	216	97	188
R25	ER-, HER2-, PR-	171	105	14
R35	HER2-	12.4	28.9	51.3
R36	HER2-	130	117	131
R37	HER2-	146	87.4	164
R39	HER2-	N/A	201	N/A
R40	HER2-	34.2	42.7	88.1
R41	ER+, HER2-, PR+	54.8	113	624
R42	ER+, HER2-, PR+	214	106	150
R43	ER+, HER2-, PR+	141	30.6	171
R45	ER+, HER2-, PR+	82.8	151	503
R47	HER2-	38.1	163	266
R49	HER2-	107	73.7	115
R51	ER+, HER2-, PR+	373	116	83.7
R52	ER+, HER2-, PR+	28.4	36.2	84.7
R53	ER+, HER2-, PR+	41.6	163	327
R54	ER+, HER2-, PR+	159	56.5	107
R56	ER+, HER2-, PR+	185	115	209
R57	ER+, HER2-, PR+	111	178	361
R58	ER+, HER2-, PR+	45.4	96.4	311
R60	ER+, HER2-, PR+	32.2	47.7	88.7
R66	ER+, HER2-, PR+	156	72.5	104
R69	ER+, HER2-, PR+	248	144	195
R71	ER+, HER2-, PR+	60	33.1	179
R79	ER+, HER2-, PR+	186	114	129
R82	ER+, HER2-, PR+	95.2	129	148
R84	ER+, HER2-, PR+	165	74.2	165
R91	ER+, HER2-, PR-	318	99.3	117
R95	ER+, HER2-, PR+	85.2	166	465
R99	ER+, HER2-, PR+	119	46.1	63
R62	Healthy example	111	100	141
SKBr3	HER2+ Cell Line (DAKO 3+)	47.8	2386	290
	HER2 - Tumor Range	360.6	172.1	610
	HER2 - Tumor Max	373	201	624
	HER2 - Tumor Min	12.4	28.9	14

**HER1-3 levels were determined by flow cytometry using the antibody listed in Table2.*
***The CELx Test positive patient samples are in red.*

Figure 2. Platform Sensitivity Enables Quantification of HER2 Signaling

HER2 ligands and inhibitors used to turn on/off HER pathways
↓
Very subtle, sub-nanometer, cell adhesion changes are measured
↓
Response quantified in Cell Index signaling units over time to detect patterns
↓
Create Pathway Dysfunction and Drug Response Scores

Typical Impedance vs. Time Data Set

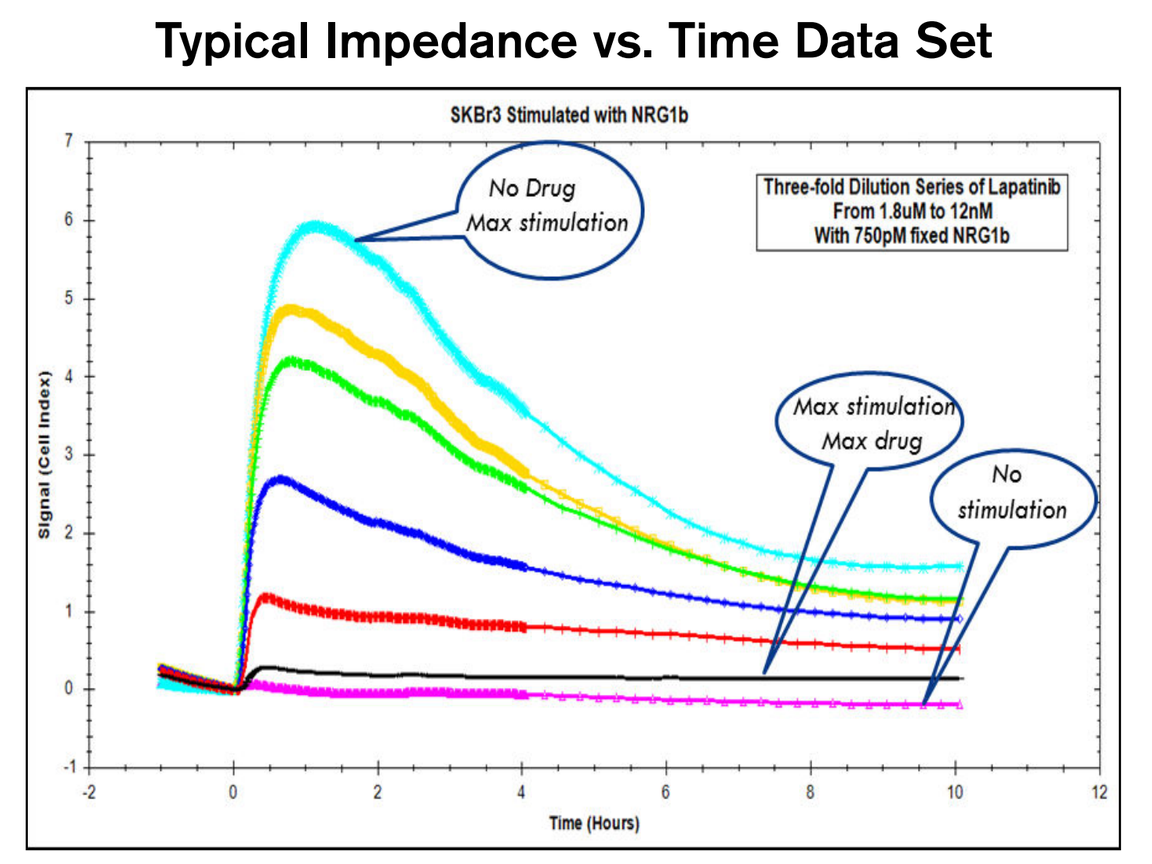
SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib
From 1.8uM to 12nM
With 750pM fixed NRG1b
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Data shown in Table 3 suggest:

- The HER1-3 expression data provided no key to determining which samples would have CELx HSF Test positive results.
- HER1 expression in all 7 CELx HSF test positive samples was below the mean and median for this group of 34 tumor samples.
- HER2 protein level in the tumor group showed no correlation with CELx HSF test positive group result even within the HER2- group (P = 0.15, R2 = 0.069).
- In the 7 CELx HSF test positive HER2- samples, the HER3 expression levels span from low to high range.

Summary of Results

- Of the HER2- breast tumor cell samples tested, 7 of 34 patients (20.5%; 95% CI = 10-37%) had net HER2 signaling activity, as determined by the CELx test, that was greater than the median $HER2_S$ of the HER2+ cell lines.
- There was no categorical correlation between HER2 IHC status (+ or -) and HER2 signaling activity (abnormal or normal) (Pearson's Chi-Square = 3.68; Phi Max = -0.78, Contingency Coefficient 0.28).
- The median $HER2_S$, or net HER2 signaling activity, was comparable for the HER2- tumors, HER2- cell lines, and the healthy patient samples (Md = 100, 117, 77, respectively). The median $HER2_S$ in HER2+ cell lines (Md = 248) is approximately 2.5-3.0 fold greater than the median of the other groups.
- A $HER2_S$ above 250 was considered abnormal or test positive, and was defined as the cut-point. Two clinical IHC HER2 test control cell lines, SKBR3 for HER2+ with IHC = 3+ and MDA-MB231 for HER2- with IHC = 0, have CELx HSF test measurands of 544 and 0, respectively.

Figure 3. Defining Test Conditions and Specificity of the CELx HSF Test Measurand

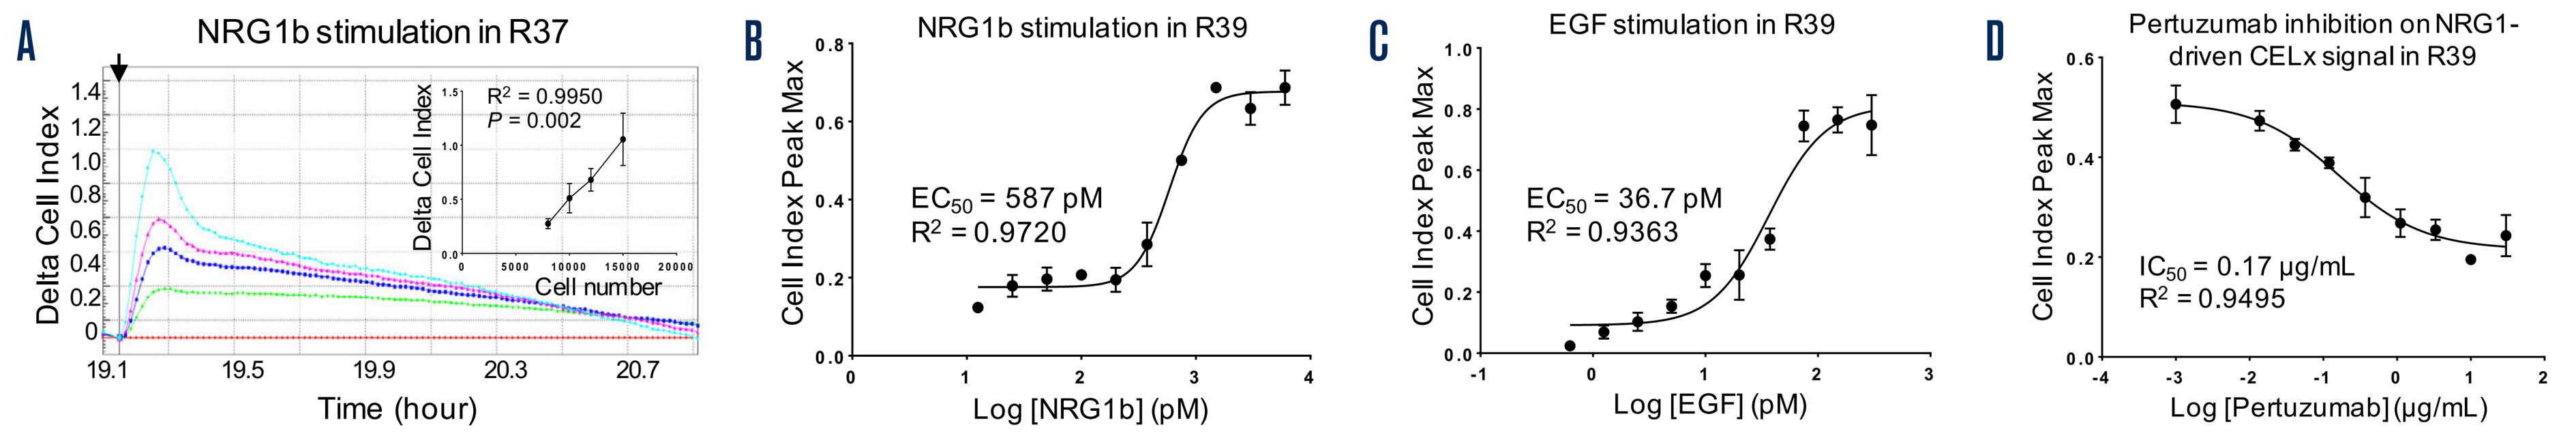
A
NRG1b stimulation in R37
R^2 = 0.9950
P = 0.002
Delta Cell Index
Cell number
Time (hour)
B
NRG1b stimulation in R39
EC_{50} = 587 pM
R^2 = 0.9720
Cell Index Peak Max
Log [NRG1b] (pM)
C
EGF stimulation in R39
EC_{50} = 36.7 pM
R^2 = 0.9363
Log [EGF] (pM)
D
Pertuzumab inhibition on NRG1-driven CELx signal in R39
IC_{50} = 0.17 µg/mL
R^2 = 0.9495
Log [Pertuzumab] (µg/mL)

(A) R37 primary cells seeded at different densities were stimulated with NRG1b (3nM). CELx curves are displayed using Delta CI values to demonstrate the relative signals normalized to the time point (arrow) when NRG1b was added. Positive correlation between cell number and NRG1-driven CELx signal is shown in the inset.
(B and C) Dose-response curves of NRG1b and EGF stimulation of CELx signals in R39 primary cells.
(D) Dose-response curve of pertuzumab showing its specific inhibitory effect on NRG1b-driven CELx signal.
Collectively, these results demonstrate that the CELx HSF test can specifically detect ligand-induced HER2-related signals and determine whether a HER2-driven test signal is sensitive or insensitive to a HER2-targeted drug (e.g., pertuzumab).

Figure 4. HER2- Abnormal Signaling by CELx HSF Test

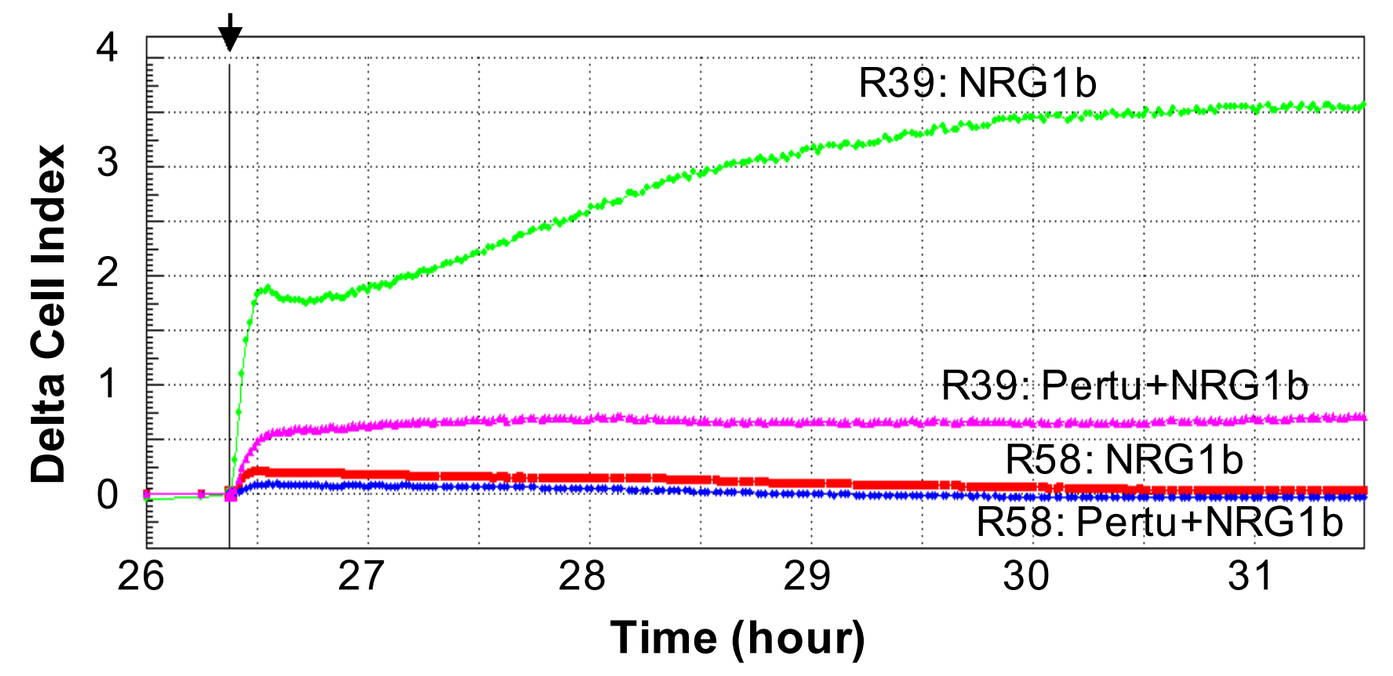
R39: NRG1b
R39: Pertu+NRG1b
R58: NRG1b
R58: Pertu+NRG1b
Delta Cell Index
Time (hour)

Representative CELx time-course curves representing a high, abnormal HER2 signaling activity in a high responder (R39) and a low HER2 signaling activity in a nonresponder (R58). In this display, curves of NRG1 stimulation in the absence versus presence of pertuzumab (10µg/mL) are presented.
The data show that the high NRG1b responder has more than 10 times greater signal than the low responder, indicating the test has a large dynamic range.

Table 4. CELx HSF Test Results of 7 Abnormal Signaling HER2- Patient Samples With Normal and HER2+ Cell Line Comparators

HER2-Negative Patient Samples	Total NRG1 Signaling	Total EGF Signaling	Total HER-dependent Signaling	Total HER2-dependent Signaling from NRG1	Total HER2-dependent Signaling from EGF	Test Measurand (*Total HER2-dependent Signaling*)	CELx Test Result
R39	634	294	928	475	88	563	Abnormal
R20	539	286	824	409	120	529	Abnormal
R160	349	229	578	332	99	430	Abnormal
R82	336	332	668	272	40	312	Abnormal
R95	250	116	366	227	44	271	Abnormal
R25	326	206	533	238	29	267	Abnormal
R71	336	211	547	228	23	252	Abnormal
R22	8	353	361	1	78	79	Normal
R62 Healthy	32	13	46	31	7	38	Normal
SKBR3 cell line	802	367	1169	401	143	544	Abnormal

CELx HSF tests were performed on 34 primary tumor cell samples from patients with breast cancer classified as HER2- to measure HER2 pathway stimulation and signal specificity. For comparative purposes, 16 primary breast epithelial cell samples from healthy patients, and DAKO IHC test standard breast cancer cell lines SKBr3 and MDA-231 were also analyzed with CELx HSF Tests. NRG1b-induced and EGF-induced CELx signals for tumor and healthy primary cells and a HER2+ reference cell line (SKBr3) (DAKO test score 3+) are summarized in the table.
The CELx HSF test identified seven HER2- patient samples having abnormally high HER2 signaling activity comparable to HER2+ cell line signaling activity.

Figure 5. HER2-Driven Signaling Activity Examples (HER2+/$HER2_S$+)

NRG1-driven activity and use of pertuzumab to ID net HER2-driven activity

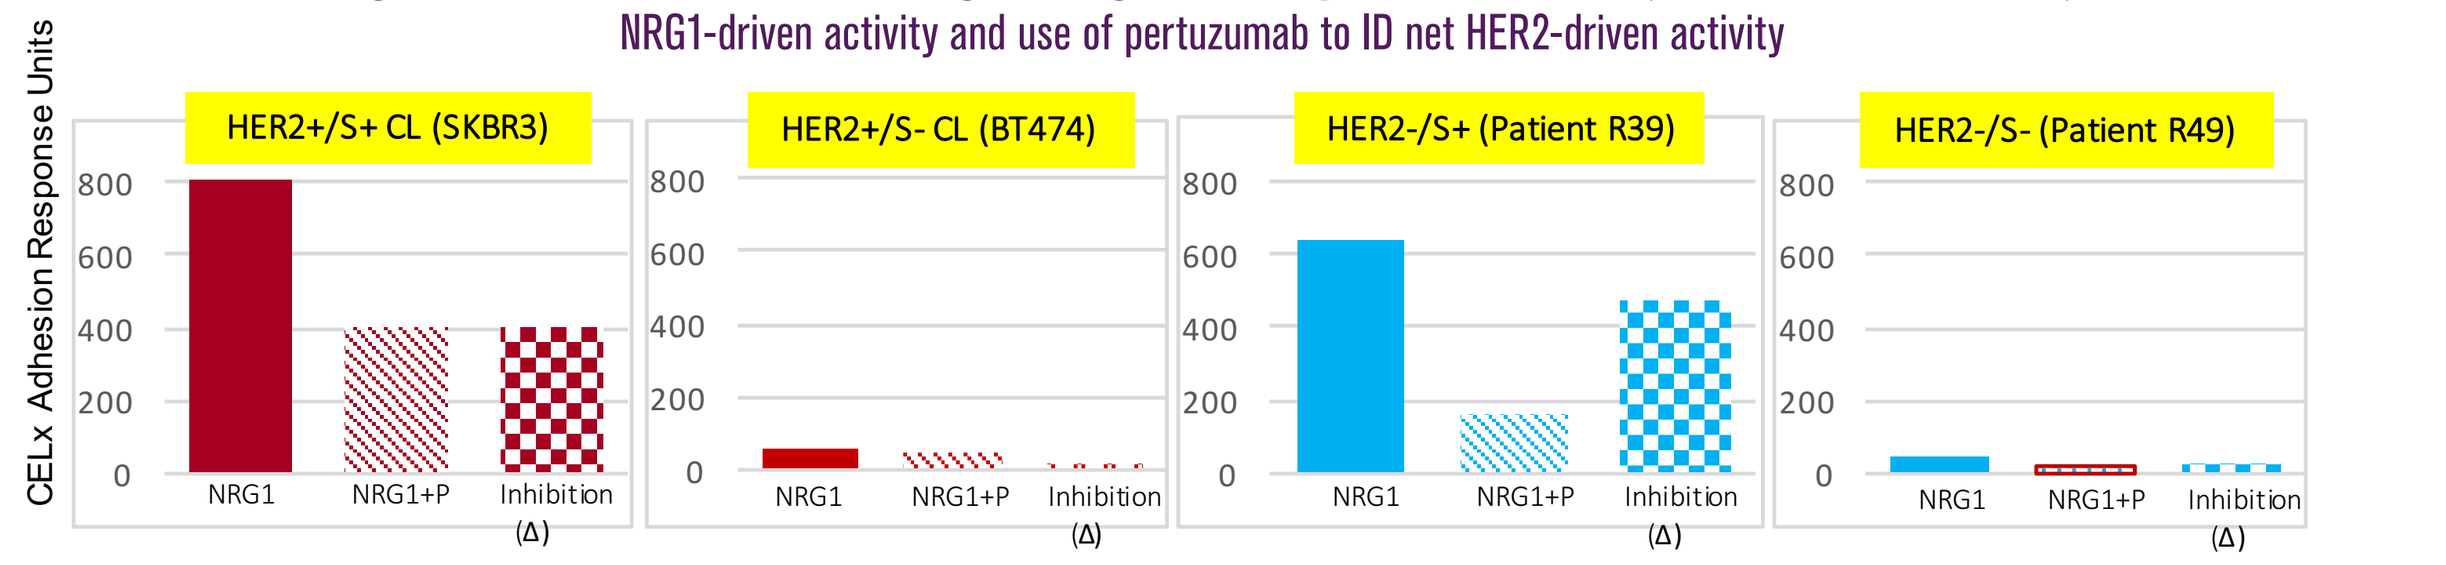
CELx Adhesion Response Units
HER2+/S+ CL (SKBR3)
HER2+/S- CL (BT474)
HER2-/S+ (Patient R39)
HER2-/S- (Patient R49)
NRG1
NRG1+P
Inhibition (Δ)

- NRG1 initiated signaling varied widely in the HER2+ samples
- SKBR3's signaling was one of highest HER2+ cell lines tested
- Difference between NRG1 and NRG1 + P above is amount of signaling involving HER2 in HER2/HER3 dimerization
- Pertuzumab inhibits 50% of the SKBR3 signal
- None of BT474's limited activity is inhibited

- HER2- primary tumor samples, R39 and R49, show a similar range of NRG1 initiated activity as the HER2+ cell lines
- The HER2-driven activity (Δ) for R39 is nearly identical to SKBR3's
- High Pz inhibition is similar to other HER2/$HER2_S$+ tumors
- R49's low signaling is typical of 80% of HER2- patient tumors

Figure 6. Comparison of CELx HSF Test Results for Different HER2 Sample Groups

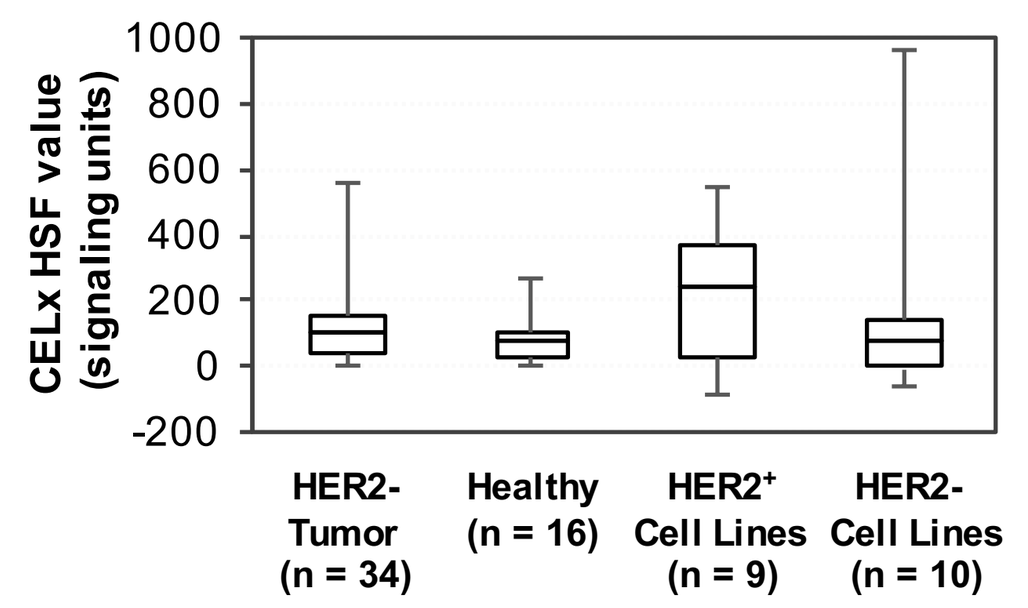
CELx HSF value (signaling units)
HER2- Tumor (n = 34)
Healthy (n = 16)
HER2+ Cell Lines (n = 9)
HER2- Cell Lines (n = 10)

Box-and-whisker plots of the CELx HSF test scores for four cell sample groups (HER2- patient-derived tumor cells and cells derived from healthy tissue are plotted with HER2+ cell lines, HER2- cell lines.

A cutoff of 250 signaling units (90% of the upper range of the healthy responses and coincidental with the median value of the HER2+ cell line population) was determined to represent an abnormally active HER2 signaling network in primary breast cancer cells.
Of the HER2- breast tumor cell samples tested, 7 of 34 patients (20.5%; 95% CI 10.0-37.1) had HER2 signaling activity that was characterized as abnormally high.

The CELx HSF Test identified at least four subtypes of the HER2 Samples:
1. HER2+/S+ (HER2+ cells having high HER2 signaling activities).
2. HER2+/S- (HER2+ cells having low HER2 signaling activities).
3. HER2-/S+ (HER2- cells having high HER2 signaling activities).
4. HER2-/S- (HER2- cells having low HER2 signaling activities).

Conclusions

- These findings provide strong evidence that measurement of HER2 signaling activity may provide clinically relevant information, particularly for HER2- breast cancer patients.
- These results suggest a new group of HER2 negative breast cancer patients with abnormal HER2 signaling may benefit from anti-HER2 therapy.
- Additional studies are underway to confirm these findings and to analytically validate the CELx HSF test.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. Cancer Res. 2004;64:8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. J Clin Oncol. 2010;28:92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. Oncologist. 2009;14:320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. Science. 1987;235:177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. Cancer Treat Rev. 2014;40:276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. N Engl J Med. 2008;358:1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. J Clin Oncol. 2010;28:4307-4315.
8. Baselga J and Swain SM. Novel anticancer targets: revisiting ERBB2 and discovering ERBB3. Nat Rev Cancer. 2009;9:463-475.
9. Campbell MR and Moasser MM. HER Targeting in HER2-Negative Breast Cancers: Looking for the HER3 Positive. Clin Cancer Res. 2015;21:2886-2888.
10. Holbro T, et al. The ErbB2/ErbB3 heterodimer functions as an oncogenic unit: ErbB2 requires ErbB3 to drive breast tumor cell proliferation. Proc Natl Acad Sci U S A. 2003;100:8933-8938.
11. Lee CY, et al. Neuregulin autocrine signaling promotes self-renewal of breast tumor-initiating cells by triggering HER2/HER3 activation. Cancer Res. 2014;74:341-352.
12. Mukohara T. PI3K mutations in breast cancer: prognostic and therapeutic implications. Breast Cancer. 2015;7:111-123.
13. Bose R et al. Activating HER2 mutations in HER2 gene amplification negative breast cancer. Cancer Discov. 2013;3:224-237.
14. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. Oncotarget. 2016; in press.
15. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. BMC Cancer. 2016; in revision.
16. Stampfer MR, et al. Culture of human mammary epithelial cells. In: Freshney RI and Freshney MG, eds. Culture of Epithelial Cells. 2002.
17. Proia DA and Kuperwasser C. Reconstruction of human mammary tissues in a mouse model. Nat Protoc. 2006;1:206-214.

P6-07-14

This presentation is the intellectual property of Celcuity LLC. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.