CELx™ | breast

HER2 Signaling Function Test

Inhibition of abnormal HER2-driven signaling by two HER2 targeted antibody drugs tested *ex vivo* in live primary HER2- breast cancer cell samples and HER2+ cell lines

Lance G Laing, David J Burns, Yao Huang, Ian A MacNeil, Benjamin E Rich, Sajjad M. Soltani, Samantha Myhre, Brian F Sullivan – Celcuity LLC, 16305 36th Ave N, Suite 450, Minneapolis, MN 55446, USA

celcuity™

www.celcuity.com

Background

HER2 gene (*ERBB2*) amplification and/or HER2 protein overexpression is detected in approximately 15-20% of breast cancers and associated with more aggressive disease progression, metastasis, and a poorer prognosis [1-4]. The use of HER2-targeting agents, such as monoclonal antibodies, trastuzumab (Herceptin) and pertuzumab (Perjeta), significantly improves clinical outcomes in HER2+ patients [4, 5]. Currently, a patient's eligibility for HER2-targeted therapies is determined by their IHC- or FISH-based HER2 testing scores [4]. However, clinical trials have indicated a weak correlation between HER2 expression levels and HER2 targeted therapy benefit [6, 7]. Other biological factors, such as HER2 signaling activity, may be important to measure, in addition to expression and amplification of HER2, when identifying patients eligible for HER2 therapies.

A new functional cellular analysis platform, the CELx HER2 Signaling Function (CELx HSF) Test, which uses a label-free impedance biosensor to measure HER2 signaling activity in live tumor cells, has been developed by Celcuity [8, 9] (Figure 1). This test has been applied to tumor cell samples from 89 patients' fresh HER2- breast cancer tissue (DAKO 0 or 1+) and found abnormal HER2 signaling activity in 16 of 89 (18%; 95% CI=11%-27%) patients. This work that has been published recently [9], which suggests a new sub-group of HER2- breast cancer patients may benefit from anti-HER2 therapy.

In order to extend the understanding of any benefit this new sub-group may receive from HER2 therapy, the current study set out to quantify the percentage of HER2-driven signaling activity two HER2 mAb's, trastuzumab (Tz) and pertuzumab (Pz) , both alone and in combination, could inhibit two groups of live cells *ex vivo*: 1) HER2- breast tumor cells with abnormal HER2-driven signaling (n=5); and 2) HER2+ breast cancer cell lines (n=4).

Methods

Specimens and Cell Culture: Epithelial cells derived from five HER2- primary tumors (IHC 0 or 1+) and HER2+ breast cancer cell lines, each with comparable levels of abnormal HER2-driven signaling, were used in this study. Methods for tissue extraction and primary cell culture are essentially as described previously [10, 11]. Modifications can be found in our recent publication [9]. All cell lines were maintained according to ATCC recommendations and authenticated by ATCC in March 2016.

Flow Cytometry: Flow cytometry was performed on a BD FACSCalibur™ system using cells harvested at the time of the CELx HSP test using FACS markers listed in Table 2. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2, ER, and PR markers that were provided for each specimen by the clinic.

CELx HSF Test and Data Analysis: Real time live cell response to specific HER2 agonists (NRG1b or EGF) and with or without an antagonist (trastuzumab, pertuzumab, or the combination of the two antibodies) was measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). Clinically relevant concentrations of agonist and each HER2 mAb were used. From these responses, the net amount of HER2 participation in HER2 signaling initiated by the HER2 agonists ("$HER2_S$") was determined (Figure 1). To classify a HER2– sample as having abnormal signaling, a test cut-off value of 250 was established [8, 9]. The percentage of inhibition of NRG1-driven HER2 CELx signal by trastuzumab alone, pertuzumab alone, or their combination in each sample was calculated and compared between the HER2- primary cell group (n=5) and the HER2+ cell line group (n=4).

Table 1. Antibodies Used in Flow Cytometry

Description of Antibody	Vendor
Mouse anti-human HER2-phycoerythrin (PE), clone 24D2	Biolegend, San Diego, CA
Mouse anti-human HER1 conjugated with AlexaFluor 647, clone EGFR.1	BD Biosciences, San Jose, CA
Mouse anti-human HER3 conjugated with PerCP-sFluor710, clone SGP1	eBioscience, San Diego, CA
Mouse anti-human EPCAM conjugated with AlexaFluor 488, clone MH99	eBioscience, San Diego, CA
Mouse anti-human Claudin4 conjugated to PE, clone 382321	R&D Systems, Minneapolis, MN
Rat anti-human CD49f conjugated to PerCP/eFluor710, clone eBioGoH3	eBioscience, San Diego, CA
Mouse anti-human CD10 conjugated to Allophycocyanin (AP), cloneHL10a	BioLegend, San Diego, CA
Rabbit polyclonal anti-human estrogen receptor alpha (ERα) conjugated to AlexaFluor488	Bioss, Woburn, MA
Mouse anti-human progesterone receptor (PRG) conjugated to eFluor660, clone KMC912	eBioscience, San Diego, CA

All epitopes were extracellular with the exceptions of ER and PR. All antibodies were purchased from companies as listed who provided empirical demonstration of each of the antibodies for our applications.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. Cancer Res. 2004; 64(23):8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. J Clin Oncol. 2010; 28(1):92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. Oncologist. 2009; 14(4):320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. Science. 1987; 235(4785):177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. Cancer Treat Rev. 2014; 40(2):276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. N Engl J Med. 2008; 358(13):1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. J Clin Oncol. 2010; 28(28):4307-4315.
8. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. BMC Cancer. 2016; in revision.
9. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. Oncotarget. 2016; in press.
10. Stampfer MR, et al. (2002). Culture of human mammary epithelial cells. In: Freshney RI and Freshney MG, eds. Culture of Epithelial Cells.
11. Proia DA and Kuperwasser C. Reconstruction of human mammary tissues in a mouse model. Nat Protoc. 2006; 1(1):206-214.

34th Annual Miami Breast Cancer Conference®

March 9–12, 2017

Results

Figure 1. Impedance Platform Sensitivity Enables Quantification of HER2 Signaling

HER2 ligands and inhibitors used to turn on/off HER pathways

Very subtle, sub-nanometer, cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Create Pathway Dysfunction and Drug Response Scores

Typical Impedance vs. Time Data Set

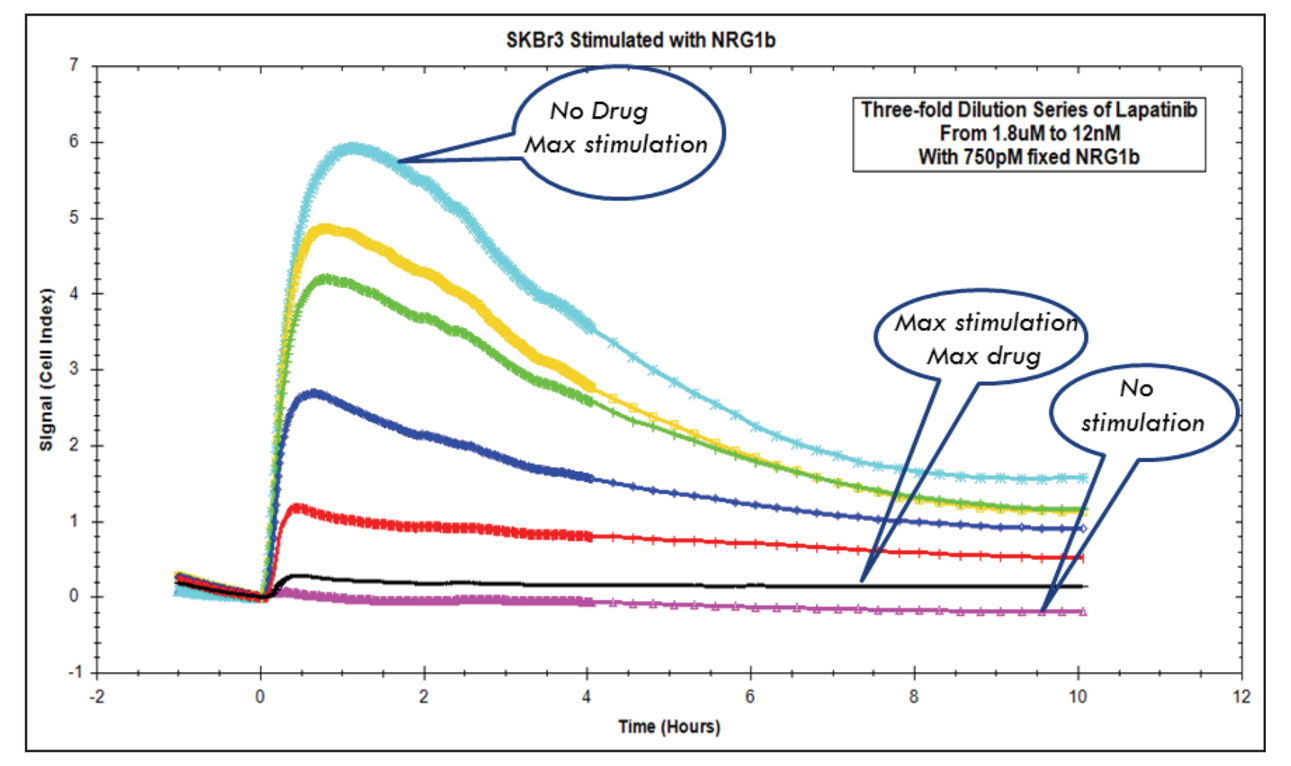
SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib
From 1.8uM to 12nM
With 750pM fixed NRG1b
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Figure 2. Characterization of Primary Epithelial Cells Derived from Patient Tissue

A

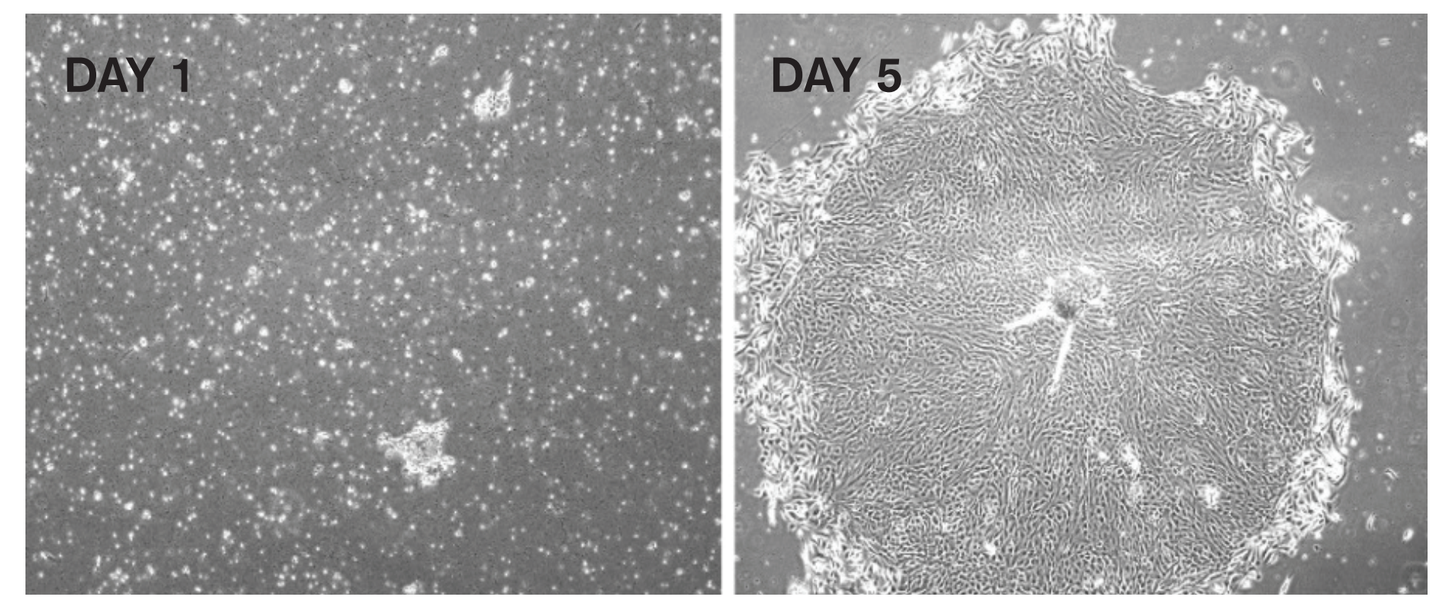
DAY 1
DAY 5

B

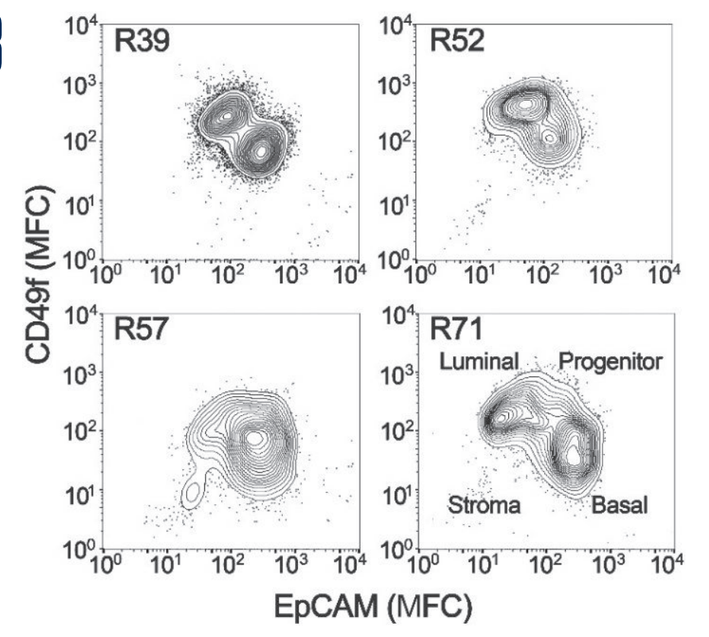
R39
R52
R57
R71
Luminal
Progenitor
Stroma
Basal
CD49f (MFC)
EpCAM (MFC)

C

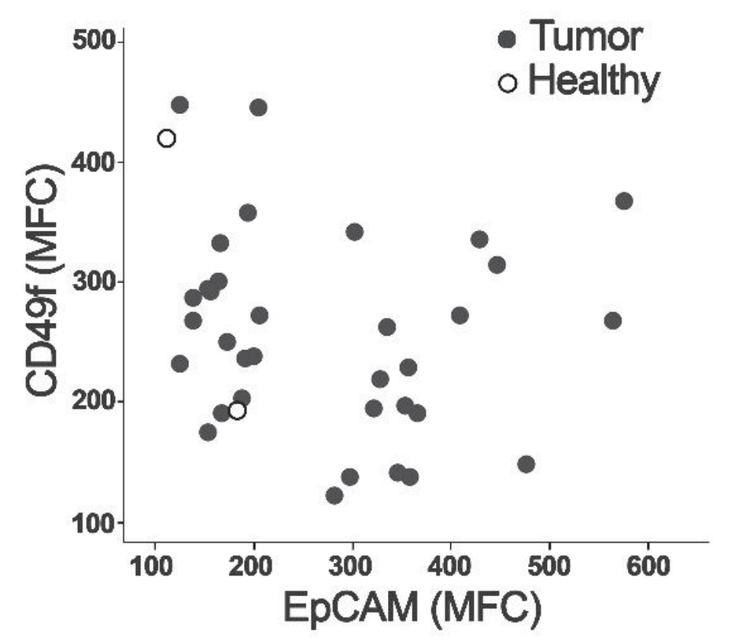
Tumor
Healthy
CD49f (MFC)
EpCAM (MFC)

D

	Fluorescence (MFC)	
	Cell Lines	Primary Cells
HER2+	1000-4000	
HER2-	30-500	30-230
ERα	25-100	50-240
PR	40-1300*	60-240

** The majority of the cell lines expressed PR between 40 and 280*

E

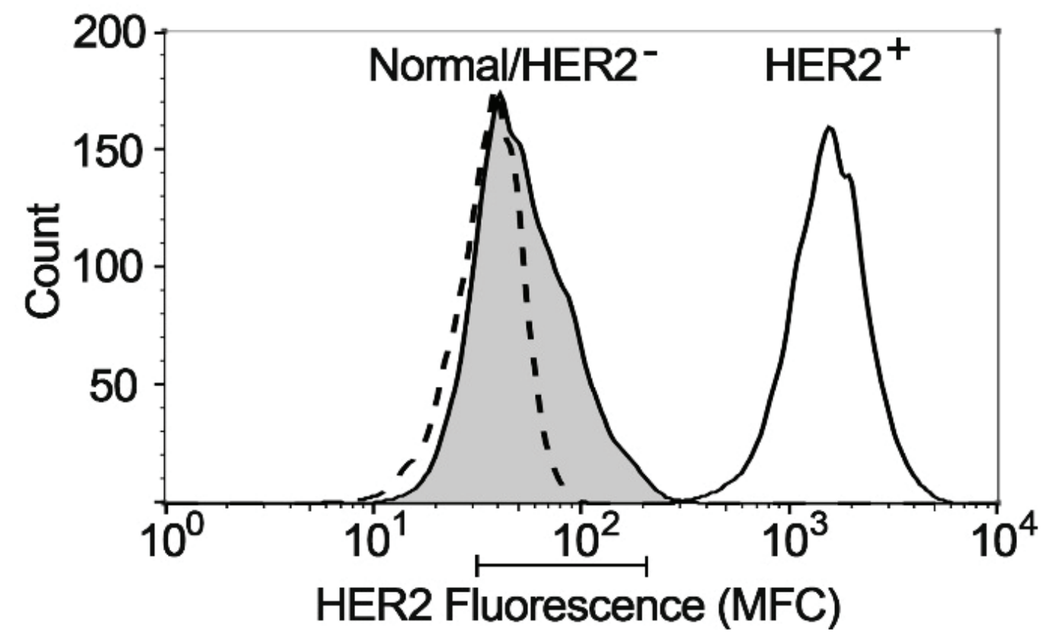
Normal/HER2⁻
HER2⁺
Count
HER2 Fluorescence (MFC)

(A) A representative culture of primary cells from a digested tumor biopsy.

The results show that colonies appear typically epithelial, marked by a tight cobblestone structure.

(B) Flow cytometric analysis of luminal ($EpCAM^+$, $Claudin4^+$) and basal ($CD49f^+$, $CD10^+$) markers on representative tumor primary cells harvested at the time of CELx HSF test.

(C) Plot showing the Mean Fluorescence Channel (MFC) of the luminal marker EpCAM (x-axis) and the basal/progenitor biomarker CD49f (y-axis) for all 34 tumor samples tested.

The results shown in B and C indicate populations containing both luminal-like and basal-like characteristics.

(D) Comparison of expression levels of HER2, ERα, and PR, determined by flow cytometry, between primary cells and cell lines ($HER2^+$ and $HER2^-$).

The results show that primary cells grown from tumor samples of the ER+/PR+ patients continue to express ER and PR.

(E) Histogram plot of HER2 expression of a representative tumor (shaded peak) compared to HER2+ cell line SKBR3 (DAKO 3+) (solid line) and HER2- cell line MDA231 (DAKO 0) (dashed line) that is coincidentally in the same range as the healthy samples. The bar below the graph represents the range of means from all the primary tumors and healthy tissue tested (MFC range 31-210).

The results shown in D and E indicate that all 34 tumor and 16 healthy samples had normal/low HER2 expression levels in the range of $HER2^-$ cell lines. This confirms the clinical pathology test results reported to us for each specimen, namely that the 34 primary tumor samples were true $HER2^-$.

Table 2. Examples of HER2-Negative ($HER2^-$) Patient Samples with Abnormally High HER2-Dependent Signaling ($HER2_s$+) Identified by CELx HSF Test

$HER2^-$ Patient ID	NRG1-driven HER2-dependent CELx Signal Units	EGF-driven HER2-dependent CELx Signal Units	Total HER2-dependent CELx Signal Units	CELx Test Result
R39 (tumor)	475	88	563	Abnormal
R20 (tumor)	409	120	529	Abnormal
R160 (tumor)	332	99	430	Abnormal
R82 (tumor)	272	40	312	Abnormal
R95 (tumor)	227	44	271	Abnormal
R25 (tumor)	238	29	267	Abnormal
R71 (tumor)	228	23	252	Abnormal
R22 (tumor)	1	78	79	Normal
R62 (healthy)	31	7	38	Normal
SKBr3 ($HER2^+$ cell line)	401	143	544	Abnormal

In one of our studies, CELx HSF tests were performed on 34 tumor cell samples from patients with breast cancer classified as $HER2^-$ to measure HER2 pathway stimulation and signal specificity [9]. For comparative purposes, 16 primary breast epithelial cell samples from healthy patients, and DAKO IHC test standard breast cancer cell lines SKBr3 (IHC 3+) and MDA-231 (IHC 0) were also subjected to CELx HSF Tests. NRG1b-induced and EGF-induced CELx signals for tumor and healthy primary cells and a $HER2^+$ reference cell line (SKBr3) (DAKO test score 3+) are summarized in the table.

In this study, the CELx HSF test identified 7 of 34 (~20%) HER2- patient samples having abnormally high HER2 signaling activities.

Figure 3. CELx Test Identifies Four Different Subgroups for HER2 Signaling

Examples of CELx HSF test results ($HER2\pm/HER2_S\pm$): use of NRG1 (ligand) and pertuzumab (antagonist) to ID net HER2-driven signaling activity. Within HSF+ subgroups differential response to drug is demonstrated.

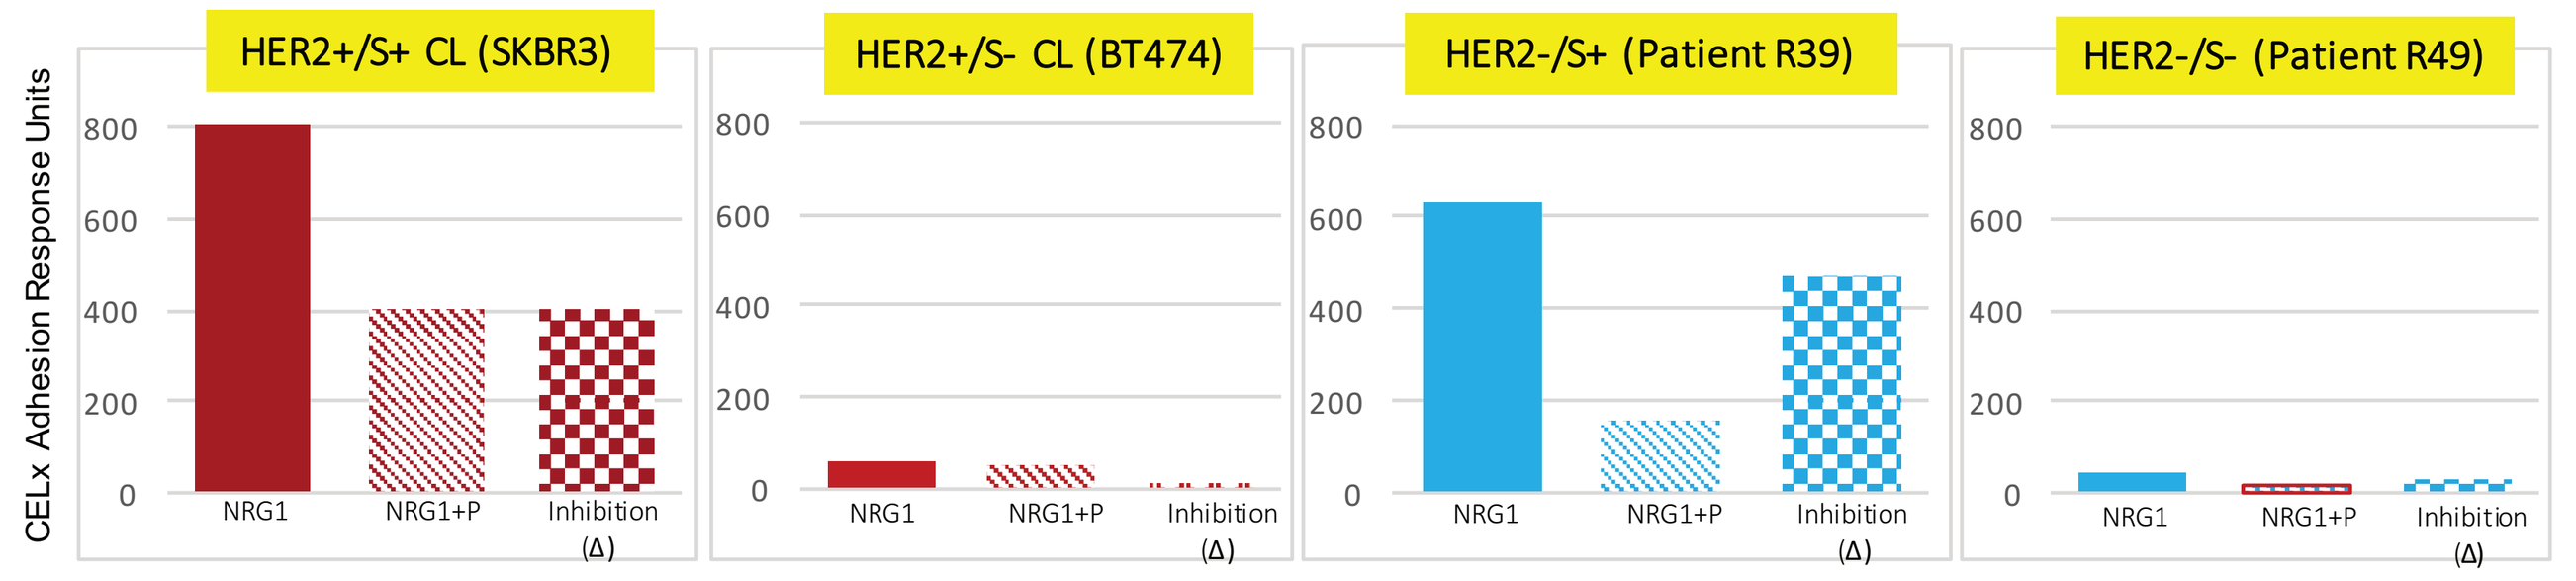
HER2+/S+ CL (SKBR3)
HER2+/S- CL (BT474)
HER2-/S+ (Patient R39)
HER2-/S- (Patient R49)
CELx Adhesion Response Units
NRG1
NRG1+P
Inhibition (Δ)

- NRG1 initiated signaling varied widely in the HER2+ samples
- SKBR3's signaling was one of highest HER2+ cell lines tested
- Difference between NRG1 and NRG1 + P above is amount of signaling involving HER2 in HER2/HER3 dimerization
- Pertuzumab inhibits 50% of the SKBR3 signal
- None of BT474's limited activity is inhibited

- HER2- primary tumor samples, R39 and R49, show a similar range of NRG1 initiated activity as the HER2+ cell lines
- The HER2-driven activity (Δ) for R39 is nearly identical to SKBR3's
- High Pz inhibition is similar to other $HER2/HER2_S$+ tumors
- R49's low signaling is typical of 80% of HER2- patient tumors

Table 3. Comparison of Inhibition of NRG1-driven HER2 CELx Signals by Trastuzumab (Tz) or Alone or in Combination

Average % Inhibition of NRGI-driven HER2 Signaling Activity		
HER2 mAb	$HER2+/HER2_S+$ Cell Lines (n=3)	$HER2-/HER2_S+$ Primaries (n=5)
Trastuzumab	19%	44%
Pertuzumab	62%	73%
Trastuzumab + Pertuzumab	87%	81%

Pertuzumab and trastuzumab alone were each more effective in the HER2- group than in HER2+ group.

Figure 4. Examples of Comparison of NRG1-driven HER2 Signaling Inhibition by Trastuzumab (Tz) or Pertuzumab (Pz) Alone or in Combination

HER2+ Cell Line	NRG1-driven HER2-dependent CELx Signals	EGF-driven HER2-dependent CELx Signals	Total HER2-dependent CELx Signals	CELx Test Results
HCC1569	295	74	369	Abnormal

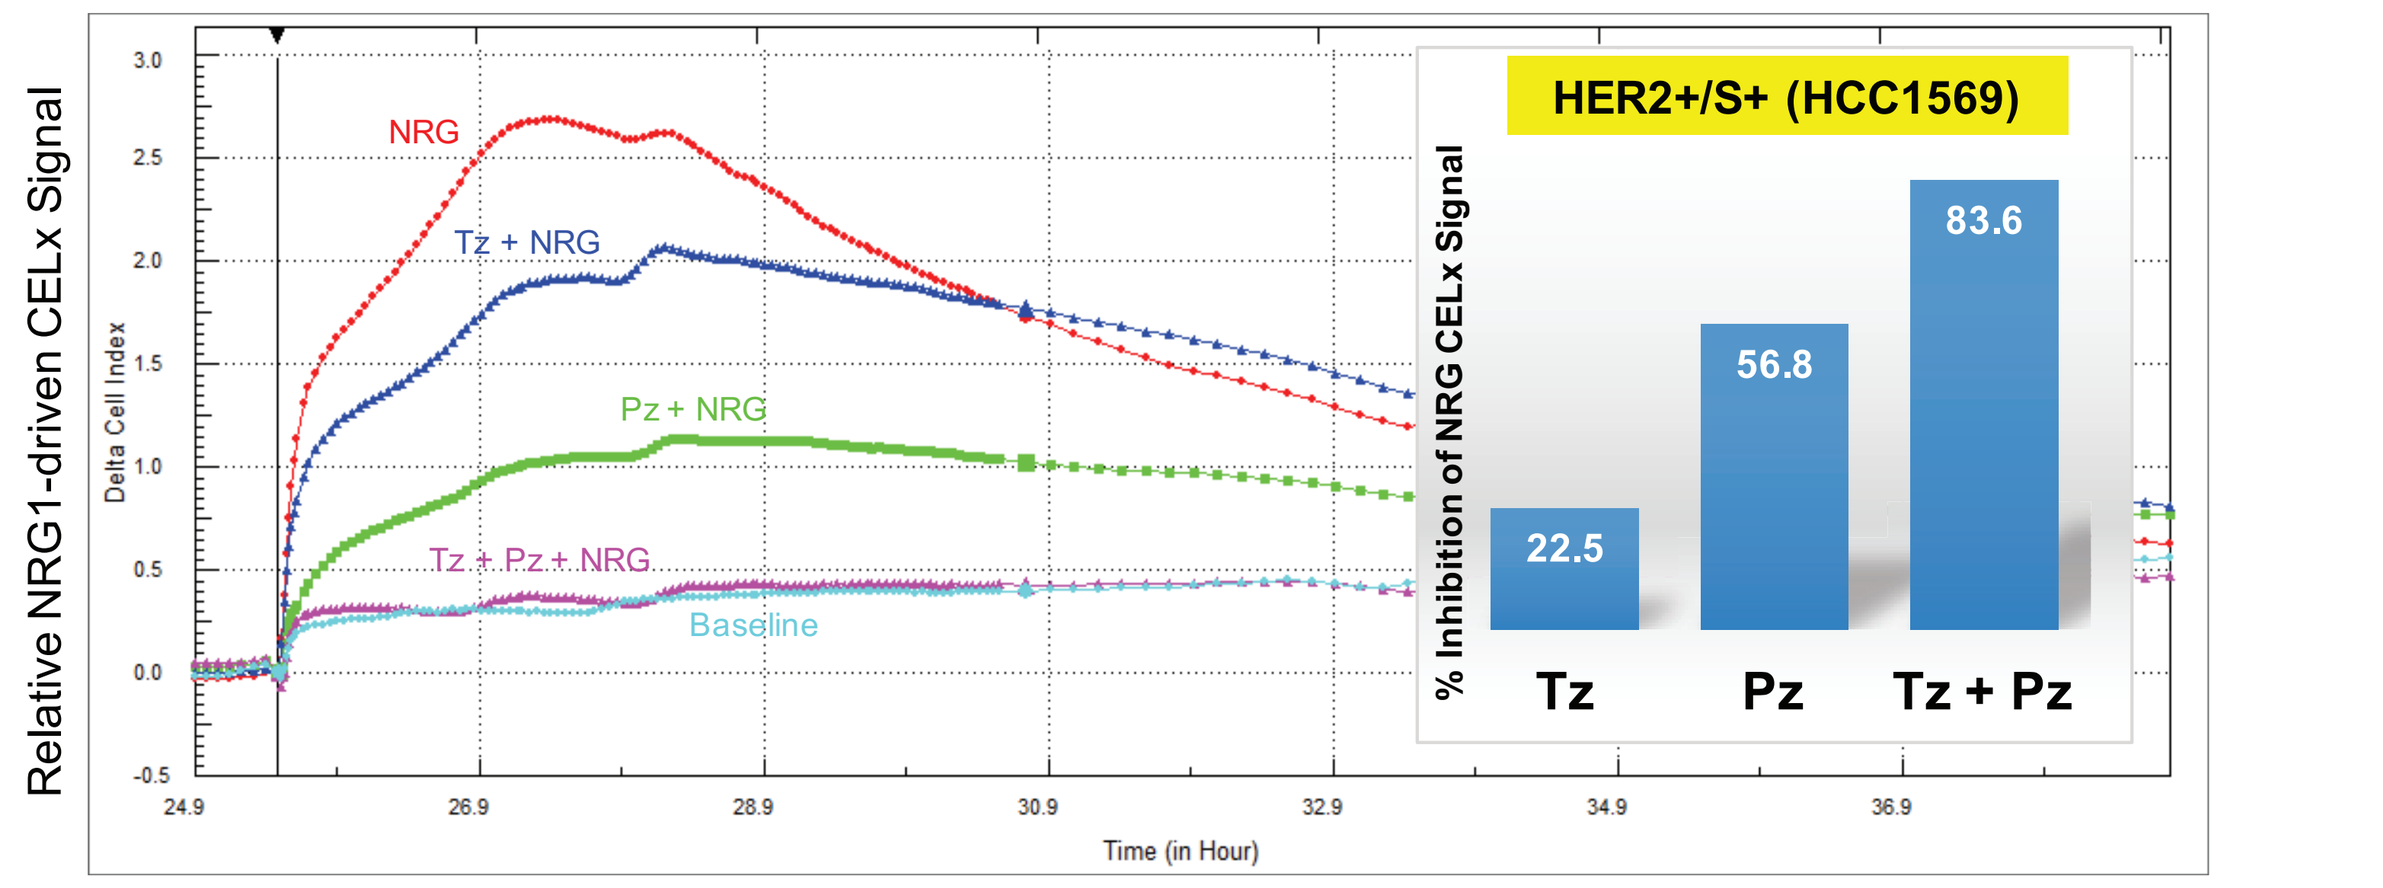
Relative NRG1-driven CELx Signal
Delta Cell Index
NRG
Tz + NRG
Pz + NRG
Tz + Pz + NRG
Baseline
Time (in Hour)
HER2+/S+ (HCC1569)
% Inhibition of NRG CELx Signal
22.5
56.8
83.6
Tz
Pz
Tz + Pz

$HER2^-$ Patient ID	NRG1-driven HER2-dependent CELx Signals	EGF-driven HER2-dependent CELx Signals	Total HER2-dependent CELx Signals	CELx Test Results
R978	748	1	749	Abnormal

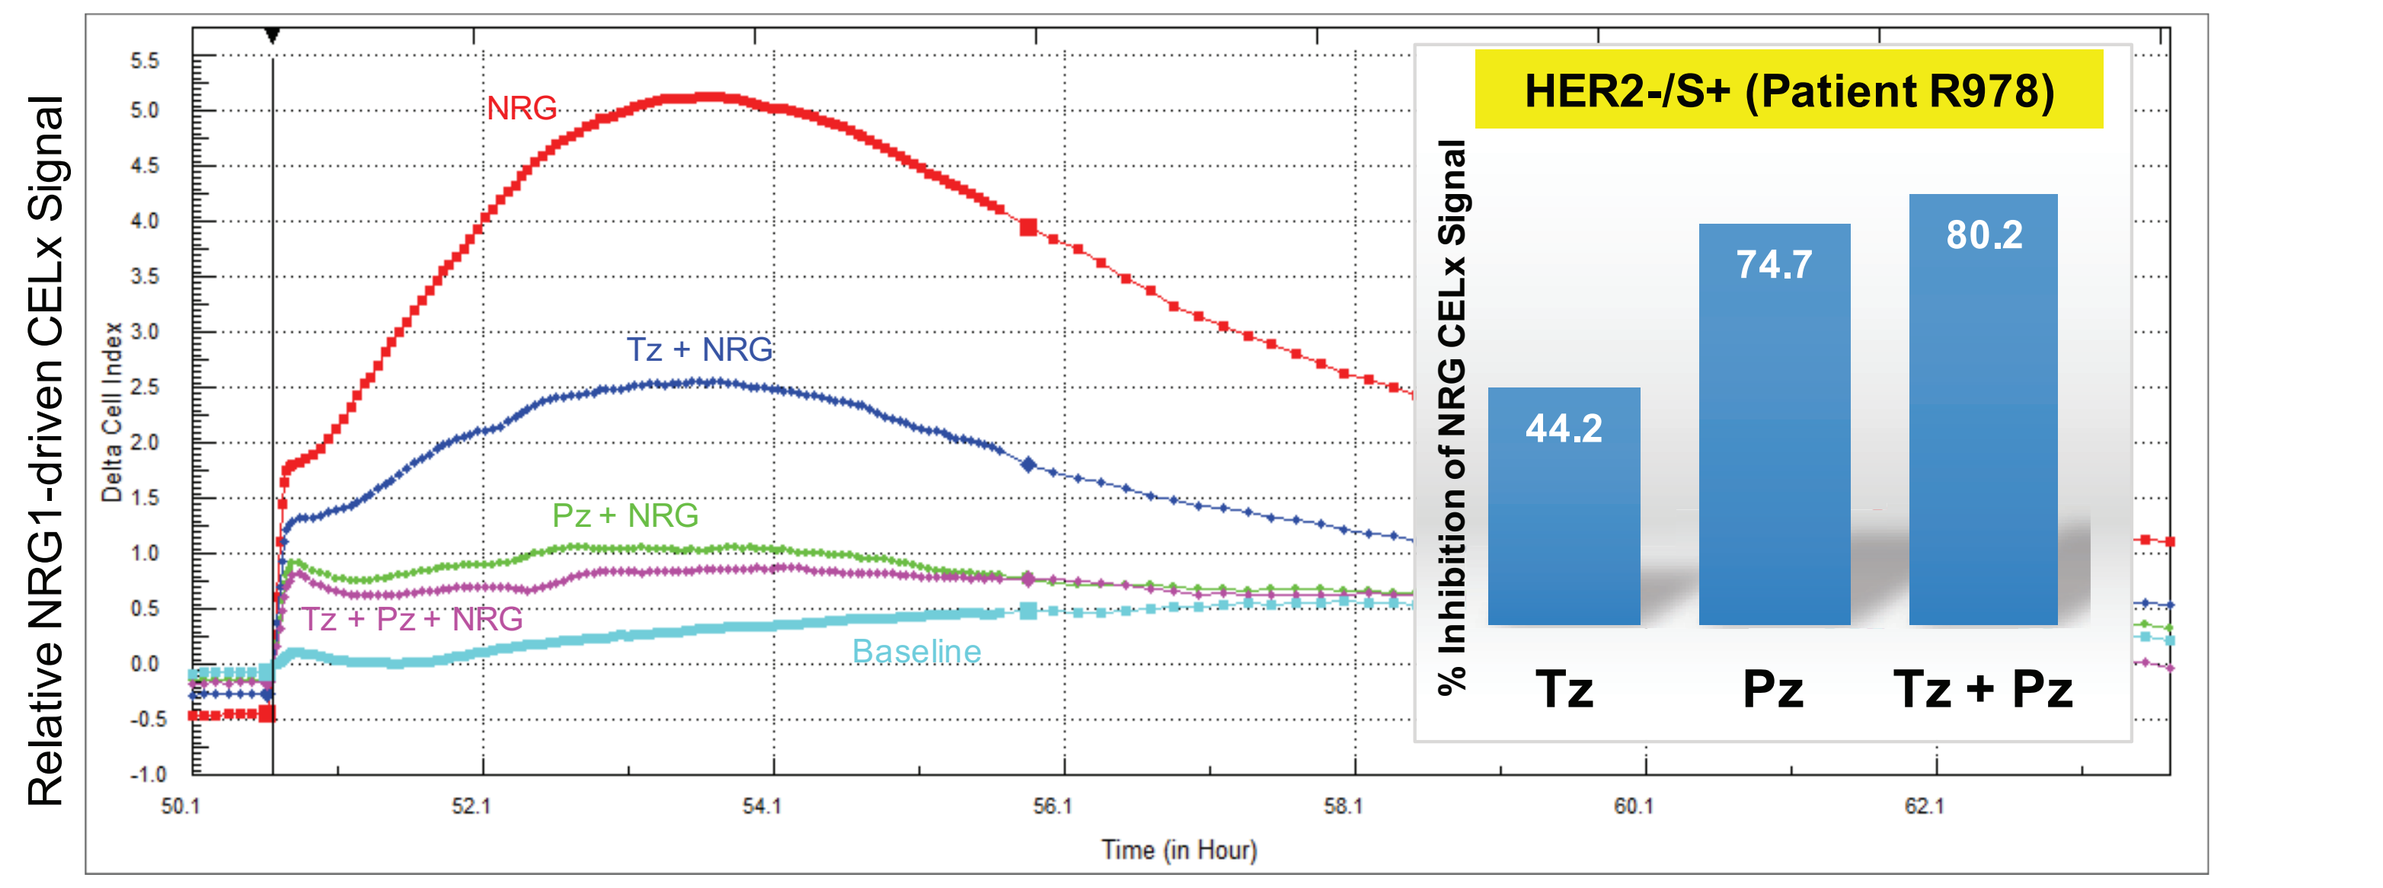
Relative NRG1-driven CELx Signal
Delta Cell Index
NRG
Tz + NRG
Pz + NRG
Tz + Pz + NRG
Baseline
Time (in Hour)
HER2-/S+ (Patient R978)
% Inhibition of NRG CELx Signal
44.2
74.7
80.2
Tz
Pz
Tz + Pz

Summary of Results

- All cell samples tested with pertuzumab and trastuzumab had comparable, and abnormal, levels of NRG1 activated HER2-driven signaling.
- In each sample, the two mAbs (trastuzumab and pertuzumab) inhibited a higher percentage of signaling in combination than either mAb alone; no interference effects between the two mAbs were detected.
- Pertuzumab and trastuzumab alone were each more effective in the HER2- cell samples than in the HER2+ ones.

Conclusions

- These findings provide strong evidence that the two HER2 mAbs (trastuzumab and pertuzumab) used to treat HER2+ breast cancer patients are as effective in blocking abnormal HER2-driven signaling function *ex vivo* in HER2- primary cells with abnormal signaling as they are in HER2+ cells with abnormal signaling.
- Overexpressed HER2 receptor may thus not be a required condition for breast cancer patient response to HER2 mAbs.
- These results suggest a new sub-group of breast cancer patients, HER2- with abnormal HER2 signaling ($HER2-/HER2_S+$), identified with the CELx HER2 Signaling Function (HSF) Test, may benefit from the addition of HER2 mAbs to current therapeutic regimens.
- Additional studies to confirm these findings are currently underway.

Abstract ID: 459

This presentation is the intellectual property of Celcuity LLC. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.