Subgroup of HER2-negative breast cancer patients with hyperactive RAS network signaling identified: dynamic pathway activity test identifies patients that may benefit from PI3K/mTOR or PI3K/mTOR/BCL inhibitors

CELsignia

Salmaan Khan, Adrish Sen, Catherine Kuzmicki, Ian MacNeil, Aaron Broege, Sarah Mutka, Kelly Brass, Ky McCracken, Laura Milligan, Kaija Kottke, Brian Sullivan, Lance G. Laing

Celcuity Inc., 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

celcuity

celcuity.com

Background

G-protein-coupled receptors (GPCRs) and their phospholipid ligands have well described links to cancer, including breast cancer (BC).[1,2] Lysophosphatidic acid (LPA) is a GPCR ligand with a long-known link to RAS/MAPK/PI3K oncogenic signaling. LPA can activate RAS that in turn activates PI3K-α to advance tumor growth.[3-5] Additionally, through several mechanisms, LPA can activate receptor tyrosine kinases (RTK) that in turn can work through PI3K-α or other PI3K isoforms to drive oncogenic signaling.[6-8] Due to the nonlinear, non-serial nature of the RAS signaling network and other complexities, identifying RAS nodes involved in oncogenic signal transduction has been challenging. Moreover, since inhibition of a single RAS node can trigger adaptation that results in activation of other RAS nodes, multiple RAS nodes and PI3K isoforms may need to be targeted to induce durable anti-tumor responses. To identify patients with dysregulated RAS signaling tumors who may respond to RAS node inhibitors, an assay using an impedance biosensor was developed. The CELsignia RAS Activity Test measures GPCR-initiated signaling activity and PI3K, mTOR, and BCL's role in transducing this activity in live tumor cells. In this test, LPA is used to stimulate multiple pathways linked to RAS activation and identify which of these RAS nodes are involved. The current study set out to characterize the prevalence of dysregulated RAS signaling initiated by LPA in HER2-negative BC patients and the role played by PI3K, mTOR and BCL.

Methods

Cell lines: The breast cancer cell lines used in this study were maintained according to ATCC recommendations and authenticated by ATCC.

Tissue specimens and patient tumor culture: A set of de-identified excess breast cancer tissue specimens was obtained from 60 patients. Methods for tumor cell extraction and culture were based on the Huang, et al. method.[9,10]

Flow cytometry: Flow cytometry on disaggregated tissue and cultured cells was performed on the Agilent Novocyte 3005. Antibody staining was performed by standard procedures. DNA staining for DNA index calculation was performed with FxCycle™ violet. Apoptosis was assessed by staining with anti-Cleaved-Caspase 3. mTOR activity was assessed by staining with an anti-RPS6 (pS235/S236) antibody.

CELsignia analysis: Dynamic live cell response to a GPCR agonist (LPA), a PI3K-α inhibitor (GDC-0077), a pan-PI3K/mTOR inhibitor (gedatolisib), and a BCL inhibitor (navitoclax) was measured using an xCELLigence impedance biosensor (Agilent Technologies). From these responses, the gross amount of GPCR-initiated signaling and corresponding participation of PI3K-α, all Class 1 PI3K-isoforms, mTORC1, and BCL was quantified and converted to a signaling score.

Statistical analysis: A data set of 60 CELsignia LPA scores from BC patient cultures was analyzed. A normal mixture model was fitted to the combined data set using the normalmixEM procedure in the R package mixtools. Two runs of the statistical analysis were made, fitting 2 and 3 components, along with a baseline single-component model.

Figure 1: CELsignia analysis uses biosensor to quantify signaling activity in real time in live cells

- CELsignia analysis leverages connections among cell adhesion, impedance, and cell signal transduction
- Live cells are attached via ECM to a microelectrode on the bottom of a 96-well impedance biosensor plate. Additionally, the tumor epithelial cells in the wells form adhesion-based gap junctions
- The cells attached to the biosensor impede the flow of electrons when mVAC current is applied and changes in impedance (mΩ) are recorded
- Signaling activity causes cell adhesion changes that affect impedance levels recorded by the biosensor

Example Impedance vs. Time Data Set

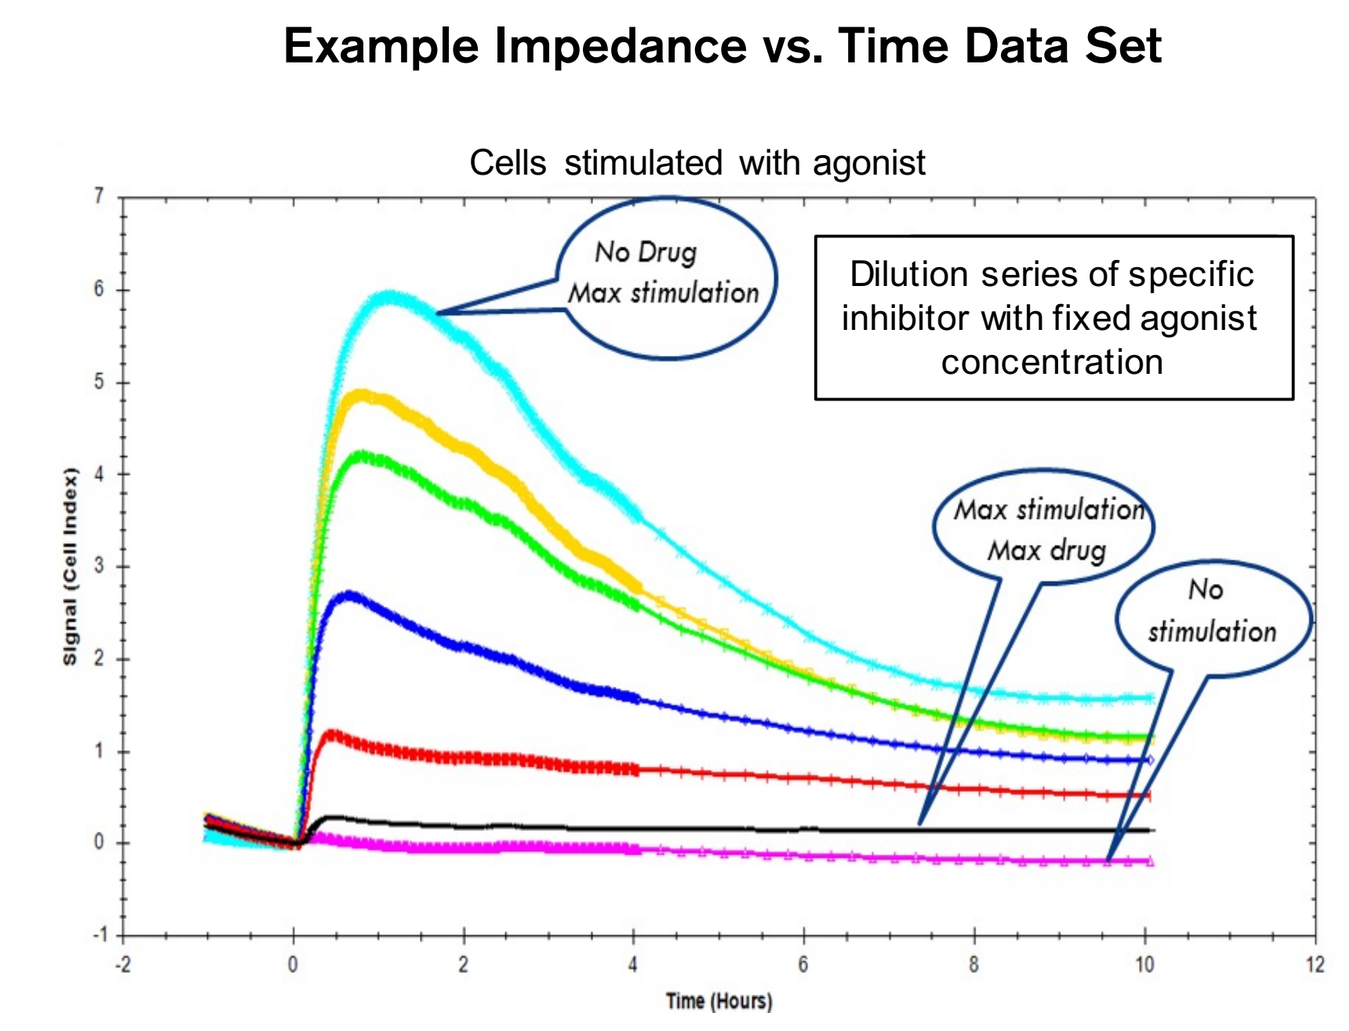

Cells stimulated with agonist
No Drug
Max stimulation
Dilution series of specific inhibitor with fixed agonist concentration
Max stimulation
Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Pathway-specific ligands and inhibitors used to turn on/off signaling pathways

▼

Very subtle cell adhesion changes are measured

▼

Response quantified as signaling units over time to characterize signaling activity

▼

Determine Signaling Score for each pathway tested

Results

Figure 2: LPA-initiated activity measured by CELsignia in BC cell lines and patient cells

A

Cell line	Tumor type & molecular classification		Mutational status: PIK3CA	PTEN	p53	CELsignia: LPA response	LPAR expression (CCLE): LPAR1	LPAR2	LPAR3
HCC1954	HER2+	Basal A-HER2	Mut	wt	Mut	2427	4.0	7.4	5.8
JIMT-1	NA	NA	Mut	wt	Mut	1193	0.1	11.2	0.1
BT-20	TNBC	Basal A	Mut	wt	Mut	1155	0.0	13.6	0.0
HCC38	TNBC	Basal B	Mut	wt	Mut	436	1.2	7.2	1.0
MDA-MB-231	TNBC	Basal B	wt	wt	Mut	352	24.0	2.7	0.0
HCC1806	TNBC	Basal A	wt	wt	wt	334	6.3	5.6	6.9
MDA-MB-415	ER+	Luminal A	wt	Mut	Mut	238	0.8	4.1	5.1
Hs578T	TNBC	Basal B	wt	wt	Mut	210	9.4	0.9	0.1
HCC1569	HER2+	Basal A-HER2	wt	Mut	Mut	158	0.0	7.1	4.5
MDA-MB-453	TNBC/HER2+	Basal A	Mut	Mut	wt	143	0.3	7.5	0.2
MFM223	NA	NA	Mut	wt	Mut	79	NA	NA	NA
HCC1428	ER+	Luminal A	wt	wt	wt	24	0.0	12.1	0.8
CAL-51	NA	NA	Mut	Mut	wt	22	0.2	9.2	0.0
SKBR3	HER2+	HER2	wt	wt	Mut	16	0.0	2.2	0.3
T47D	ER+	Luminal A	Mut	wt	Mut	14	4.7	7.3	0.0
MCF-7	ER+	Luminal A	Mut	wt	wt	1	0.0	13.6	0.0
BT-483	ER+	Luminal B	Mut	wt	Mut	0	0.1	4.3	0.0
AU565	HER2+	HER2	wt	wt	Mut	-2	0.0	2.2	0.1
CAMA-1	ER+	Luminal A	wt	Mut	Mut	-5	0.2	12.1	0.0
MDA-175-vii	ER+	Luminal B	wt	wt	wt	-241	1.4	12.2	0.0

B CELsignia LPA response vs. LPAR expression in BC cell lines

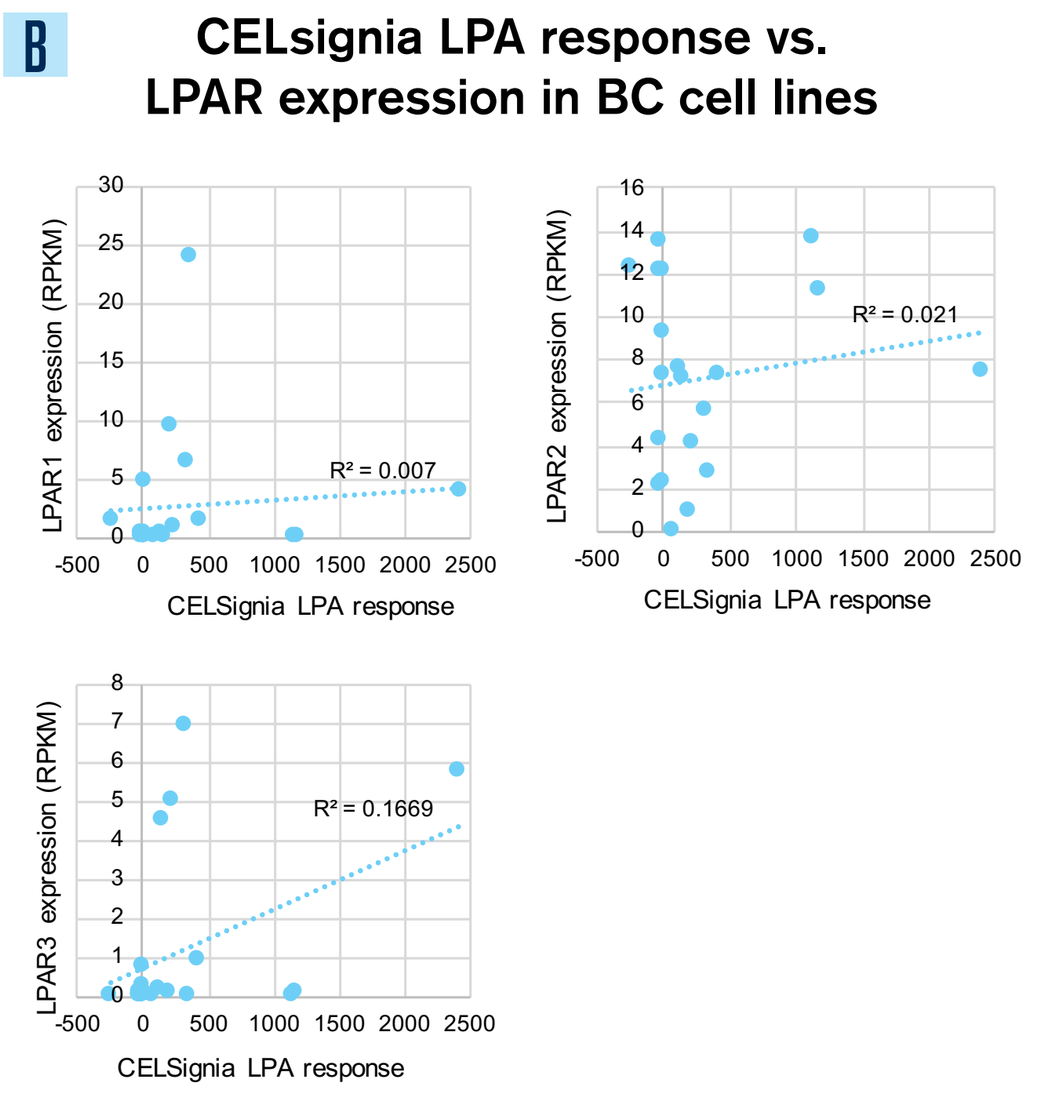

LPAR1 expression (RPKM)
R² = 0.007
LPAR2 expression (RPKM)
R² = 0.021
LPAR3 expression (RPKM)
R² = 0.1669
CELsignia LPA response

C C1974–5-day culture

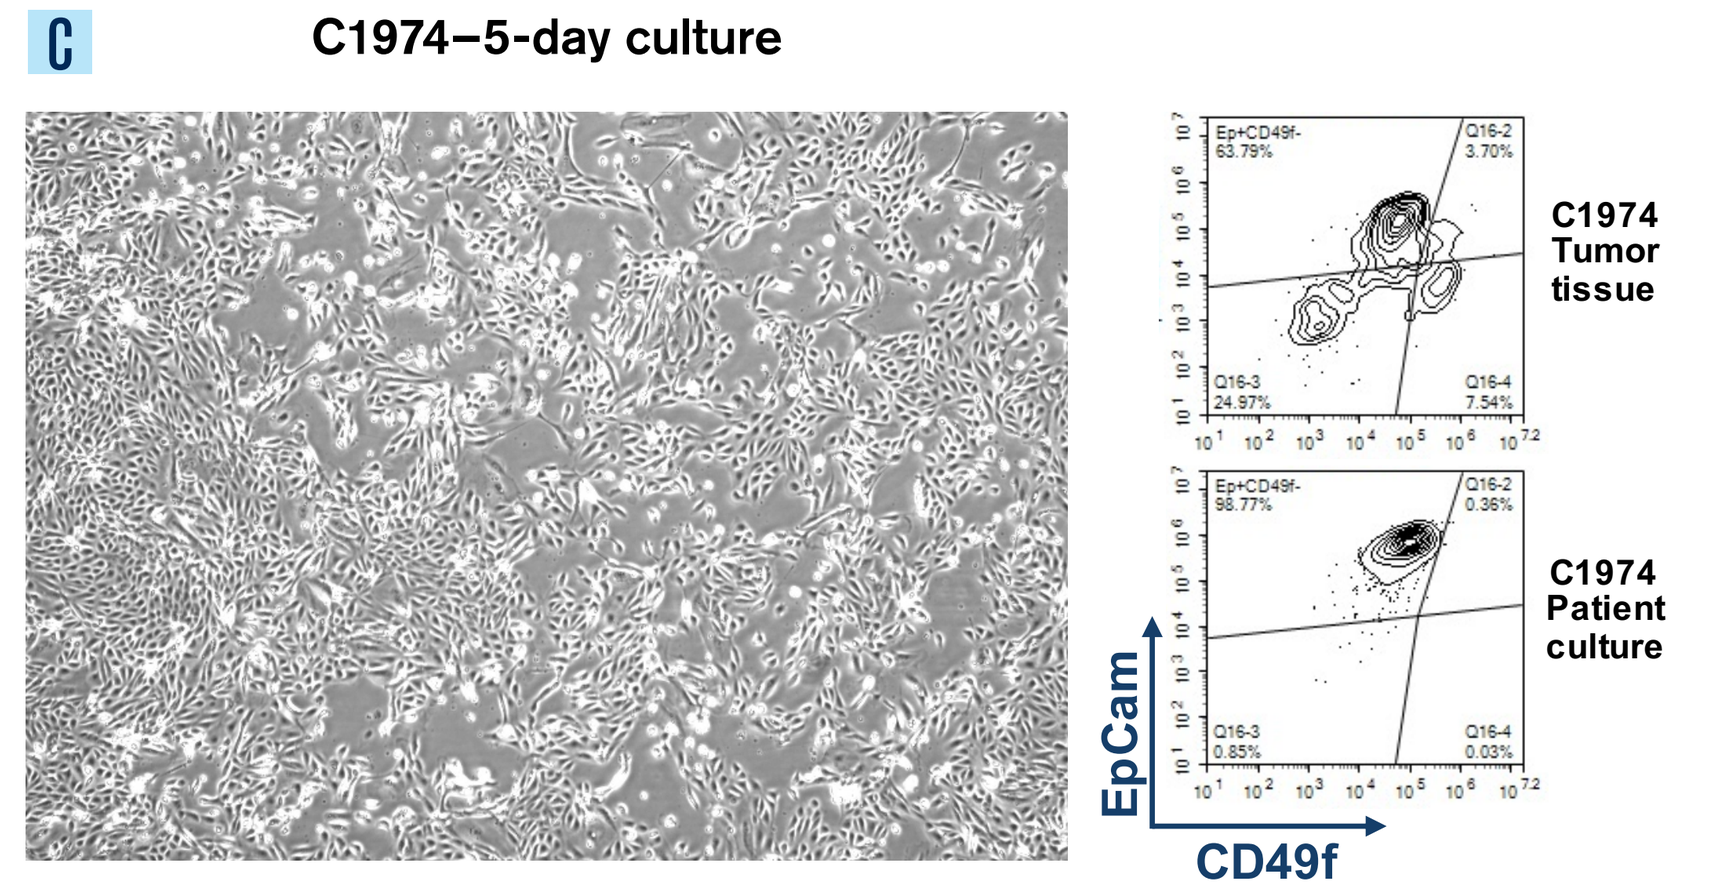

C1974 Tumor tissue
C1974 Patient culture
EpCam
CD49f

D CELsignia LPA response vs. LPAR expression in BC patient cultures

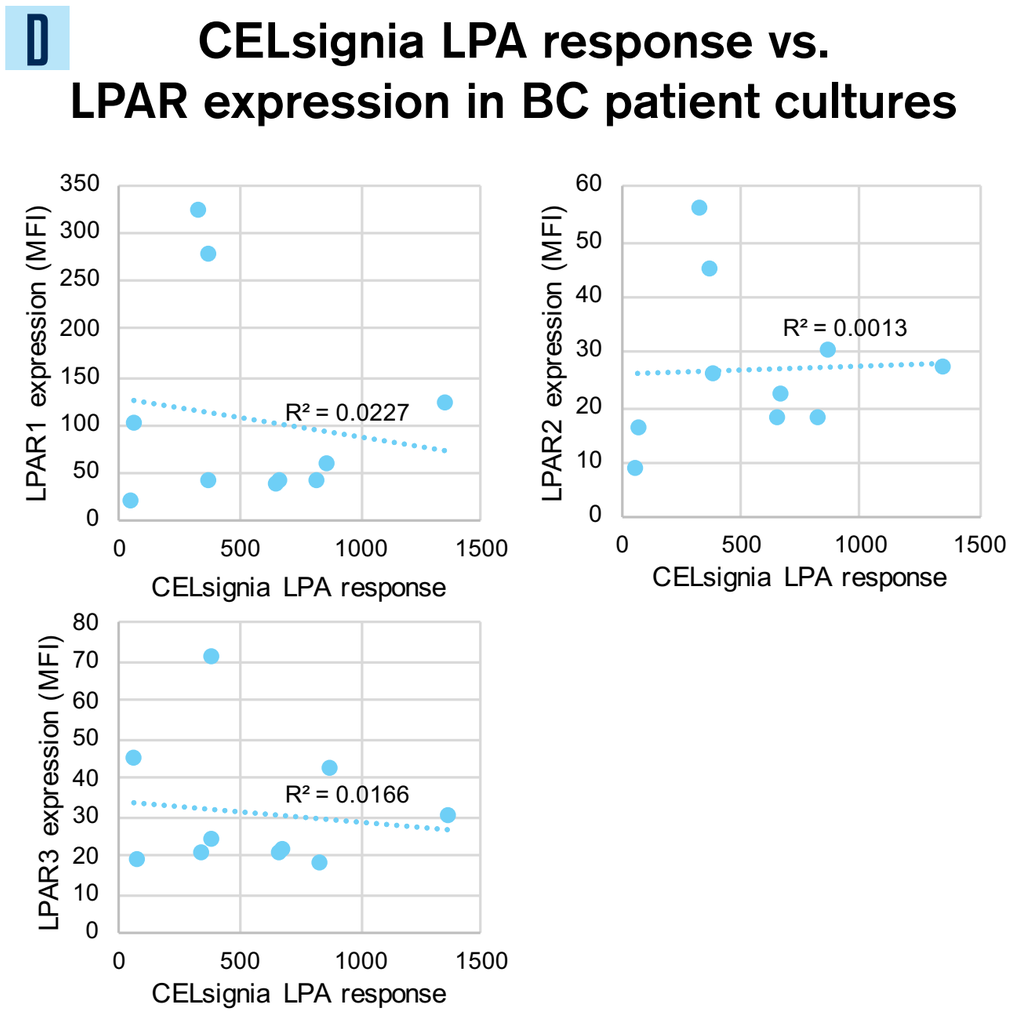

LPAR1 expression (MFI)
R² = 0.0227
LPAR2 expression (MFI)
R² = 0.0013
LPAR3 expression (MFI)
R² = 0.0166
CELsignia LPA response

(A) Table showing molecular characteristics, mutational status, LPA signaling activity and LPA receptors (LPAR1-3) expression in a panel of 20 BC cell lines. LPAR expression data show RPKM from the Cancer Cell Line Encyclopedia (CCLE). Conditional formatting was applied to all LPARs columns. NA = not available; mut = mutated; wt = wild type. **(B)** Scatter plots showing that lack of correlation between CELsignia LPA response and LPAR1, LPAR2 and LPAR3 expression. **(C)** Representative culture of BC patient cells from a digested tumor biopsy showing epithelial cells with a tight cobblestone structure (left). Representative FACS analysis of a disaggregated tissue and the resulting cultured cells showing that processing effectively isolates and expands the luminal tumor cells (EpCam+/CD49f-) for the CELsignia test (right). **(D)** Analysis of LPA receptor (LPAR) expression (FACS) and LPA signaling in 10 BC patient cultures showing no correlation. MFI = mean fluorescence intensity.

These results show that:

1. LPA signaling response in BC cells does not correlate with PIK3CA, PTEN, and P53 mutations or LPA receptor expression

Table 1: Patients screened with the CELsignia test

Characteristics	N. of patients	%	CELsignia score: Mean	SD	Above cutoff
Total patients	60	100%	147	168	12 (20%)
Age, years					
Mean	58.3	NA	NA	NA	NA
Range	27-94	NA	NA	NA	NA
Ethnicity					
African American	11	18%	118	142	2 (18%)
Caucasian	45	75%	147	166	9 (20%)
Other	4	7%	226	262	1 (25%)
Estrogen Receptor Status					
ER+	53	88%	156	173	11 (21%)
ER-	7	12%	80	104	1 (14%)

Characteristics	N. of patients	%	CELsignia score: Mean	SD	Above cutoff
Histology					
DCIS, ductal invasive	42	70%	148	174	9 (21%)
LCIS, lobular invasive	9	15%	114	90	1 (11%)
DCIS/LCIS, ductal/lobular inv.	7	12%	212	218	2 (29%)
DCIS/other	2	3%	35	12	0 (0%)
Lymph Status					
Positive	28	47%	162	181	7 (25%)
Negative	29	48%	132	162	4 (14%)
pNx	3	5%	147	120	1 (33%)

The table shows relevant clinical features of 60 HER2-negative BC patients screened with the CELsignia RAS test and used to estimate the CELsignia PI3K/mTOR cutoff in Figure 3. The CELsignia score mean and standard deviation (SD) for the different patient subgroups is calculated from the LPA (150nM) + Gedatolisib (150 nm) CELsignia scores. The % of patients with a CELsignia score above the 250 cutoff (see Figure 3) is indicated for each patient subgroup. There is no apparent correlation with LPA hypersignaling and patient characteristics.

Figure 3: Estimating PI3K/mTOR Test signal cutoff

Density curves 150 nM LPA + 150 nM Gedatolisib

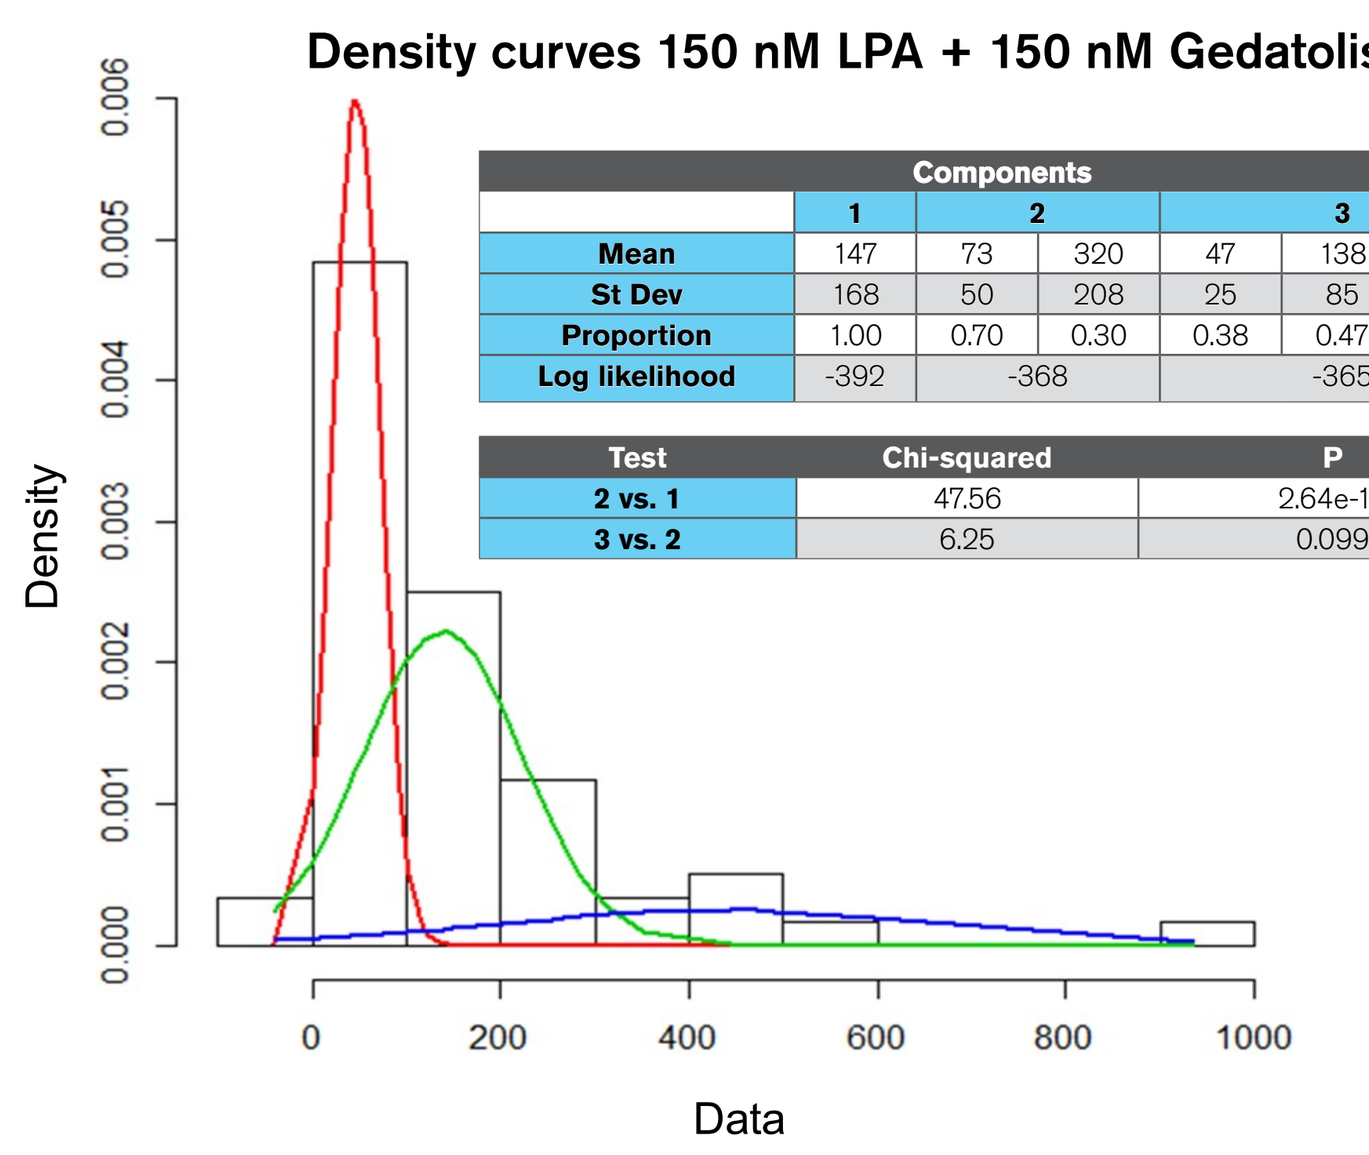

Density
Data

Components	1	2		3		
Mean	147	73	320	47	138	435
St Dev	168	50	208	25	85	234
Proportion	1.00	0.70	0.30	0.38	0.47	0.15
Log likelihood	-392	-368		-365		

Test	Chi-squared	P
2 vs. 1	47.56	2.64e-10
3 vs. 2	6.25	0.099

Sensitivity & FP cutoff analysis

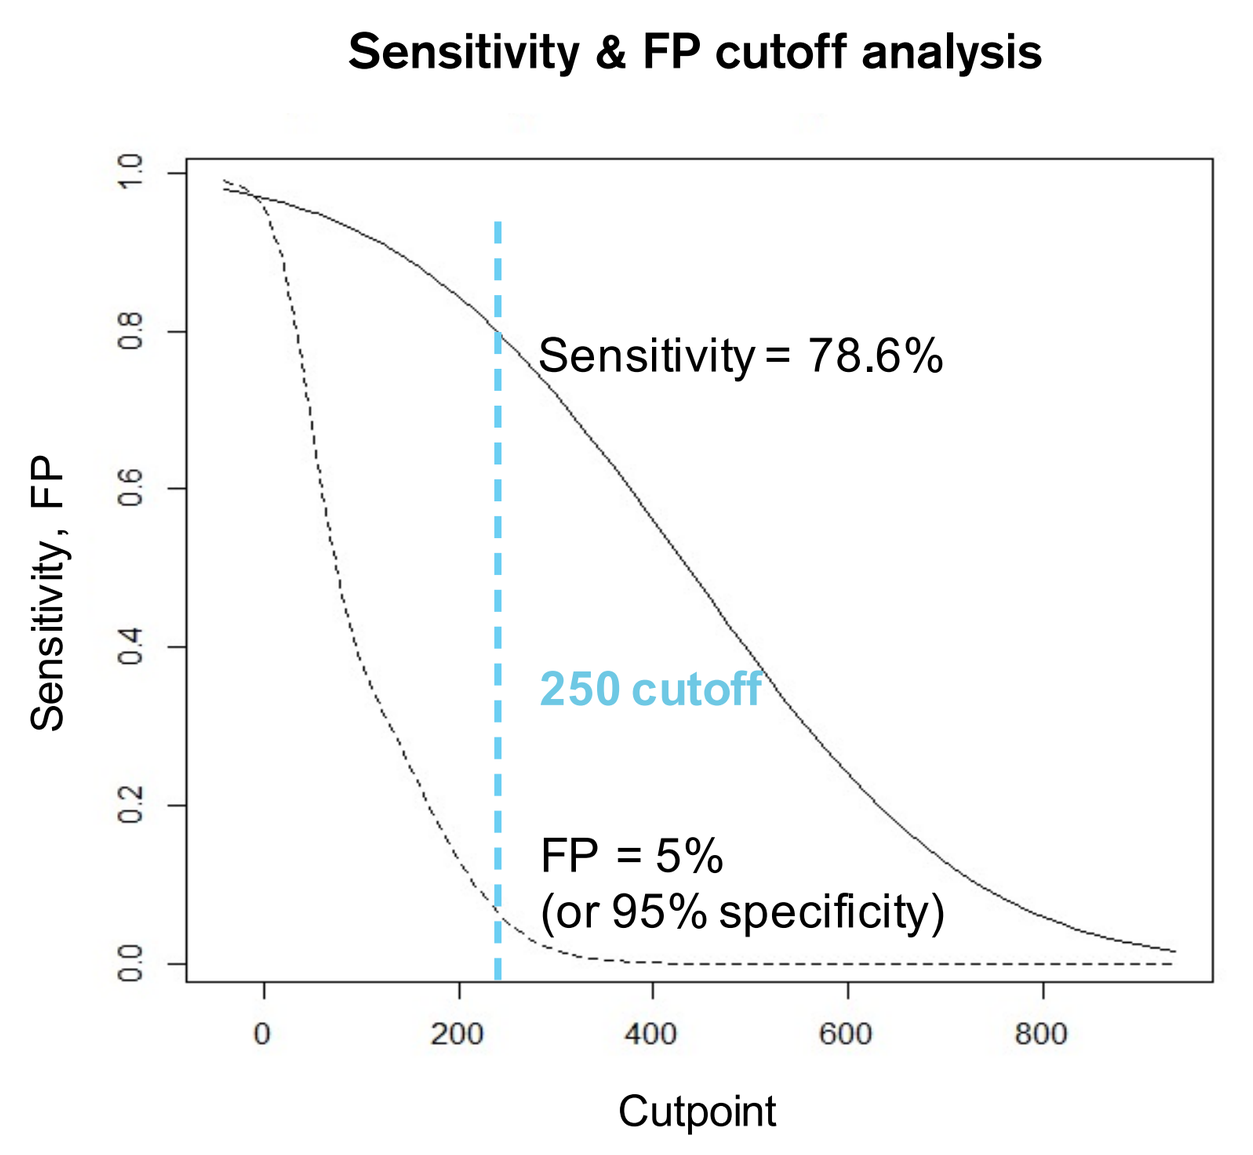

Sensitivity = 78.6%
250 cutoff
FP = 5%
(or 95% specificity)
Sensitivity, FP
Cutpoint

The charts and relative tables show the statistical analysis of the LPA CELsignia scores from 60 BC patient cultures.

These results show that:

1. A test score cutoff of 250 for PI3K/mTOR signaling activated by LPA can separate BC patients into two distinct populations (component 3 vs. components 1 & 2), where component 3 has abnormally high active PI3K/mTOR signaling

2. A test score cutoff of 250 has specificity >95% and sensitivity >78%

3. Using the 250 cutoff, 12/60 (20%) of patients in this random population have hyperactive PI3K/mTOR involved signaling

Figure 4: Hyperactive RAS signaling involves the PI3K, mTOR, & BCL nodes

A

Sample	LPA 150 nM	% LPA signaling inhibition: Gedatolisib 150 nM	GDC-0077 150 nM	% LPA signaling inhibition: Gedatolisib 30 nM	Gedato 30 nM + navitoclax 100 nM
BT-20	1042	94%	71%	66%	97%
C1441	1111	90%	4%	61%	106%
C3730	1075	87%	-10%	46%	80%
C3311	922	65%	2%	23%	40%
C1061	850	42%	25%	21%	50%
C1033	782	88%	6%	54%	97%
C1838	768	93%	8%	56%	103%
C3337	566	83%	25%	59%	81%
C2991	506	97%	11%	78%	100%
C1816	502	89%	-4%	17%	76%
C3377	468	54%	-34%	24%	77%
C3873	403	87%	30%	74%	92%
C1536	398	67%	16%	38%	70%
C1986	362	82%	17%	28%	83%
C1076	355	92%	16%	78%	101%
C3091	350	90%	17%	58%	75%
C1918	341	75%	11%	61%	82%
C4005	322	89%	-4%	67%	100%
C1518	312	70%	5%	48%	75%
C3101	281	81%	0%	53%	95%
C2169	269	99%	13%	90%	109%
C2483	262	77%	43%	50%	29%

BC cell line with PIK3CA mutations (P539R and H1047R)

21 patient BC cultures with hyperactive RAS signal

Gedatolisib (150 nM) vs. GDC-0077 (150 nM)
- Geda (150 nM) average LPA attenuation = 81%
- GDC-0077 (150 nM) average LPA attenuation = 9%

Gedatolisib (30 nM) vs. GDC0077 (150 nM)
- Geda (30 nM) average LPA attenuation = 52%
- GDC-0077 (150 nM) average LPA attenuation = 9%

Geda (30 nM) + navitoclax (100 nM) vs. Geda (30 nM)
- Geda + navitoclax average LPA attenuation = 86%
- Geda (30 nM) average LPA attenuation = 52%

B BT20

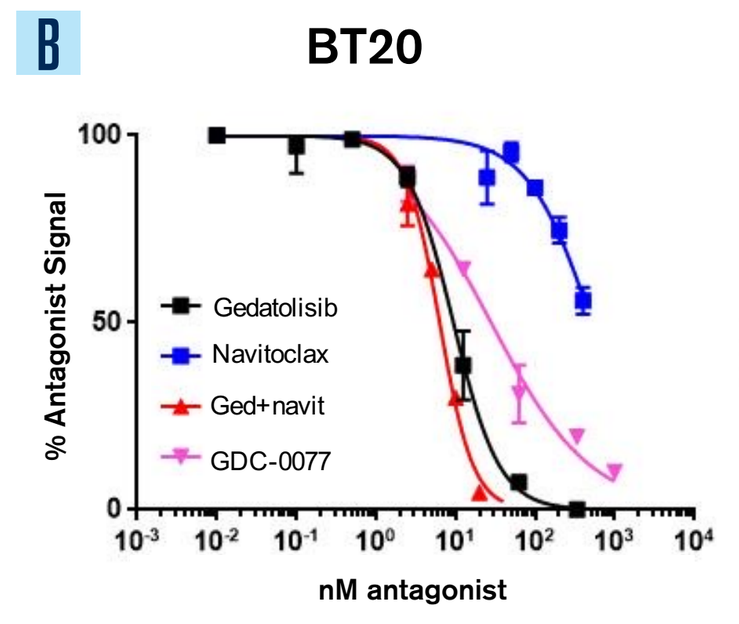

% Antagonist Signal
Gedatolisib
Navitoclax
Ged+navit
GDC-0077
nM antagonist

	gedatolisib	navitoclax	Gedato+navito	GDC-0077
IC50	9.4	520.5	6.3	64.2

C BT20

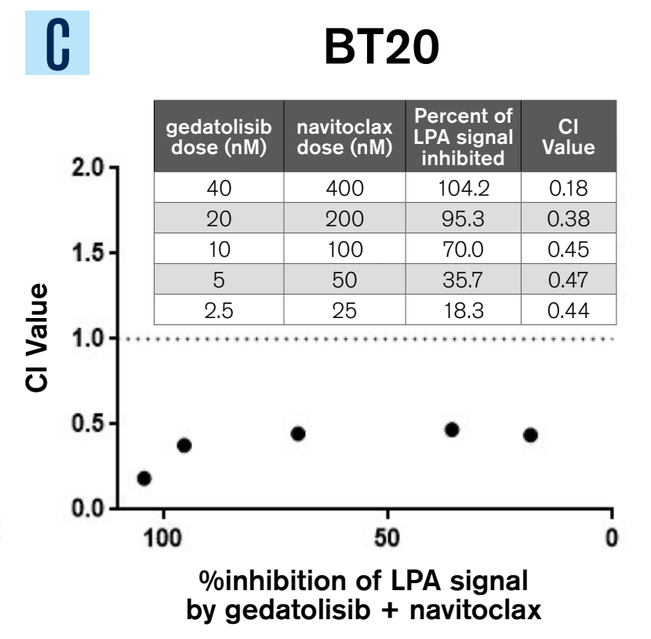

CI Value
%inhibition of LPA signal by gedatolisib + navitoclax

gedatolisib dose (nM)	navitoclax dose (nM)	Percent of LPA signal inhibited	CI Value
40	400	104.2	0.18
20	200	95.3	0.38
10	100	70.0	0.45
5	50	35.7	0.47
2.5	25	18.3	0.44

D C1441

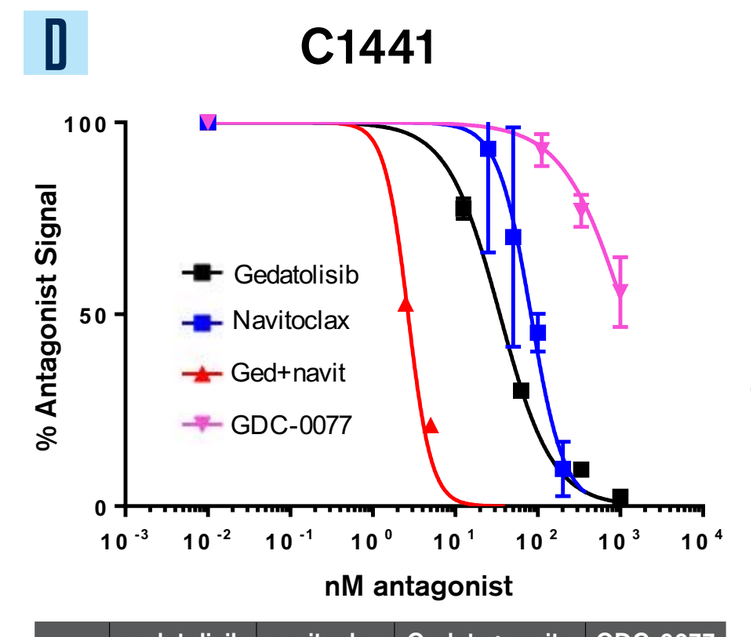

% Antagonist Signal
Gedatolisib
Navitoclax
Ged+navit
GDC-0077
nM antagonist

	gedatolisib	navitoclax	Gedato+navito	GDC-0077
IC50	34.5	82.0	2.7	1159

E C1441

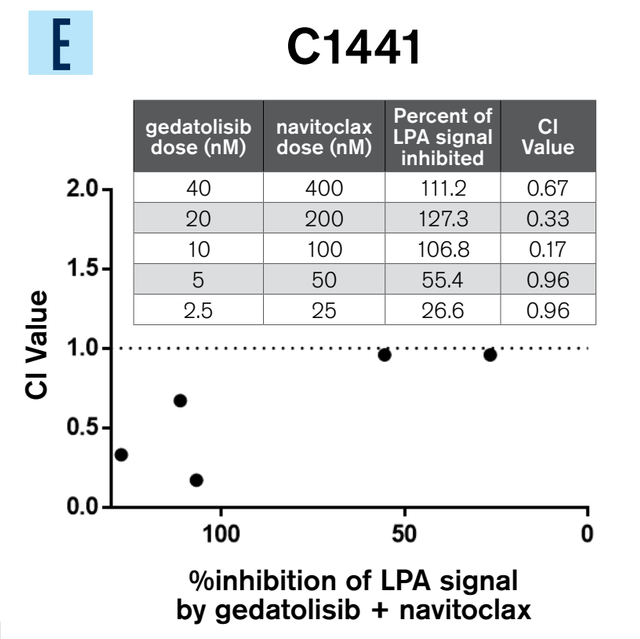

CI Value
%inhibition of LPA signal by gedatolisib + navitoclax

gedatolisib dose (nM)	navitoclax dose (nM)	Percent of LPA signal inhibited	CI Value
40	400	111.2	0.67
20	200	127.3	0.33
10	100	106.8	0.17
5	50	55.4	0.96
2.5	25	26.6	0.96

(A) Table showing the response of 21 no/low passage BC patient cultures with hyperactive RAS signaling to a PI3K-α inhibitor (GDC-0077), a pan-PI3K/mTOR inhibitor (gedatolisib) and gedatolisib with a BCL inhibitor (navitoclax). Two concentrations of gedatolisib were selected to isolate and measure the pan-PI3K/mTOR signal, as well as measure the effect of combining with BCL inhibitor, navitoclax. **(B)** Dose response curves for gedatolisib, GDC-0077, navitoclax and gedatolisib + navitoclax (at a 10-fold ratio to gedatolisib) in BT-20 cells. **(C)** Chou-Talalay (C&T) synergy analysis of combinations of gedatolisib and navitoclax in BT-20, a PI3K-α mutant. **(D)** Dose response curves and **(E)** C&T synergy analysis of combinations of gedatolisib and navitoclax in a BC patient cell sample. C&T Combination Index (CI) values less than 1 are indicative of synergy.

These results suggest that:

1. RAS hypersignaling initiated by LPA primarily involves the PI3K and mTOR nodes

2. Attenuation of LPA hypersignaling with a PI3K-α inhibitor was 9-fold less effective than a PI3K/mTOR inhibitor

3. RAS hypersignaling may involve synergistic cooperation between BCL and the PI3K and mTOR nodes

Figure 5: The effects of PI3K, mTOR, and BCL inhibitors on CELsignia RASs+ tumors correlate with changes in cell physiology markers

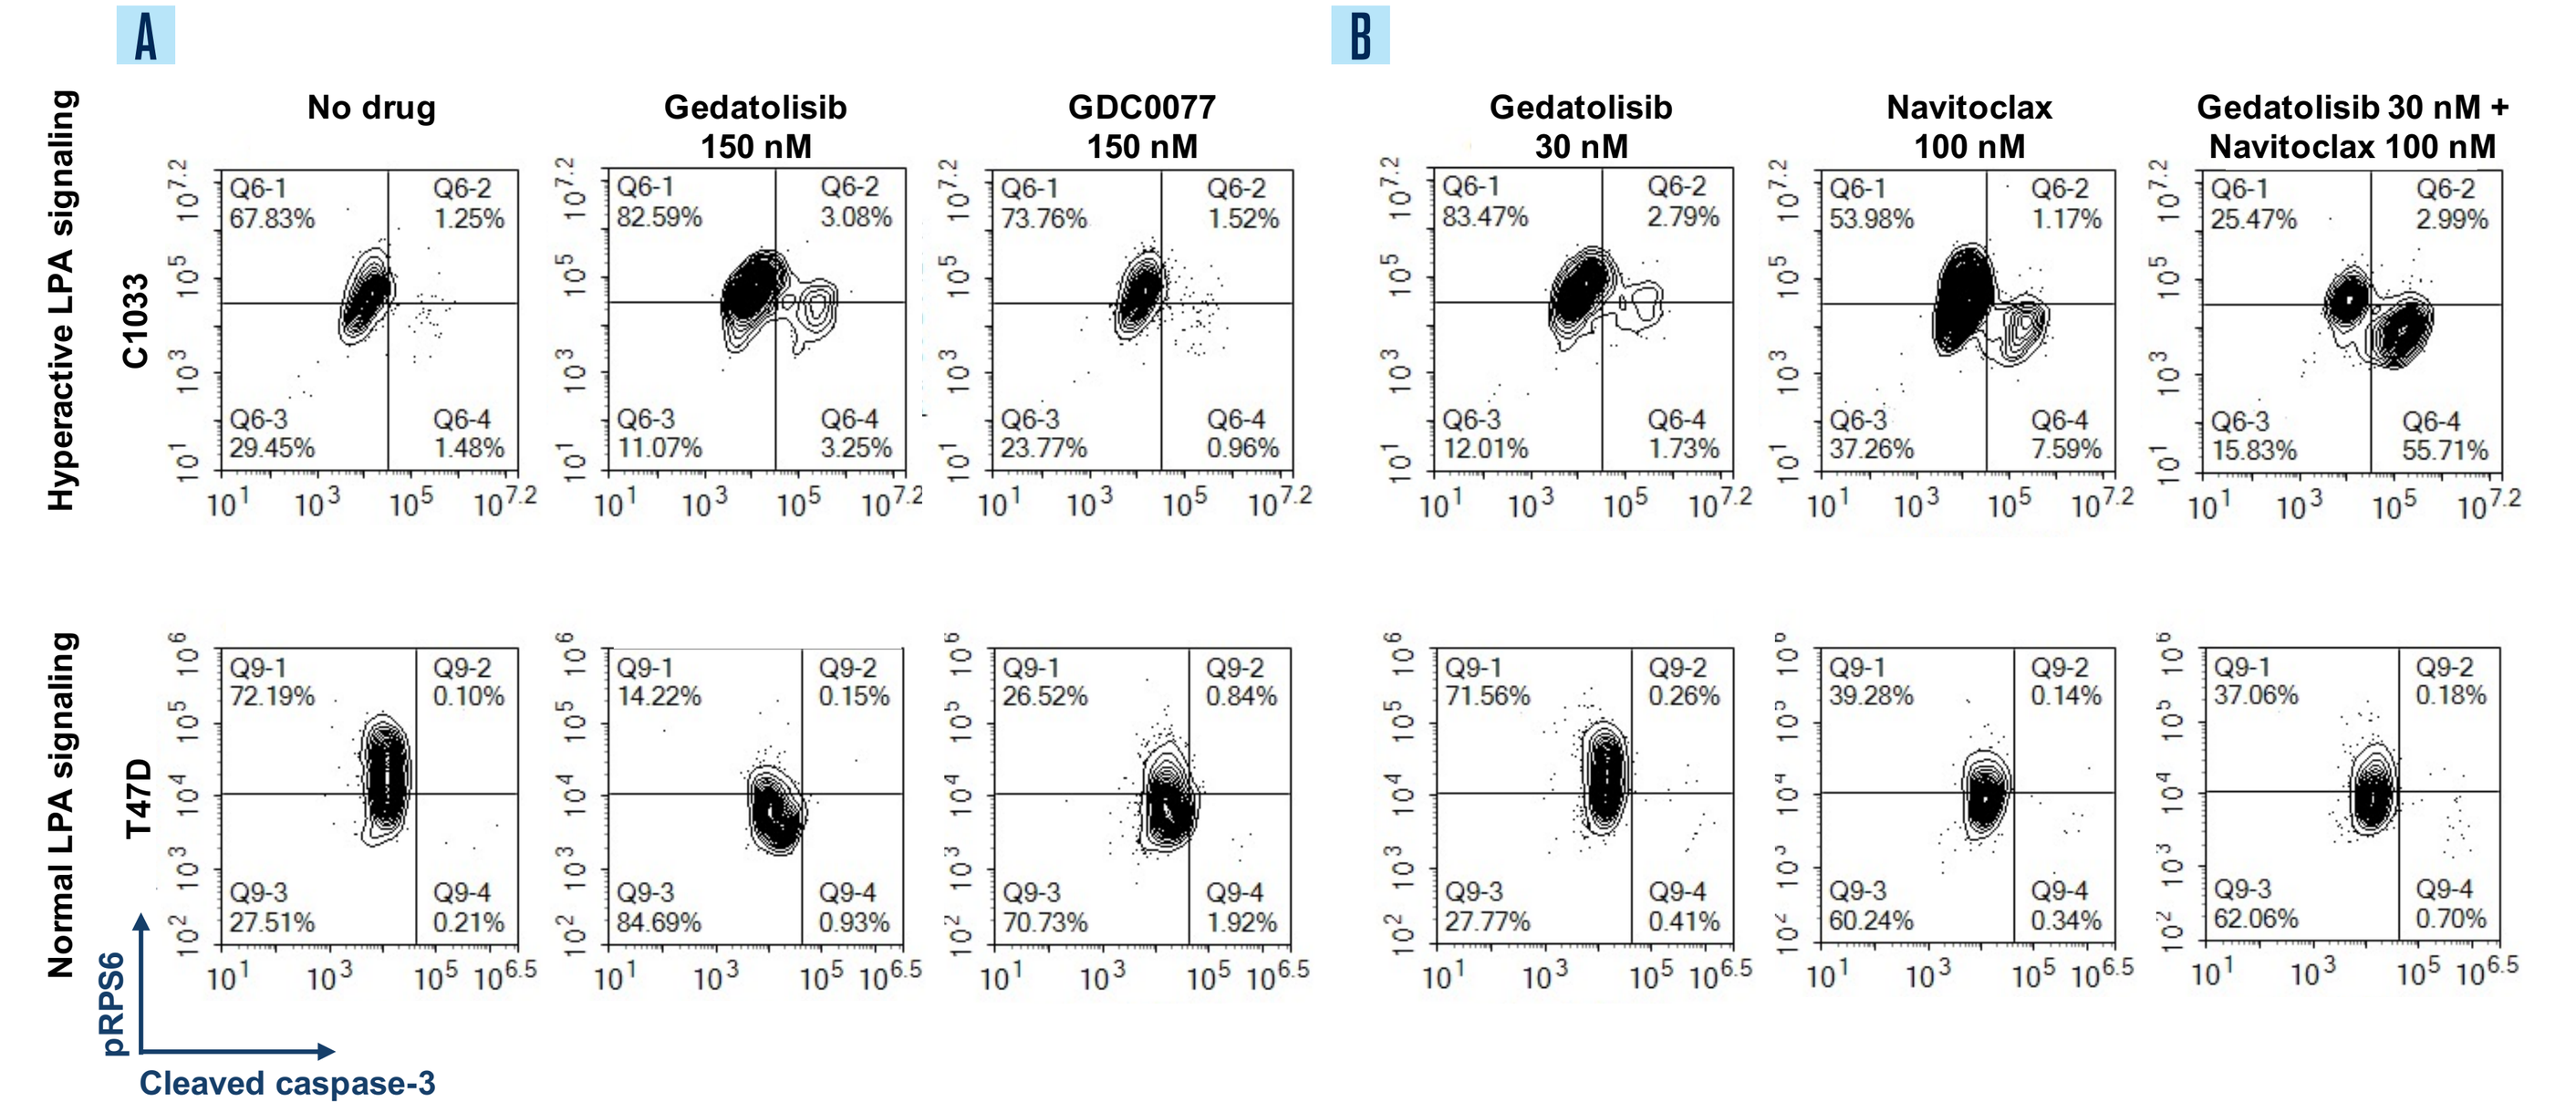

A
B
No drug
Gedatolisib 150 nM
GDC0077 150 nM
Gedatolisib 30 nM
Navitoclax 100 nM
Gedatolisib 30 nM + Navitoclax 100 nM
Hyperactive LPA signaling
C1033
Normal LPA signaling
T47D
pRPS6
Cleaved caspase-3

FACS analysis of apoptosis (assessed by cleaved caspase 3) and mTOR activation (assessed by pRPS6) in these BC patient cells treated with **(A)** either gedatolisib or GDC0077 or **(B)** gedatolisib, navitoclax, and gedatolisib + navitoclax.

The results with these markers for these BC patient cells correlate with the CELsignia LPA response in C1033, which had hyperactive LPA signaling, when assessed with gedatolisib (150 nM) or the combination of gedatolisib (30nM) + navitoclax (100 nM) (see CELsignia response to these drugs in Figure 4A). The T47D cell line, which has a PIK3-α mutation and had normal CELsignia LPA signaling (response to LPA 150 nM = 58), did not show induction of apoptosis with these drugs.

These results provide evidence that hyperactive RAS signaling detected by the CELsignia RAS test is oncogenic.

1. Consistent with the CELsignia analysis, inhibiting PI3K/mTOR with gedatolisib induces more apoptosis than inhibiting PI3K-α with GDC-0077

2. The greater level of apoptosis induced when BCL and PI3K/mTOR inhibitors are combined is consistent with the synergy found between BCL and PI3K/mTOR signaling by the CELsignia analysis

Summary of Results

- The CELsignia RAS Activity Test identified patients with hyperactive RAS signaling regardless of LPAR expression of mutational status of RAS signaling-related genes
- The CELsignia RAS Activity Test identified 12 of 60 (20%) BC patients with hyperactive RAS signaling
- Hyperactive RAS signaling is always more effectively inhibited with a PI3K/mTOR inhibitor (gedatolisib) than a PI3K-α inhibitor (GDC-0077)
- Synergistic cooperation between BCL and PI3K/mTOR was detected, suggesting that addition of a BCL inhibitor to a PI3K/mTOR inhibitor may induce a greater anti-tumor effect
- The attenuation of hyperactive RAS signaling by gedatolisib or gedatolisib + navitoclax correlates with reduced mTOR signaling, and induction of apoptosis

Conclusions

These findings suggest that a significant subgroup of BC patients have a RAS-involved oncogenic signaling driver that is responsive *ex vivo* to pan-PI3K/mTOR and pan-PI3K/mTOR/BCL inhibitors. A clinical trial to evaluate treatment response of this patient subgroup to combined PI3K/mTOR or PI3K/mTOR/BCL inhibitors is warranted.

References

1. Bar-Shavit, et al. G protein-coupled receptors in cancer. *Int J Mol Sci.* 2016;17(8):1320.
2. Predescu, et al. G protein-coupled receptors (GPCRs)-mediated calcium signaling in ovarian cancer. *Int J Mol Sci.* 2019;20(22):5568.
3. Rubio, et al. RAS activation in response to lysophosphatidic acid requires a permissive input from the epidermal growth factor receptor. *Biochem J.* 2003;376(Pt 3):571-576.
4. Downward. Role of receptor tyrosine kinases in G-protein-coupled receptor regulation of RAS: transactivation or parallel pathways? *Biochem J.* 2003;376(Pt 3):e9-10.
5. Rodriguez-Viciana, et al. Activation of phosphoinositide 3-kinase by interaction with RAS and by point mutation. *EMBO.* 1996;15(10):2442-2451.
6. Herrlich, et al. Ligand-independent activation of platelet-derived growth factor receptor is a necessary intermediate in lysophosphatidic, acid-stimulated mitogenic activity in L cells. *PNAS.* 1998;95(15): 8985-8990.
7. Hu, et al. Lysophosphatidic acid induction of vascular endothelial growth factor expression in human ovarian cancer cells. *J Natl Cancer Inst.* 2001;93(10):762-768.
8. Fang, et al. Mechanisms for lysophosphatidic acid-induced cytokine production in ovarian cancer cells. *J Biol Chem.* 2004;279(10):9653-9661.
9. Huang, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016;7(48):78577-78590.
10. Huang, et al. Development of a test that measures real-time HER2 signaling function in live breast cancer cell lines and primary cells. *BMC Cancer.* 2017;17:199.

Abstract #: 342

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.