CELx™ | breast

HER2 Signaling Function Test

Trial in Progress: A phase 2 study of neoadjuvant chemotherapy plus trastuzumab and pertuzumab in HER2-negative breast cancer patients with abnormal HER2-driven signaling transduction (NCT03412643)

Eleftherios P. Mamounas, MD, MPH; Lance G Laing, PhD – NSABP Foundation and Orlando Health UF Cancer Center, Orlando, FL; Celcuity Inc., Minneapolis, MN

celcuity™

www.celcuity.com

Background

Biological factors, such as HER2 signaling activity, may be important to measure in addition to expression and amplification of HER2 when identifying patients eligible for HER2 therapies. HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15–20% of breast cancers and is associated with more aggressive disease progression, metastasis and a poorer prognosis [1-4]. Agents targeting HER2, such as trastuzumab, lapatinib, and pertuzumab, significantly improve clinical outcomes in HER2+ patients [4, 5]. Currently, a patient's eligibility for HER2-targeted therapies is determined using IHC or FISH HER2 tests [4]. However, clinical trials have indicated a weak correlation between HER2 expression or amplification levels and HER2 targeted therapy benefit [6, 7].

To overcome the limitation of measuring only receptor levels to diagnose HER2 breast cancer, a new assay using an impedance biosensor, the CELx HER2 Signaling Function (CELx HSF) Test, was developed [8, 9]. This test measures functional HER2 signaling in live patient tumor cells to identify patients who have abnormal HER2-driven signaling activity despite having normal HER2 receptor levels. Celcuity found that 20% of HER2-negative breast cancer patients have the same level of abnormal HER2 signaling activity as the abnormal HER2 signaling activity found in HER2-positive breast cancer cell lines. This is currently an undiagnosable sub-type of HER2-related breast cancer that may be responsive to treatment with anti-HER2 therapies.

To assess the potential clinical benefit HER2 therapies may offer these patients, a prospective, single-arm, open label interventional neoadjuvant Phase 2 study was initiated.

Trial Design

This is a prospective, single arm, open label, multicenter interventional study designed to evaluate the efficacy of neoadjuvant doxorubicin plus cyclophosphamide followed by weekly paclitaxel plus trastuzumab and pertuzumab in early stage HER2-negative breast cancer patients who have abnormal HER2 signaling activity determined by a live cell HER2 signaling function test.

- The primary objective of the study is to evaluate whether patients with HER2-negative breast cancers who have abnormal HER2-driven signaling pathways and receive HER2-targeted therapy with neoadjuvant chemotherapy will have a higher rate of pCR in the breast and lymph nodes than has been found historically in patients with HER2-negative breast cancer who have received neoadjuvant chemotherapy.
- Patients will be required to have a prescreening research core needle biopsy to procure a fresh tumor specimen that will be analyzed to assess whether their HER2 signaling activity is abnormal or normal. Patients must have abnormal HER2 signaling activity to be enrolled.
- It is expected that approximately 270 patients will need to be prescreened in order to enroll 54 patients (26 ER+/HER2- and 28 ER-/HER2-) who have abnormal HER2 signaling activity.
- The sample size calculations for the ER+ and ER- subgroups assumed the historical pCR rate is 11% and 34%, respectively.
- In each of these two subgroups, a Sargent two-stage three-outcome optimal design has been used where the type I error is set at 0.05, the type II error is set at 0.1 and the probabilities of a true outcome (positive or negative) are both set at 0.8.

Figure 1. NSABP FB-12 Schema

HER2-Negative Invasive Breast Cancer Diagnosed by Core Needle Biopsy

Pretreatment research core biopsy (2 cores) of primary tumor to procure tumor tissue for submission to Celcuity for assessment of HER2 signaling function

AC* x 1 cycle may be administered while waiting on results

Tumor determined to have abnormal HER2 signaling activity by CELx HSF testing

STUDY ENTRY
AC* x 3 cycles if 1st cycle of AC was administered prior to receipt of CELx HSF results
OR
AC* x 4 cycles if CELx HSF results are known prior to initiating chemotherapy

Weekly Paclitaxel** + Trastuzumab† + Pertuzumab†

SURGERY
Lumpectomy or mastectomy and axillary staging

* *Doxorubicin (A) 60 mg/m² IV + cyclophosphamide (C) 600 mg/m2 IV Day 1 every 2 weeks or every 3 weeks at investigator's discretion.*

** *Weekly Paclitaxel (WP): 80 mg/m² IV weekly for 12 doses.*

† *Trastuzumab + Pertuzumab: Trastuzumab (administer a loading dose of 8 mg/kg IV; then 6 mg/kg IV every 3 weeks for 4 cycles) with pertuzumab (administer a loading dose of 840 mg IV; then 420 mg IV every 3 weeks for 4 cycles).*

35th Annual Miami Breast Cancer Conference — March 8–11, 2018

Identifying HER2- Patients With Abnormal HER2 Signaling

CELx HER2 Signaling Function Test

The CELx HSF Test measures real-time live cell response to specific HER2 agonists (NRG1b and EGF) with or without an antagonist (2C4, a HER2 dimer blocker using an xCELLigence RTCA impedance biosensor (ACEA Biosciences)). From these responses, the net amount of HER2 participation in HER-family signaling initiated by HER receptor agonists (HER2S) was determined [8]. Samples with HER2 signaling activity levels above a previously determined cutoff value of 250 signaling units that was attenuatable with a HER2 dimer blocker were identified as abnormal.

Figure 2. Impedance Platform Sensitivity Enables Quantification of HER2 Signaling

HER2 ligands and inhibitors used to turn on/off HER pathways

Very subtle, sub-nanometer cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Create Pathway Dysfunction and Drug Response Scores

Typical Impedance vs. Time Data Set

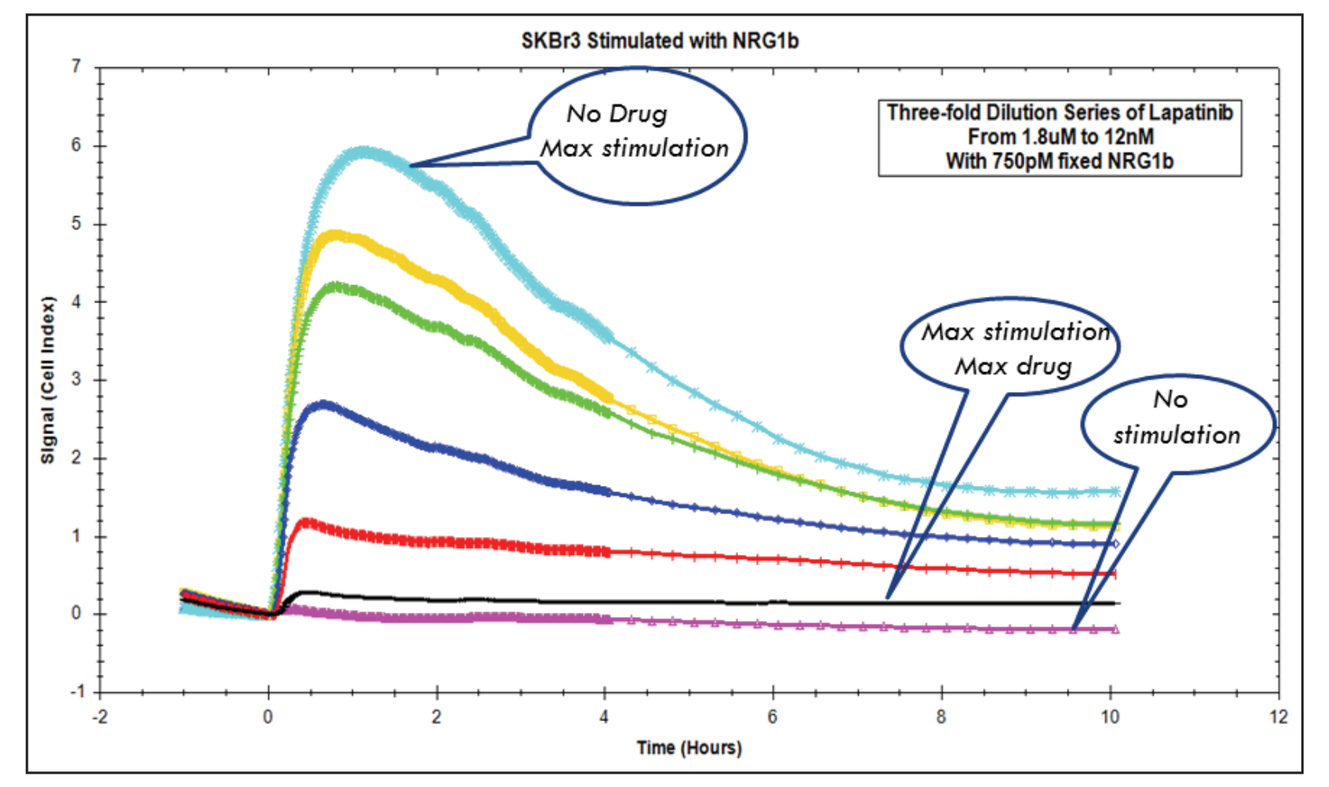

SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib
From 1.8uM to 12nM
With 750pM fixed NRG1b
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Analytical Validation

Celcuity has completed analytical validation studies in accordance with applicable FDA guidance and Clinical and Laboratory Standards Institute (CLSI) standards in its CLIA/CAP certified laboratory to characterize the performance of the CELx HSF test. A summary of the results is below in Table 1.

Table 1. Summary of CELx HSF Test Performance Characteristics

Performance Characteristics	Results
Analytical Precision (Qualitative)	
Analytical Sensitivity (95% CI)	95.8–100% (88/88)
Analytical Specificity (95% CI)	95.8–100% (88/88)
Detection Limits	
Limit of Blank	0.0020 cell attachment units
Limit of Detection	0.0099 cell attachment units
Limit of Quantification	0.1000 cell attachment units
Cutoff Characterization	250 signaling units

References

1. *Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer.* Cancer Res. *2004; 64(23):8534-8540.*
2. *Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review.* J Clin Oncol. *2010;28(1):92-98.*
3. *Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine.* Oncologist. *2009;14(4):320-368.*
4. *Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene.* Science. *1987;235(4785):177-182.*
5. *Perez EA, et al. HER2 testing: current status and future directions.* Cancer Treat Rev. *2014;40(2):276-284.*
6. *Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer.* N Engl J Med. *2008;358(13):1409-1411.*
7. *Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial.* J Clin Oncol. *2010;28(28):4307-4315.*
8. *Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells.* BMC Cancer. *2016; in revision.*
9. *Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity.* Oncotarget. *2016; in press.*

Prevalence of Abnormal HER2 Signaling in HER2- Patients

Table 2. Summary of Patient Characteristics

Characteristic	No.	Percentage (%)
Total Patients	114	100
Age, years		
Mean	58.6	
Range	36–85	
Clinical Stage		
I	23	20
II	62	54
III	24	22
N/A*	4	4
Histology		
DCIS only	0	0
Invasive only	24	21
Invasive Ductal/DCIS mixed	55	48
Lobular/other	35	31
Lymph Status		
Positive	56	49
Negative	46	40
pNx or N/A[1]	12	11
Estrogen Receptor Status		
ER+	96	84
ER-	18	16
HER2 IHC score/FACS		
IHC 0, 1+ or FISH not amp[2]	101	89
IHC 2+ and FISH not amp	13	11

CELx HSF tests were performed on 114 primary tumor cell samples from patients with breast cancer classified as HER2- to measure HER2 pathway stimulation and signal specificity.

For comparative purposes, healthy patients and DAKO IHC test standard breast cancer cell lines SKBr3 and MDA-231 were also analyzed with CELx HSF Tests.

The CELx HSF test identified 27 of 114 HER2- patient samples (23.7%; 95% CI = 17–32%) having abnormally high HER2 signaling activity comparable to HER2+ cell line signaling activity.

Table 2. Patient characteristics of a random set of tumors collected from clinical cancer patients with a distribution of stage, histology and age to study a new test for identifying pathway dysfunction in HER2- breast cancer.

Figure 3. CELx Test Identifies Four Different Subgroups for HER2 Signaling

Examples of CELx HSF test results (HER2±/HER2$_S$±): use of NRG1 (ligand) and pertuzumab (antagonist) to ID net HER2-driven signaling activity. Within HSF+ subgroups differential response to drug is demonstrated.

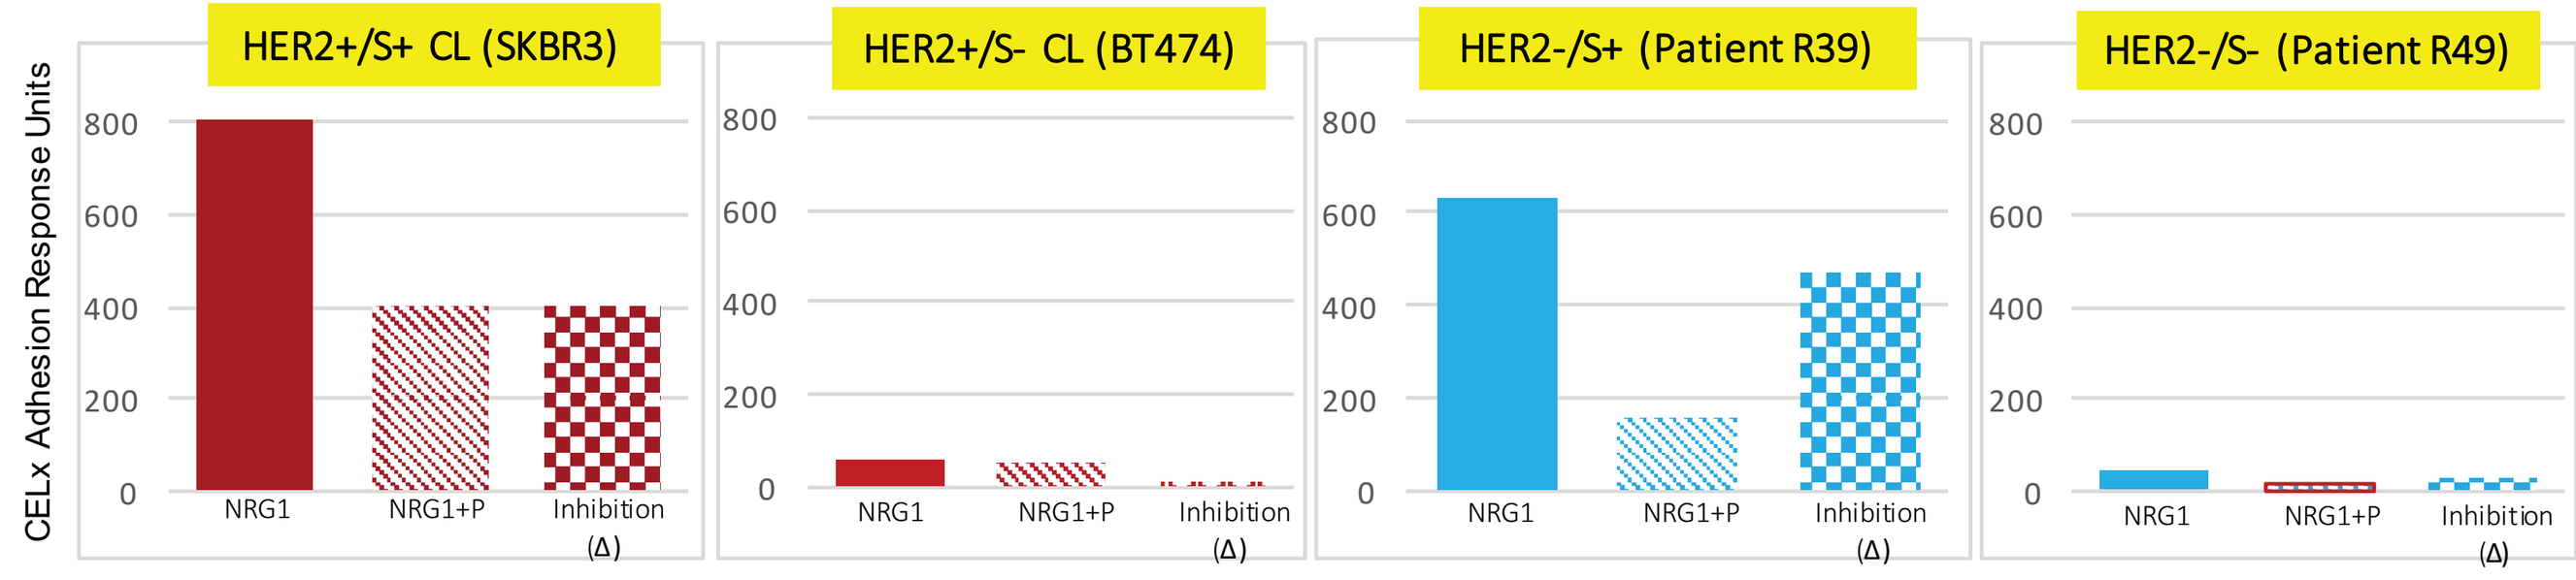

HER2+/S+ CL (SKBR3)
HER2+/S- CL (BT474)
HER2-/S+ (Patient R39)
HER2-/S- (Patient R49)
CELx Adhesion Response Units
800
600
400
200
0
NRG1
NRG1+P
Inhibition (Δ)

- NRG1 initiated signaling varied widely in the HER2+ samples
- SKBR3's signaling was one of highest HER2+ cell lines tested
- Difference between NRG1 and NRG1 + P above is amount of signaling involving HER2 in HER2/HER3 dimerization
- Pertuzumab inhibits 50% of the SKBR3 signal
- None of BT474's limited activity is inhibited

- HER2- primary tumor samples, R39 and R49, show a similar range of NRG1 initiated activity as the HER2+ cell lines
- The HER2-driven activity (Δ) for R39 is nearly identical to SKBR3's
- High Pz inhibition is similar to other HER2/HER2$_S$+ tumors
- R49's low signaling is typical of 80% of HER2- patient tumors

Table 3. Comparison of Inhibition of NRG1-driven HER2 CELx Signals by Trastuzumab (Tz) or Pertuzumab Alone or in Combination

Average % Inhibition of NRGI-driven HER2 Signaling Activity		
HER2 mAb	**HER2+/HER2$_S$+ Cell Lines (n = 3)**	**HER2-/HER2$_S$+ Primaries (n = 5)**
Trastuzumab	19%	44%
Pertuzumab	62%	73%
Trastuzumab + Pertuzumab	87%	81%

Pertuzumab and trastuzumab alone were each more effective in the HER2- group than in HER2+ group.

HER2 Signaling Status vs. IHC HER2 Status in Xenografts

Two breast cancer cell lines were studied: HCC1954, a HER2+ cell line with normal HER2 signaling according to the CELx HSF Test, and BT483, a HER2- cell line with abnormally high HER2 signaling according to the CELx HSF Test. Thus, the HER2 receptor status of each cell line was opposite its HER2 signaling status. For each cell line, twenty 4–5-week-old female NSG mice were injected with two million cells. Mice were randomly assigned to either a control group that received Captisol® or a treatment group that received lapatinib at a dose of 20 mg/kg daily for 16 days.

Table 4. Xenograft Results With HER2+ and HER2- Cell Lines

Cell Line	HCC1954	BT483
HER2 Receptor Expression (IHC)	HER2+ (3+)	HER2- (0)
HER2 Signaling Status (CELx)	Normal	Abnormal
Lapatinib Inhibition (Xenograft)	13% (p = 0.34)	49% (p = 0.01)

HCC1954 (HER2+, normal HER2 signaling) cells

There was no significant difference in tumor volume between the control and lapatinib-treated groups in the mice injected with the HCC1954 (HER2+, normal HER2 signaling) cells. Average tumor sizes reached 1028.7 ± 166.9 mm3 for the control group and 893.8 ± 111.5 mm3 for the lapatinib-treated group (P = 0.285) by the end of the study.

BT483 (HER2-, abnormal HER2 signaling) cells

With the BT483 (HER2-, abnormal HER2 signaling) cells, lapatinib treatment significantly slowed down the increase in tumor size. Average tumor sizes reached 328.3 ± 54.9 mm3 for the control group and 192.4 ± 19.4 mm3 for the lapatinib-treated group (p = 0.0049).

Conclusions:

The results demonstrate that functional HER2-driven signaling status in live tumor cells is more correlative to response to lapatinib in mouse xenograft tumors than HER2 expression level. These findings provide strong evidence that HER2- breast cancer patients with abnormal HER2 signaling may respond to anti-HER2 therapies.

Justification for Clinical Trial Design

Celcuity's findings strongly support the hypothesis that a subgroup of HER2-negative patients may benefit from treatment with HER2-targeted therapies. These findings include:

- No categorical correlation between HER2 IHC status (+ or -) and HER2 signaling activity (abnormal or normal) (Pearson's Chi-Square = 3.68; Phi Max = -0.78, Contingency Coefficient 0.28).
- Approximately 20% of HER2-negative breast cancer patients have the same level of abnormal HER2 signaling activity as the abnormal HER2 signaling activity found in HER2-positive breast cancer cell lines.
- The two HER2 mAbs (trastuzumab and pertuzumab) used to treat HER2+ breast cancer patients are as effective in blocking abnormal HER2-driven signaling function ex vivo in HER2- primary cells with abnormal signaling as they are in HER2+ cell lines. Overexpressed HER2 receptor may thus not be a required condition for breast cancer patient response to HER2 mAbs.
- Functional HER2-driven signaling status in live tumor cells is more correlative to response to lapatinib in mouse xenograft tumors than HER2 expression level.

To assess the potential clinical benefit HER2 therapies may offer these patients, a prospective, single-arm, open label interventional neoadjuvant Phase 2 study was thus initiated.

Abstract ID: 636

This presentation is the intellectual property of Celcuity LLC. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.