Phase Ib expansion study of gedatolisib in combination with palbociclib and endocrine therapy in women with ER+ advanced breast cancer

celcuity™

www.celcuity.com

SAN ANTONIO BREAST CANCER SYMPOSIUM®

DECEMBER 7–10, 2021

Rachel M. Layman[1], Robert Wesolowski[2], Hyo Han[3], Jennifer M. Specht[4], Erica M. Stringer-Reasor[5], E. Claire Dees[6], Peter Kabos[7], Ingrid A. Mayer[8], Ulka Vaishampayan[9], Janice Lu[10], Keerthi Gogineni[11], Aditya Bardia[12], Anne F. Schott[13], Maysa M. Abu-Khalaf[14], Doug Hawkins[15], Brian Sullivan[15], Igor Gorbatchevsky[15], Hope S. Rugo[16].

[1]The University of Texas MD Anderson Cancer Center, Houston, TX; [2]James Cancer Hospital and the Ohio State University Comprehensive Cancer Center, Columbus, OH; [3]Moffit Cancer Center, Tampa, FL; [4]Fred Hutch University of Washington Cancer Consortium, Seattle, WA; [5]University of Alabama, Birmingham, AL; [6]UNC Lineberger Comprehensive Cancer Center, Chapel Hill, NC; [7]University of Colorado Hospital- Anschutz Caner Pavilion (ACP), Aurora, CO; [8]Vanderbilt University Medical Center, Vanderbilt-Ingram Cancer Center, Nashville, TN; [9]Karmanos Cancer Institute, Detroit, MI; [10]USC Norris Comprehensive Cancer Center, Los Angeles, CA; [11]Emory University Hospital, Atlanta, GA; [12]Massachusetts General Hospital, Harvard Medical School, Boston, MA; [13]University of Michigan, Ann Arbor, MI; [14]Sidney Kimmel Cancer Center at Jefferson Health, Philadelphia, PA; [15]Celcuity, Minneapolis, MN; [16]University of California San Francisco Comprehensive Cancer Center, San Francisco, CA

BACKGROUND

The addition of CDK4/6, PI3K-α, or mTOR inhibitors to endocrine therapy (ET) improves progression free survival (PFS) in first and later lines of therapy for estrogen receptor positive (ER+), HER2-negative advanced breast cancer (ABC).[1-5] Gedatolisib is a potent pan-PI3K/mTOR dual inhibitor (Table 1), and, in a preclinical ER+/HER2- xenograft model, gedatolisib combined with palbociclib and fulvestrant was superior to any single agent or dual combination (Figure 1). We hypothesized that simultaneous inhibition of these pathways would improve treatment efficacy. We conducted a Phase Ib study of triplet therapy with gedatolisib (**G**), palbociclib (**P**) a CDK4/6 inhibitor, and ET with letrozole (**L**) or fulvestrant (**F**) in women with ER+/HER2-negative ABC. Manageable toxicity and preliminary antitumor activity were observed in 35 patients enrolled on the dose escalation portion of the study (ASCO 2018[6]). We now report results from the dose expansion study arms at the recommended Phase II dose of 180 mg administered intravenously.

Table 1: Specificity and Potency of Gedatolisib

Gedatolisib is potent against all Class I PI3K isoforms and mTORC1/2, with a superior mechanism of action that minimizes potential for activation of resistance mechanisms. No other pan-PI3K/mTOR inhibitor is known to be under active development in solid tumors.

IC_{50} (nM; cell-free biochemical dose response analysis)

Inhibitor	PI3K-α (m)	PI3K-α (WT)	PI3K-β	PI3K-γ	PI3K-δ	mTORC1	mTORC2
Gedatolisib[7]	0.6	0.4	6.0	5.4	6.0	1.6	1.6
Alpelisib (Piqray)[8]	~4.0	4.6	1156	250	290	-	-
Everolimus (Afinitor)[9]	-	-	-	-	-	~2.0	-

Figure 1: Xenograft Data Consistent with Treatment Hypothesis

Gedatolisib (G) with palbociclib (P) and fulvestrant (F) was evaluated in an ER+, HER2-, PIK3CA[mut] breast cancer mouse xenograft model. During the dosing period, the G+P+F arm and G+P arm induced 90% and 80% tumor regression, respectively. G+F induced 75% tumor growth inhibition. Furthermore, 65 days post dosing, tumor growth suppression was sustained in the G+P+F and G+P arms.

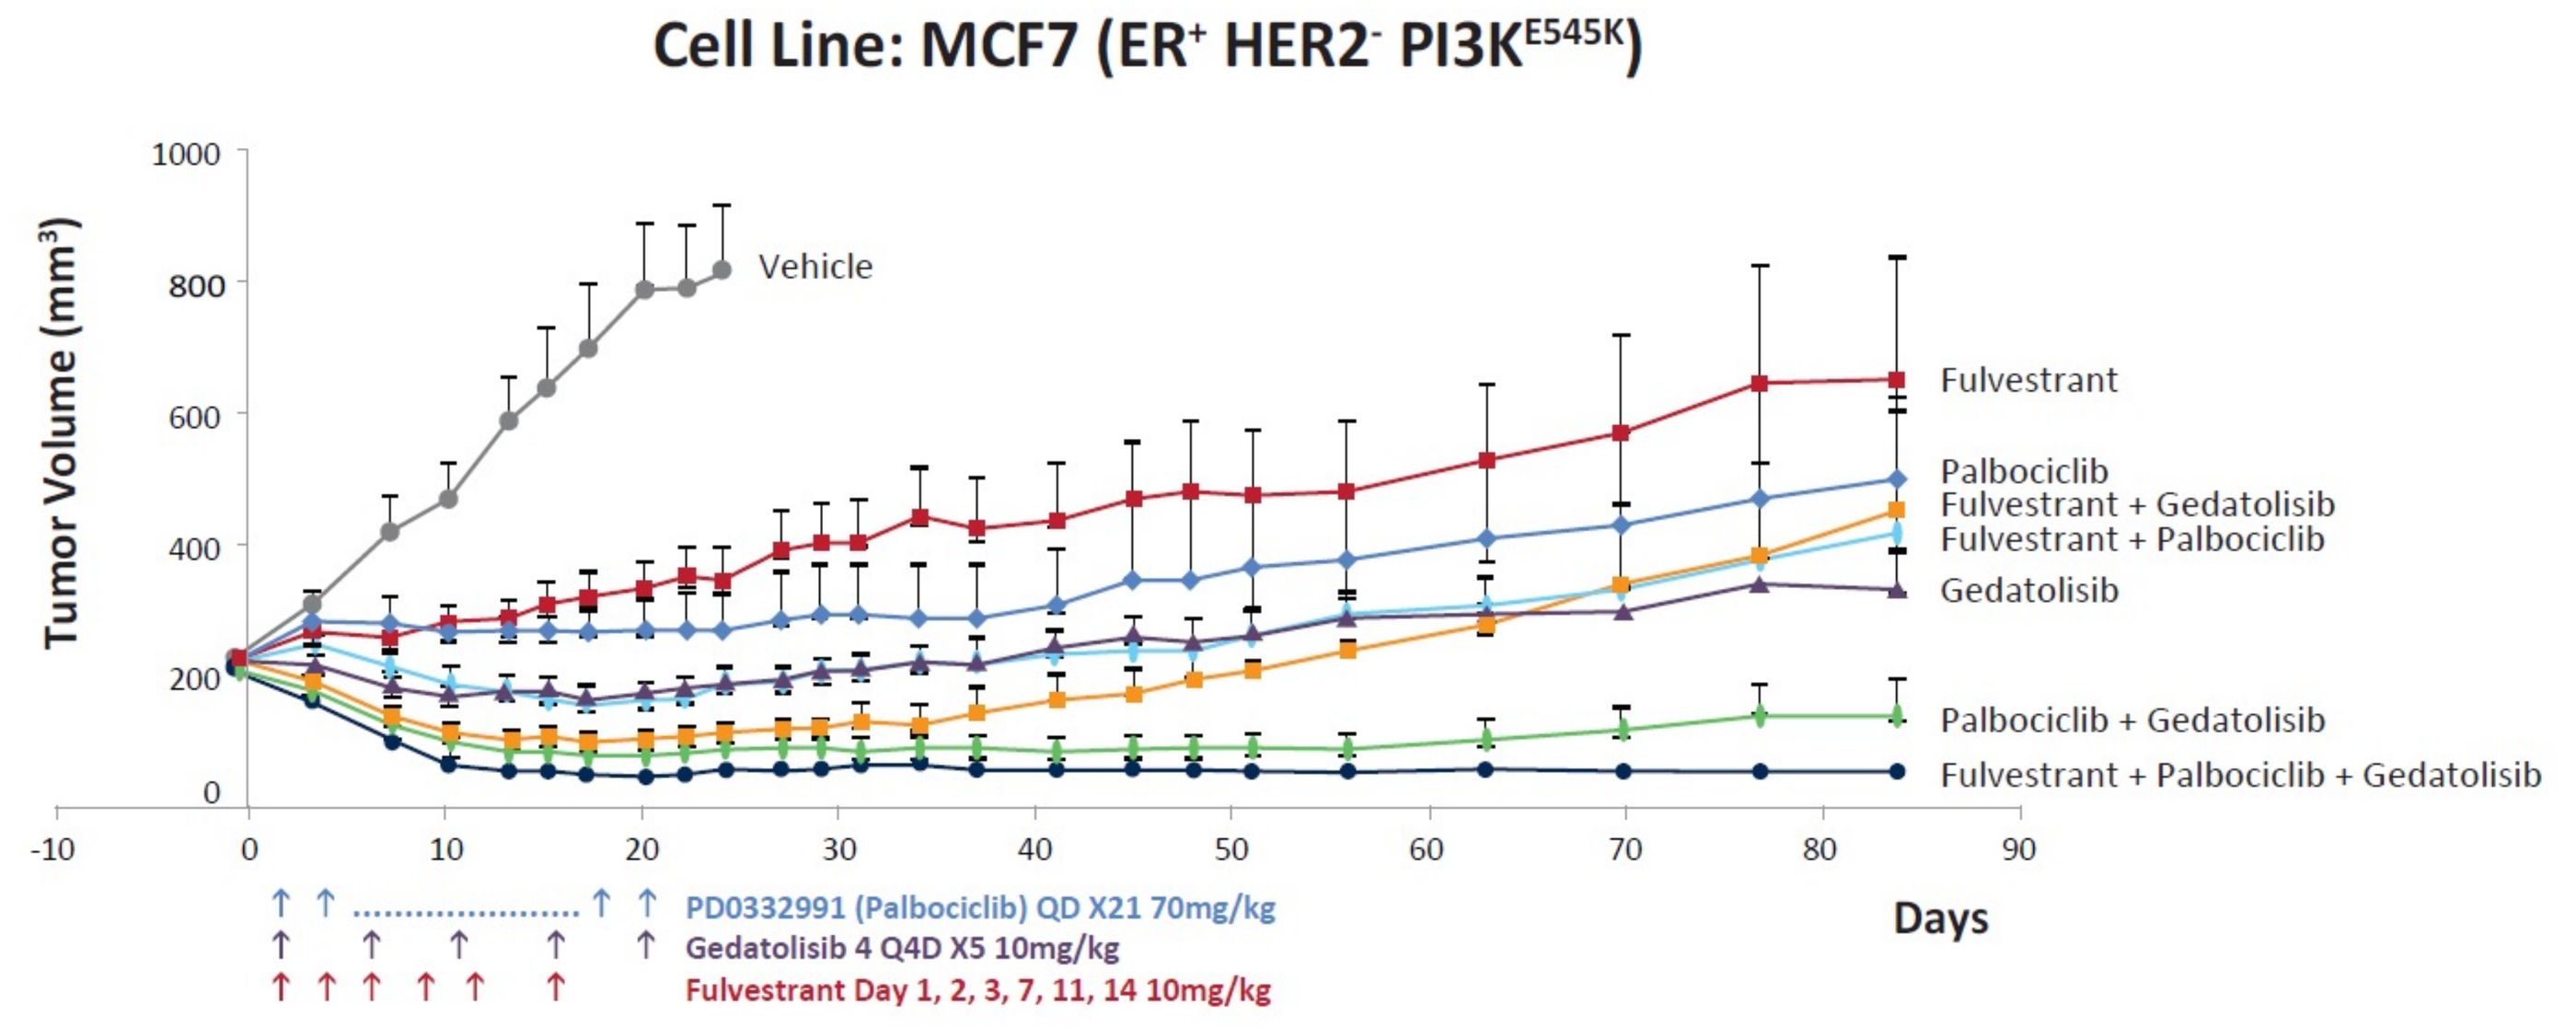

Cell Line: MCF7 (ER+ HER2- PI3K E545K)
Tumor Volume (mm3)
1000
800
600
400
200
0
-10
0
10
20
30
40
50
60
70
80
90
Days
Vehicle
Fulvestrant
Palbociclib
Fulvestrant + Gedatolisib
Fulvestrant + Palbociclib
Gedatolisib
Palbociclib + Gedatolisib
Fulvestrant + Palbociclib + Gedatolisib
PD0332991 (Palbociclib) QD X21 70mg/kg
Gedatolisib 4 Q4D X5 10mg/kg
Fulvestrant Day 1, 2, 3, 7, 11, 14 10mg/kg

STUDY DESIGN

Figure 2: Study Schema and Treated Patients

- Patients with ER+/HER2- ABC were treated in four-arms as shown in "Expansion" panel. Pre-/peri-menopausal women received ovarian suppression.
- Dosing information: Palbociclib: 125 mg/day in a 3 week on, 1 week off schedule; Letrozole : 2.5 mg/day; Fulvestrant: 500 mg intramuscular injection on cycle 1 days 1 and 15, and every 28 days ± 3 days thereafter; Gedatolisib: 180 mg IV weekly (Arms A, B, and C) or three weeks on/one week off (Arm D).
- Tumor assessment performed at baseline and every 12–16 weeks until progression or start of a new anti-cancer therapy.
- Endpoints: Primary - objective response assessed by the investigator; Secondary - safety, duration of response (DOR), and PFS.

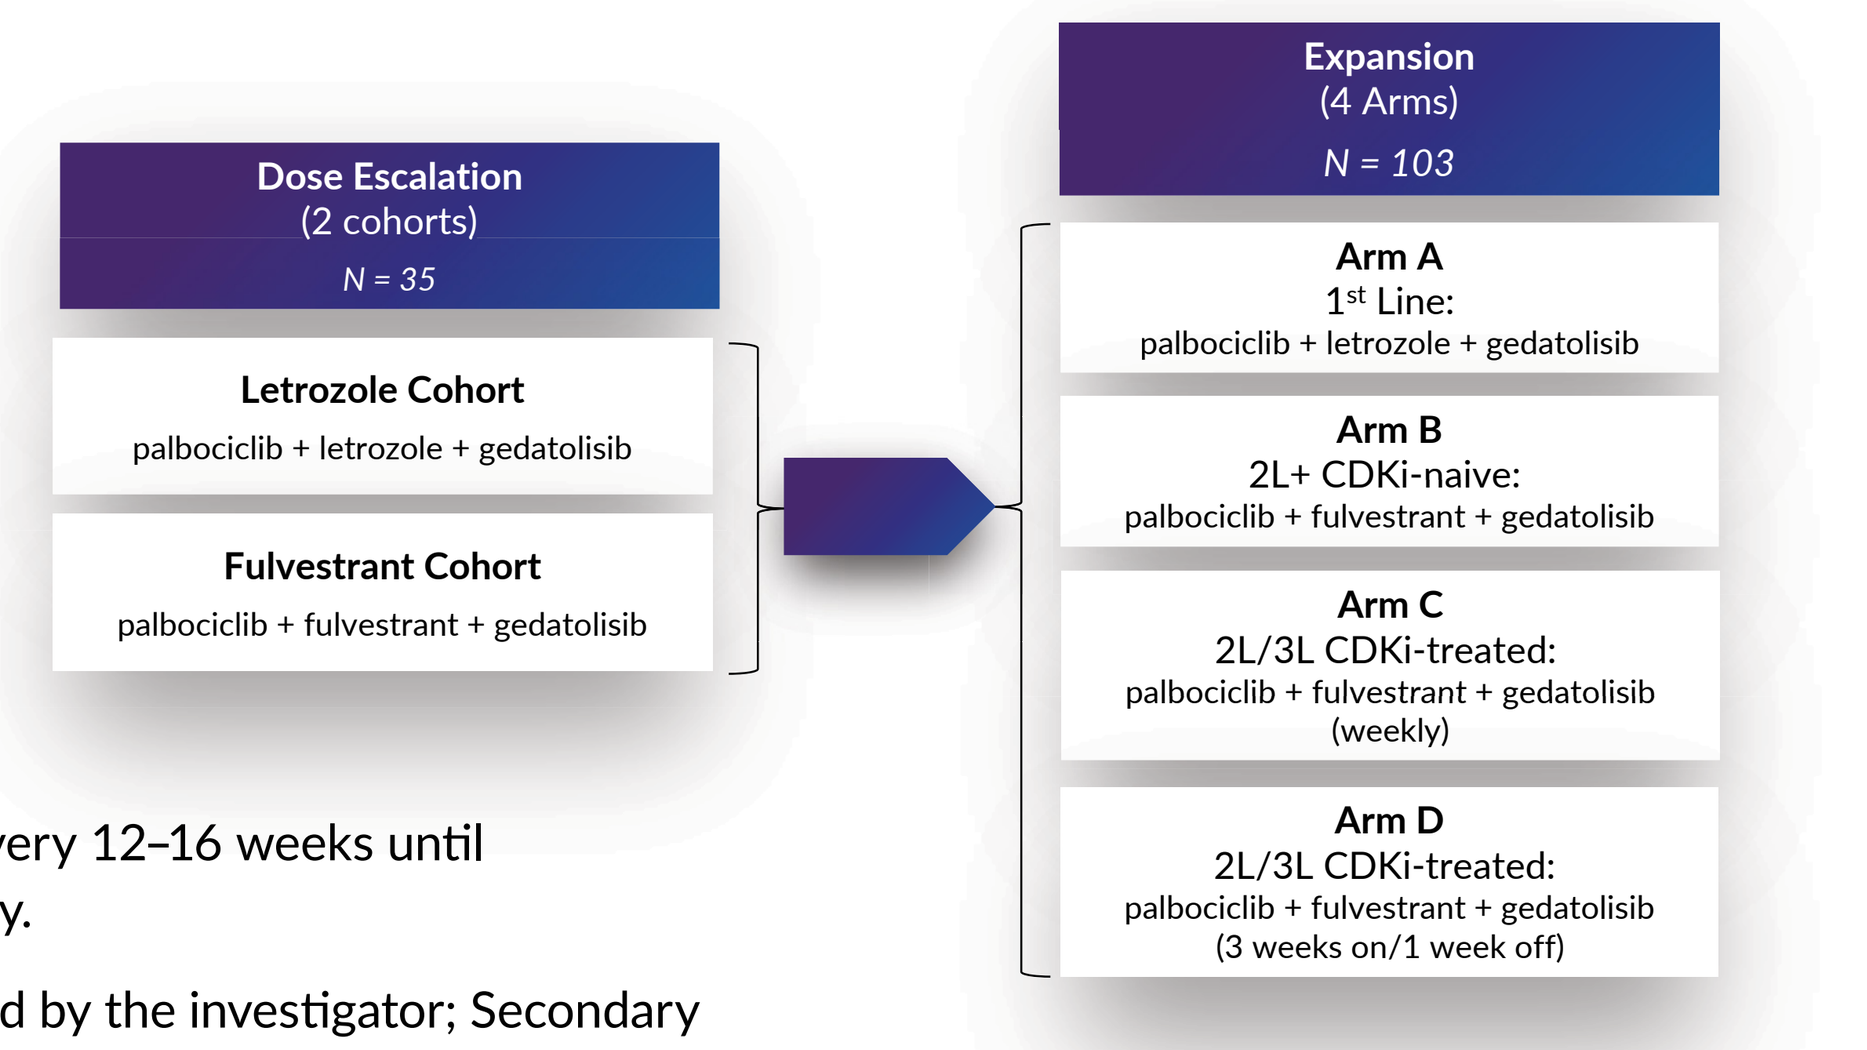

Dose Escalation
(2 cohorts)
N = 35
Letrozole Cohort
palbociclib + letrozole + gedatolisib
Fulvestrant Cohort
palbociclib + fulvestrant + gedatolisib
Expansion
(4 Arms)
N = 103
Arm A
1st Line:
palbociclib + letrozole + gedatolisib
Arm B
2L+ CDKi-naive:
palbociclib + fulvestrant + gedatolisib
Arm C
2L/3L CDKi-treated:
palbociclib + fulvestrant + gedatolisib
(weekly)
Arm D
2L/3L CDKi-treated:
palbociclib + fulvestrant + gedatolisib
(3 weeks on/1 week off)

RESULTS

Preliminary data as of 10-May-2021 data cut-off, representing a database snapshot, and may change based on ongoing routine data monitoring

Table 2: Patient Characteristics at Baseline

	Arm A (n=31)	Arm B (n=13)	Arm C (n=32)	Arm D (n=27)
Age				
Median	54.0	62.0	59.5	59.0
Range	28-78	41-71	41-74	34-79
Prior Therapies – All Stages				
Prior Chemotherapy	15 (48%)	11 (85%)	29 (91%)	16 (59%)
Prior SERD or SERM Therapy	18 (58%)	10 (77%)	23 (72%)	21 (78%)
Prior Aromatase Inhibitor Therapy	10 (32%)	10 (77%)	32 (100%)	23 (85%)
Prior CDK4/6 inhibitor	0 (0%)	0 (0%)	32 (100%)	25 (93%)
Prior Therapies - Advanced Breast Cancer				
1	1 (3%)	8 (62%)	11 (34%)	17 (63%)
2	0	2 (15%)	17 (53%)	9 (33%)
3 or more	0	0	4 (13%)	0
Number of Metastases				
1 – 3	14 (45%)	5 (39%)	14 (43%)	11 (40%)
4 or more	17 (55%)	8 (61%)	18 (57%)	16 (60%)

SAFETY

Table 3: Patient Treatment Discontinuation

	Arm A (N=31)		Arm B (N=13)		Arm C (N=32)		Arm D (N=27)	
Treatment Discontinuation	N	%	N	%	N	%	N	%
Discontinuation of Study Treatment	20	(64.5)	10	(76.9)	32	(100.0)	24	(88.9)
Reasons other than AEs	17	(54.8)	8	(61.5)	28	(87.5)	22	(81.5)
Global deterioration of health status	2	(6.5)	0	-	2	(6.3)	2	(7.4)
No longer willing to participate in study	3	(9.7)	0	-	4	(12.5)	0	-
Objective progression or relapse	10	(32.3)	8	(61.5)	22	(68.8)	17	(63.0)
Other	1	(3.2)	0	-	0	-	3	(11.1)
Protocol violation	1	(3.2)	0	-	0	-	0	-
Adverse Event Related to Study Drug	3	(9.7)	1	(7.7)	4	(12.5)	1	(3.7)
Adverse Event Not Related to Study Drug	0	-	1	(7.7)	0	-	0	-
Death as reason of treatment discontinuation	0	(0.0)	0	(0.0)	0	(0.0)	1	(3.7)
Ongoing at date of cut-off	11	(32.3)	3	(23.1)	0	(0.0)	3	(11.1)

Table 4: Treatment Related and Emergent Adverse Events

	All Expansion Arms (n=103) TRAEs > 20%		
Adverse Event	All Grades %	Grade 3 %	Grade 4 %
Stomatitis[1]	81	27	-
Neutropenia/Neutrophil count decrease[2]	80	53	14
Nausea	76	-	-
Fatigue	68	11	-
Dysgeusia	46	-	-
Vomiting	45	1	-
Anemia	40	12	-
Diarrhea	34	4	-
Decreased appetite	32	0	-
Leukopenia	32	13	3
Dry Mouth	27	-	-
Constipation	25	1	-
Headache	25	-	-
Pruritus	25	5	-
Rash	24	7	-
Rash maculo-papular	24	14	-
Hyperglycemia	23	5	2
Infusion related reaction	23	-	-
Lymphocyte count decreased	20	12	1

[1] Prophylactic treatment for stomatitis not initially included; Stomatitis Gr3: A(32%); B(39%); C(22%); D(22%); [2] Neutropenia and neutrophil count decrease were reported interchangeably for many patients; except for Arm B, the total for each neutrophil related category alone in all arms is well below levels typically reported for patients treated with palbociclib.

Table 5: Combined AEs of Interest by Arm

	Arm A (n=31)		Arm B (n=13)		Arm C (n=32)		Arm D (n=27)	
	Any Grade	G3-4	Any Grade	G3-4	Any Grade	G3-4	Any Grade	G3-4
Stomatitis or Mucosal inflammation	87%	32%	100%	38%	88%	22%	89%	22%
Neutropenia or Neutrophil count decreased	71%	58%	100%	85%	66%	56%	81%	67%
Hyperglycemia or Blood glucose increased	32%	3%	31%	8%	25%	9%	26%	7%
Rash (includes maculo-papular, pruritic, pustular, and papular)	81%	45%	62%	38%	41%	3%	44%	7%
AST or ALT increased	32%	10%	38%	8%	9%	3%	19%	4%
Diarrhea	61%	6%	31%	0%	31%	6%	52%	7%
Colitis	3%	0%	0 %	0%	0%	0%	0%	0%
Pneumonitis	6%	3%	0%	0%	3%	0%	4%	4%

No grade 5 AEs occurred in any Arm. Treatment emergent AEs start on/after the first study dose and up to 28 days from the last study dose.

CLINICAL ACTIVITY

Table 6: Efficacy Summary

	Phase Ib (N=103)			
Arm	A (N=31)	B (N=13)	C (N=32)	D (N=27)
Patients	1L: CDKi-Naïve	2L+: CDKi-naïve	2L/3L: CDKi-pretreated	2L/3L: CDKi-pretreated
Study Treatment	P + L+ G (weekly)	P + F+ G (weekly)	P + F + G (weekly)	P + F + G **(3 weeks on/1 week off)**
# of Evaluable Patients	27	13	28	27
ORR[1] (95% CI)	**85%** (66%-96%)	**77%**[2] (46%-95%)	**32%**[2,3] (16%-52%)	**63%**[2,3] (42%-81%)
CBR[4] (95% CI)	**96%** (81%-~100%)	**100%** (75%-100%)	**79%** (59%-92%)	**96%** (81%-~100%)
Median PFS (mos) (95% CI)	**31.1** (16.9, NR)	**11.9** (3.7, NR)	**5.1** (3.4, 7.5)	**12.9** (7.4, 16.7)

[1] ORR is objective response rate in evaluable patients. ORR represents PR, except in Arm A, which had 1 CR. Responses by Physician Assessment per RECIST 1.1; [2] Includes 2 unconfirmed partial responses; [3] ORR was superior in Arm D relative to Arm C in patients regardless of the number of prior therapies for ABC. In Arm C and Arm D, ORR for patients receiving 1 prior line of therapy was 33% and 56% respectively and for ≥2 prior lines of therapy it was 32% and 78%; [4] CBR is clinical benefit rate; 95% confidence interval calculated by Clopper- Pearson Exact method; NR is not reached

Figure 3: Progression Free Survival (PFS) Kaplan-Meier Curves

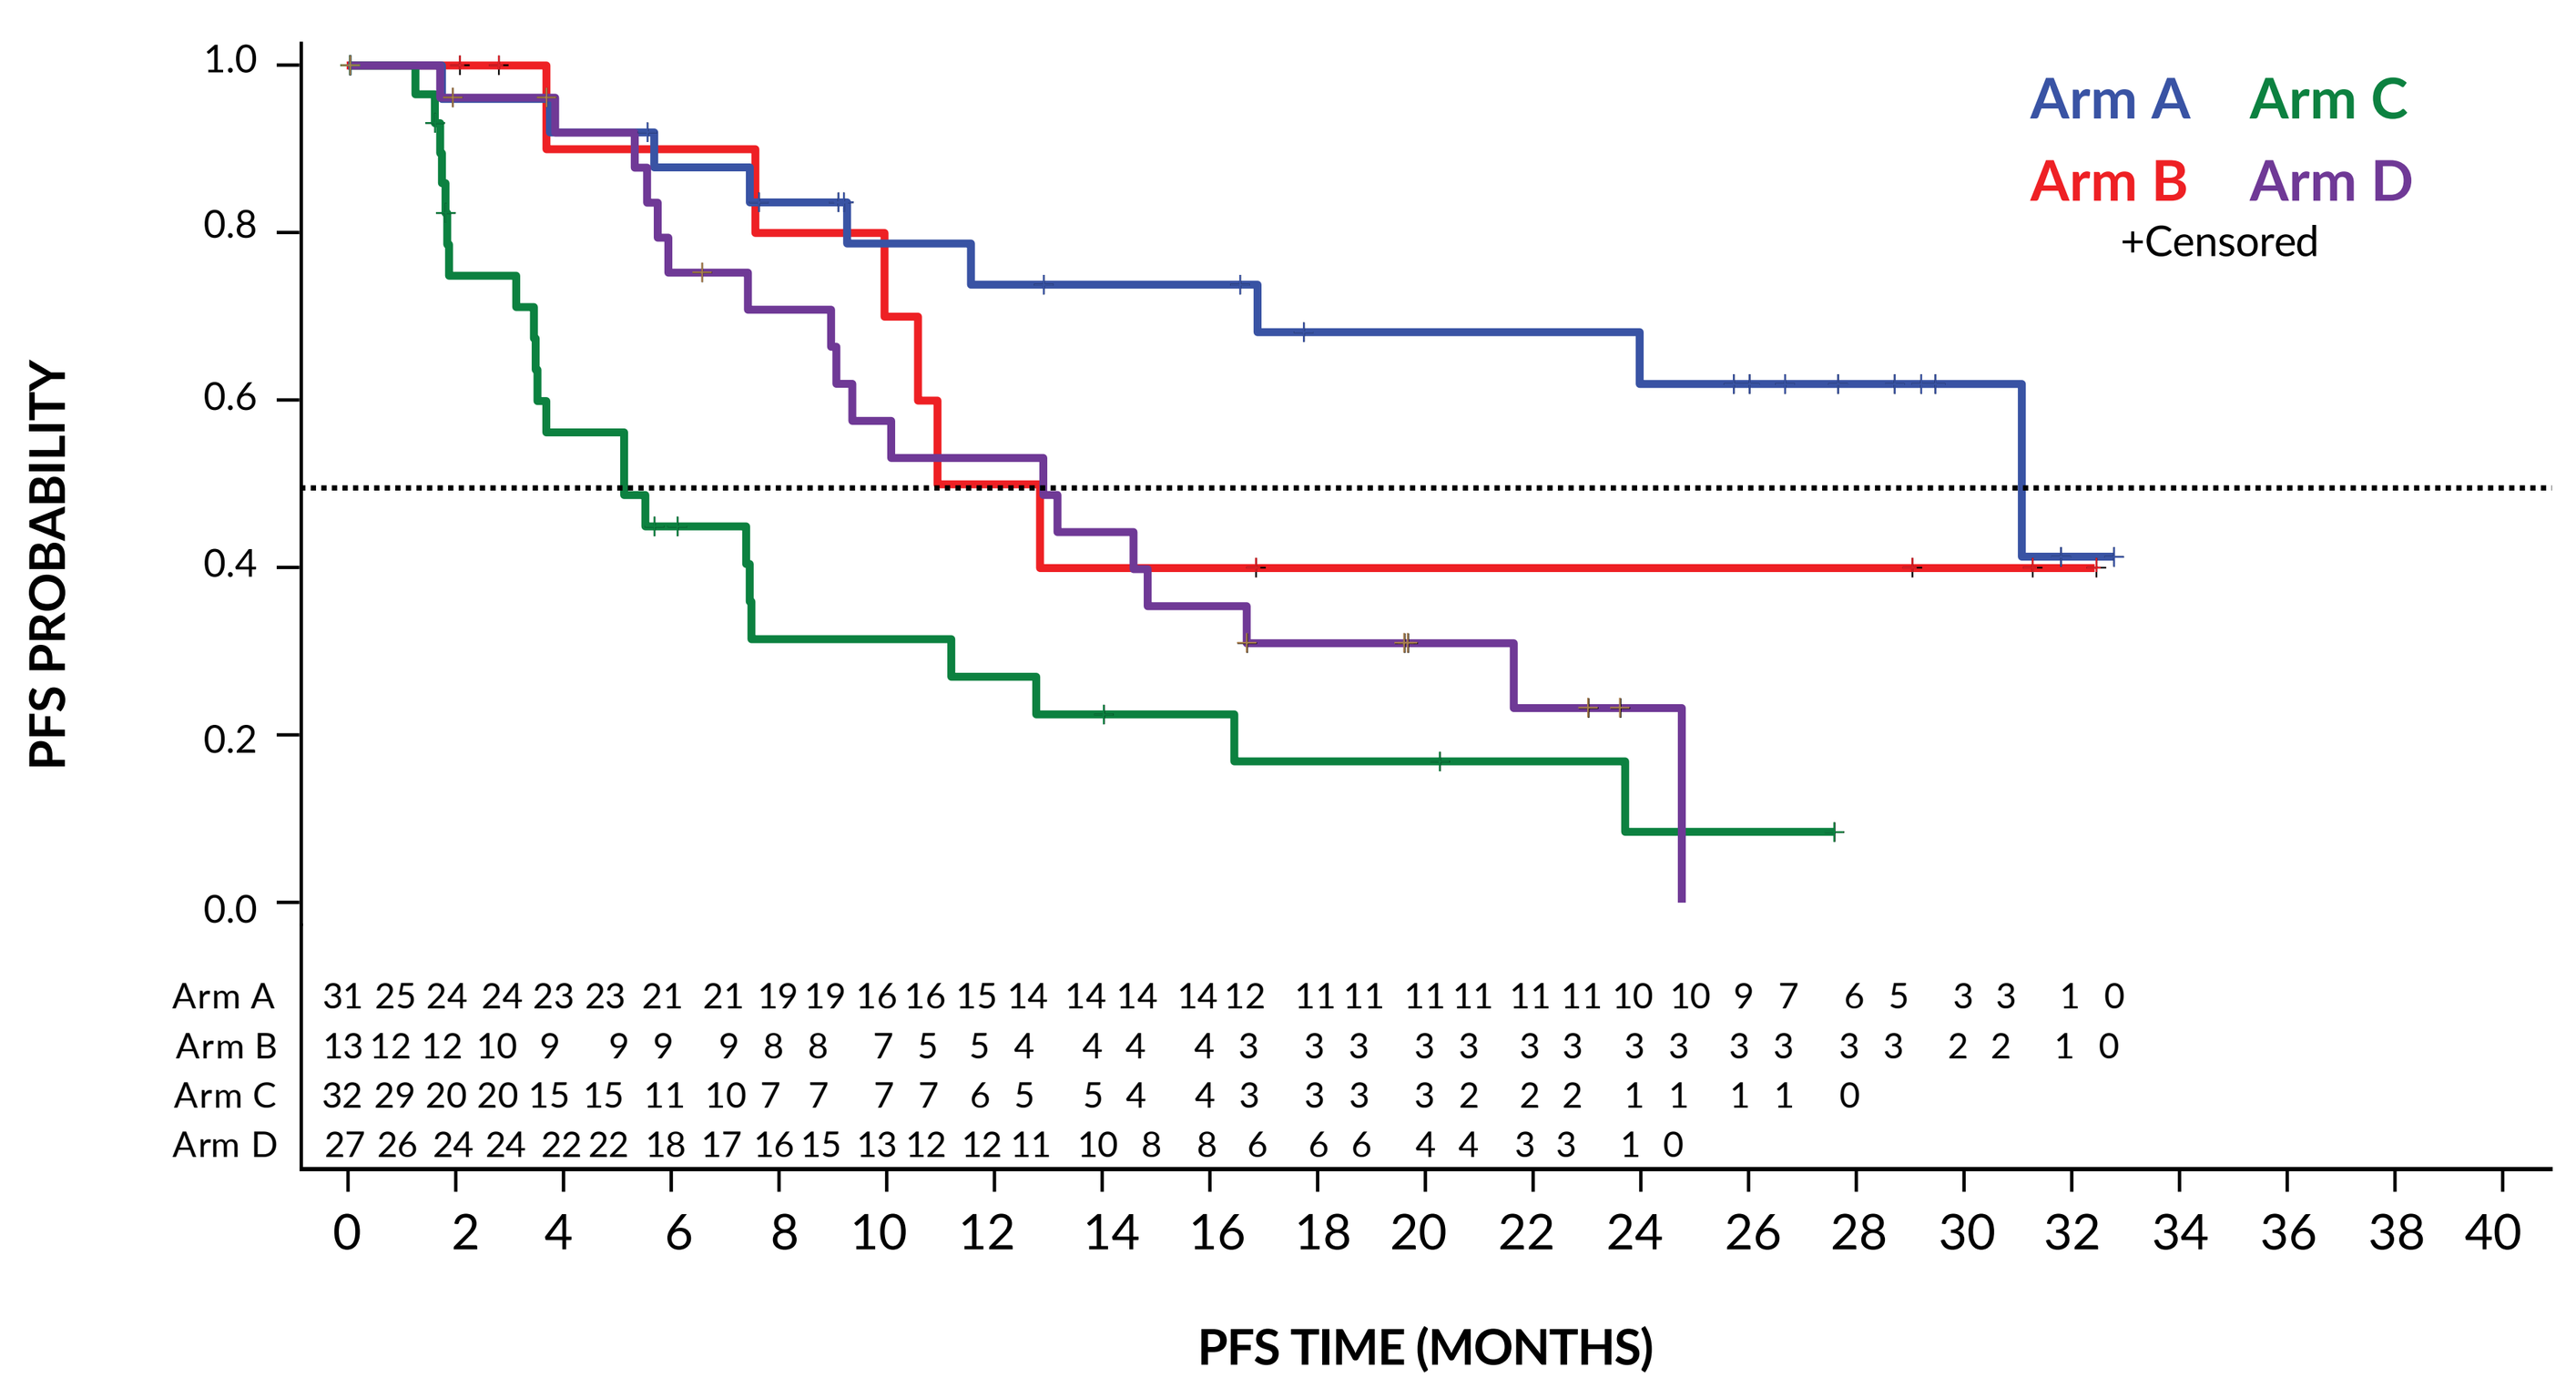

Arm A
Arm C
Arm B
Arm D
+Censored
PFS PROBABILITY
1.0
0.8
0.6
0.4
0.2
0.0
Arm A 31 25 24 24 23 23 21 21 19 19 16 16 15 14 14 14 14 12 11 11 11 11 11 11 10 10 9 7 6 5 3 3 1 0
Arm B 13 12 12 10 9 9 9 9 8 8 7 5 5 4 4 4 4 3 3 3 3 3 3 3 3 3 3 3 3 3 2 2 1 0
Arm C 32 29 20 20 15 15 11 10 7 7 7 7 6 5 5 4 4 3 3 3 3 2 2 2 1 1 1 1 0
Arm D 27 26 24 24 22 22 18 17 16 15 13 12 12 11 10 8 8 6 6 6 4 4 3 3 1 0
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40
PFS TIME (MONTHS)

Table 7: Comparison of Patient Characteristics: Arm C vs Arm D

	Arm C (N=32)	Arm D (N=27)
Duration of Immediate Prior Therapy (DIPT) (weeks)	Mean 41 Median 28 Min, Max 7, 150	Mean 69 Median 59 Min, Max 4, 219
Duration of Study Treatment (DST) (weeks)	Mean 29 Median 15 Min, Max 1, 139	Mean 52 Median 41 Min, Max 7, 112
ORR (%) / Median PFS (m)	32% / 5.1 months	63% / 12.9 months

Table 8: Exploratory Analysis of Arm C and Arm D Patient Characteristics

	Duration of Immediate Prior Treatment (DIPT)			
	DIPT <180 Days		DIPT <365 Days	
Arm	C	D	C	D
# Evaluable patients with DIPT <185 or 365 days (% of evaluable)	12 (44%)	7 (27%)	20 (74%)	11 (42%)
Gedatolisib Dosing Schedule	Weekly	**3 wk on / 1 wk off***	Weekly	**3 wk on / 1 wk off***
Median DIPT (days)	97	106	146	155
Median Duration of Study Treatment (DST, days)	81	270	131	276
Ratio of median DST vs. DIPT	0.8	2.6	0.9	1.8
Overall Response Rate to Study Treatment (95% CI)	**0%** (0%-26%)	**71%** (29%-96%)	**15%** (3%-38%)	**73%** (39%-94%)
	(p = 0.0018)		(p = 0.0043)	

DIPT: Duration of Prior Therapy; DST: Duration of Study Treatment; Responses by Physician Assessment per RECIST 1.1; 2 by 2 contingency table using Fisher's exact test for ORR p-value; *gedatolisib and palbociclib dosing schedule synchronized at 3 weeks on/1 week off

Figure 4: Arm D Best Response – Tumor Size

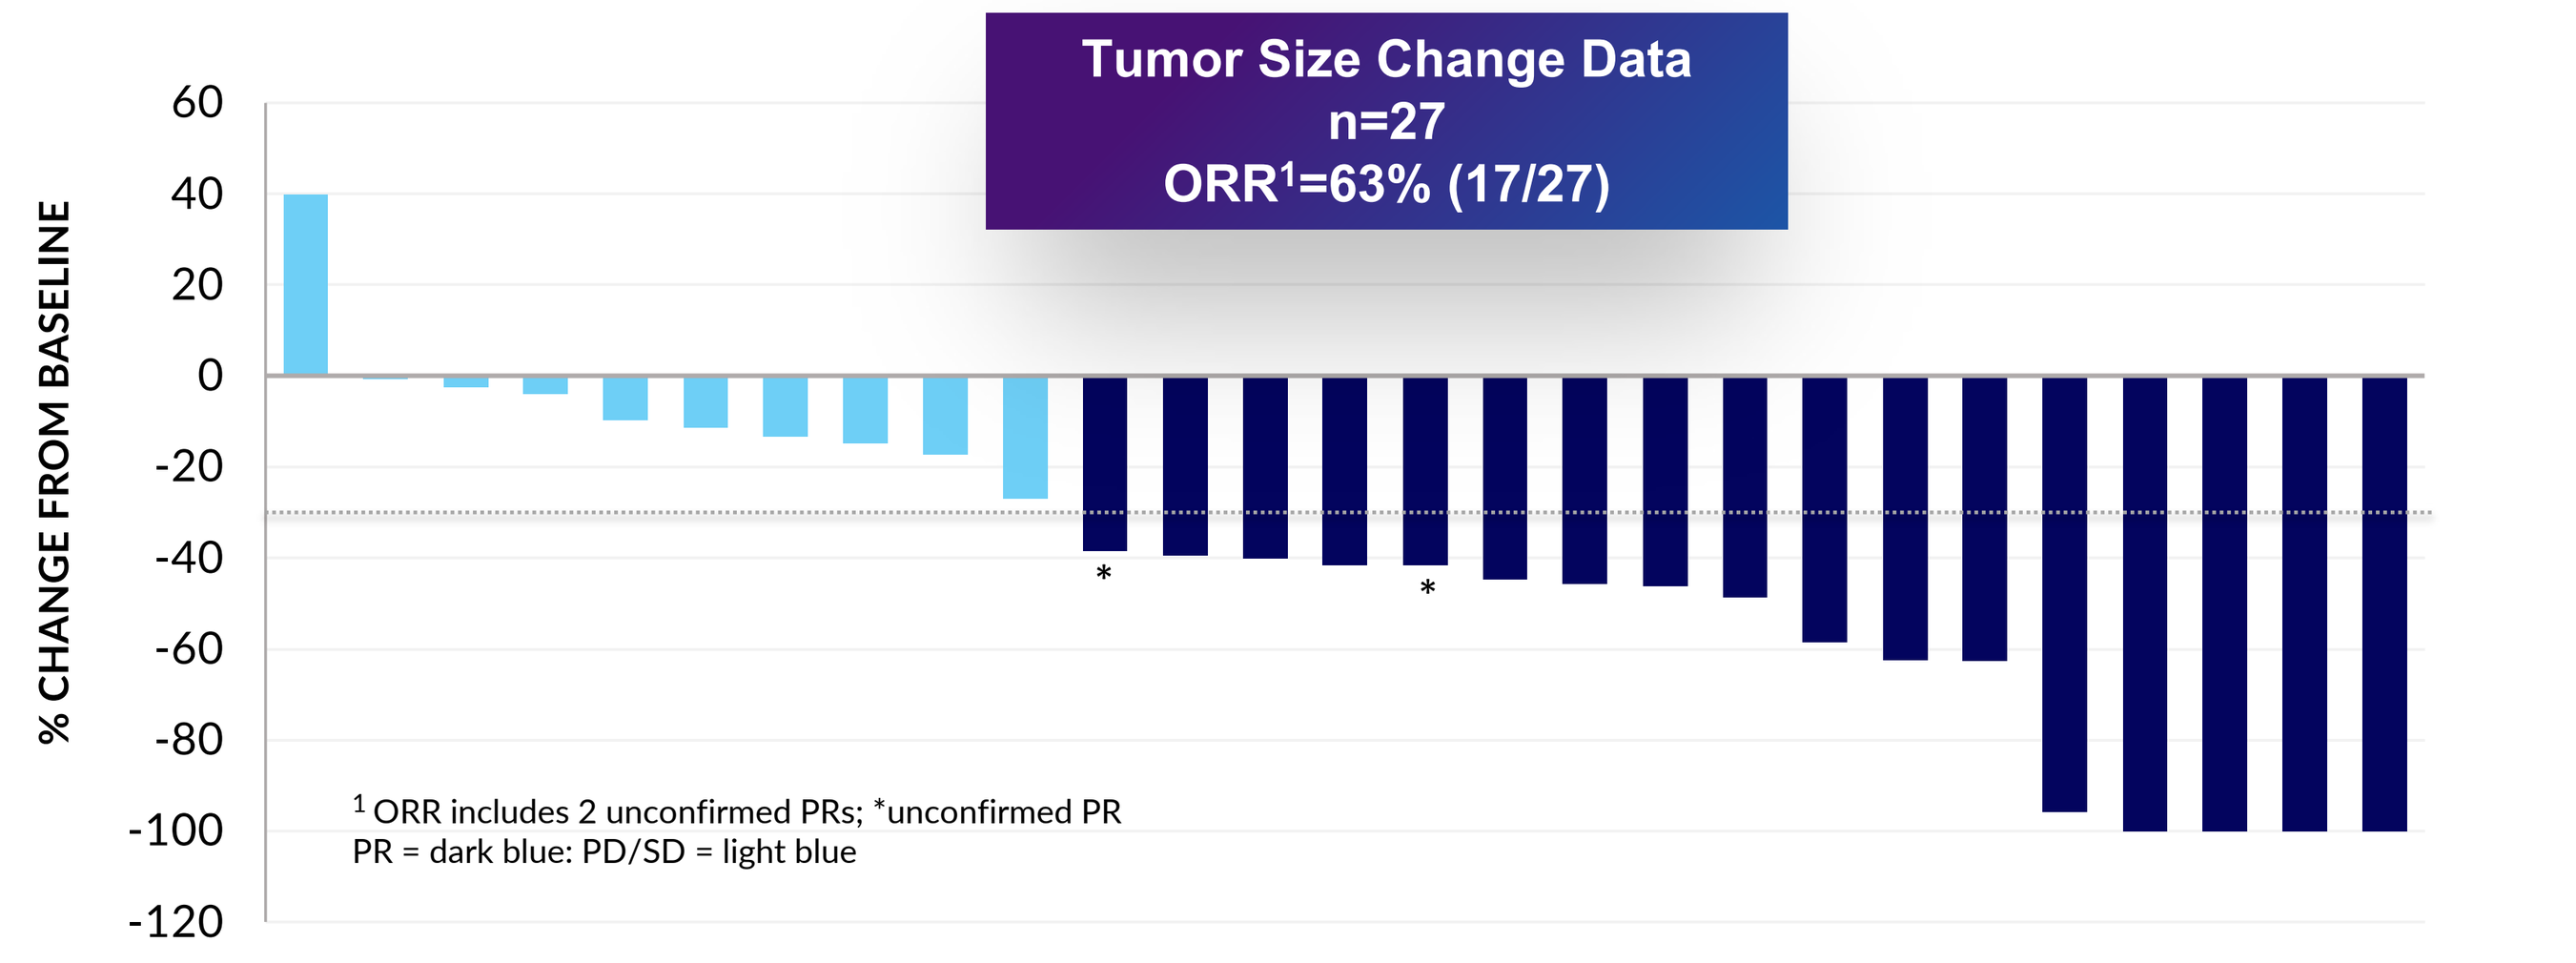

Tumor Size Change Data
n=27
ORR1=63% (17/27)
% CHANGE FROM BASELINE
60
40
20
0
-20
-40
-60
-80
-100
-120
1 ORR includes 2 unconfirmed PRs; *unconfirmed PR
PR = dark blue: PD/SD = light blue

Figure 5: Arm D Time to First Response, Duration of Response & Treatment

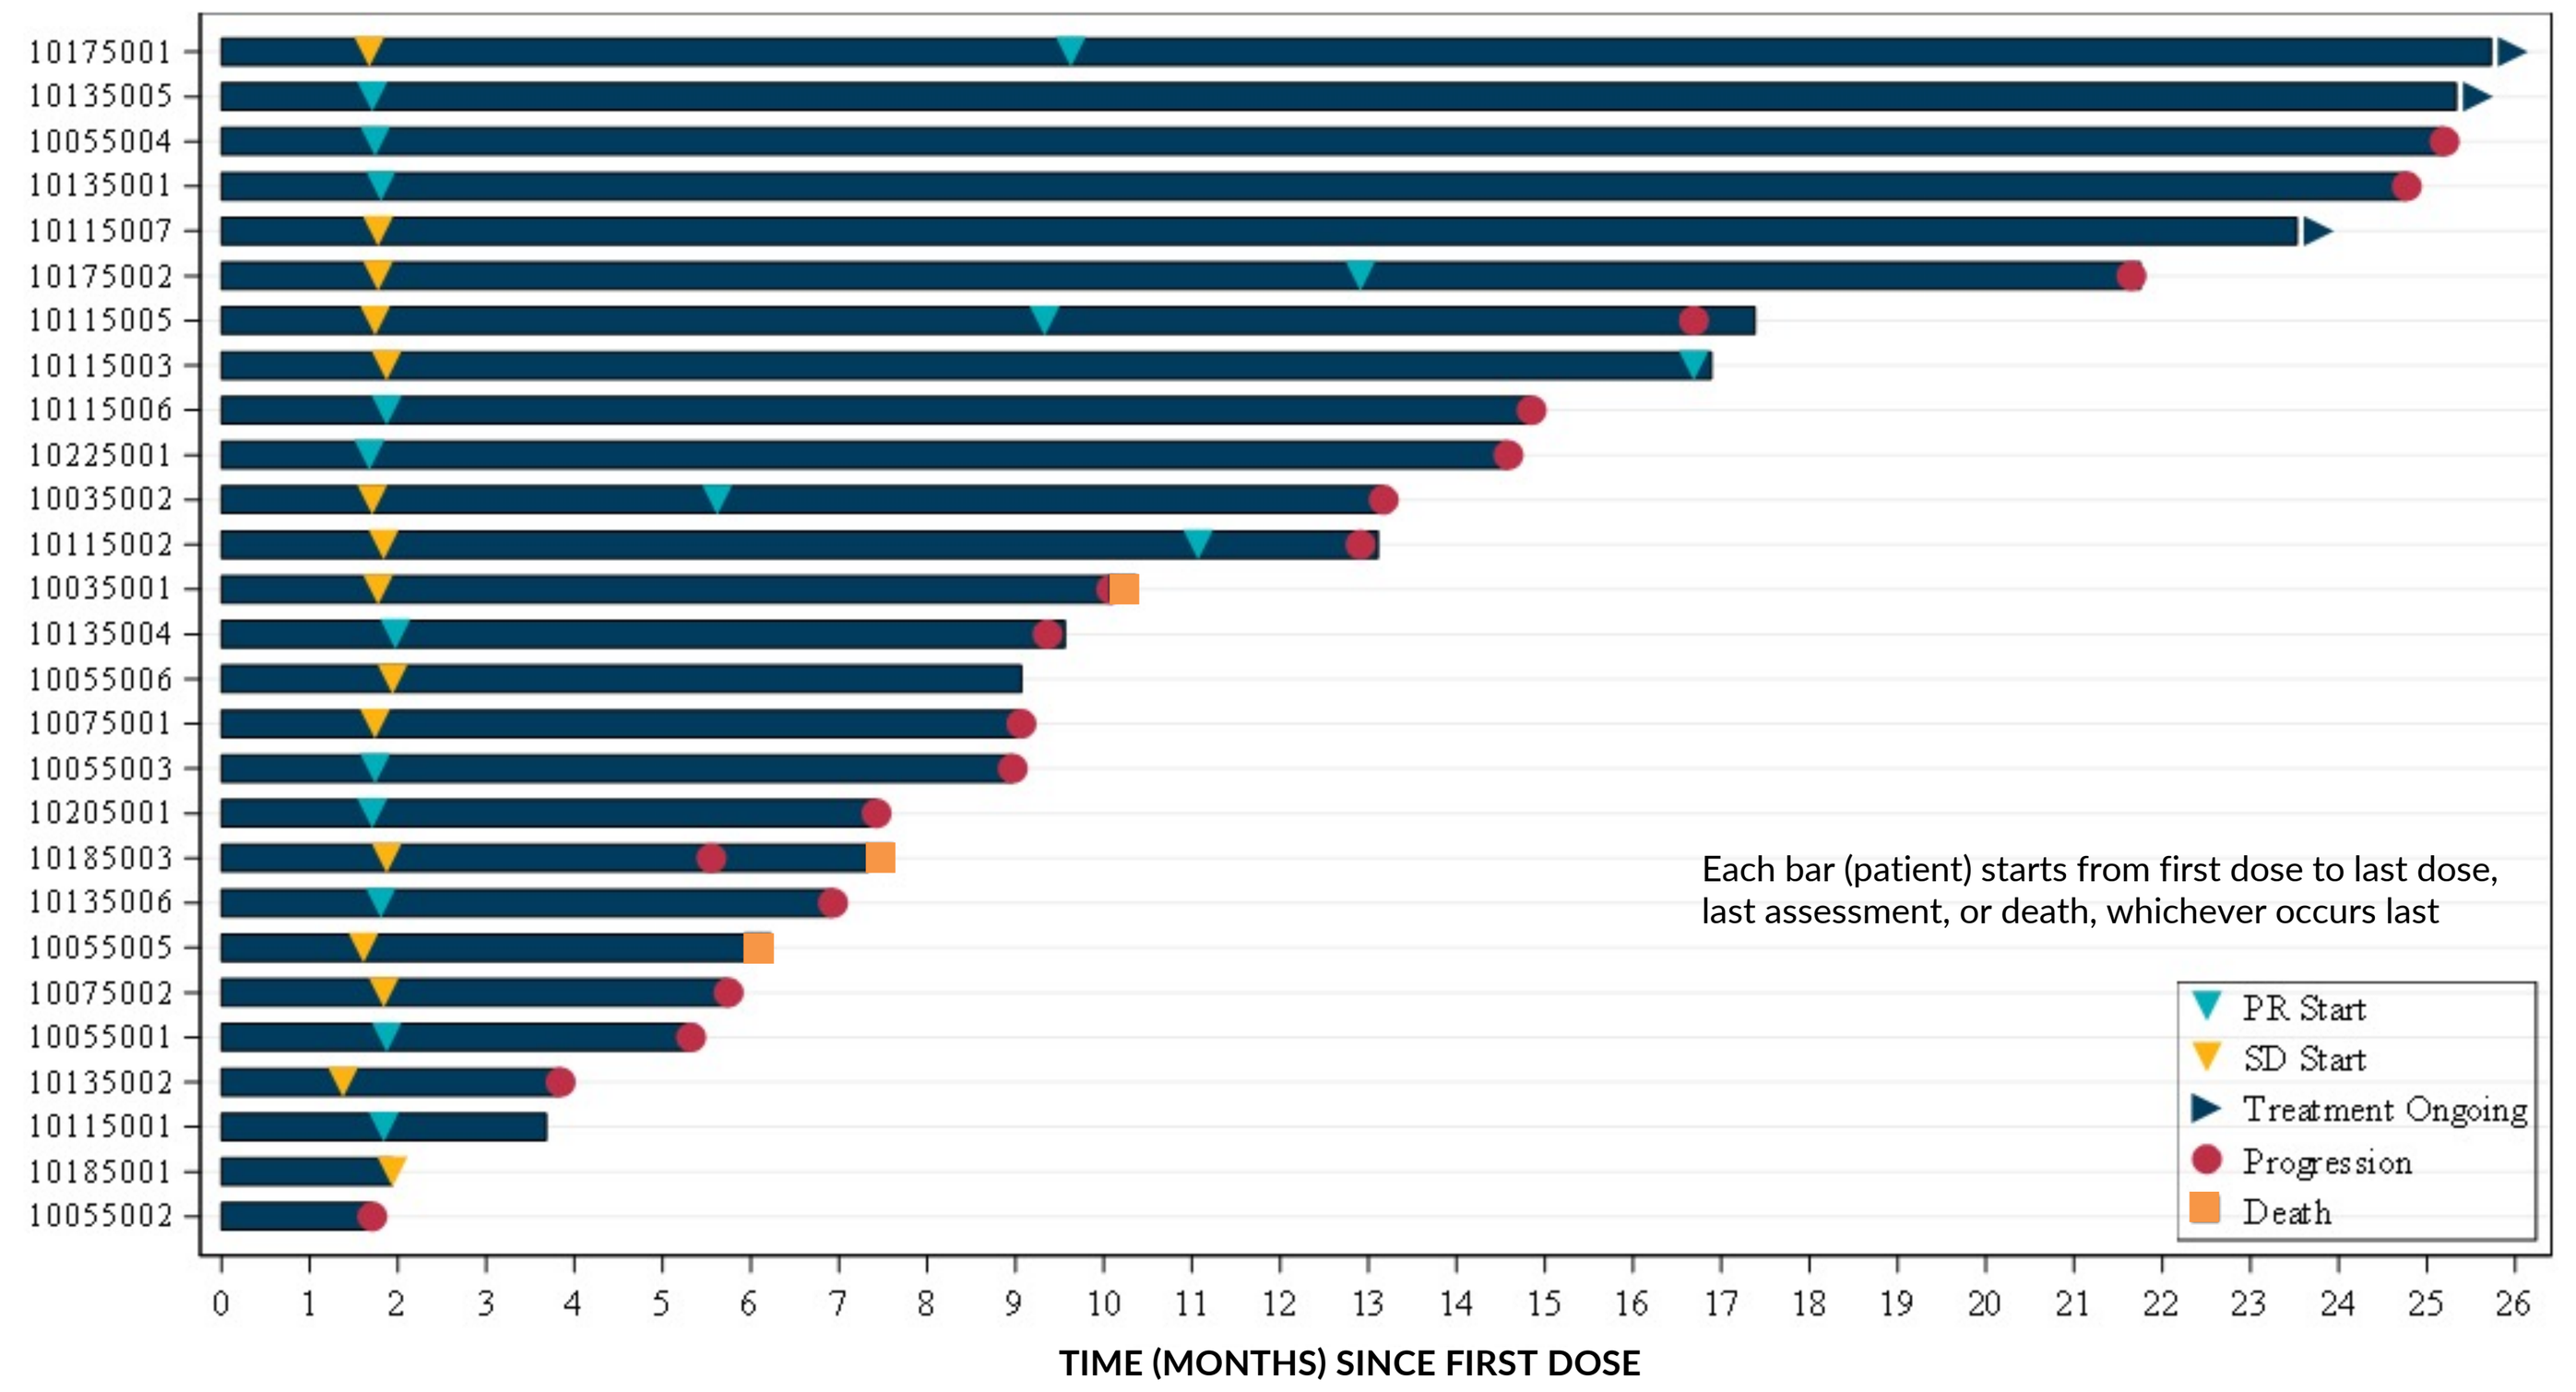

10175001
10135005
10055004
10135001
10115007
10175002
10115005
10115003
10115006
10225001
10035002
10115002
10035001
10135004
10055006
10075001
10055003
10205001
10185003
10135006
10055005
10075002
10055001
10135002
10115001
10185001
10055002
Each bar (patient) starts from first dose to last dose, last assessment, or death, whichever occurs last
PR Start
SD Start
Treatment Ongoing
Progression
Death
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26
TIME (MONTHS) SINCE FIRST DOSE

CONCLUSIONS AND FUTURE DIRECTIONS

- This Phase Ib study of gedatolisib in combination with palbociclib and ET demonstrated a favorable safety profile with manageable toxicity and a low number of treatment discontinuations due to related AEs. The most common AE was stomatitis; hyperglycemia was lower compared to other PI3K inhibitors reflecting differentiation of PK properties.
- Promising antitumor activity in both first and later line settings for ER+ ABC that compared favorably to historical controls.
- Responses were also observed in cancers that were refractory to the last treatment.
- Two factors may explain the lower ORR and PFS in Arm C compared to Arm D: patient characteristics (see Table 7) and treatment schedule
 - Arm C included twice as many patients who had received 2 or more prior therapies as Arm D (66% vs. 33%).
 - Exploratory analysis of ORR and duration of treatment (Table 8) found significantly higher ORR and longer duration of treatment in Arm D vs. Arm C patients who progressed within 6 months and within 12 months. In addition, ORR was superior in Arm D relative to Arm C in patients regardless of the number of prior therapies for ABC.
 - This suggests that Arm D's gedatolisib/palbociclib synchronized dosing schedule (3 wks on/1 wk off) may have played a significant role in the different outcomes between the Arms.
- A phase 3 study evaluating gedatolisib in patients with ER+/HER2- ABC is planned.

References

1. Turner NC, et al. (2015) doi: 10.1056/NEJMoa1505270
2. Andre F, et al. (2019) doi: 10.1056/NEJMoa1813904
3. Rugo HS, et al. (2020) doi: 10.1200/JCO.2020.38.15_suppl.1006
4. Baselga J, et al.(2012) doi: 10.1056/NEJMoa1109653
5. Dhakal A, et al. (2020). doi: 10.1177/1178223420944864
6. Forero-Torres, A., et al. (2018). J Clin Oncol 36(15_suppl): 1040-1040
7. Venkatesan 2010 DOI: 10.1021/jm901830p
8. Fritsch 2014 DOI: 10.1158/1535-7163.MCT-13-0865
9. Schuler 1997 DOI: 10.1097/00007890-199707150-00008

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

Abstract #1017

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.