CELx™ | breast

HER2 Signaling Function Test

Evaluating contribution of hyperactive c-Met and HER (ErbB) signaling to tumor progression in mouse breast tumor xenografts: an *in vivo* study of c-Met and HER/ErbB targeted therapies

Laing LG, Burns DJ, Khan SE, MacNeil IA, Kharbush S, Soltani SM, Brass K, Sullivan BF – Celcuity Inc., 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 4–8, 2018

celcuity™
www.celcuity.com

Background

HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15–20% of breast cancers and is associated with more aggressive disease progression, metastasis, and a poorer prognosis.[1-4] Agents targeting HER2, such as trastuzumab, lapatinib, and pertuzumab, significantly improve clinical outcomes in HER2+ patients.[4,5] Currently, a patient's eligibility for HER2 targeted therapies is determined using IHC or FISH HER2 tests.[4] However, clinical trials have indicated a weak correlation between HER2 expression or amplification levels and HER2 targeted therapy benefit.[6,7]

c-Met is the cognate receptor for Hepatocyte Growth Factor (HGF). MET amplification and HGF overexpression have emerged as mechanisms by which cancers become resistant to HER family therapies.[8,9] c-Met and HER family signaling pathways also participate in extensive cross-talk that can drive cancer progression.[10] Phase II trials with c-Met targeting therapeutics in combination with the HER1 (EGFR) inhibitor erlotinib showed promising results in progression-free survival.[11] However, Phase III clinical trials using MET overexpression as an indicator for c-Met targeted therapy and studies in which patients were randomly selected to receive combination c-Met/HER treatment failed to demonstrate clinical efficacy.[11] Because c-Met expression level using IHC as a clinical pathology diagnostic marker fails to identify a population responsive to c-Met targeted therapies, an alternative approach is required to identify patients with dysfunctional c-Met signaling who will respond to existing therapies.

We previously developed a test to identify patients with abnormal HER2 signaling.[12,13] To elucidate the role of c-Met signaling and its involvement with HER family signaling as a cancer driver, a new assay using an impedance biosensor and live cells derived from each patient's tumor, the CELx multi-pathway signaling function (CELx MP) test, was developed. The CELx MP test measures a patient's *ex vivo* live tumor cell response in real-time to specific HER family and c-Met agonists to diagnose breast cancer tumors with hyperactive HER1, HER2, HER3, HER4, and c-Met signaling activity.[14]

In this study, to further elucidate the role of c-Met signaling and its potential involvement with HER family signaling as a cancer driver, we studied the response to a c-Met inhibitor (tepotinib), a HER1 inhibitor (erlotinib), a pan-HER inhibitor (neratinib), and a combination of these therapies using both the CELx MP test and a tumor xenograft model using the HCC1954 breast cancer cell line.

Methods

Breast Cancer Cell Lines: HCC1954, a HER2+ cell line with hyperactive c-Met and HER1 signaling and normal HER3 and HER2-driven signaling, as determined by CELx testing

Cell Culture: Cell lines were maintained in RPMI media + 10% FBS according to ATCC recommendations and authenticated by ATCC STR profiling.

CELx MP Test: HCC1954 cells were activated with 0.3 nM EGF, 3 nM NRG, and 30 pM HGF ± erlotinib, tepotinib, neratinib, erlotinib + tepotinib, or neratinib + tepotinib in a CELx multi-pathway signaling function test.[14]

Xenograft: 4–5-week-old female NSG (NOD scid gamma; NOD.Cg-Prkdc^{scid} Il2rg^{tm1Wjl}/SzJ) mice were injected subcutaneously in the left mammary fat pad with two million cells in 150 µL of a 50% Matrigel solution. Mice were randomly assigned to either control groups (10% Captisol) or a treatment group: neratinib (40 mg/kg), tepotinib (50 mg/kg), erlotinib (25 mg/kg), erlotinib + tepotinib, or neratinib + tepotinib (n=8–10 per group). After the average tumor volume reached 150 mm³, mice were dosed by oral gavage with 100 µL dosing solution daily (QD) for 17 days. Tumor size and body weight were measured at least twice weekly. Mice were euthanized for necropsy and sample/tissue collection at the study endpoint or when tumor size was ≥2 cm³.

Figure 1. Platform Biosensor Sensitivity Enables Quantification of HER and c-Met Signaling Real-Time in Live Cells

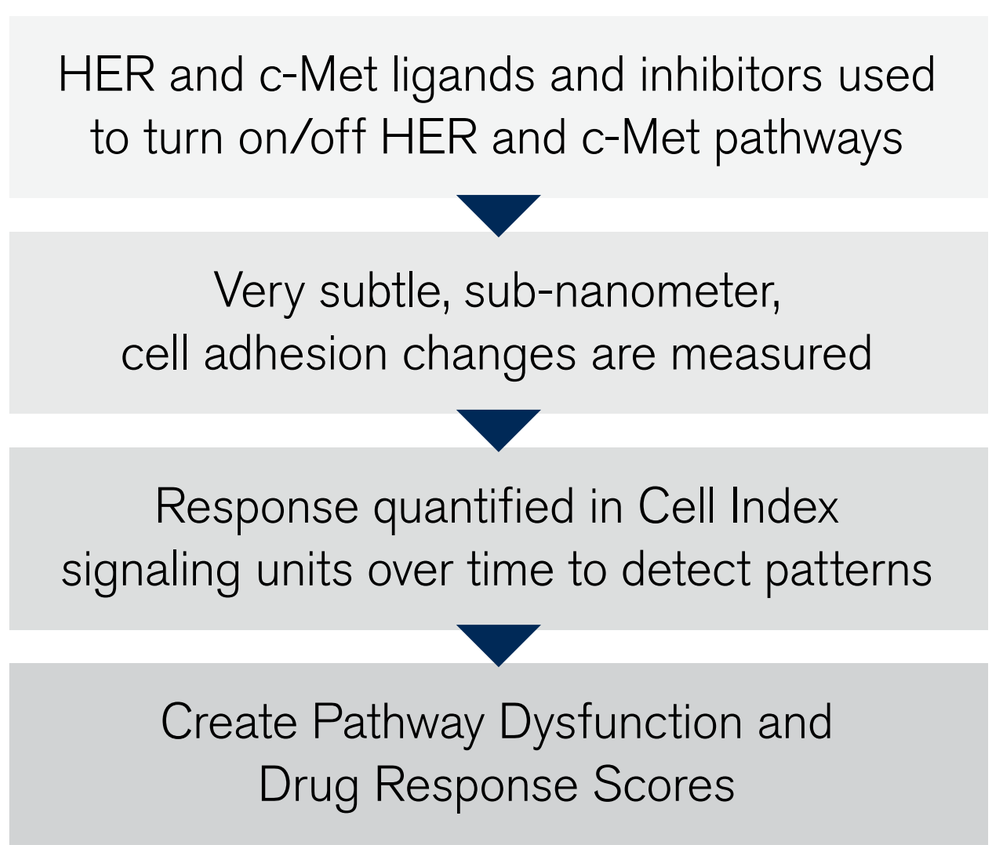
HER and c-Met ligands and inhibitors used to turn on/off HER and c-Met pathways
Very subtle, sub-nanometer, cell adhesion changes are measured
Response quantified in Cell Index signaling units over time to detect patterns
Create Pathway Dysfunction and Drug Response Scores

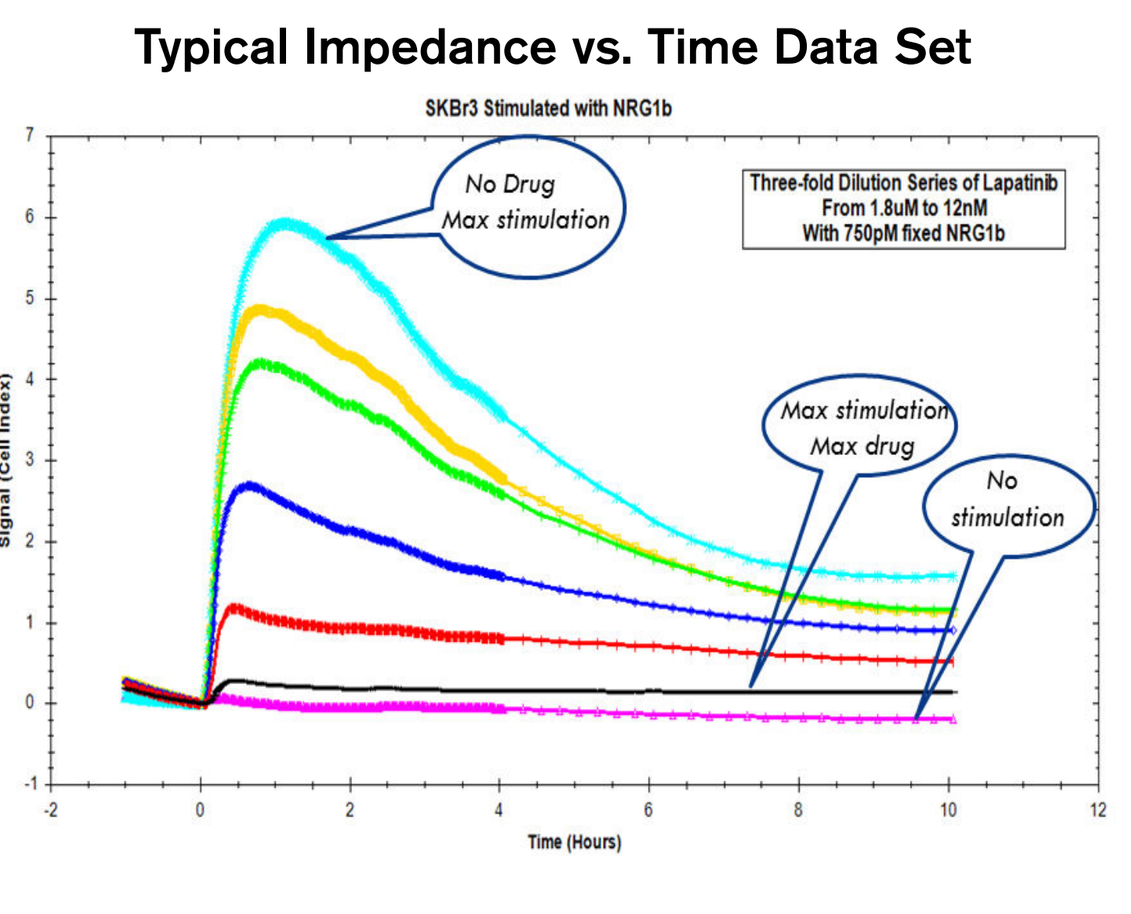
Typical Impedance vs. Time Data Set
SKBr3 Stimulated with NRG1b
No Drug Max stimulation
Three-fold Dilution Series of Lapatinib From 1.8uM to 12nM With 750pM fixed NRG1b
Max stimulation Max drug
No stimulation
Signal (Cell Index)
Time (Hours)

Figure 2. HER2⁻ Abnormal Signaling Identified by CELx Test

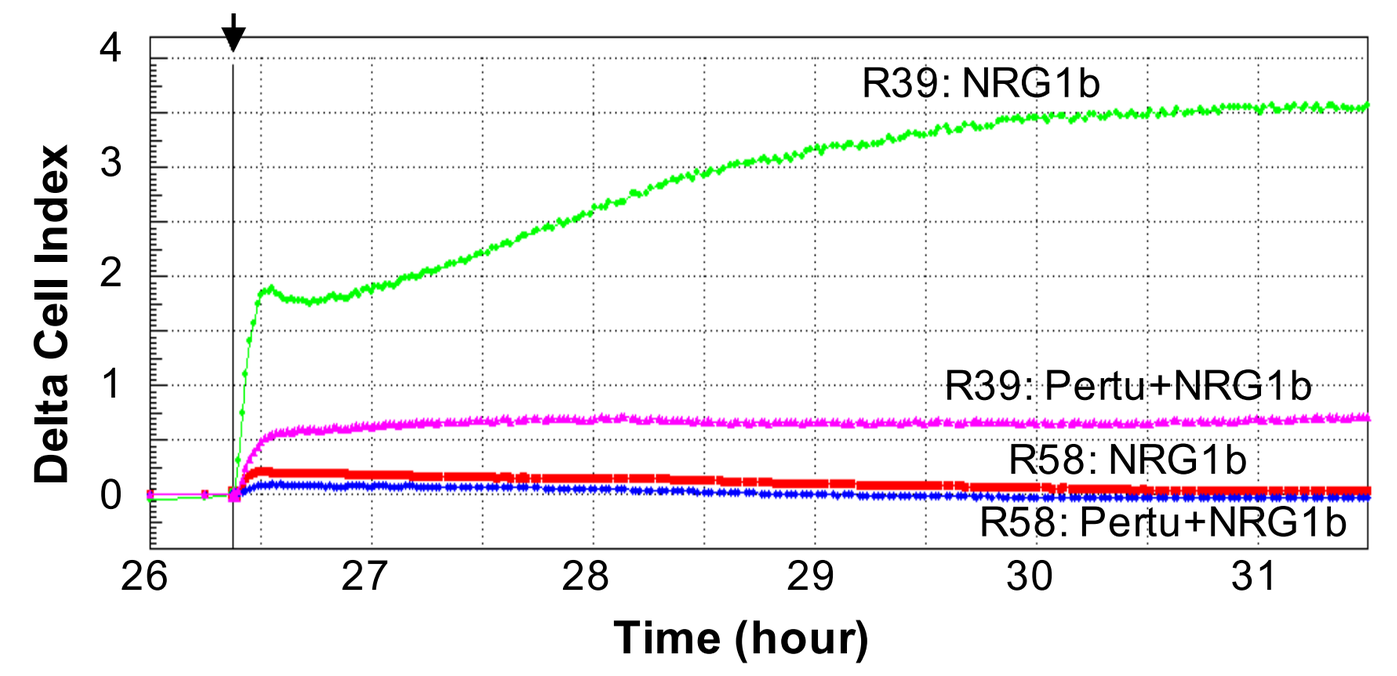
R39: NRG1b
R39: Pertu+NRG1b
R58: NRG1b
R58: Pertu+NRG1b
Delta Cell Index
Time (hour)

CELx time-course curves representing a high, abnormal HER2 signal in a high responder (R39) and a low HER2 signal in a non-responder (R58). In this display, curves of NRG1 stimulation in the absence versus presence of HER2 dimer blocker (10µg/mL) are presented.

The data show that the high NRG1b responder has more than 10 times greater signal than the low responder, indicating the test has a large dynamic range.

Results

Table 1. Real-Time Live Cell CELx Multi-Pathway Signaling Inhibition Analysis of HCC1954 Cells

Pan-HER Inhibitors	HER/c-Met Inhibition (%)
Erlotinib (HER1i)	9.2 ± 10.3 (n=2)
Tepotinib (c-MeTi)	-8.8 ± 12.8 (n=5)
Neratinib (pan-HER)	14.6 ± 29.6 (n=4)
Erlotinib + Tepotinib	47.6 ± 0.1 (n=2)
Neratinib + Tepotinib	82.5 ± 20.1 (n=4)

HCC1954 cells were activated with EGF, NRG, and HGF in the presence or absence of HER and c-Met inhibitors and percent of signal inhibition was measured real-time in live cells using the CELx MP test.

- Tepotinib (c-Met inhibitor) and erlotinib (HER1 inhibitor) have limited effectiveness when HER and c-Met pathways are co-activated.
- Neratinib (pan-HERi) or erlotinib + tepotinib inhibitors enhance inhibition, but it is still limited.
- Neratinib + tepotinib combined completely inhibits co-activated HER and c-Met pathways.

Table 2: HCC1954 Xenograft Model: Experimental Design

Experiment	Cohort	n	Drug	Dose (mg/kg)	Dosing Frequency	Number of Doses
A	1	10	vehicle	0	QD	21
	2	10	neratinib	40	QD	21
	3	10	tepotinib	50	QD	21#
	4	9*	neratinib + tepotinib	40 + 50	QD	21
B	1	7^	vehicle	0	QD	21
	2	8	erlotinib	25	QD	21
	3	8	erlotinib + tepotinib	25 + 50	QD	21

* 1 mouse experienced weight loss and diarrhea and was found dead on treatment day 13.
\# Due to weight loss, 1 mouse given 3-day dose holiday due, and 1 mouse given 7-day dose holiday.
^ 1 mouse euthanized on treatment day 8 due to severe weight loss.

Figure 3. HCC1954 Xenograft Model: Mouse Body Weight

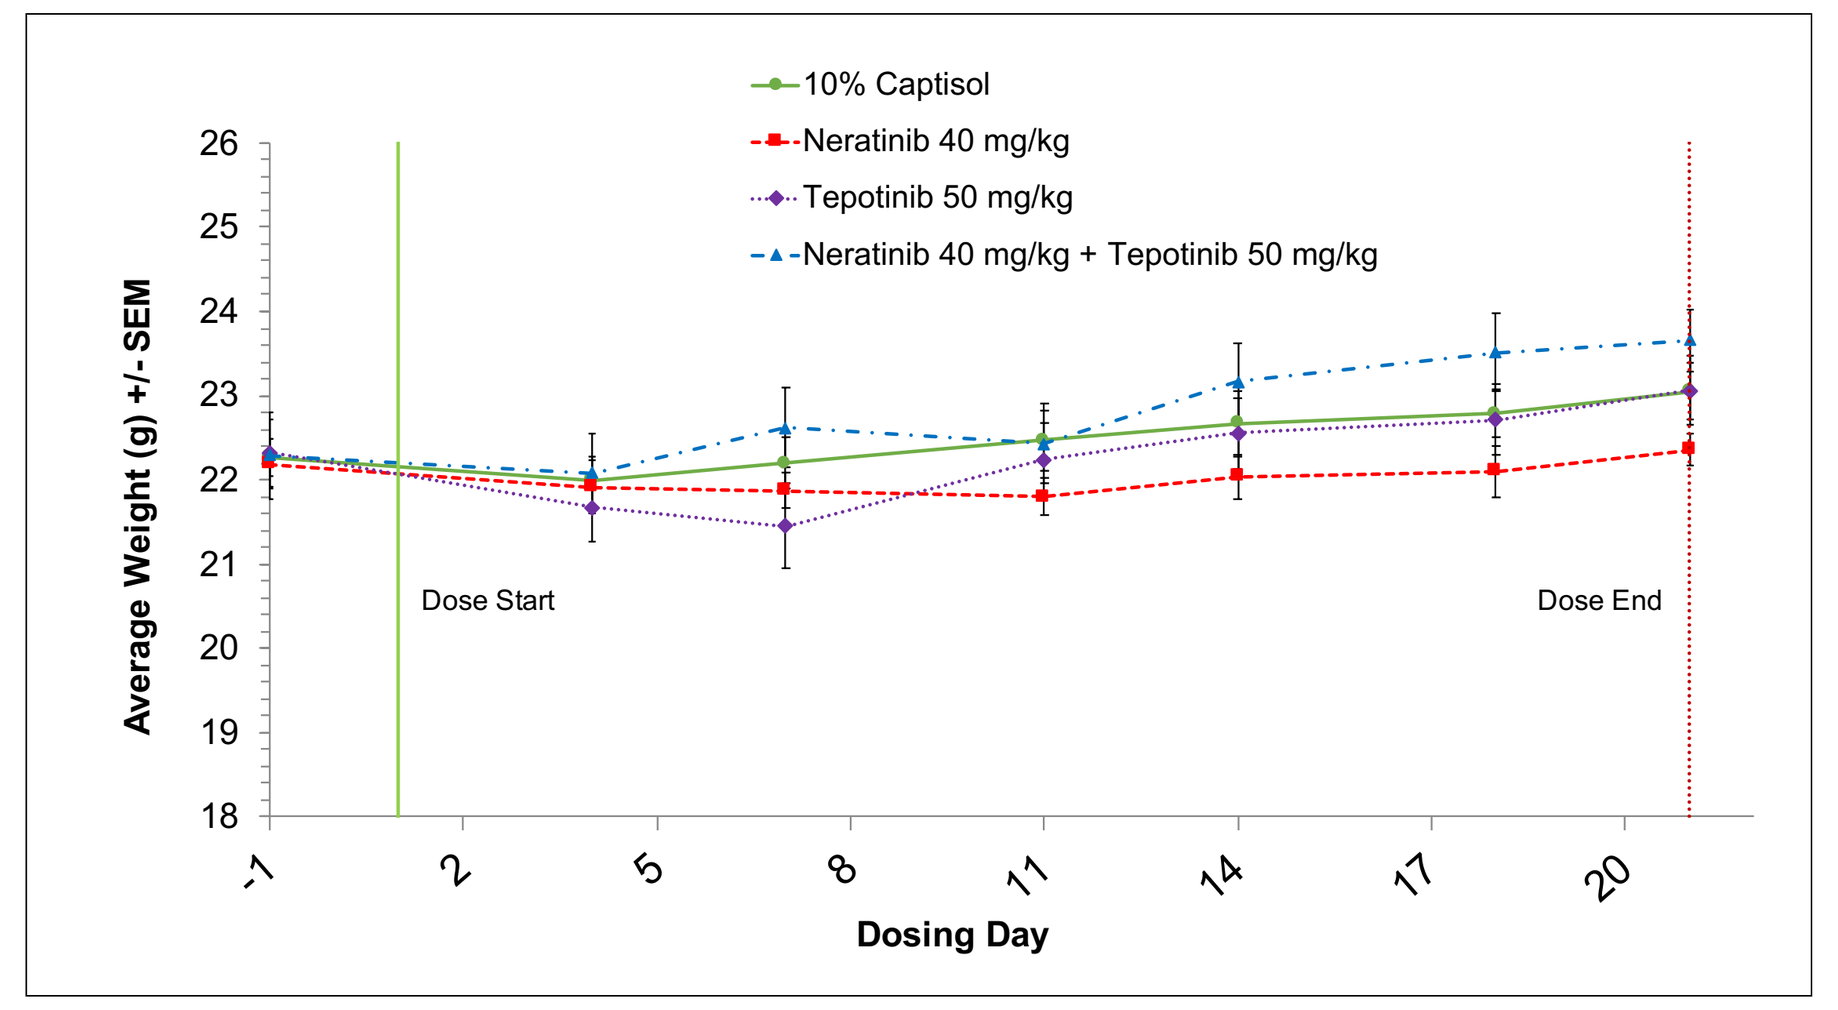
10% Captisol
Neratinib 40 mg/kg
Tepotinib 50 mg/kg
Neratinib 40 mg/kg + Tepotinib 50 mg/kg
Dose Start
Dose End
Average Weight (g) +/- SEM
Dosing Day

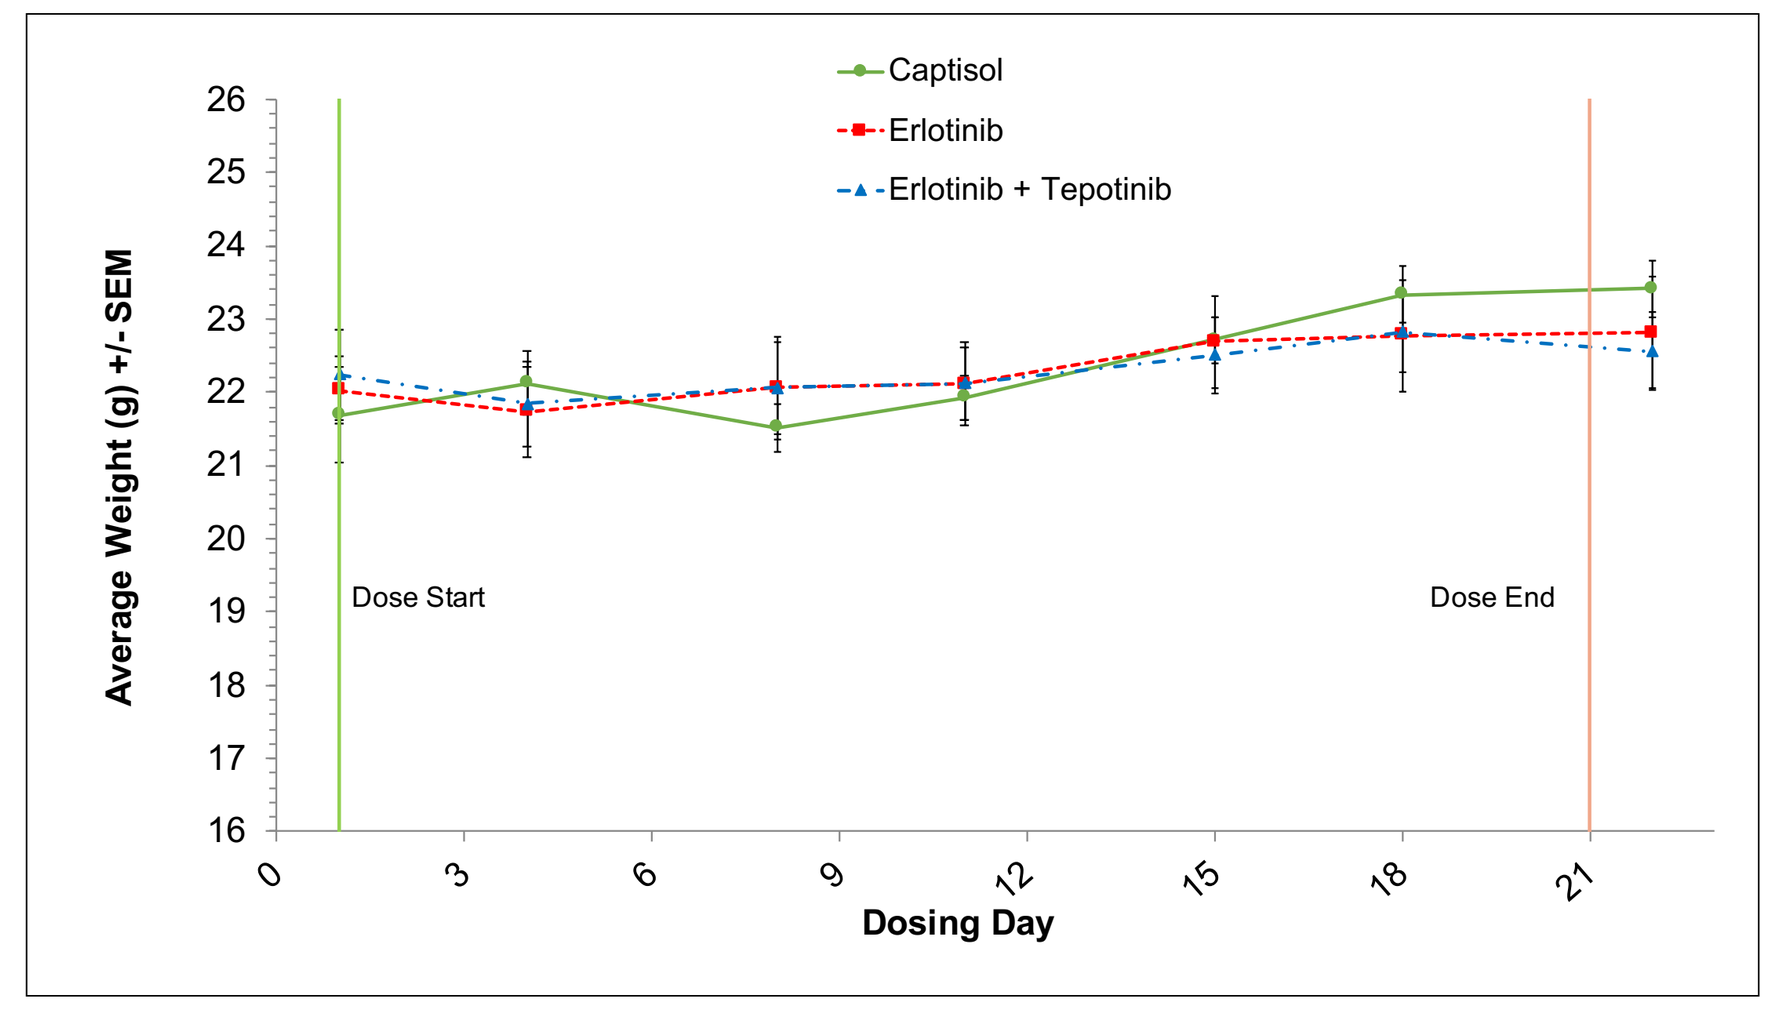
Captisol
Erlotinib
Erlotinib + Tepotinib
Dose Start
Dose End
Average Weight (g) +/- SEM
Dosing Day

Results

Figure 4. HCC1954 Xenograft Drug Response is Consistent with CELx MP Signaling Results

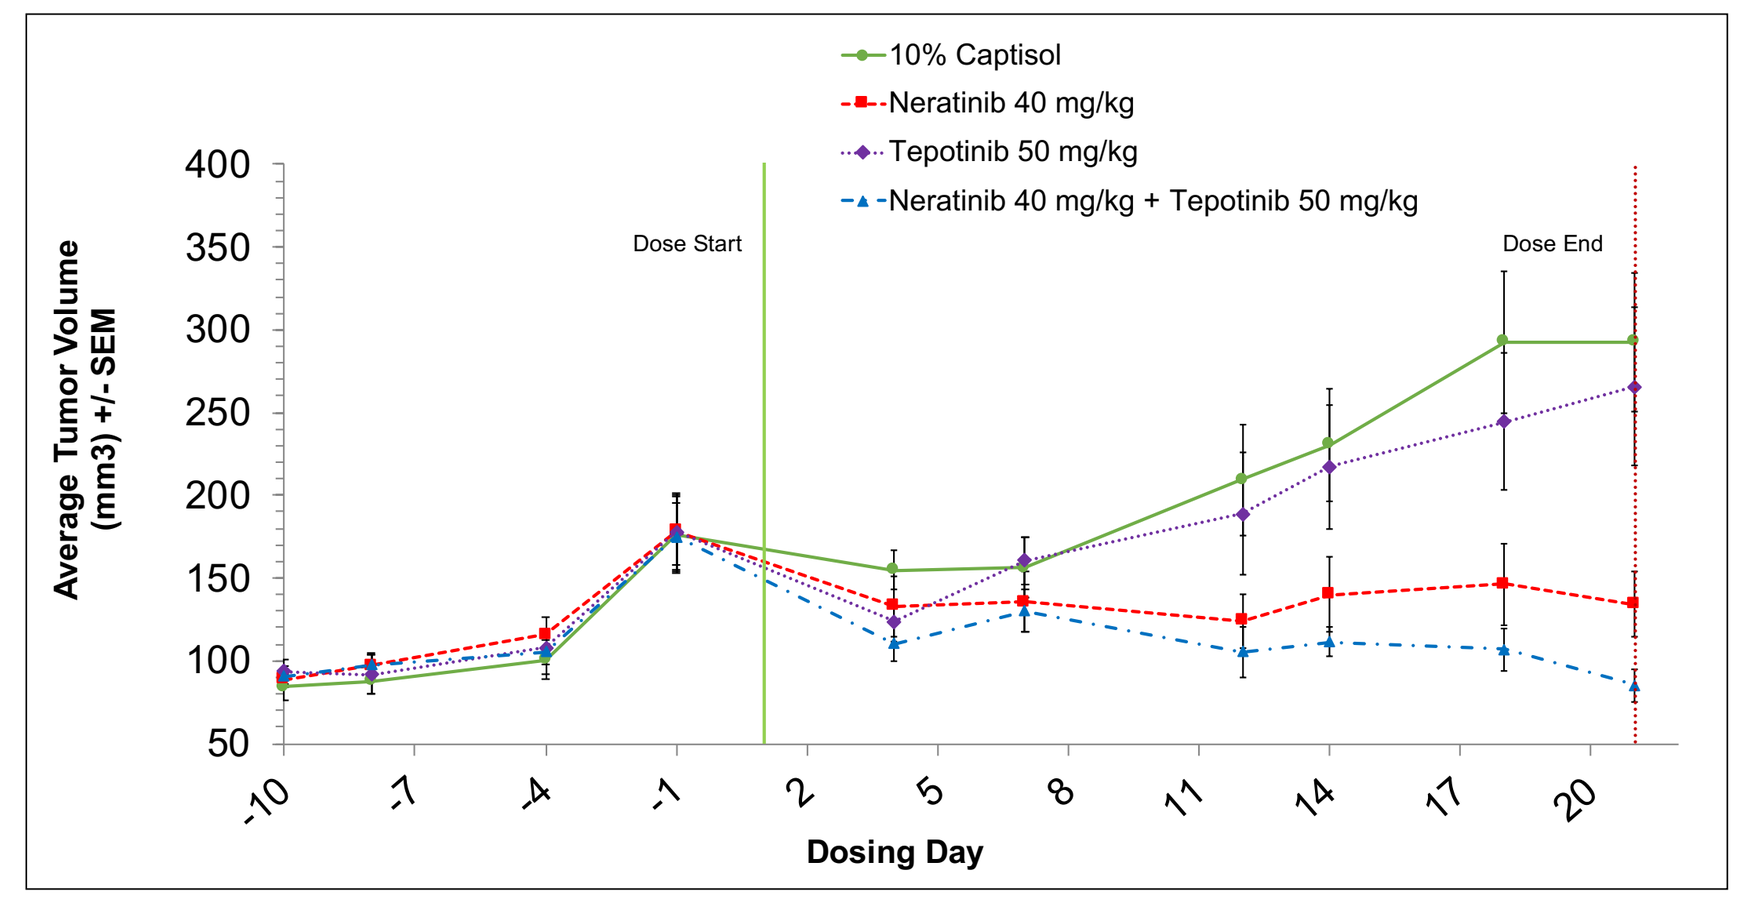
10% Captisol
Neratinib 40 mg/kg
Tepotinib 50 mg/kg
Neratinib 40 mg/kg + Tepotinib 50 mg/kg
Dose Start
Dose End
Average Tumor Volume (mm3) +/- SEM
Dosing Day

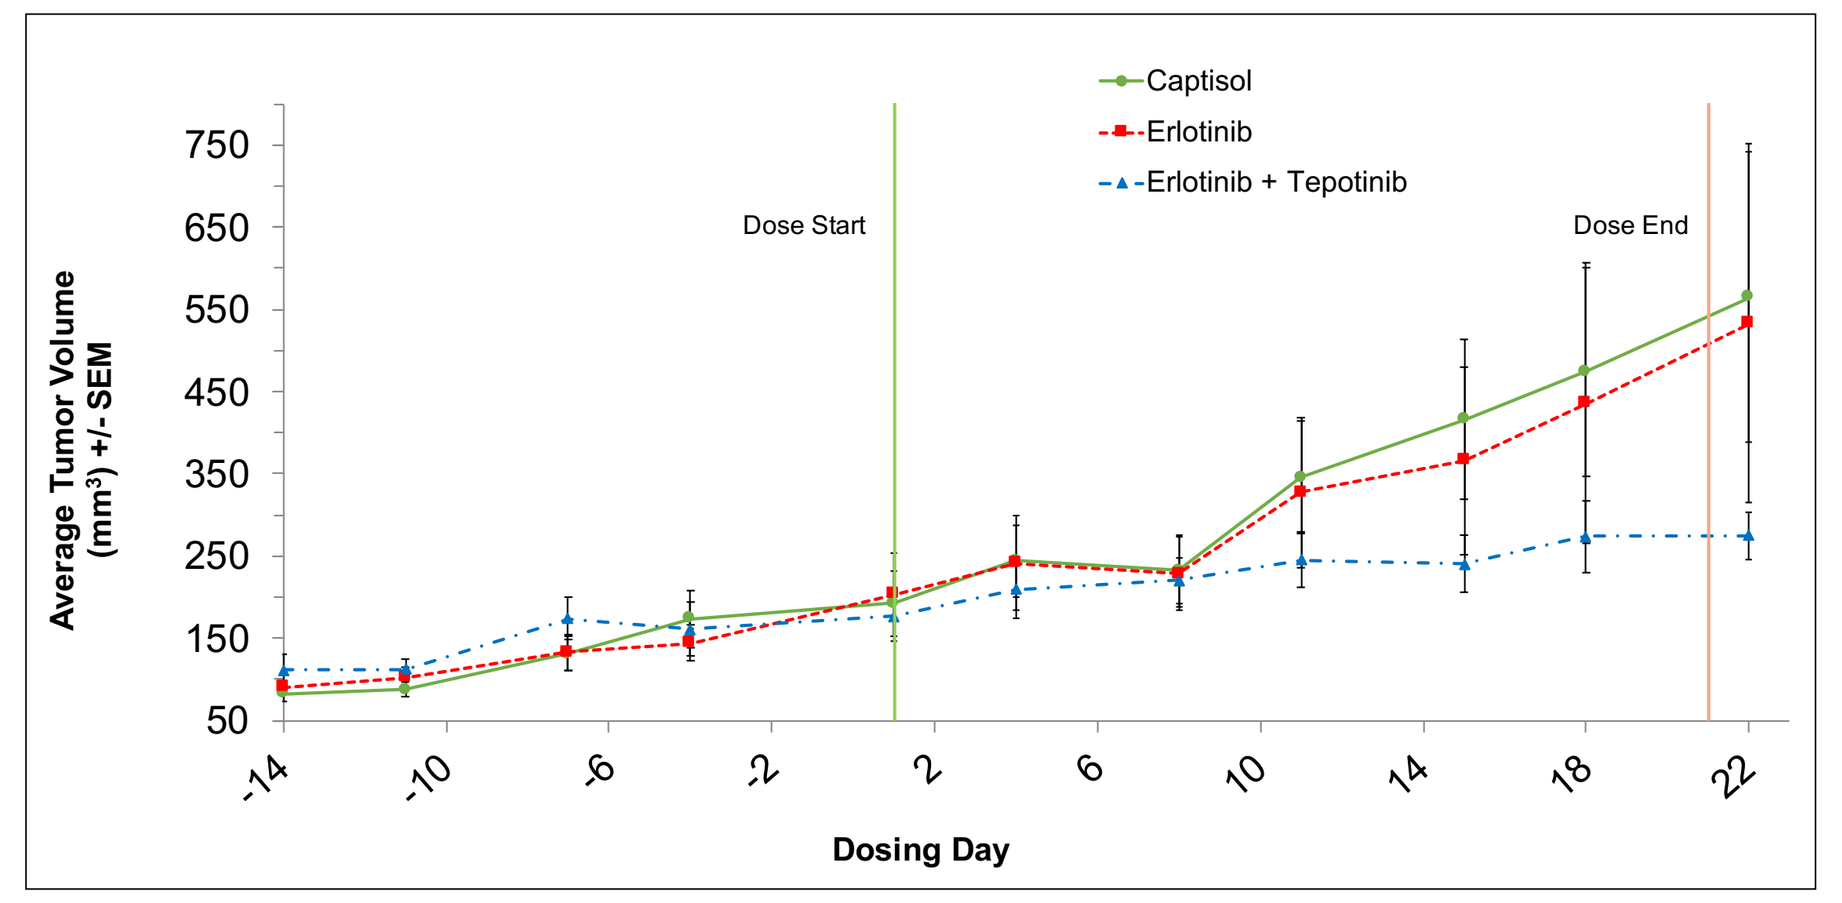
Captisol
Erlotinib
Erlotinib + Tepotinib
Dose Start
Dose End
Average Tumor Volume (mm³) +/- SEM
Dosing Day

Drug Arm vs. Comparator Arm	Tumor Reduction (t-test)
Tepotinib vs. Control	10% (p=0.780)
Neratinib vs. Control	54% (p=0.0026)
Neratinib + Tepotinib vs. Control	71% (p=0.0003)
Neratinib + Tepotinib vs. Neratinib	37% (p=0.05)

Drug Arm vs. Comparator Arm	Tumor Reduction (t-test)
Erlotinib vs. Control	5% (p=0.870)
Erlotinib + Tepotinib vs. Control	51% (p=0.110)

Summary of Results

- HER1 or c-Met inhibitors have limited single agent effectiveness when HER and c-Met pathways are co-activated either in vitro (real-time live cell CELx MP Test) or *in vivo* (Xenograft mouse model).
- Combination of c-Met + HER1 inhibitors OR a pan-HER inhibitor alone can more effectively inhibit co-activation of HER and c-Met pathways, but full inhibition requires the combination of pan-HER + c-Met inhibitors.

Conclusions

- Hyperactive and coincident c-Met and HER signaling contributes to the progression of certain HER2- negative breast cancers.
- The real-time live cell CELx MP test results are consistent with xenograft data and suggest that c-Met and HER pathways are co-activated and must both be inhibited in HCC1954 tumor models.
- This HER2-negative breast cancer sub-type is more responsive to treatment with a combination of c-Met inhibitor plus a pan-HER inhibitor versus a c-Met inhibitor plus an HER1 inhibitor or any of the single agents studied.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. *Cancer Res.* 2004;64:8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. *J Clin Oncol.* 2010;28:92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. *Oncologist.* 2009;14:320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. *Science.* 1987;235:177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. *Cancer Treat Rev.* 2014;40:276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. *N Engl J Med.* 2008;358:1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. *J Clin Oncol.* 2010;28:4307-4315.
8. Ko B, et al. MET/HGF pathway activation as a paradigm of resistance to targeted therapies. *Ann Transl Med.* 2017;5(1):4.
9. Engelman JA, et al. MET amplification leads to gefitinib resistance in lung cancer by activating ERBB3 signaling. *Science.* 2007;316(5827):1039-1043.
10. Breindel J, et al. EGF receptor activates MET through MAPK to enhance non-small cell lung carcinoma invasion and brain metastasis. *Cancer Res.* 2013;73(16):50
11. Mo HN, et al. Targeting MET in cancer therapy. *Chronic Diseases and Translational Medicine.* 2017;3:148-153.
12. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016; in press.
13. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. *BMC Cancer.* 2016; in revision.
14. Laing et al., Evaluation of pan-HER and c-MET inhibitors tested in primary HER2- breast cancer cells with hyperactive c-METand ErbB family signaling, SABCS, 2018, Abstract # P2-05-05.

P3-10-15

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.