CELx™ | breast

HER2 Signaling Function Test

Use of a functional signal profiling test to determine the prevalence of abnormal HER2-driven signaling activity in the HER2-negative breast cancer patient population: **New patient group may benefit from anti-HER2 therapy**

Lance G Laing, David J Burns, Ian A MacNeil, Benjamin E Rich, Sajjad M Soltani, Samantha Myhre, Brian F Sullivan – Celcuity LLC, 16305 36th Ave N, Suite 450, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 5–9, 2017

celcuity™
www.celcuity.com

Background

Biological factors, such as HER2 signaling activity, may be important to measure in addition to expression and amplification of HER2 when identifying patients eligible for HER2 therapies.

HER2 gene *(ERBB2)* amplification and/or HER2 protein overexpression is detected in approximately 15–20% of breast cancers and is associated with more aggressive disease progression, metastasis and a poorer prognosis.[1-4] Agents targeting HER2, such as trastuzumab, lapatinib and pertuzumab, significantly improve clinical outcomes in HER2+ patients.[4,5] Currently, a patient's eligibility for HER2-targeted therapies is determined using IHC or FISH HER2 tests.[4] However, clinical trials have indicated a weak correlation between HER2 expression or amplification levels and HER2-targeted therapy benefit.[6,7]

The catalytically inactive HER receptor family member, HER3, is a preferred dimerization partner for HER2 and couples active HER2 to the PI3K/AKT pathway to drive tumor cell growth and survival in breast tumors.[8-11] Numerous other mechanisms and associated biomarkers have been examined (e.g., PI3K activating mutation[12] and HER2 mutants[13] to assess correlation with drug efficacy, but to date they have not been clinically adopted.

To measure the HER2-driven signaling activity of a patient's tumor cells, a new assay using an impedance biosensor, the CELx HER2 Signaling Function (CELx HSF) Test, was developed.[14,15]

The CELx HSF Test measures HER2 signaling activity in live tumor cells using a label-free impedance biosensor to identify HER2-negative breast cancer patients likely to be responsive to treatment with anti-HER2 therapies. Previous studies quantified HER2-driven signaling activity in a training set (N=34) of primary tissue samples from HER2-negative breast cancer patients and found 21% of the samples had abnormal HER2 signaling. Other studies confirmed that anti-HER2 therapies, such as trastuzumab, pertuzumab, afatinib and neratinib, are as effective in inhibiting HER2-driven signaling activity in HER2- tumor cells as they are in HER2+ tumor cells. This study set out to confirm the prevalence of abnormal HER2 signaling among a new population of HER2-negative breast cancer patients in a larger sample (N=114) and to characterize the potential sensitivity and specificity of the CELx HSF Test.

This study provides an analytical validation of the CELx test, specifically to accomplish the following:

1. Quantify HER2-driven signaling activity ($HER2_S$) in primary breast tumor cells
2. Verify the cut-point between normal and abnormal HER2 signaling function in the HER2-negative patient population
3. Estimate the proportion of HER2- primary breast cancer tumors with abnormal HER2 signaling

Methods

Reference Breast Cancer Cell Lines: A set were tested previously, including the two cell lines used as 0 and 3+ controls in IHC HER2 clinical tests.

Specimens: A training set of de-identified fresh breast tissue specimens was obtained from 114 patients diagnosed with HER2- breast cancer. See Summary of 114 HER2- tumor patient characteristics in Table 1.

Cell Culture: Methods for tissue extraction and primary cell culture are essentially as described previously [16,17]. Cell lines were maintained according to ATCC recommendations and authenticated by ATCC in March 2016.

Flow Cytometry: Flow cytometry of all cell samples was performed on a BD FACSCalibur using cells harvested at the time of the CELx HSF Test. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2 that were provided for each specimen by the clinic.

CELx HSF Test: Real-time live cell response to specific HER2 agonists (NRG1b and EGF) with or without an antagonist (2C4, a HER2 dimer blocker) was measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, the net amount of HER2 participation in HER-family signaling initiated by HER receptor agonists ("$HER2_S$") was determined.[15,16] Samples with HER2 signaling activity levels above a previously determined cutoff value of 250 signaling units that was attenuatable with a HER2 dimer blocker were identified as abnormal.

Statistical Analyses: A data set of 114 CELx scores from HER2-negative patients collected between July 2015 and January 2017 and tested in 2016 and January 2017 was analyzed. A normal mixture model was fitted to the combined data set using the normalmixEM procedure in the R package mix tools. Two runs of the statistical analysis were made, fitting 2 and 3 components, along with a baseline single-component model.

Figure 1. Platform Biosensor Sensitivity Enables Quantification of HER2 Signaling

HER2 ligands and inhibitors used to turn on/off HER pathways

Very subtle, sub-nanometer, cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Create Pathway Dysfunction and Drug Response Scores

Typical Impedance vs. Time Data Set

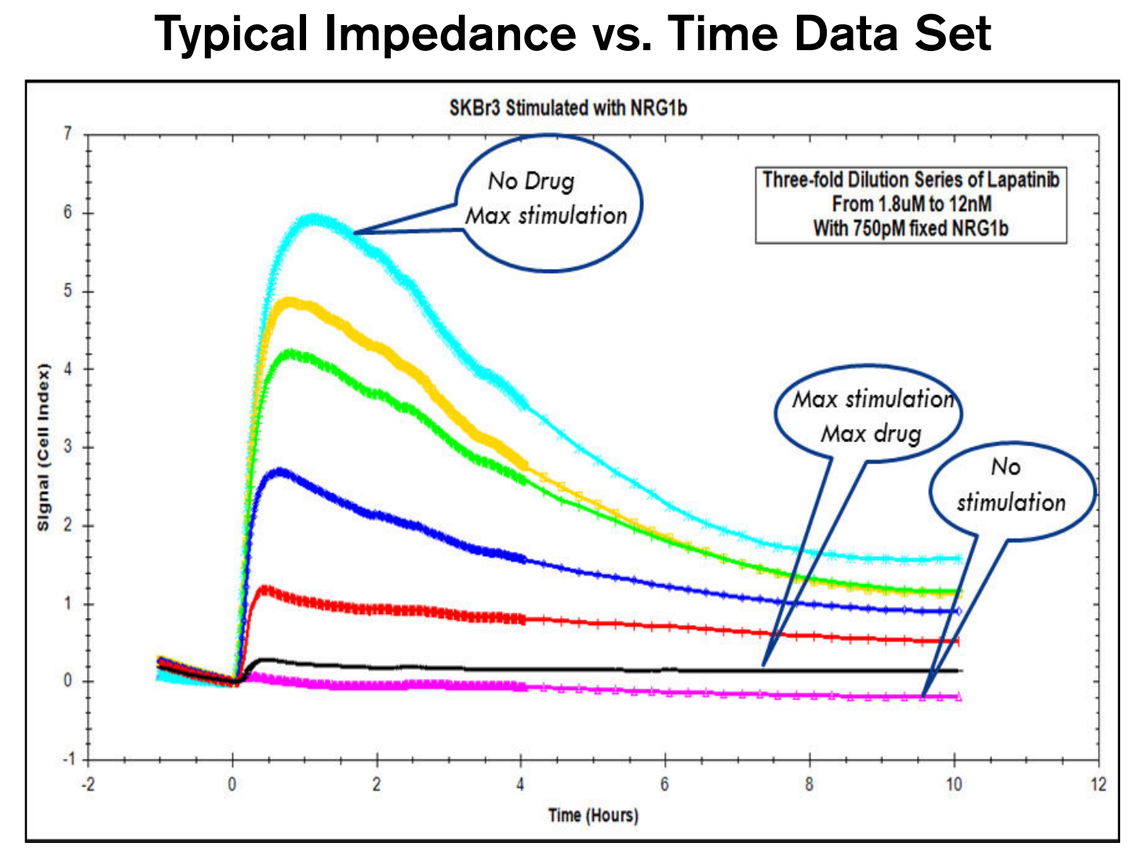
SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib From 1.8uM to 12nM With 750pM fixed NRG1b
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Figure 2. HER2- Abnormal Signaling by CELx HSF Test

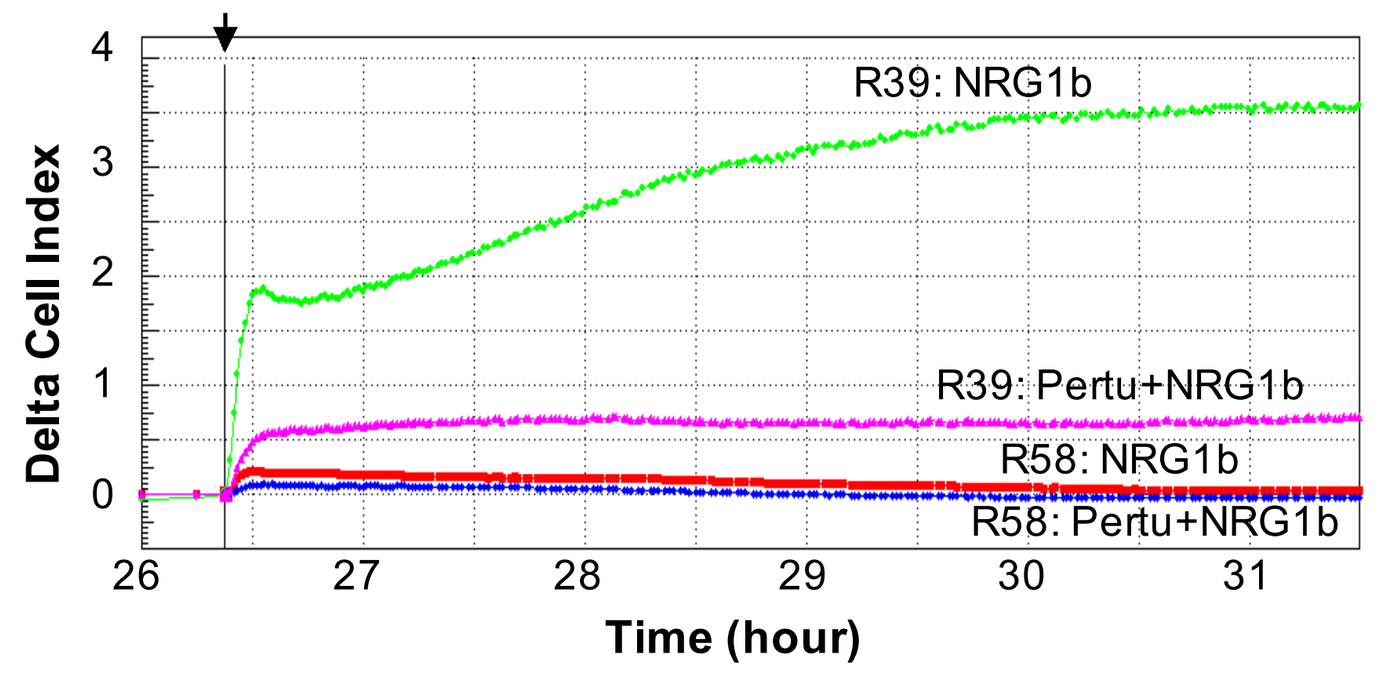
R39: NRG1b
R39: Pertu+NRG1b
R58: NRG1b
R58: Pertu+NRG1b
Delta Cell Index
Time (hour)

CELx time-course curves representing a high, abnormal HER2 signal in a high responder (R39) and a low HER2 signal in a non-responder (R58). In this display, curves of NRG1 stimulation in the absence versus presence of HER2 dimer blocker (10µg/mL) are presented.

The data show that the high NRG1b responder has more than 10 times greater signal than the low responder, indicating the test has a large dynamic range.

Results

Figure 3. Characterization of Primary Epithelial Cells Derived From Patient Tissue

A

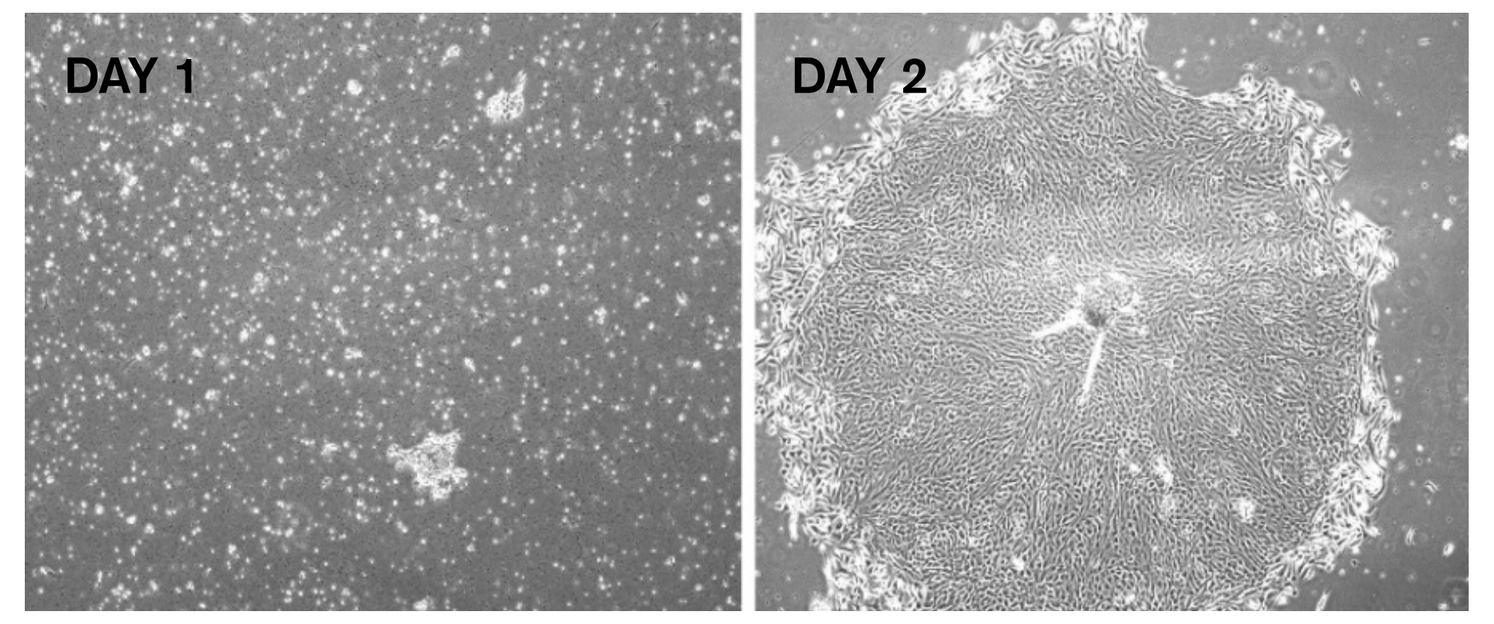
DAY 1
DAY 2

(A) A representative culture of primary cells from a digested tumor biopsy.

The results show that cells appear typically epithelial, marked by a tight cobblestone structure.

B

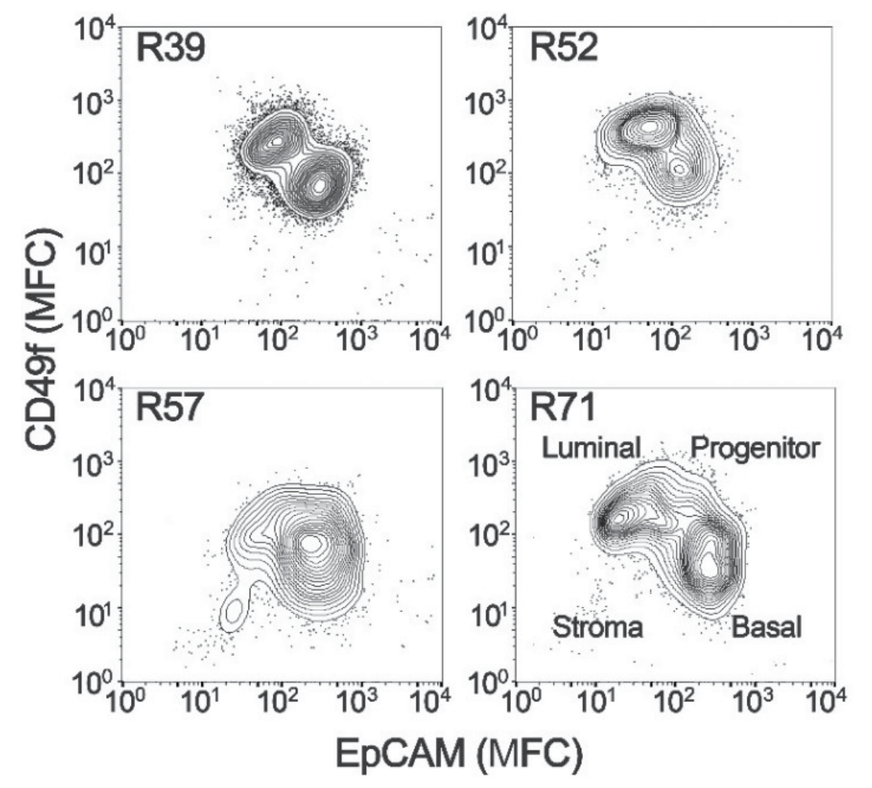
R39
R52
R57
R71
Luminal
Progenitor
Stroma
Basal
CD49f (MFC)
EpCAM (MFC)

C

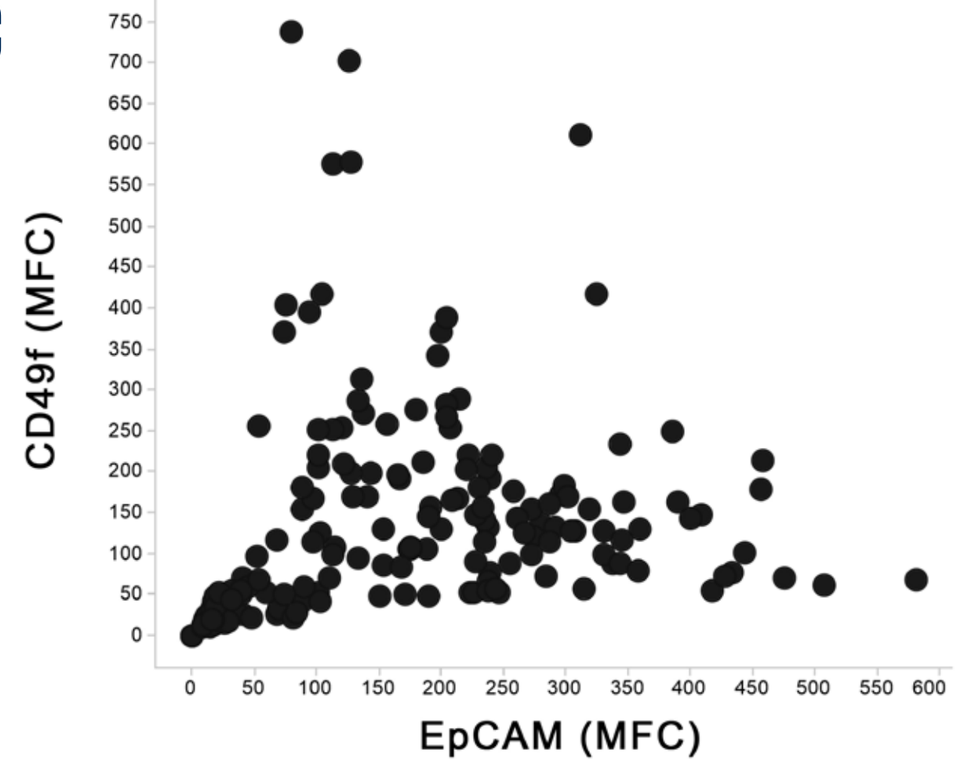
CD49f (MFC)
EpCAM (MFC)

(B) Flow cytometric analysis of luminal (EpCAM+, Claudin4+) and basal (CD49f+) markers on representative tumor primary cells harvested at the time of CELx HSF Test.

(C) Plot showing the Mean Fluorescence Channel (MFC) of the luminal marker EpCAM (x-axis) and the basal/progenitor biomarker CD49f (y-axis) for all 114 tumor samples tested.

The results shown in B and C indicate populations containing both luminal-like and basal-like characteristics.[18]

D

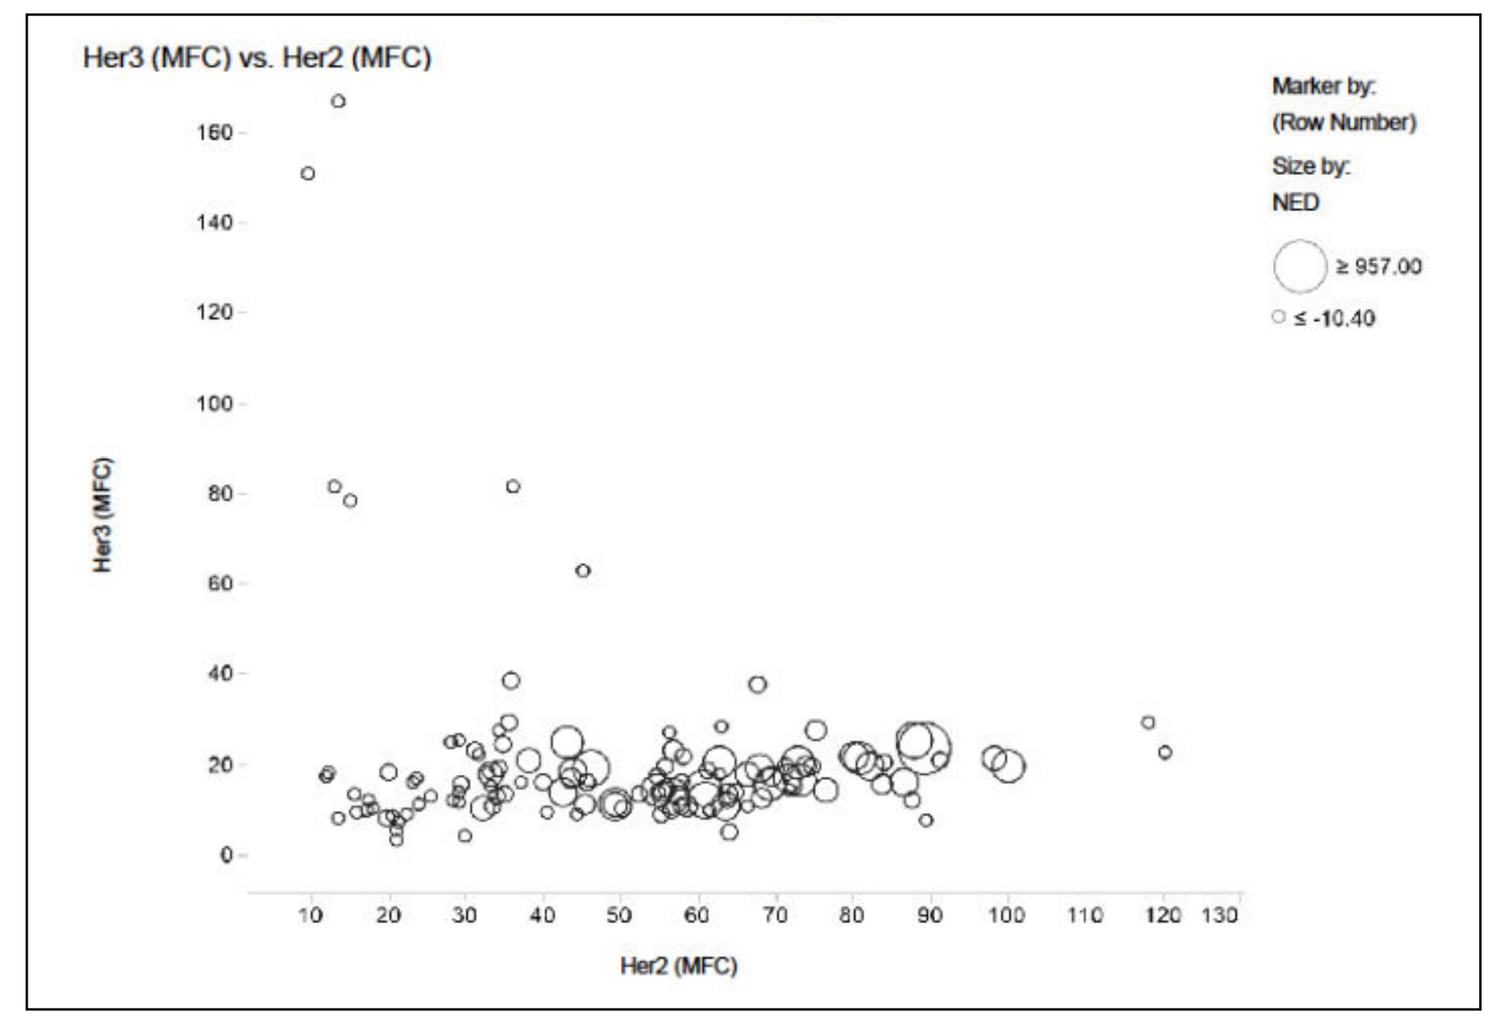
Her3 (MFC) vs. Her2 (MFC)
Marker by: (Row Number)
Size by: NED
≥ 957.00
≤ -10.40
Her3 (MFC)
Her2 (MFC)

(D) Comparison of expression levels of HER2, HER3 determined by flow cytometry with test measurand.

E

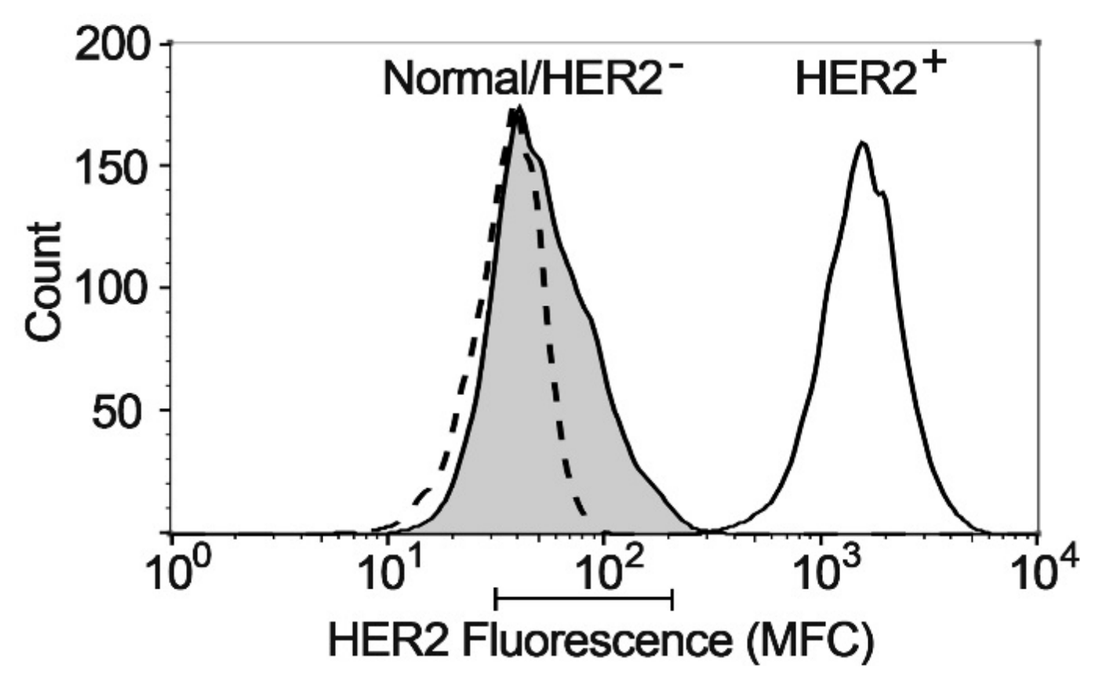
Normal/HER2⁻
HER2⁺
Count
HER2 Fluorescence (MFC)

(E) Histogram plot of HER2 expression of a representative tumor (shaded peak) compared to HER2+ cell line SKBR3 (DAKO 3+) (solid line) and HER2- cell line MDA231 (DAKO 0) (dashed line) that is coincidentally in the same range as the healthy samples. The bar below the graph represents the range of means from all the HER2- primary tumors (MFC range 31-210) compared to HER2+ cell line.

The results shown in D and E indicate that all 114 tumors had normal/low HER2 expression levels (>10x below HER2+). The patients also have normal HER3 expression levels. This confirms the clinical pathology test results reported to us for each specimen, namely that the 114 primary tumor samples were true HER2-negative.

Table 1. Summary of Patient Characteristics

Characteristic	Number of Patients	Percentage (%)
Total Patients	114	100
Age, years		
Mean	58.6	
Range	36–85	
Clinical Stage		
I	23	20
II	62	54
III	24	22
IV	4	4
Histology		
DCIS only	0	0
Invasive only	24	21
Invasive ductal/DCIS mixed	55	48
Lobular/other	35	31
Lymph Status		
Positive	56	49
Negative	46	40
pNX or N/A*	12	11
Estrogen Receptor Status		
ER+	96	84
ER-	18	16
HER2 IHC score/FACS		
IHC 0, 1+ or FISH not amp**	101	89
IHC 2+ and FISH not amp	13	11

* Information not available due to nature of some de-identified surplus tissue used in this study.
** Some labs only performed FISH, which does not discriminate 0, +1, etc.

CELx HSF tests were performed on 114 primary tumor cell samples from patients with breast cancer classified as HER2⁻ to measure HER2 pathway stimulation and signal specificity.

For comparative purposes, a healthy patient's and DAKO IHC test standard breast cancer cell lines SKBr3 and MDA-231 were also analyzed with CELx HSF Tests.

The CELx HSF Test identified 27 of 114 HER2- patient samples (23.7%; 95% CI=17–32%) having abnormally high HER2 signaling activity comparable to HER2+ cell line signaling activity.

Table 1. Patient characteristics of a random set of tumors collected from clinical cancer patients with a distribution of stage, histology and age to study a new test for identifying pathway dysfunction in HER2- breast cancer.

Statistical Analysis of Results

Fitting Cutoff Determination and Prevalence Data From 114-Patient Test Set

A first step was to test the current data set for compatibility with an earlier collection of 34 patients. This was done using the Kolmogorov-Smirnov two-sample test for identity of the distributions. The test statistic obtained was D=0.16642, with a P value of 0.4485, indicating that there is no significant difference between the CELx scores of these two groups.

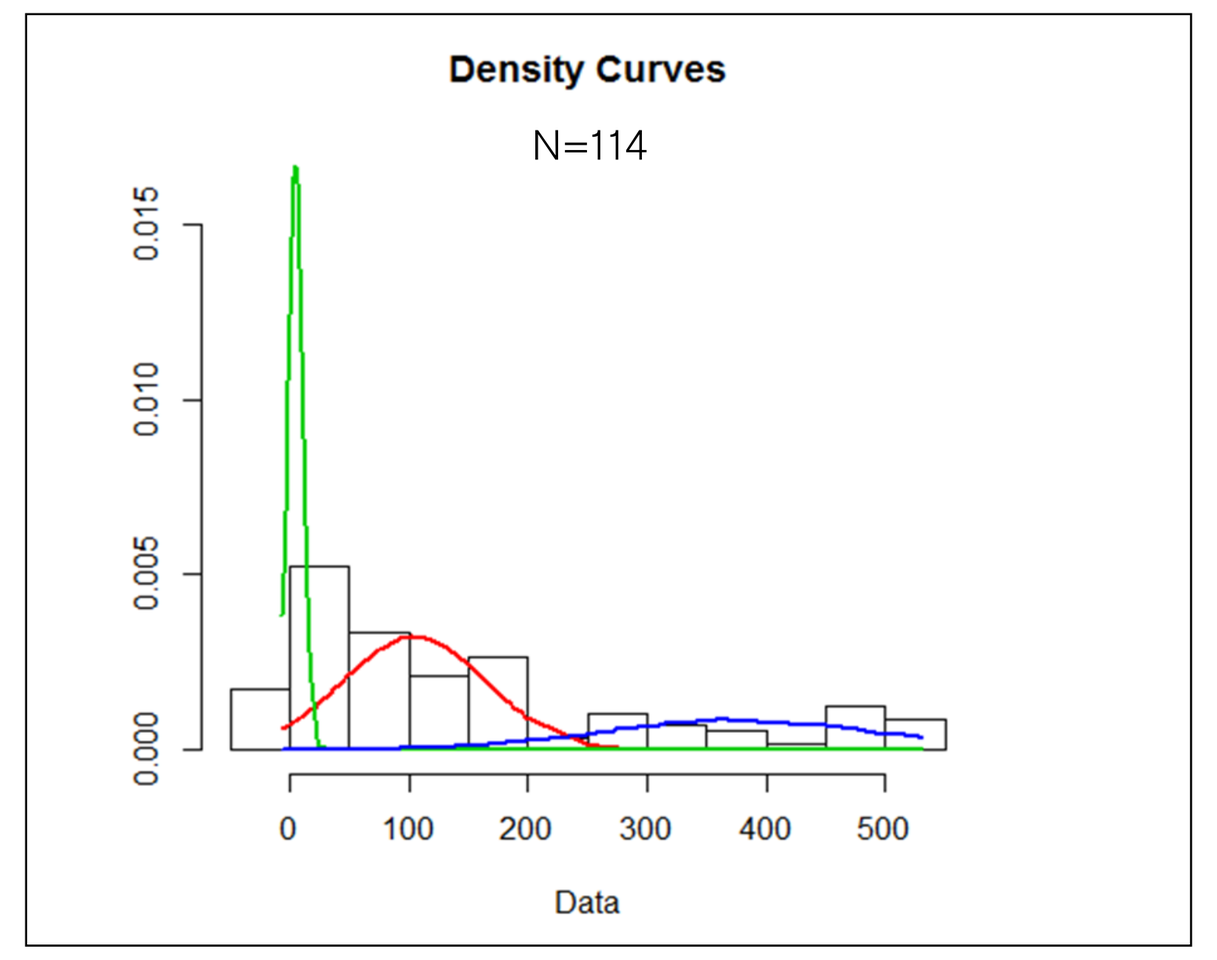
Density Curves
N=114
Data

	Components					
	1	2		3		
Mean	145.7	25.1	237.6	4.9	104.4	376.6
StDev	156.6	27.8	150.8	6.3	60.1	114.1
Proportion	1.00	0.43	0.57	0.26	0.49	0.25
Loglikelihood	-737.416	-703.955		-676.7		

The likelihood ratio test for the number of components gave the following parameters. For the 3-component model, the means of components 2 & 3 are more than 4 standard deviations apart.

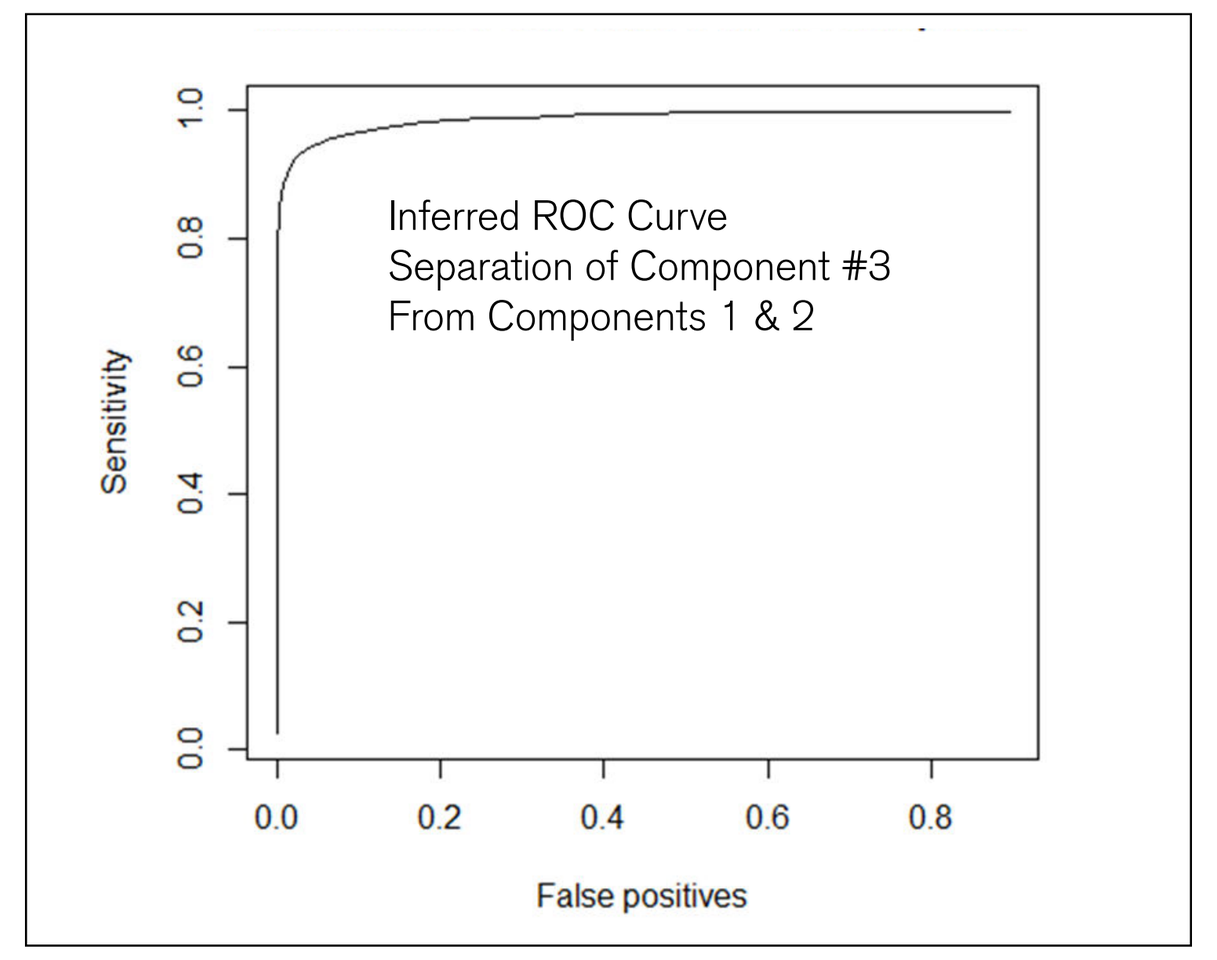
Inferred ROC Curve
Separation of Component #3
From Components 1 & 2
Sensitivity
False positives

Test	Cell Lines	Primary Cells
2 vs 1	66.92	1.8e-14
3 vs 2	54.59	8.7e-12

Formal significance testing shows that a 2-component mixture fits much better than a common normal distribution, and a 3-component mixture fits much better than 2.

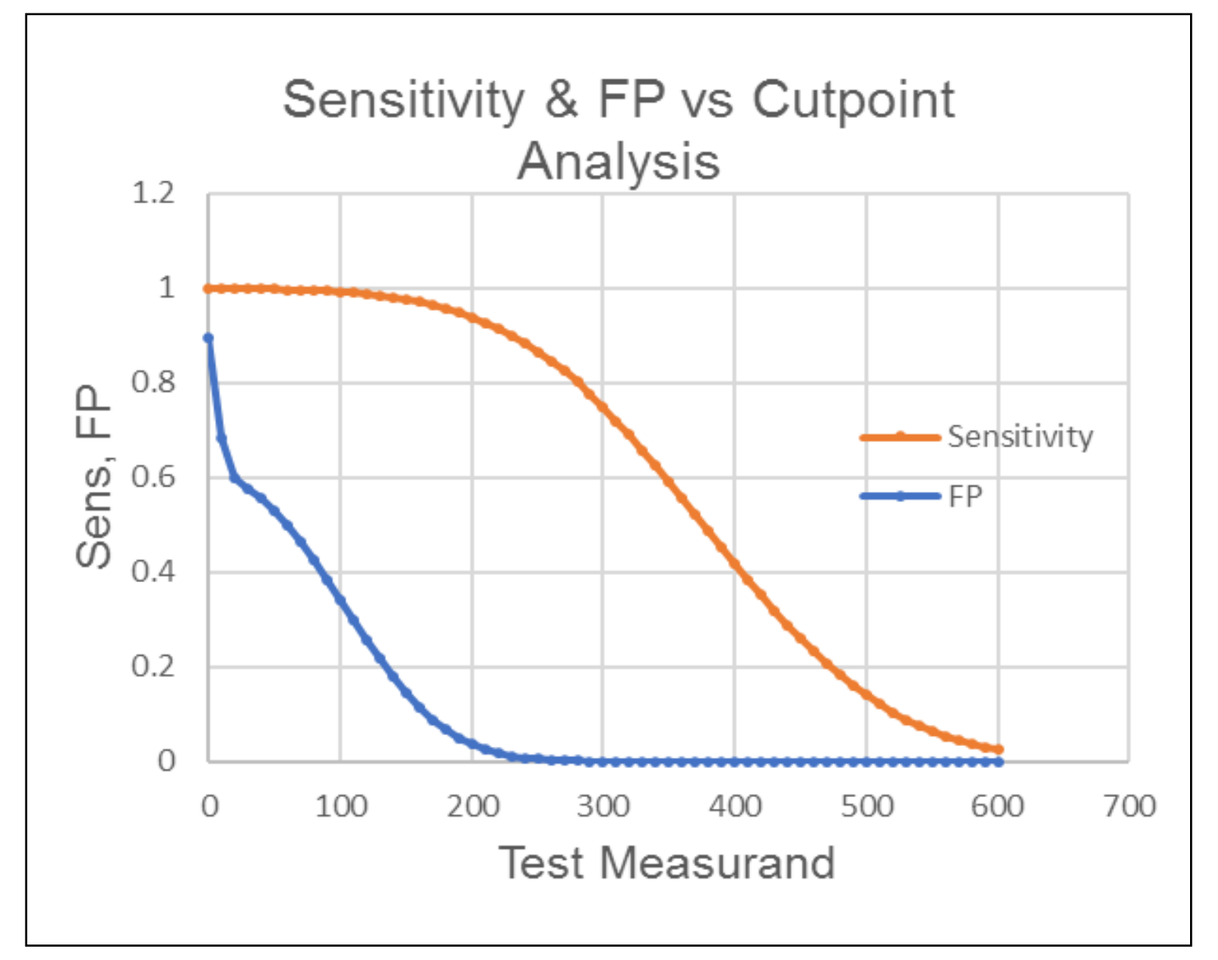
Sensitivity & FP vs Cutpoint Analysis
Sensitivity
FP
Sens, FP
Test Measurand

The graph above displays the numbers underlying the ROC plot: the sensitivity (orange line) and false positives (blue line) as a function of the cutoff used. At 250 signaling units, specificity is >99% (FP<1%) and sensitivity is 87%, indicative of an accurate test.

Collectively, these results demonstrate that the CELx HSF Test can specifically detect ligand-induced HER2-related signals and determine whether a HER2-driven test signal is sensitive or insensitive to a HER2-targeted drug (e.g., pertuzumab).

A cutoff of 250 signaling units (coincidental with the median value of the HER2⁺ cell line population) was determined to separate HER2-negative breast cancer patients into two distinct populations, one of which has abnormally active HER2 signaling activity in primary breast cancer cells.

Summary of Results

- A $HER2_S$ above 250 was considered abnormal or test positive, and was defined as the cut-point. Two clinical IHC HER2 test control cell lines, SKBR3 for HER2+ with IHC=3+ and MDA-MB231 for HER2- with IHC=0, have CELx HSF Test measurands of 544 and 0, respectively.
- Of the HER2– breast tumor cell samples tested, 27 of 114 patients (23.7%; 95% CI=17–32%) had net HER2 signaling activity, as determined by the CELx test, that was greater than the median $HER2_S$ of the HER2+ cell lines.
- At a cutoff value of 250 signaling units, specificity is >99% (FP<1%) and sensitivity is 87%, indicative of an accurate test.
- Previous results from 34 patients[15,16] are in agreement for this cut-point and 3-component population distribution.

Conclusions

- These findings provide strong evidence that measurement of HER2 signaling activity may provide clinically relevant information, particularly for HER2-breast cancer patients.
- These results suggest HER2-negative breast cancer patients with abnormal HER2 signaling may benefit from anti-HER2 therapy.
- Further analysis of this data will be performed to investigate definition of narrower subtypes that refine selection of drug treatment populations.
- Additional analytical studies are underway to further characterize independent HER1 and HER3 driven signaling activity.
- Additional clinical studies are underway to confirm these findings and to clinically validate the CELx HSF Test.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. *Cancer Res.* 2004;64:8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. *J Clin Oncol.* 2010;28:92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. *Oncologist.* 2009;14:320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. *Science.* 1987;235:177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. *Cancer Treat Rev.* 2014;40:276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. *N Engl J Med.* 2008;358:1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. *J Clin Oncol.* 2010;28:4307-4315.
8. Baselga J, Swain SM. Novel anticancer targets: revisiting ERBB2 and discovering ERBB3. *Nat Rev Cancer.* 2009;9:463-475.
9. Campbell MR, Moasser MM. HER targeting in HER2-negative breast cancers: looking for the HER3 positive. *Clin Cancer Res.* 2015;21:2886-2888.
10. Holbro T, et al. The ErbB2/ErbB3 heterodimer functions as an oncogenic unit: ErbB2 requires ErbB3 to drive breast tumor cell proliferation. *Proc Natl Acad Sci USA.* 2003;100:8933-8938.
11. Lee CY, et al. Neuregulin autocrine signaling promotes self-renewal of breast tumor-initiating cells by triggering HER2/HER3 activation. *Cancer Res.* 2014;74:341-352.
12. Mukohara T. PI3K mutations in breast cancer: prognostic and therapeutic implications. *Breast Cancer.* 2015;7:111-123.
13. Bose R, et al. Activating HER2 mutations in HER2 gene amplification negative breast cancer. *Cancer Discov.* 2013;3:224-237.
14. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016; in press.
15. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. *BMC Cancer.* 2016; in revision.
16. Stampfer MR, et al. Culture of human mammary epithelial cells. In: Freshney RI, Freshney MG, eds. *Culture of Epithelial Cells.* 2002.
17. Proia DA, Kuperwasser C. Reconstruction of human mammary tissues in a mouse model. *Nat Protoc.* 2006;1:206-214.
18. Sarrio D, et al. Epithelial and mesenchymal subpopulations within normal basal breast cell lines exhibit distinct stem cell/progenitor properties. *Stem Cells.* Feb 2012;30(2):292-303.

P6-07-14

This presentation is the intellectual property of Celcuity LLC. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.