CELx™ | breast

HER2 Signaling Function Test

Sub-group of HER2-negative breast cancer patients with hyperactive and co-involved c-Met and HER (ErbB) pathways identified: functional signaling profiling test identifies patient group that may benefit from c-Met and pan-HER combination therapy

Laing LG, Burns DJ, Khan SE, Kharbush S, Soltani SM, MacNeil IA, Rich BE, Brass K, Broege AM, Mutka S, Sullivan BF – Celcuity Inc., 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 4–8, 2018

celcuity™
www.celcuity.com

Background

Biological factors other than c-Met status, such as c-Met and HER family signaling activity, may be important to measure when identifying patients eligible for c-Met therapies. A new assay using an impedance biosensor was developed to measure c-Met and HER family signaling activity in live tumor cells. The CELx multi-pathway signaling function (CELx MP) test measures an individual patient's *ex vivo* live tumor cell response in real-time to specific HER and c-Met agonists and antagonists to diagnose breast tumors with hyperactive HER1, HER2, HER3, and c-Met signaling.

HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15–20% of breast cancers and is associated with more aggressive disease progression, metastasis, and a poorer prognosis.[1-4] Agents targeting HER2, such as trastuzumab, lapatinib, and pertuzumab, significantly improve clinical outcomes in HER2$^+$ patients.[4,5] Currently, a patient's eligibility for HER2 targeted therapies is determined using IHC or FISH HER2 tests.[4] However, clinical trials have indicated a weak correlation between HER2 expression or amplification levels and HER2 targeted therapy benefit.[6,7]

c-Met is the cognate receptor for Hepatocyte Growth Factor (HGF). Co-involvement of c-Met and HER family signaling in cancer progression is well-established. MET amplification and HGF overexpression have emerged as mechanisms by which cancers become resistant to HER family therapies.[8,9] HER family and c-Met receptors form heterodimers that activate downstream signaling pathways that drive cancer progression.[10-12] Phase II trials with c-Met targeted therapies, either cabozantinib or onartuzumab, in combination with erlotinib, showed promising improvement in PFS.[13] However, Phase III clinical trials studying c-Met targeted therapies using MET overexpression as an enrollment criteria and studies in which patients were randomly selected to receive combination c-Met/HER1 treatment failed to demonstrate clinical efficacy.[13] Because c-Met expression level using IHC as a clinical pathology diagnostic marker fails to identify a population responsive to c-Met targeted therapies, an alternative approach is required to identify patients with dysfunctional c-Met signaling who will respond to these therapies.

We previously developed a test to identify patients with abnormal HER2 signaling.[14,15] To elucidate the role of c-Met signaling and its involvement with HER family signaling as a cancer driver, a new assay using an impedance biosensor and live cells derived from each patient's tumor, the CELx MP test, was developed.

The CELx MP test measures *ex vivo* real-time live cell response to specific HER family and c-Met agonists to diagnose breast tumors with hyperactive HER1, HER2, HER3, HER4, and c-Met signaling activity. A recent study quantified c-Met and HER-driven signaling activity in epithelial cell samples derived from fresh breast tumor specimens obtained from 79 HER2-negative breast cancer patients. Of the cell samples tested, 19 of 79, (24.1%; 95% CI=16–35%) had both hyperactive c-Met signaling and at least one hyperactive HER-family receptor signal.

This study provides an analytical validation of the CELx test, specifically to accomplish the following:

1. Determine the prevalence of hyperactive c-Met and HER family signaling among HER2-negative breast cancer patients.
2. Characterize potential cross-talk between c-Met and HER pathways.

Methods

Specimens: A training set of de-identified fresh breast tissue specimens was obtained from 79 patients diagnosed with HER2-negative breast cancer. See Summary of 79 HER2-negative tumor patient characteristics in Table 1.

Cell Culture: Methods for tissue extraction and primary cell culture* are essentially as described previously.[16,17]

Flow Cytometry: Flow cytometry of all cell samples was performed on a BD FACSCalibur using cells harvested at the time of the CELx MP test. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2 that were provided for each specimen by the clinic.

CELx MP Test: Real-time live cell responses to specific HER3 and HER1 agonists (NRG1b and EGF) or c-Met agonist (HGF) with or without antagonists were measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, the net amount of HER2 participation in HER-family signaling initiated by HER receptor agonists was determined.[15,16] Samples with HER2 signaling activity levels above a previously determined cut-off value of 250 signaling units that were attenuated with a HER2 dimer blocker were identified as abnormal. Samples with c-Met signaling activity above 250 were identified as abnormal.

Statistical Analyses: A data set of 79 CELx MP test scores from HER2-negative patients collected between July 2013 and March 2017 and tested in 2017 was analyzed. A normal mixture model was fitted to the combined data set using the normalmixEM procedure in the R package mixtools. Two runs of the statistical analysis were made, fitting 2 and 3 components, along with a baseline single-component model.

Figure 1. Platform Biosensor Sensitivity Enables Quantification of HER and c-Met Signaling Real-Time in Live Cells

HER and c-Met ligands and inhibitors used to turn on/off HER and c-Met pathways

Very subtle, sub-nanometer, cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Determine Signaling Score for each pathway tested

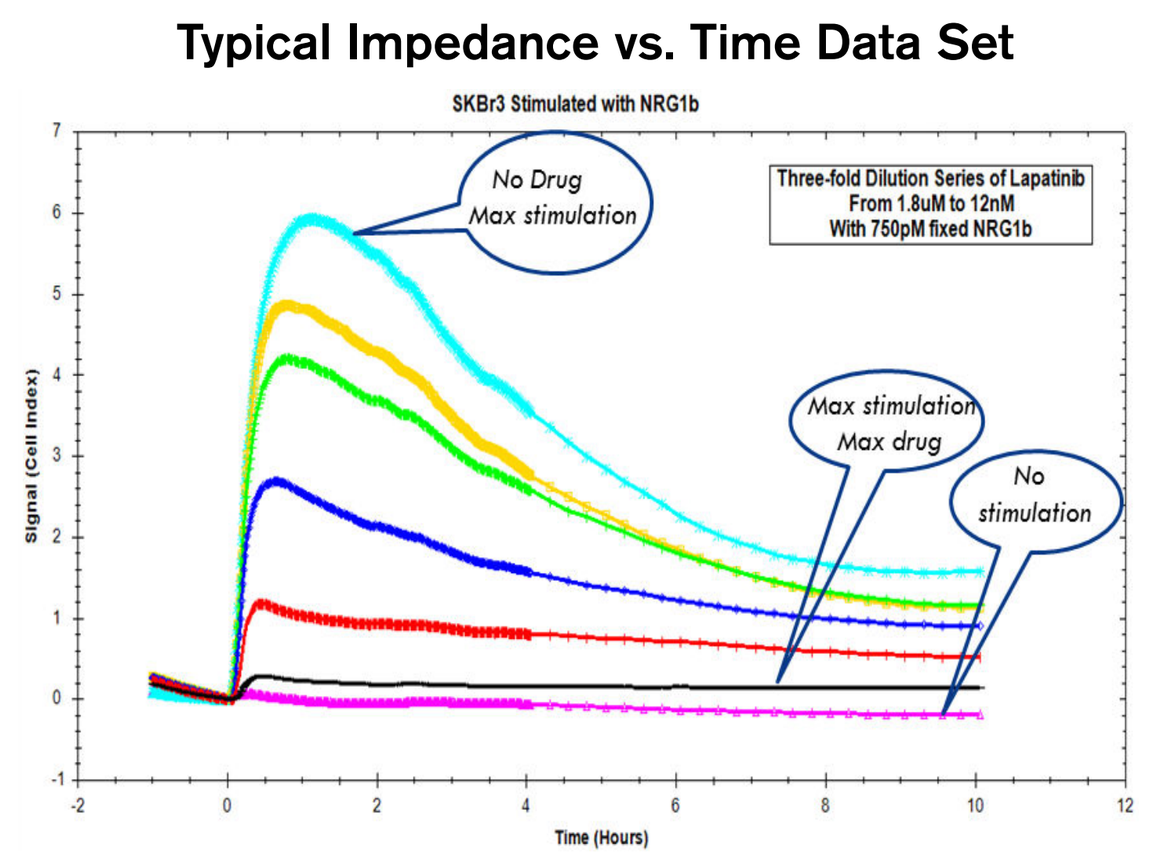
Typical Impedance vs. Time Data Set
SKBr3 Stimulated with NRG1b
No Drug Max stimulation
Three-fold Dilution Series of Lapatinib From 1.8uM to 12nM With 750pM fixed NRG1b
Max stimulation Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

* De-identified excess surgically resected human breast cancer tissue was received from multiple clinical sites located across the United States. Liberty IRB (Columbia, MD) determined that this research did not involve human subjects as defined under 45 CFR 46.102(f) and granted an IRB exemption. Liberty IRB has full accreditation with the Association for the Accreditation of Human Research Protection Programs (AAHRPP).

Figure 2. HER2⁻ Abnormal Signaling by CELx HSF Test

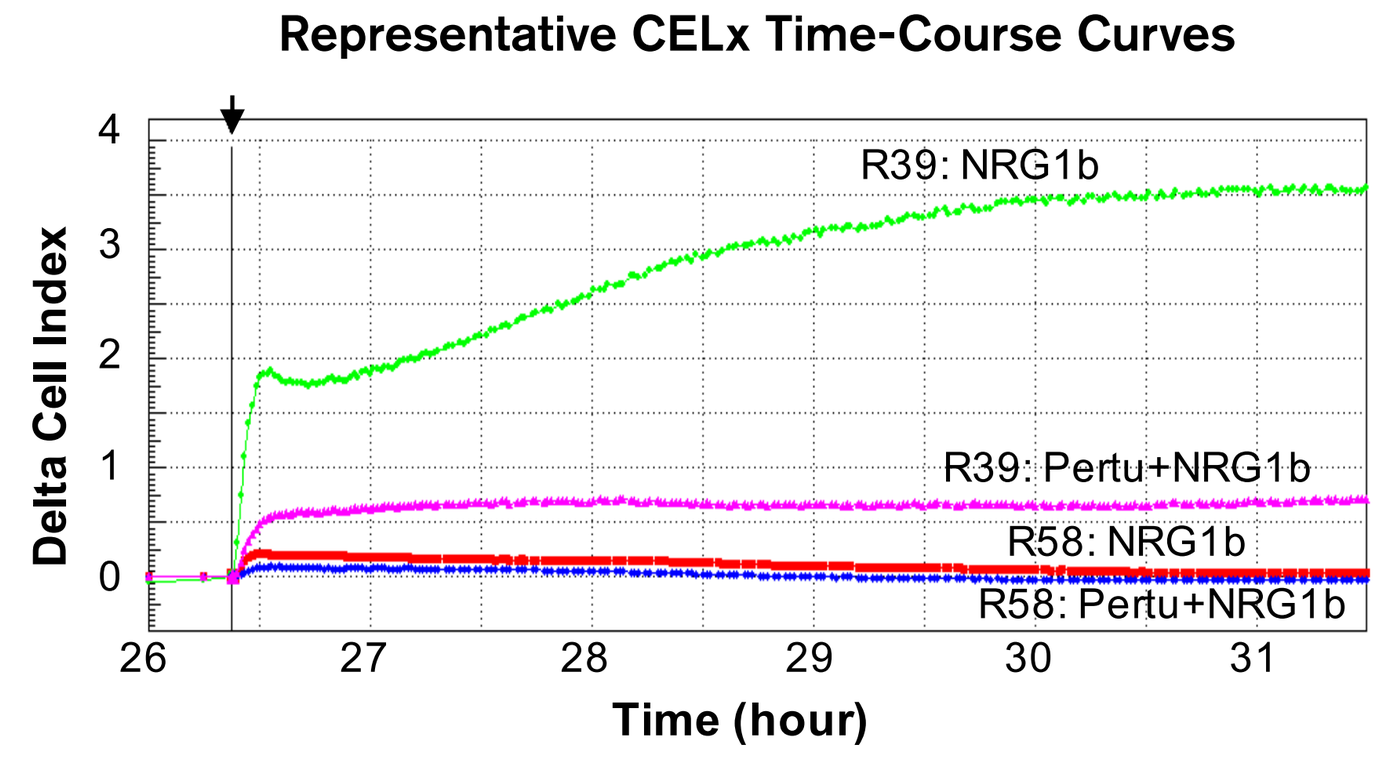
Representative CELx Time-Course Curves
R39: NRG1b
R39: Pertu+NRG1b
R58: NRG1b
R58: Pertu+NRG1b
Delta Cell Index
Time (hour)

CELx time-course curves representing a high, abnormal HER2 signal in a high responder (R39) and a low HER2 signal in a non-responder (R58). In this display, curves of NRG1 stimulation in the absence versus presence of HER2 dimer blocker (10μg/mL) are presented.

The data show that the high NRG1b responder has more than 10 times greater signal than the low responder, indicating the test has a large dynamic range.

Results

Fitting c-MET Cutoff Determination and Prevalence Data From 79-Patient Test Set

A first step was to test the current data set for compatibility with an earlier collection of 114 patients. This was done using the Kolmogorov-Smirnov two-sample test for identity of the distributions with respect to HER2 test scores. The test statistic obtained was D=0.10256, with a P-value of 0.7146, indicating that there is no significant difference between the CELx HSF scores of these two groups.

Figure 3A

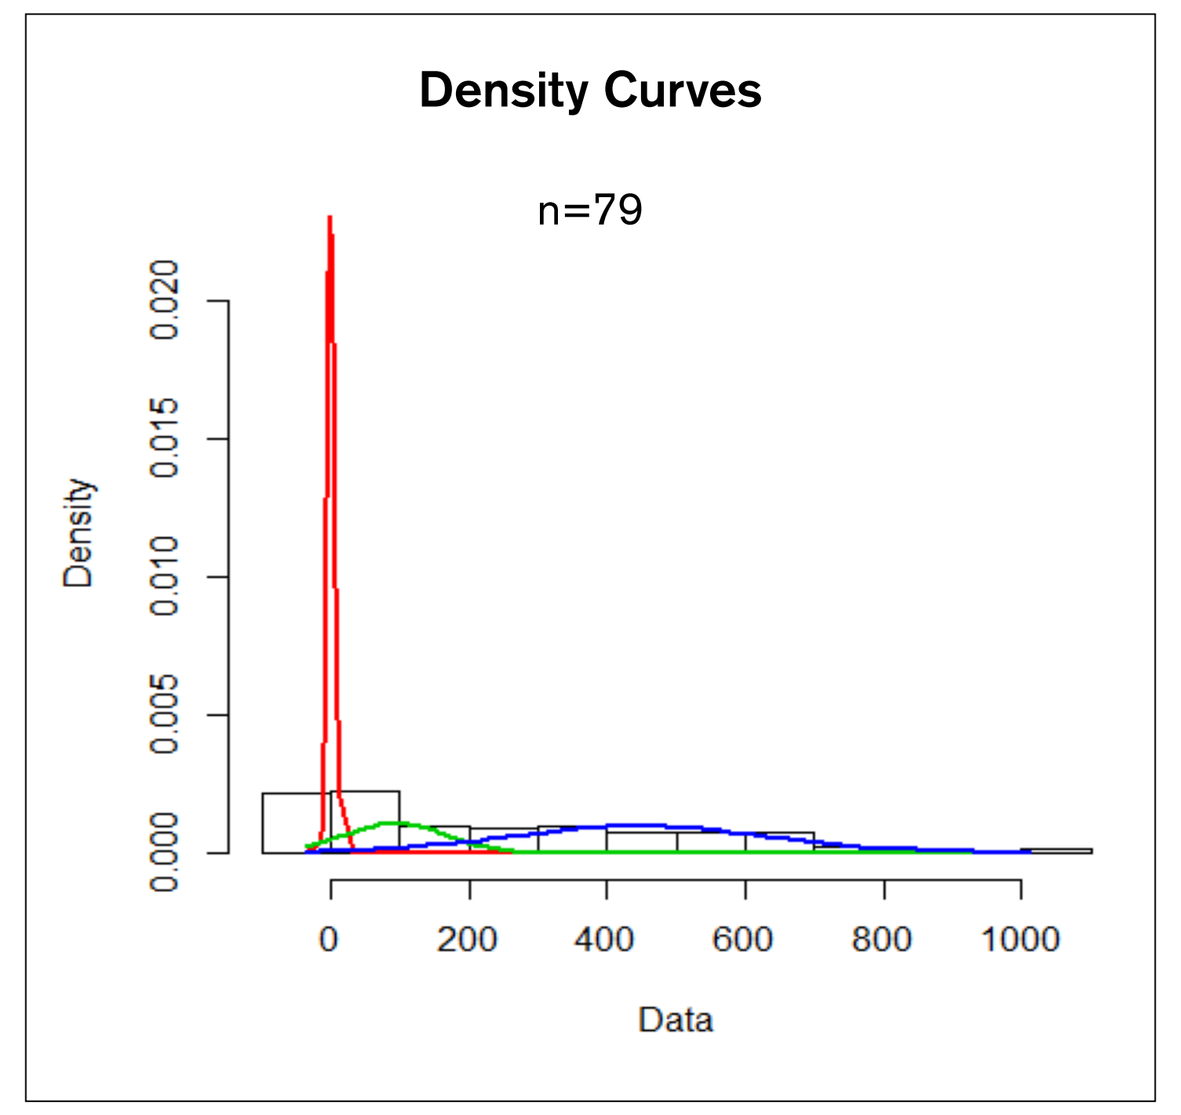
Density Curves
n=79
Density
Data

	Components				
	2		3		
Mean	-0.13	345.00	-0.06	93.70	446.30
StDev	5.70	231.70	5.50	74.20	195.40
Proportion	0.32	0.68	0.32	0.20	0.48

The likelihood ratio test for the number of components gave the following parameters: For the 3 component model, the components 2 (middle) and 3 (right) means are more than 4 standard deviations apart.

Figure 3B

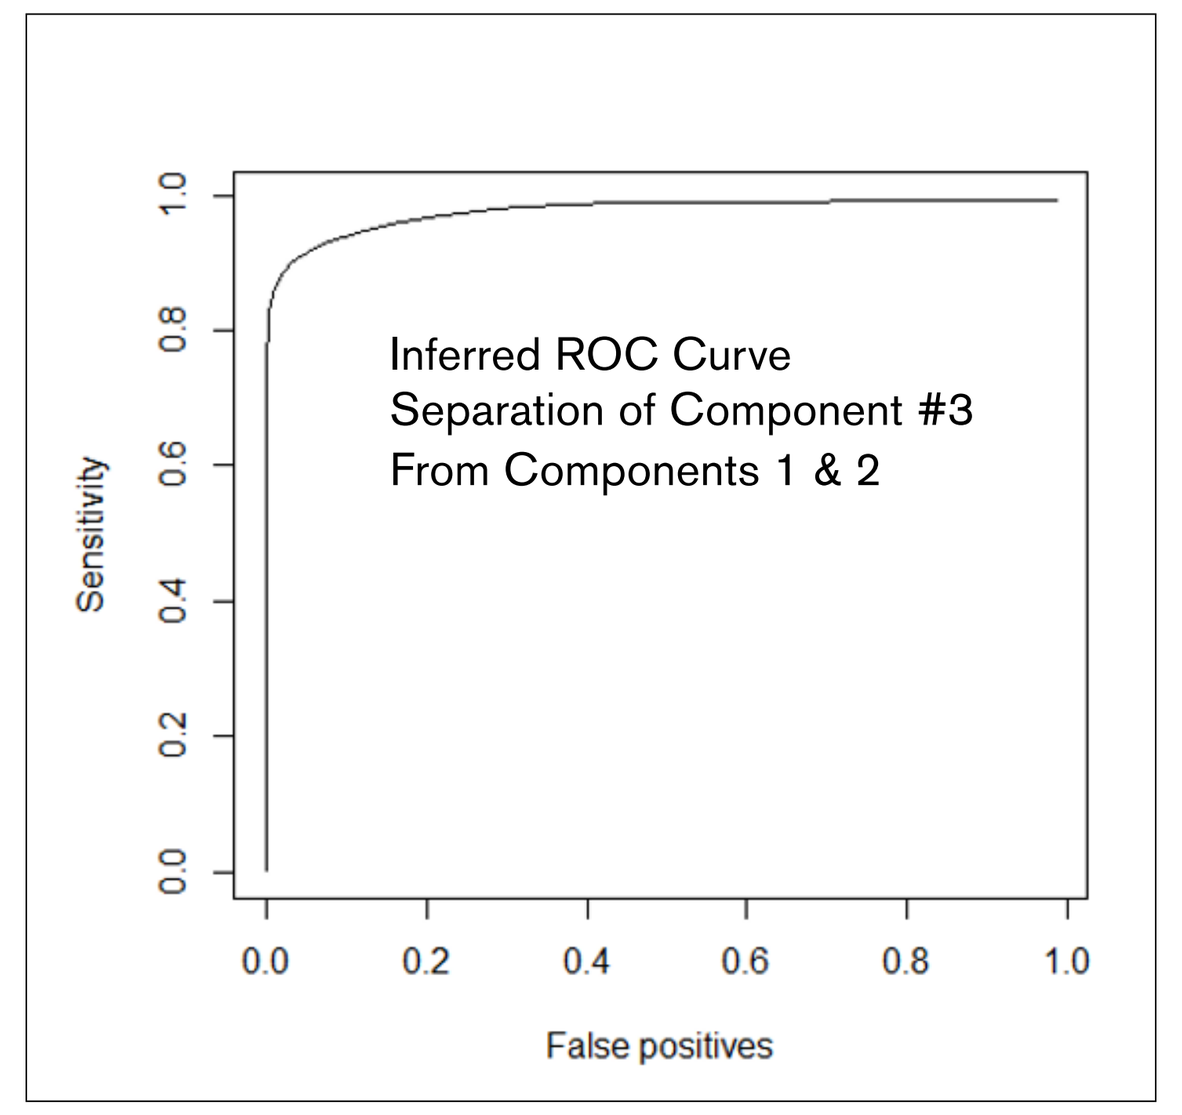
Inferred ROC Curve
Separation of Component #3
From Components 1 & 2
Sensitivity
False positives

Test	Chi-Squared	P
2 vs. 1	110.98	6.80E-24
3 vs. 2	8.19	0.04

Formal significance testing shows that a two-component mixture fits much better than a common normal distribution, and a three-component mixture fits better than two.

Figure 3C

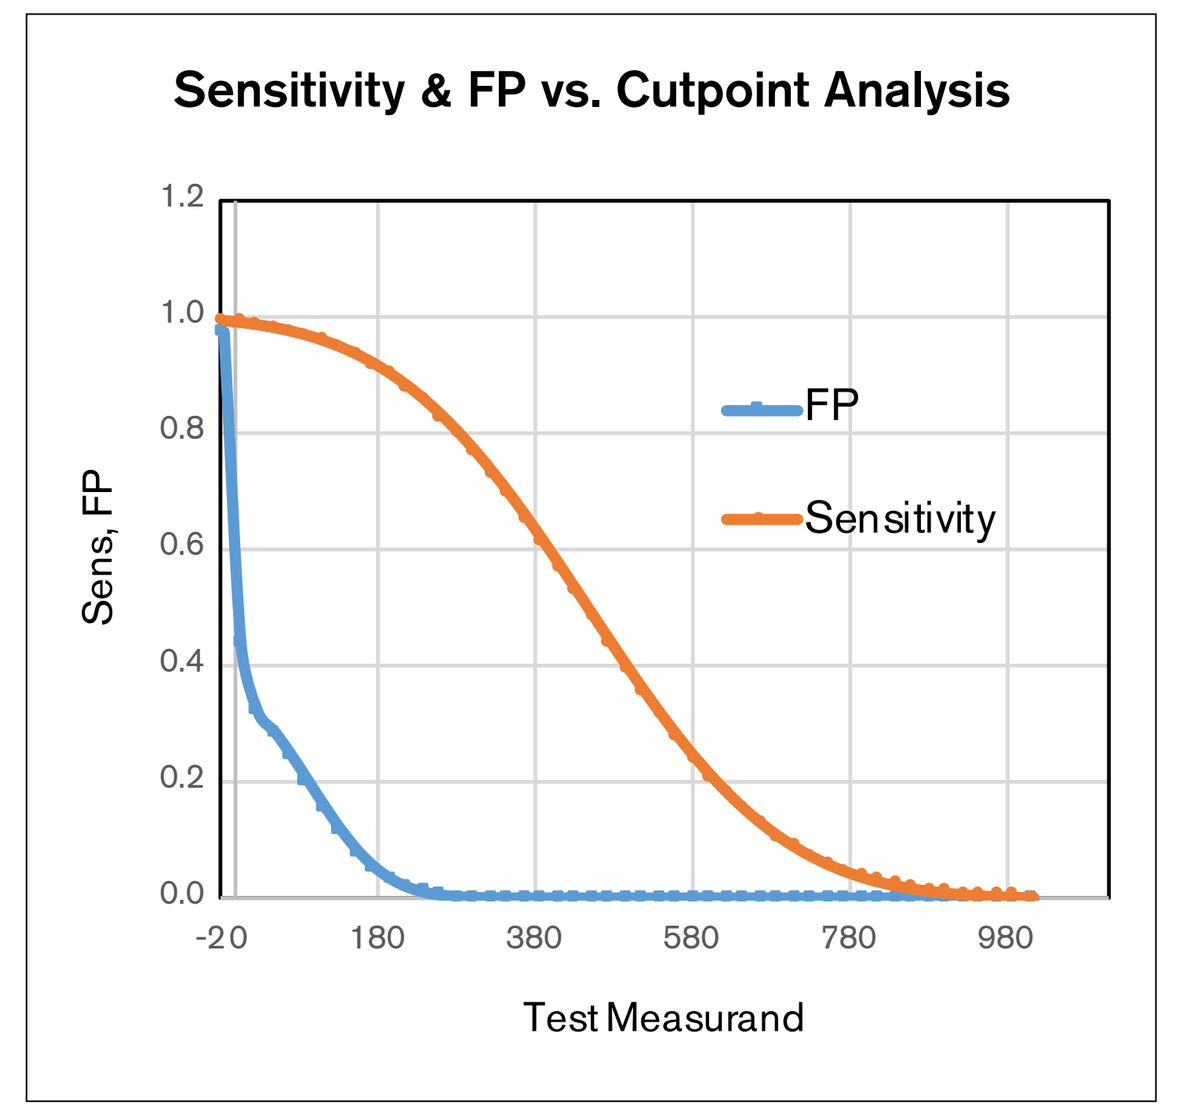
Sensitivity & FP vs. Cutpoint Analysis
FP
Sensitivity
Sens, FP
Test Measurand

The graph above displays the numbers underlying the ROC plot: the sensitivity (orange line) and false positives (blue line) as a function of the cutoff used. At 250 signaling units, specificity is >99% (FP<1%) and sensitivity is 84%, indicative of an accurate test.

Table 1. Summary of Patient Characteristics

Characteristic	Number of Patients	Percentage (%)
Total Patients	79	100%
Age, Years		
Mean	56.4	
Range	34-87	
Clinical Stage		
I	12	15%
II	49	62%
III	15	19%
IV	3	4%
Histology		
DCIS only	1	1%
Invasive only	14	18%
Invasive ductal/DCIS mixed	45	57%
Lobular/other	19	24%
Lymph Status		
Positive	40	46%
Negative	36	51%
pNX or N/A	3	4%
Estrogen Receptor Status		
ER+	69	87%
ER-	10	13%

CELx MP tests were performed on 79 primary tumor cell samples from patients with breast cancer classified as HER2-negative to measure HER family pathway and c-Met pathway stimulation and signal specificity.

For comparative purposes, the DAKO IHC test standard breast cancer cell line SKBr3 was also analyzed with the CELx MP test.

The CELx MP test identified 19 of 79 HER2-negative patient samples (24.1%; 95% CI=16–32%) with both hyperactive c-Met signaling and at least one hyperactive signaling HER-family receptor.

Table 1. Patient characteristics of a random set of tumors collected from clinical cancer patients with a distribution of stage, histology, and age to study a new test for identifying pathway dysfunction in HER2-negative breast cancer.

Results

Figure 4. Combinations of Pan-HER and c-MET Inhibitors Effectively Block NRGb1/EGF/HGF Agonist Cocktail in Real-Time Live Cell CELx MP Test

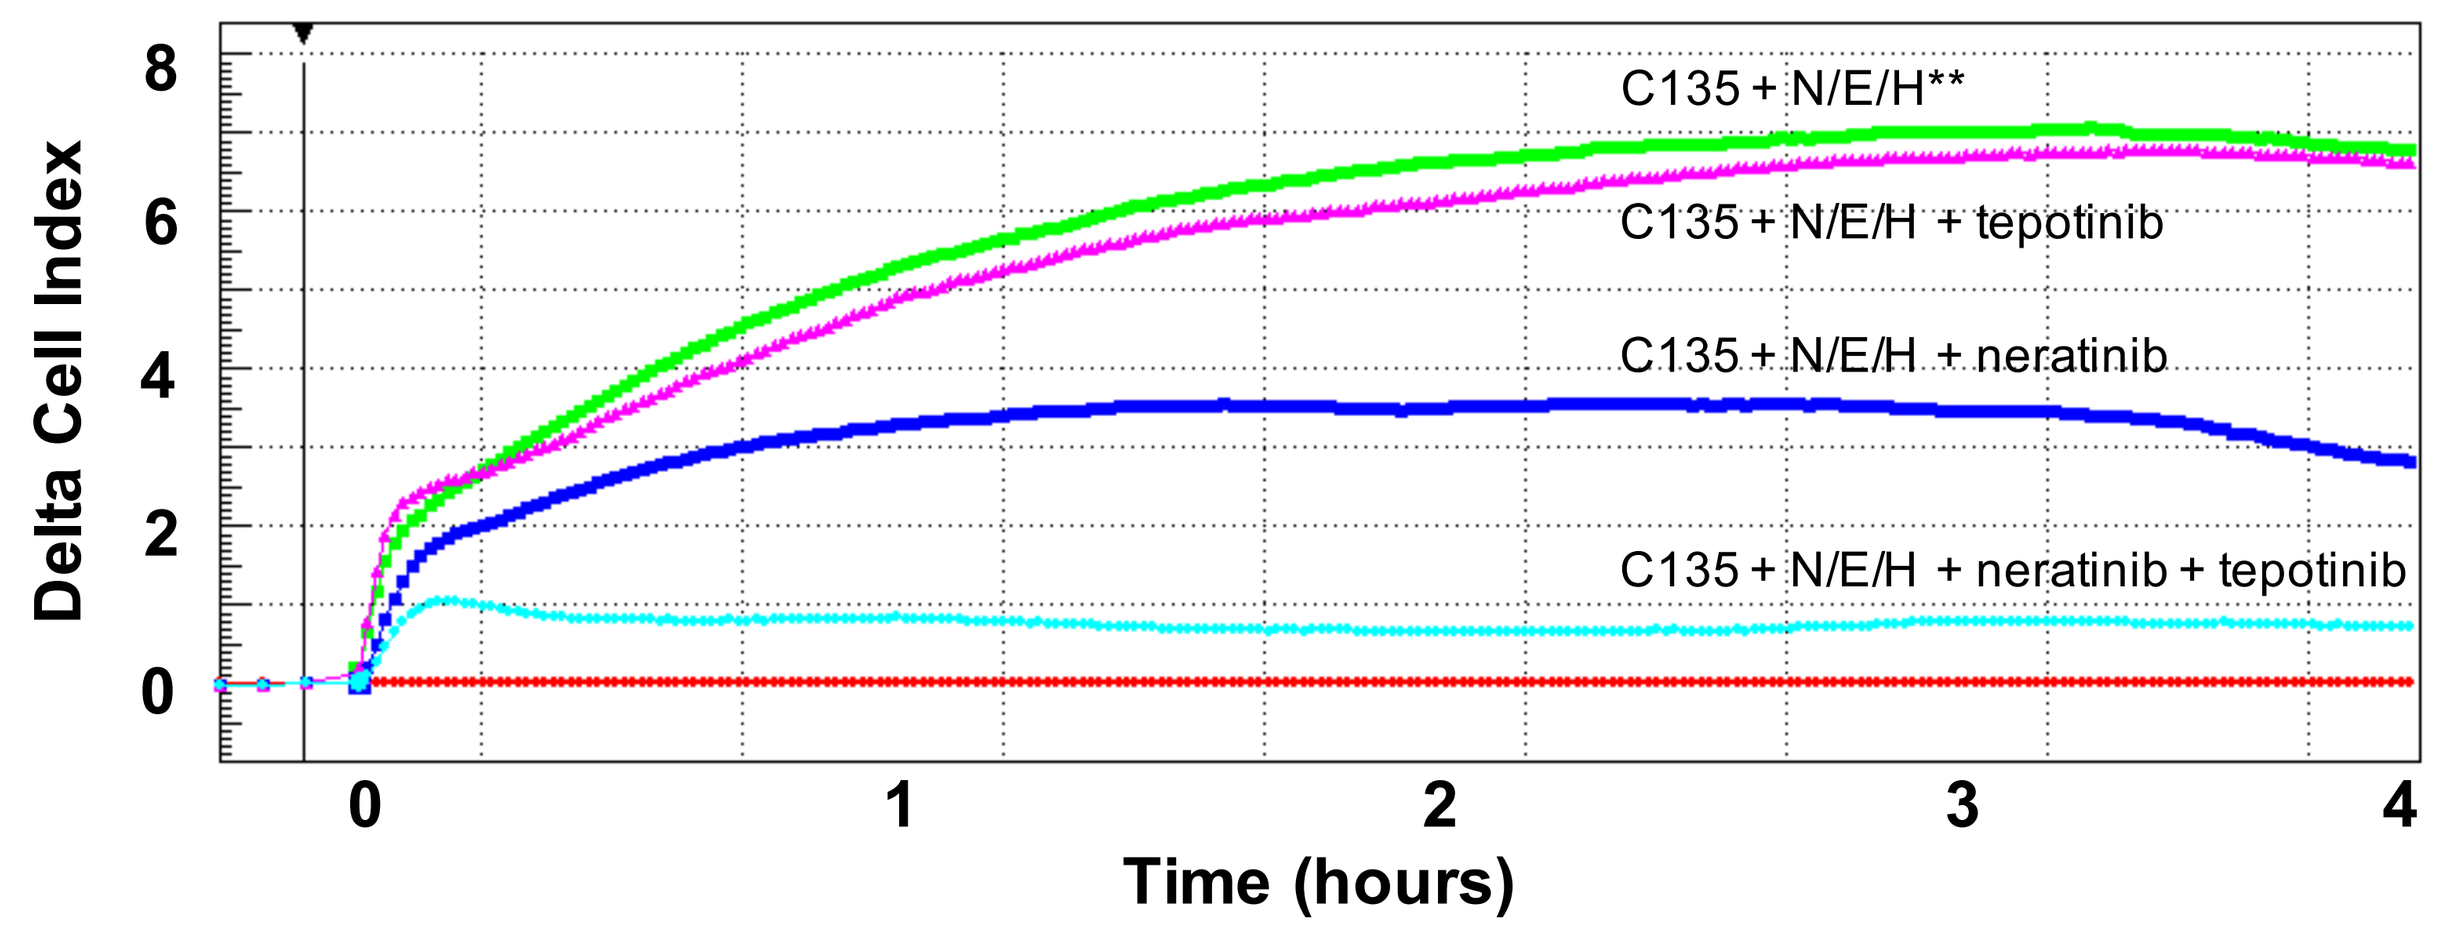
C135 + N/E/H**
C135 + N/E/H + tepotinib
C135 + N/E/H + neratinib
C135 + N/E/H + neratinib + tepotinib
Delta Cell Index
Time (hours)

A representative patient primary breast cancer cell stimulated with a growth factor cocktail of NRG1b, EGF, and HGF in the CELx MP test. Signaling units are determined over a 4-hour period following growth factor addition. Neratinib (pan-HER inhibitor), tepotinib (c-Met inhibitor), or the combination of the two was added prior to growth factor addition. Stimulation is relative to baseline (red, no factor addition).

**N/E/H: NRGb1+EGF+HGF agonist cocktail

Figure 5. Cross-Talk Analysis of Individual Antagonists Against EGF, NRG, and c-Met Illustrates Pathway Co-Involvement

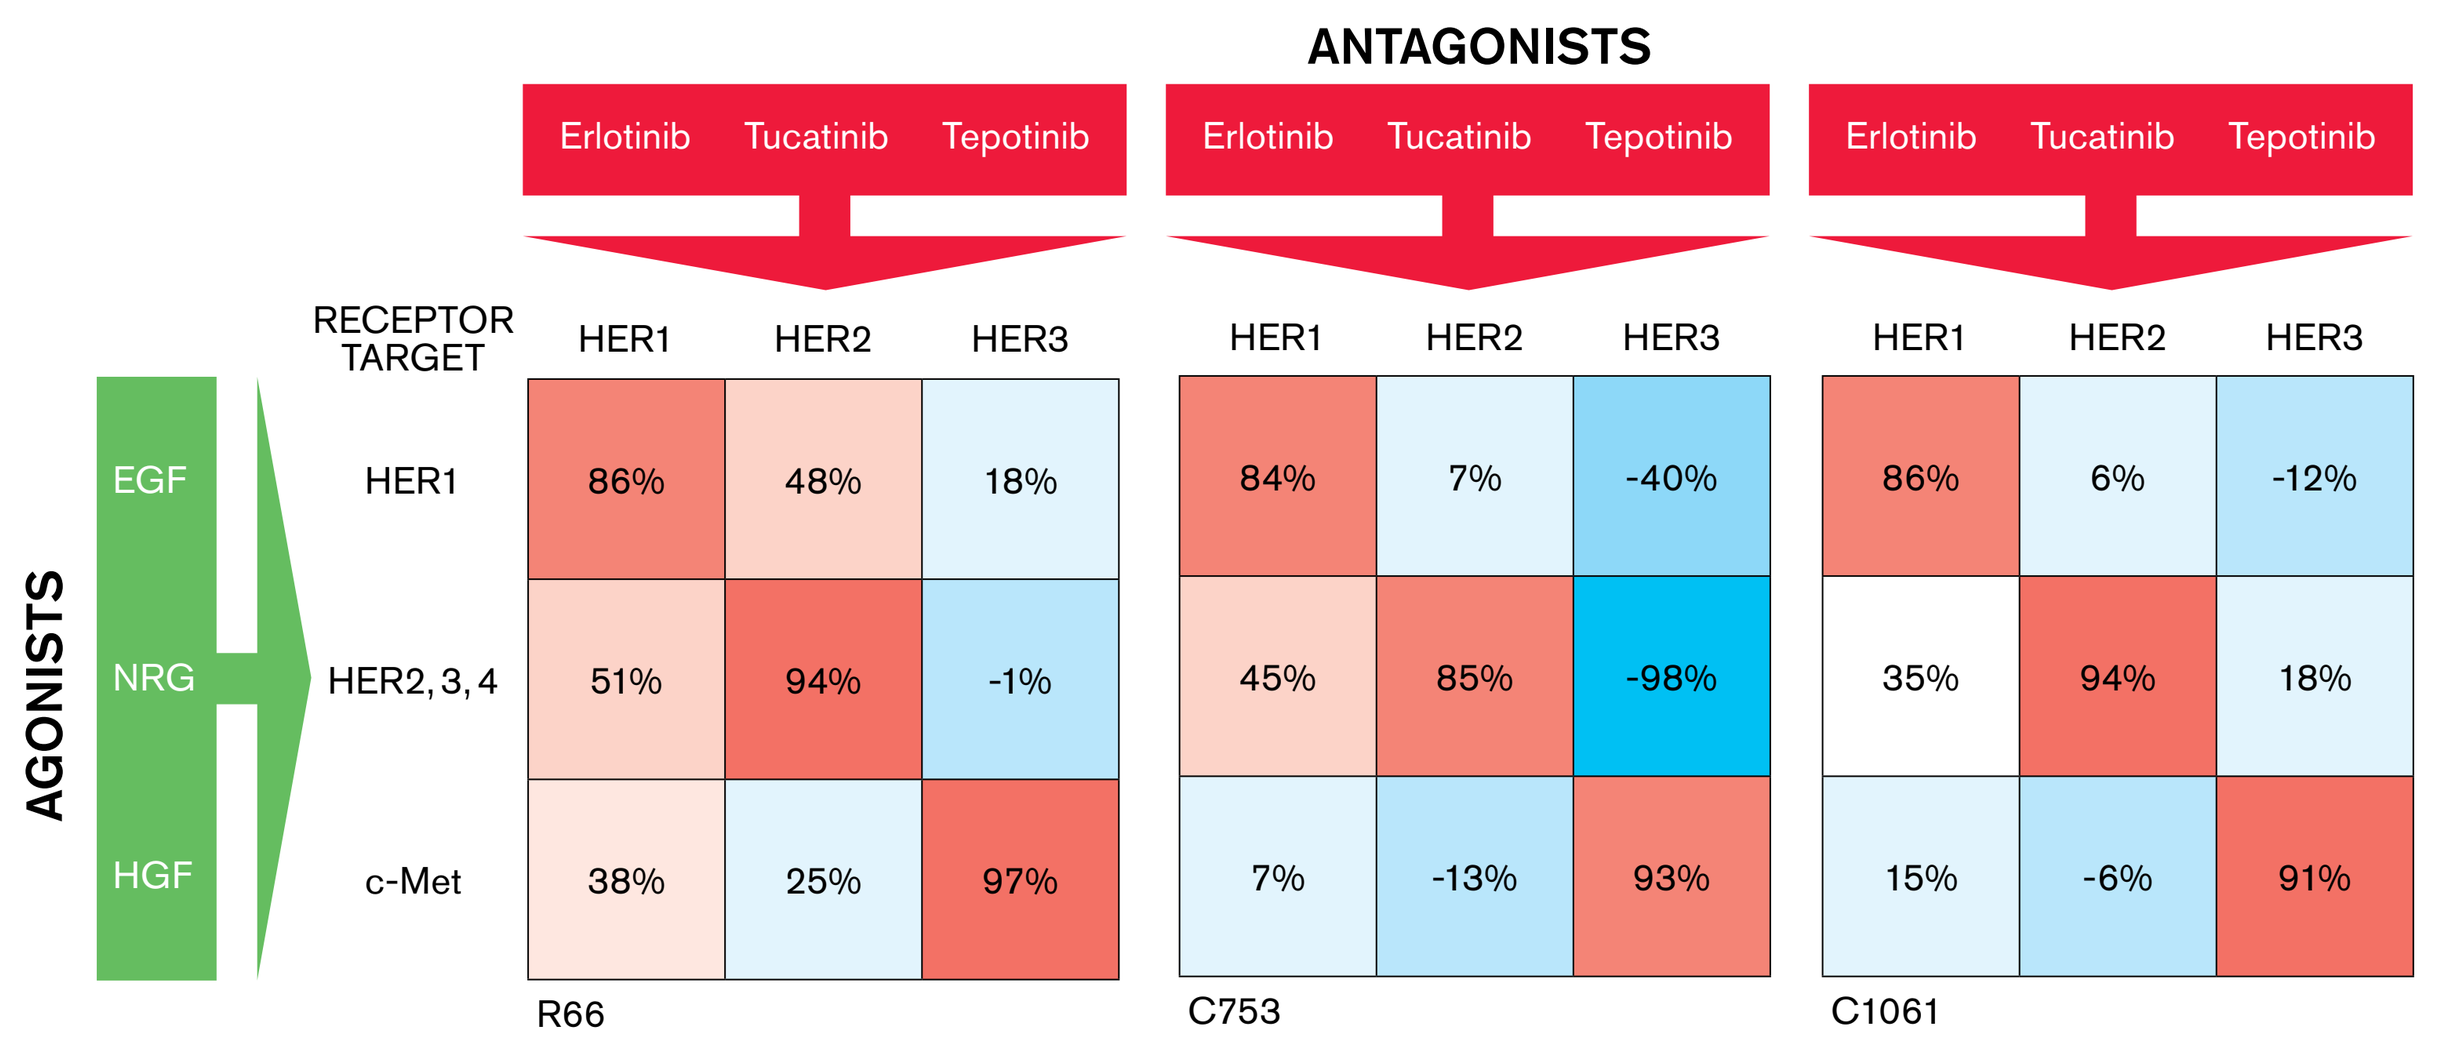
ANTAGONISTS
Erlotinib Tucatinib Tepotinib
RECEPTOR TARGET
HER1 HER2 HER3
AGONISTS
EGF HER1
NRG HER2, 3, 4
HGF c-Met
R66: 86% 48% 18% / 51% 94% -1% / 38% 25% 97%
C753: 84% 7% -40% / 45% 85% -98% / 7% -13% 93%
C1061: 86% 6% -12% / 35% 94% 18% / 15% -6% 91%

Three primary breast cancer samples were treated with either EGF, NRG, or HGF in combination with either a HER1 inhibitor, a HER2 inhibitor, or a c-Met inhibitor. Percent-inhibition of the respective growth factor signal over a 4-hour period by each antagonist was determined. A negative value for percent inhibition indicates that the antagonist enhanced the growth factor response.

Summary of Results

- CELx MP test analysis of primary breast cancer cells from 79 patients revealed that 19 of 79, (24.1%; 95% CI=16–35%) had both hyperactive c-MET signaling and at least one hyperactive signaling ErbB-family receptor.
- A signaling cutoff of 250 units for c-Met functional signaling activity provides a test with analytical specificity >99% and analytical sensitivity >84% for hyperactive c-Met signaling.
- A combination of pan-HER and c-Met TKI inhibits the functional signaling response *ex vivo* better than either antagonist alone.
- Crosstalk analysis supports reports that c-Met and HER family signaling pathways are co-involved.

Conclusions

- The CELx MP test found a significant subset of HER2-negative breasts cancer patients with coincidental hyperactive c-Met and HER family signaling tumors that respond *ex vivo* to a combination of pan-HER and c-Met TKIs.
- A clinical trial to evaluate treatment response of this patient subset to combine c-Met and pan-HER inhibitors is warranted.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. *Cancer Res.* 2004;64:8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. *J Clin Oncol.* 2010;28:92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. *Oncologist.* 2009;14:320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. *Science.* 1987;235:177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. *Cancer Treat Rev.* 2014;40:276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. *N Engl J Med.* 2008;358:1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. *J Clin Oncol.* 2010;28:4307-4315.
8. Ko B, et al. MET/HGF pathway activation as a paradigm of resistance to targeted therapies. *Ann Transl Med.* 2017;5(1):4.
9. Engelman JA, et al. MET amplification leads to gefitinib resistance in lung cancer by activating ERBB3 signaling. *Science.* 2007;316(5827):1039-1043.
10. Tanizaki J, et al. Differential roles of trans-phosphorylated EGFR, HER2, HER3, and RET as heterodimerization partners of MET in lung cancer with MET amplification. *British Journal of Cancer.* 2011;105:807-813.
11. Werbin JL, et al. Multiplex exchange-PAINT imaging reveals ligand-dependent EGFR and Met interactions in the plasma membrane. *Scientific Reports.* 2017;7: 2150.
12. Breindel J, et al. EGF receptor activates MET through MAPK to enhance non-small cell lung carcinoma invasion and brain metastasis. *Cancer Res.* 2013;73(16):5053.
13. Mo HN, et al. Targeting MET in cancer therapy. *Chronic Diseases and Translational Medicine.* 2017;3:148-153.
14. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016; in press.
15. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. *BMC Cancer.* 2016; in revision.
16. Stampfer MR, et al. Culture of human mammary epithelial cells. In: Freshney RI, Freshney MG, eds. *Culture of Epithelial Cells.* 2002.
17. Proia DA, et al. Reconstruction of human mammary tissues in a mouse model. *Nat Protoc.* 2006;1:206-214.

P3-10-20

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.