CELx™ | breast

HER2 Signaling Function Test

Evaluation of pan-HER and c-Met inhibitors tested *ex vivo* in live primary HER2- breast cancer cells with hyperactive c-Met and (HER) ErbB family signaling

Laing LG, Burns DJ, Khan SE, Kharbush S, Soltani SM, MacNeil IA, Rich BE, Brass K, Broege AM, Mutka S, Sullivan B – Celcuity Inc., 16305 36th Ave N, Suite 100, Minneapolis, MN 55446, USA

SAN ANTONIO BREAST CANCER SYMPOSIUM®
December 4–8, 2018

celcuity™
www.celcuity.com

Background

Biological factors, such as HER2 signaling activity, may be important to measure in addition to expression and amplification of HER2 when identifying patients eligible for HER2 therapies.

HER2 gene (ERBB2) amplification and/or HER2 protein overexpression is detected in approximately 15–20% of breast cancers and is associated with more aggressive disease progression, metastasis, and a poorer prognosis.[1-4] Agents targeting HER2, such as trastuzumab, lapatinib, and pertuzumab, significantly improve clinical outcomes in HER2+ patients.[4,5] Currently, a patient's eligibility for HER2 targeted therapies is determined using IHC or FISH HER2 tests.[4] However, clinical trials have indicated a weak correlation between HER2 expression or amplification levels and HER2 targeted therapy benefit.[6,7]

c-MET is the cognate receptor for Hepatocyte Growth Factor (HGF). MET amplification and HGF overexpression have emerged as mechanisms by which cancers become resistant to HER family therapies.[8,9] c-Met and HER family signaling pathways also participate in extensive cross-talk that can drive cancer progression.[10] Phase II trials with c-Met targeted therapies, either cabozantinib or onartuzumab, in combination with erlotinib, showed promising improvement in PFS.[11] However, Phase III clinical trials studying c-Met targeted therapies using MET overexpression as an enrollment criteria and studies in which patients were randomly selected to receive combination c-Met/HER1 treatment failed to demonstrate clinical efficacy.[11] Because c-Met expression level using IHC as a clinical pathology diagnostic marker fails to identify a population responsive to c-Met targeted therapies, an alternative approach is required to identify patients with dysfunctional c-Met signaling who will respond to these therapies.

We previously developed a test to identify patients with abnormal HER2 signaling.[12,13] To elucidate the role of c-Met signaling and its involvement with HER family signaling as a cancer driver, a new assay using an impedance biosensor and live cells derived from each patient's tumor, the CELx multi-pathway signaling function (CELx MP) test, was developed.

The CELx MP test measures *ex vivo* real-time live cell response to specific HER family and c-Met agonists to diagnose breast tumors with hyperactive HER1, HER2, HER3, HER4, and c-Met signaling activity. A recent study quantified c-Met and HER-driven signaling activity in epithelial cell samples derived from fresh breast tumor specimens obtained from 79 HER2-negative breast cancer patients. Of the cell samples tested, 19 of 79, (24.1%; 95% CI=16–35%) had both hyperactive c-Met signaling and at least one hyperactive signaling HER-family receptor.

Using primary breast cancer cells with hyperactive c-Met and HER family signaling and the CELx MP test, the current study set out to:

1. Determine the IC50 values of six pan-HER inhibitors and five c-Met inhibitors.
2. Characterize the efficacy of combinations of each pan-HER inhibitor with each c-Met inhibitor.

Methods

Specimens: Epithelial cells from six HER2-negative tumor specimens with hyperactive c-Met and HER-driven signaling were obtained.

Cell Culture: Methods for tissue extraction and primary cell culture* are essentially as described previously.[14,15]

Flow Cytometry: Flow cytometry of all cell samples was performed on a BD FACSCalibur using cells harvested at the time of the CELx MP test. Flow cytometry results are 100% concordant to the standard clinical IHC test evaluations for HER2 that were provided for each specimen by the clinic.

CELx MP Test: Real-time live cell responses to specific HER3 and HER1 agonists (NRG1b and EGF) or c-Met agonist (HGF) with and without antagonists were measured and quantified using an xCELLigence RTCA impedance biosensor (ACEA Biosciences). From these responses, the net amount of HER2 participation in HER-family signaling initiated by HER receptor agonists was determined.[13,14] Samples with HER2 signaling activity levels above a previously determined cut-off value of 250 signaling units that was attenuatable with a HER2 dimer blocker were identified as abnormal. Samples with a c-Met signal greater than 250 were characterized as hyperactive.

Determining IC50 values: A 1,000-fold, 5-point dose response of a panel of antagonists for HER-family and c-Met was run against the corresponding agonist stimulus (NRG1b/EGF for HER-family activation and HGF for c-Met activation) in a live-cell CELx assay. Response as a percent of maximum response of the agonist without an antagonist was plotted in GraphPad Prism software and IC50 values were derived using nonlinear regression. IC50 values were determined in six primary breast cancer cells and the values were averaged.

Figure 1. Platform Biosensor Sensitivity Enables Quantification of HER and c-Met Signaling Real-Time in Live Cells

HER and c-Met ligands and inhibitors used to turn on/off HER and c-Met pathways

Very subtle, sub-nanometer, cell adhesion changes are measured

Response quantified in Cell Index signaling units over time to detect patterns

Determine Signaling Score for each pathway tested

Typical Impedance vs. Time Data Set

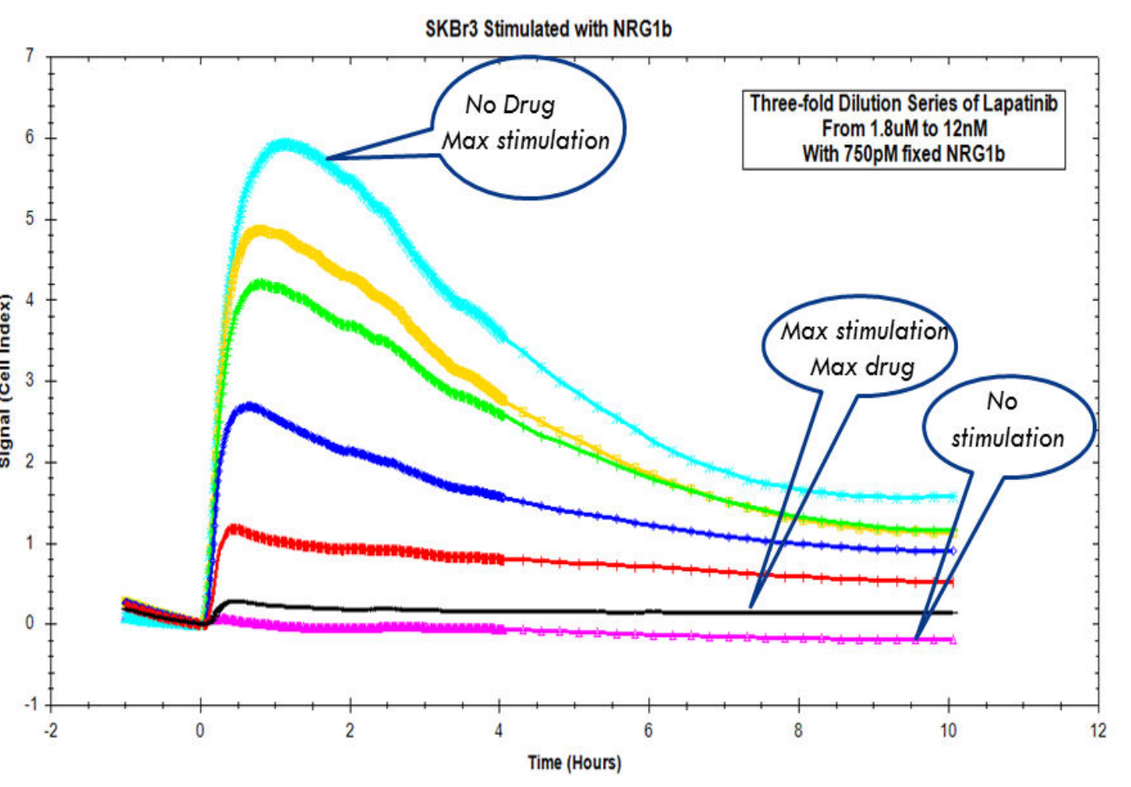
SKBr3 Stimulated with NRG1b
No Drug
Max stimulation
Three-fold Dilution Series of Lapatinib
From 1.8uM to 12nM
With 750pM fixed NRG1b
Max stimulation
Max drug
No
stimulation
Signal (Cell Index)
Time (Hours)

* De-identified excess surgically resected human breast cancer tissue was received from multiple clinical sites located across the United States. Liberty IRB (Columbia, MD) determined that this research did not involve human subjects as defined under 45 CFR 46.102(f) and granted an IRB exemption. Liberty IRB has full accreditation with the Association for the Accreditation of Human Research Protection Programs (AAHRPP).

Figure 2. HER2⁻ Abnormal Signaling by CELx HSF Test

Representative CELx Time-Course Curves

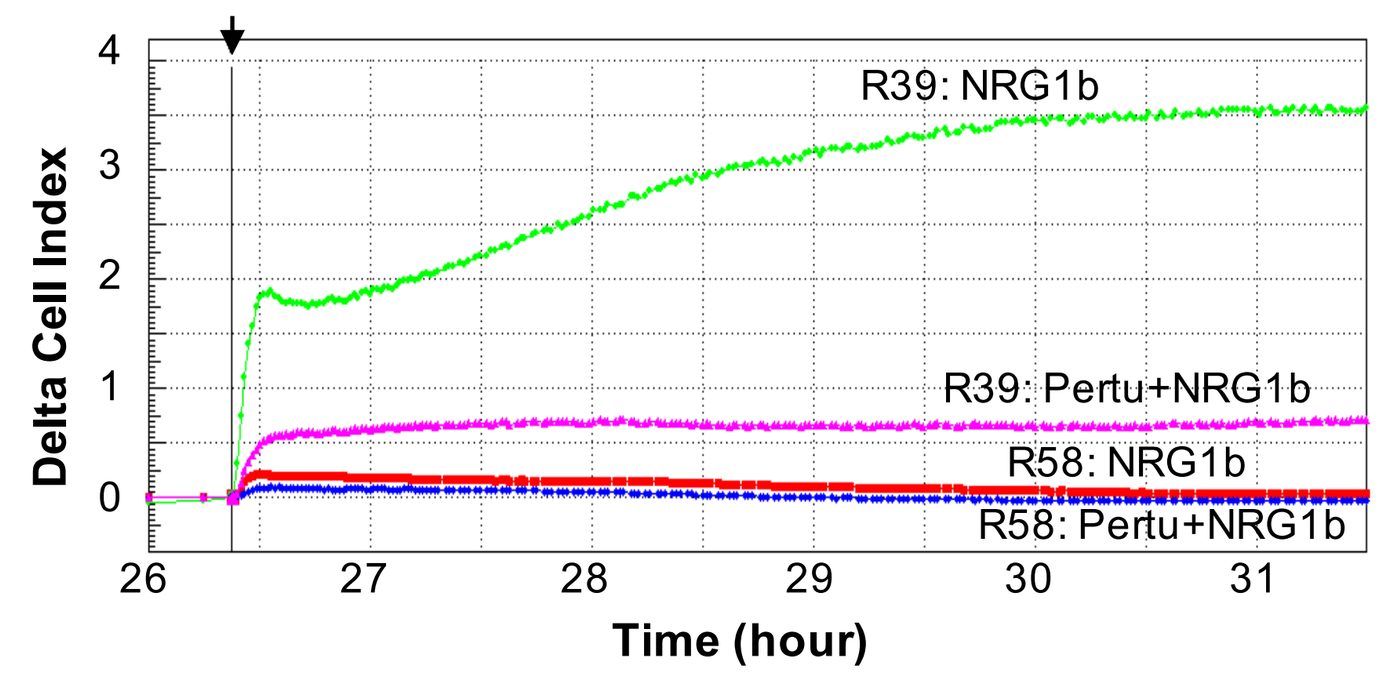
R39: NRG1b
R39: Pertu+NRG1b
R58: NRG1b
R58: Pertu+NRG1b
Delta Cell Index
Time (hour)

CELx time-course curves representing a high, abnormal HER2 signal in a high responder (R39) and a low HER2 signal in a non-responder (R58). In this display, curves of NRG1 stimulation in the absence versus presence of HER2 dimer blocker (10μg/mL) are presented.
The data show that the high NRG1b responder has more than 10 times greater signal than the low responder, indicating the test has a large dynamic range.

Results

Figure 3. Determining the IC50 Values of Pan-HER Inhibitors Against NRGb1/EGF Agonist Combination in a Real-Time Live Cell Assay

A

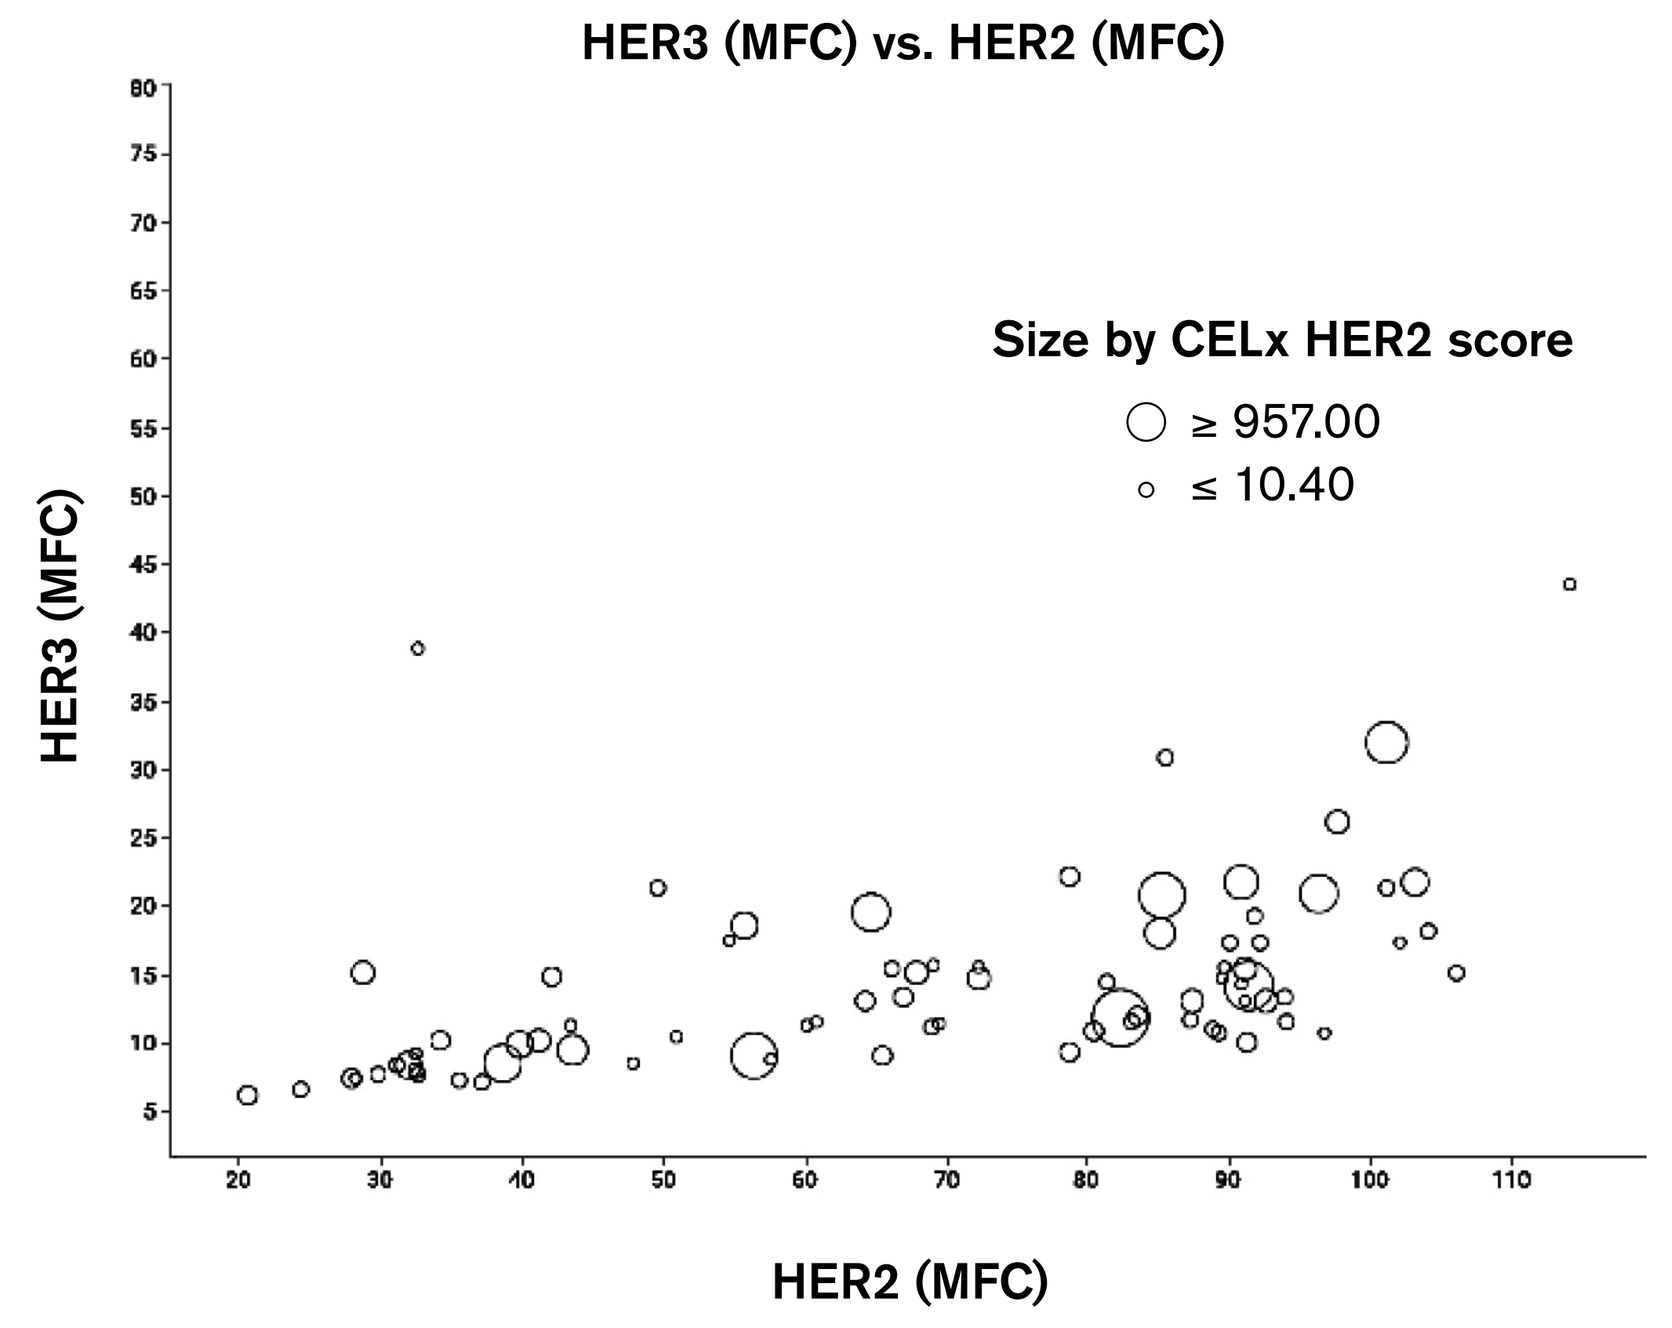
HER3 (MFC) vs. HER2 (MFC)
Size by CELx HER2 score
≥ 957.00
≤ 10.40
HER3 (MFC)
HER2 (MFC)

B

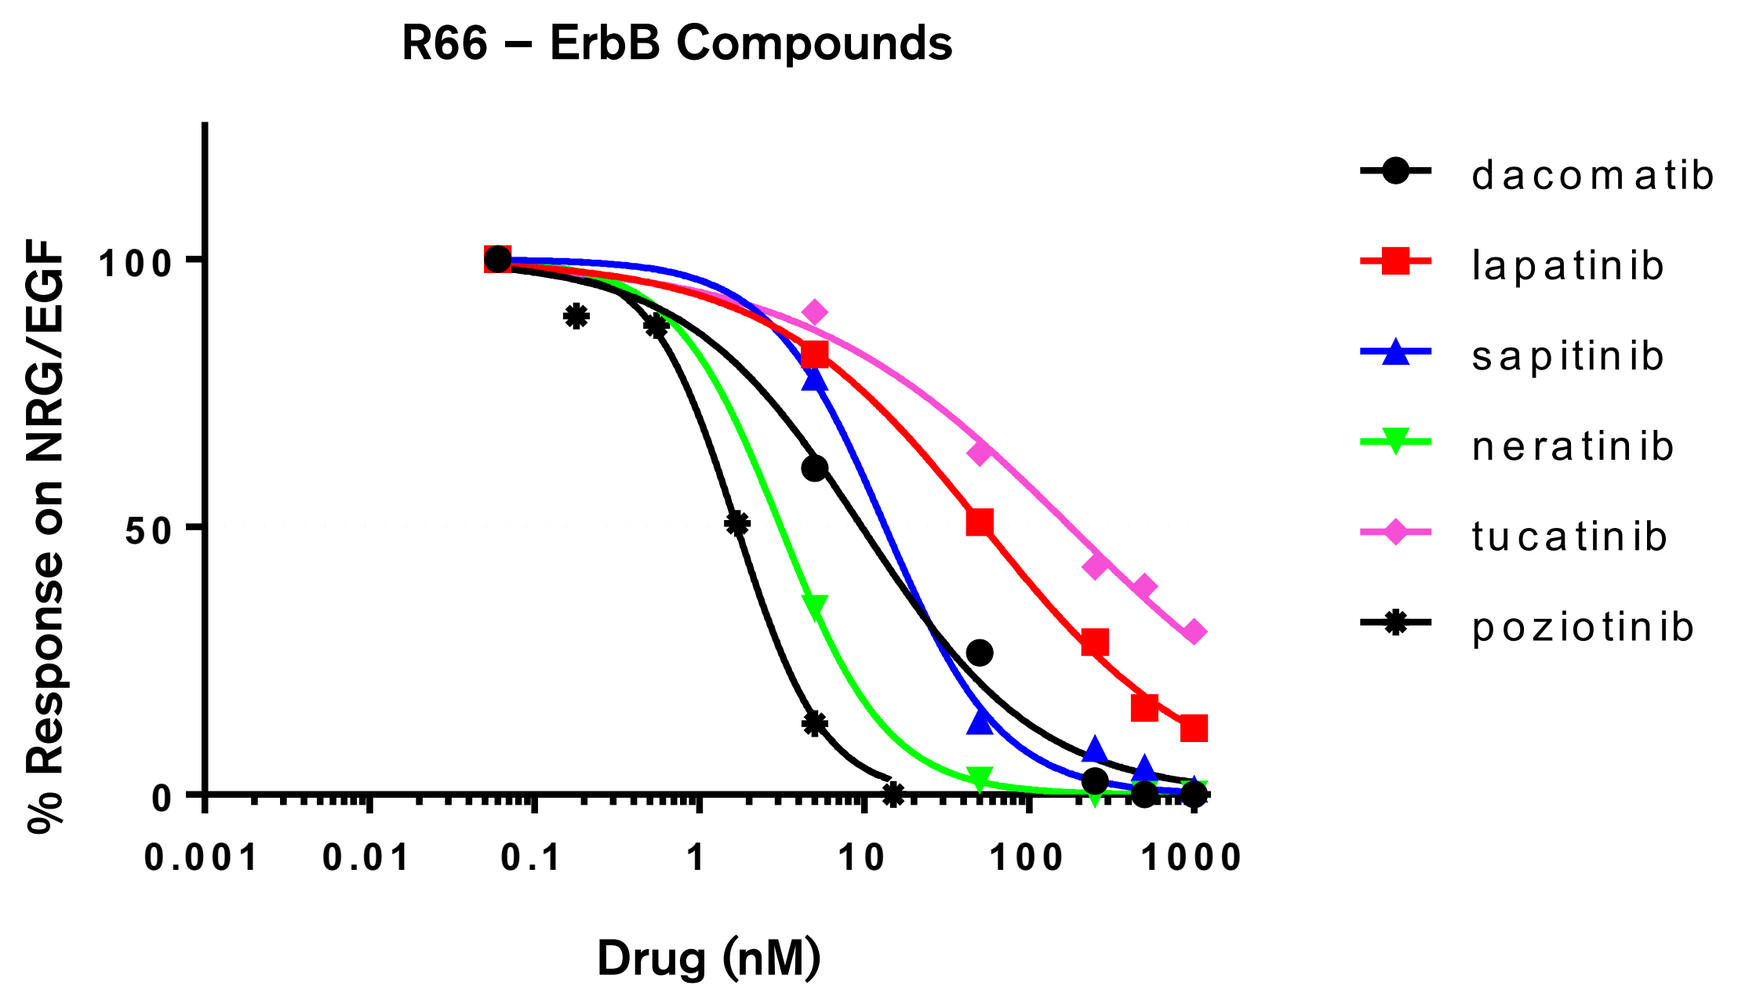
R66 – ErbB Compounds
dacomatib
lapatinib
sapitinib
neratinib
tucatinib
poziotinib
% Response on NRG/EGF
Drug (nM)

(A) Comparison of expression levels of HER2, HER3 determined by flow cytometry with CELx test score for 79 patient primary breast cancer cells. Fluorescence values for HER2 DAKO 3+ cells is approximately 2,000 MFC units.
(B) Percent response of stimulation of NRG1b/EGF signal (two-growth factor cocktail) when treated with a 5-point, 1,000-fold titration of the six indicated pan-HER antagonists. (Table 1, right) Average IC_{50} (nM) of pan-HER inhibitors from six primary breast cancer cell samples found with hyperactive HER-family and c-Met signaling.
IC50 values for pan-HER antagonists against the NRG1b/EGF stimulation determined from real-time live cell assay align with what is reported in cell-free assays.

Pan-HER Inhibitors	IC_{50} (nM)
Poziotinib	1.23
Neratinib	4.81
Ibrutinib	13.10
Dacomitinib	22.06
Sapitinib	41.28
Lapatinib	137.27

*Poziotinib average IC50 derived from dose response on three primary breast cancer cell samples.

Figure 4. Determining IC50 Values of c-Met Inhibitors Against HGF Agonist in a Real-Time Live Cell Assay

A

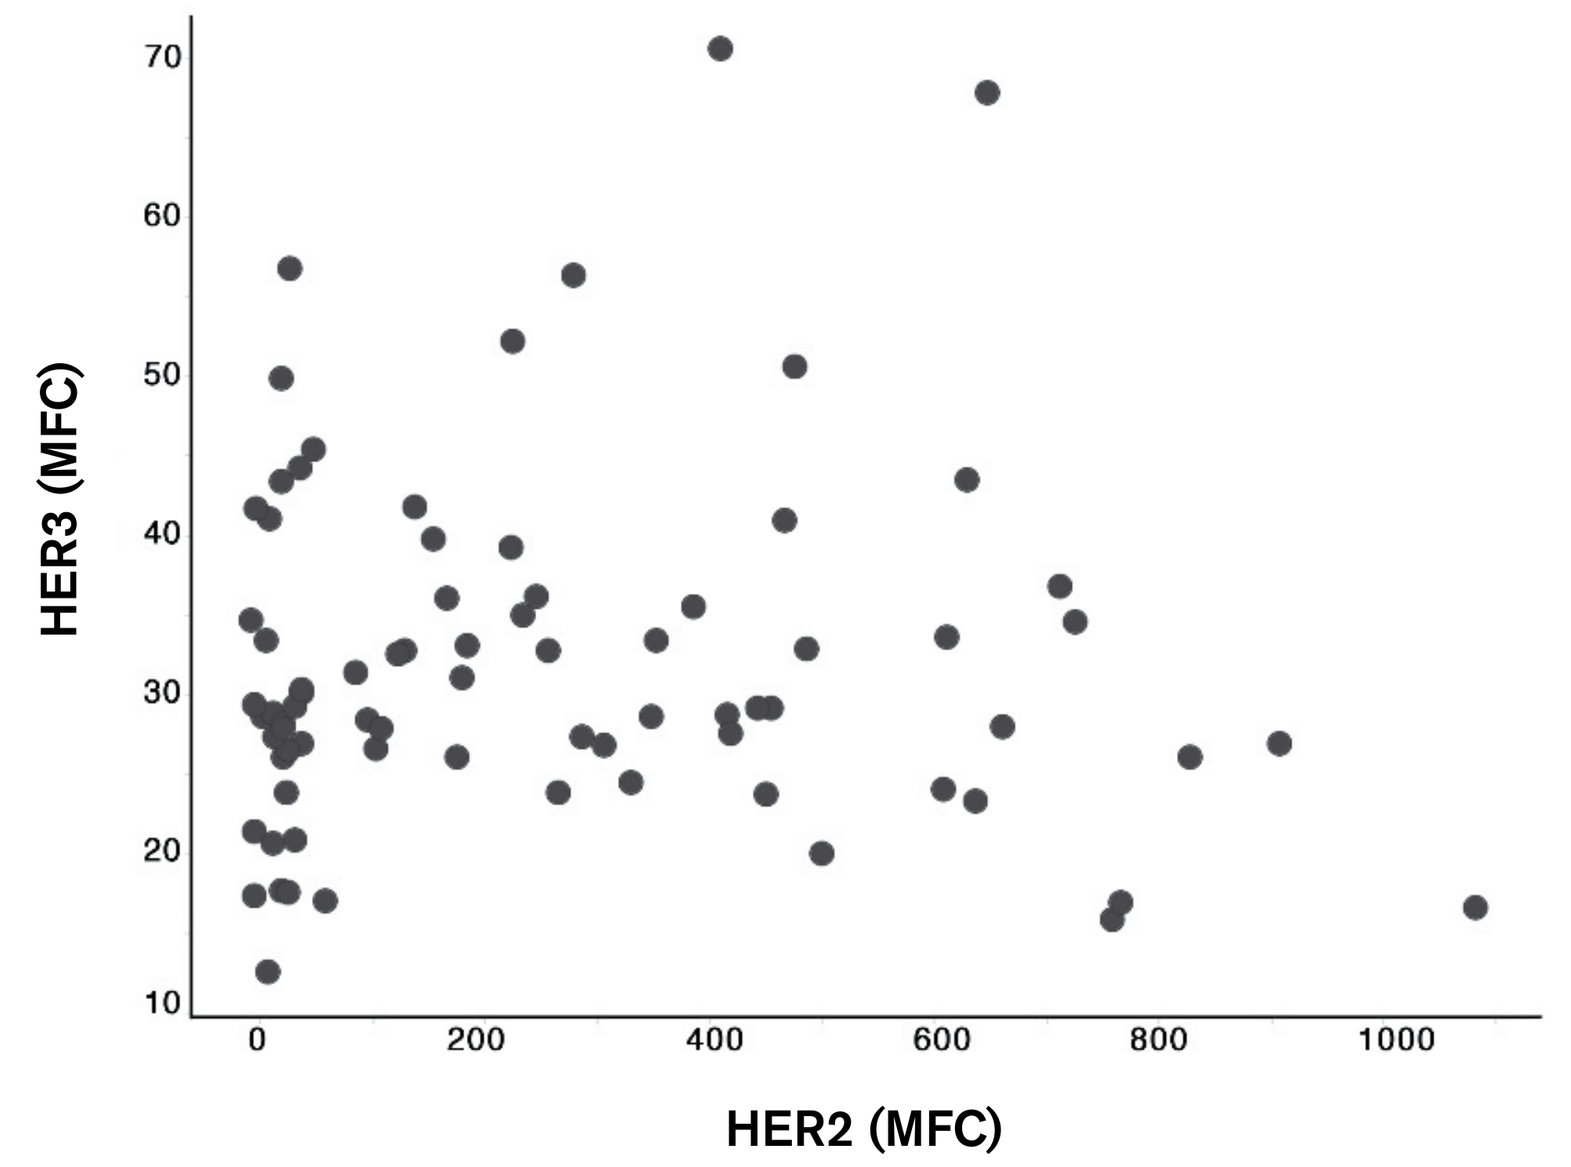
HER3 (MFC)
HER2 (MFC)

B

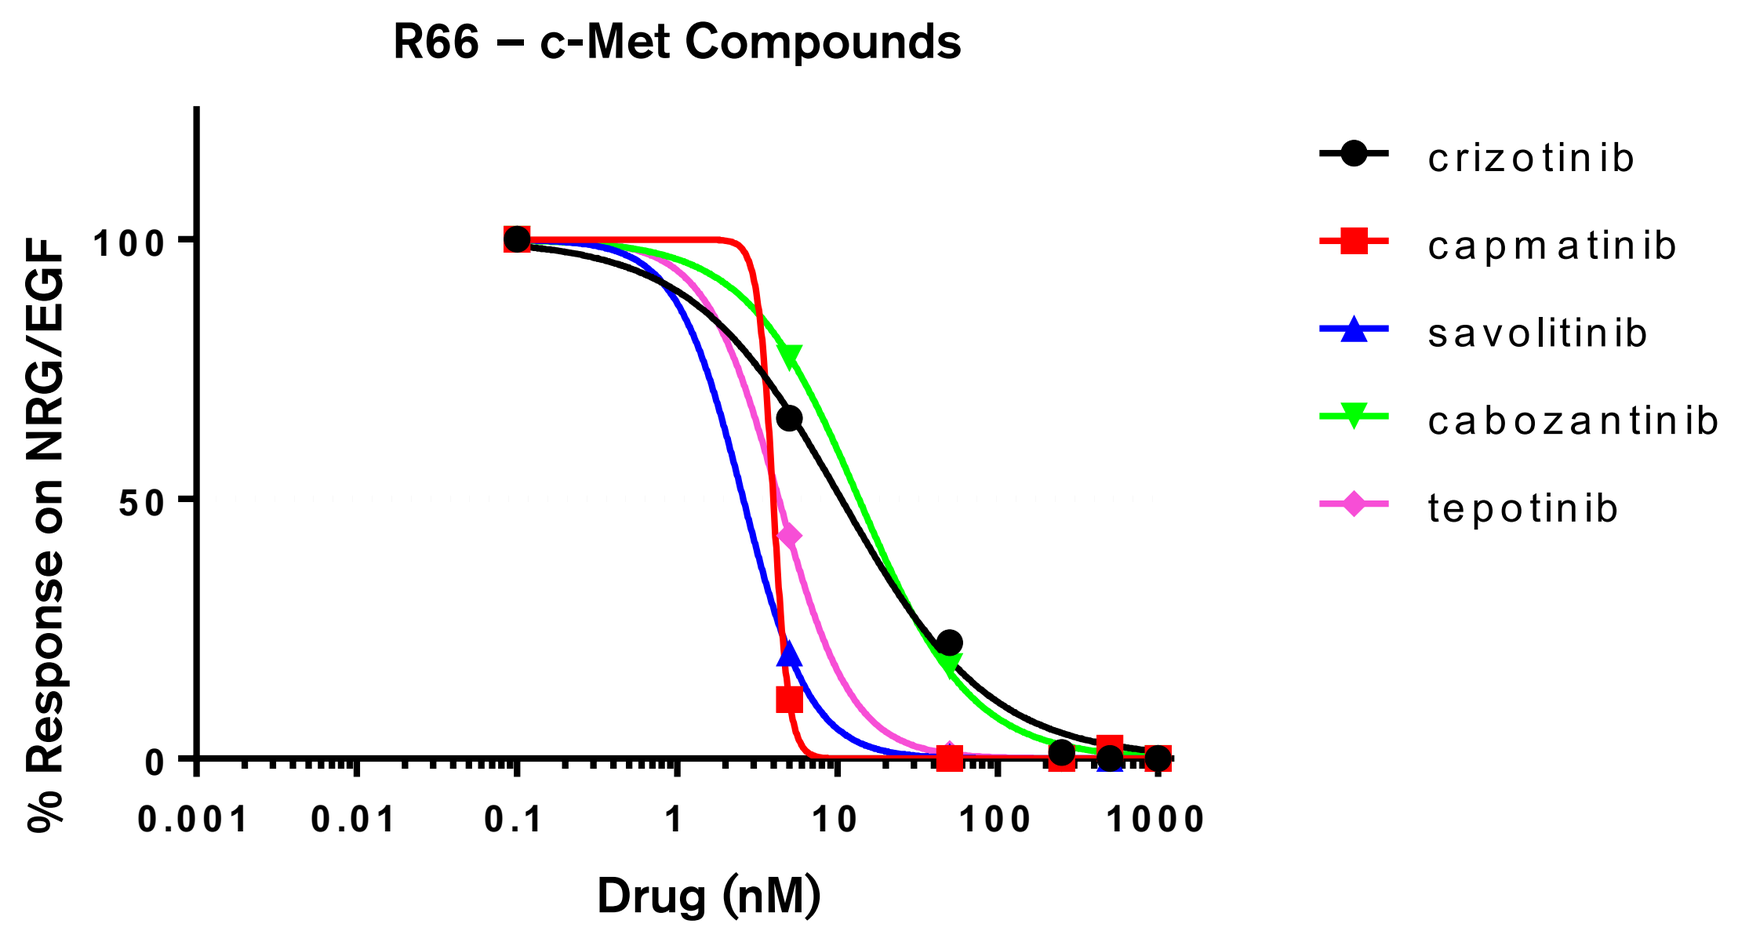
R66 – c-Met Compounds
crizotinib
capmatinib
savolitinib
cabozantinib
tepotinib
% Response on NRG/EGF
Drug (nM)

(A) Comparison of expression levels of HGF receptor (c-Met) determined by flow cytometry with HGF test score from a set of primary breast cancer cells from 79 patients.
(B) Percent response of stimulation of HGF signal when treated with a 5-point, 1,000-fold dose response of the five indicated c-Met antagonists. (Table 2, right) Average IC_{50} (nM) of c-Met inhibitors from six primary breast cancer cells with hyperactive ErbB-family and c-Met signaling.
IC50s for c-Met antagonists against the HGF response determined from real-time live cell assay align with what is reported in cell-free assays.

c-Met Inhibitors	IC_{50} (nM)
Capmatinib	3.10
Savolitinib	3.56
Tepotinib	14.70
Cabozantinib	27.36
Crizotinib	28.21

Results

Figure 5. Combinations of Pan-HER and c-Met Inhibitors Effectively Block NRG1b/EGF/HGF Agonist Cocktail in Real-Time Live Cell CELx MP Test

A

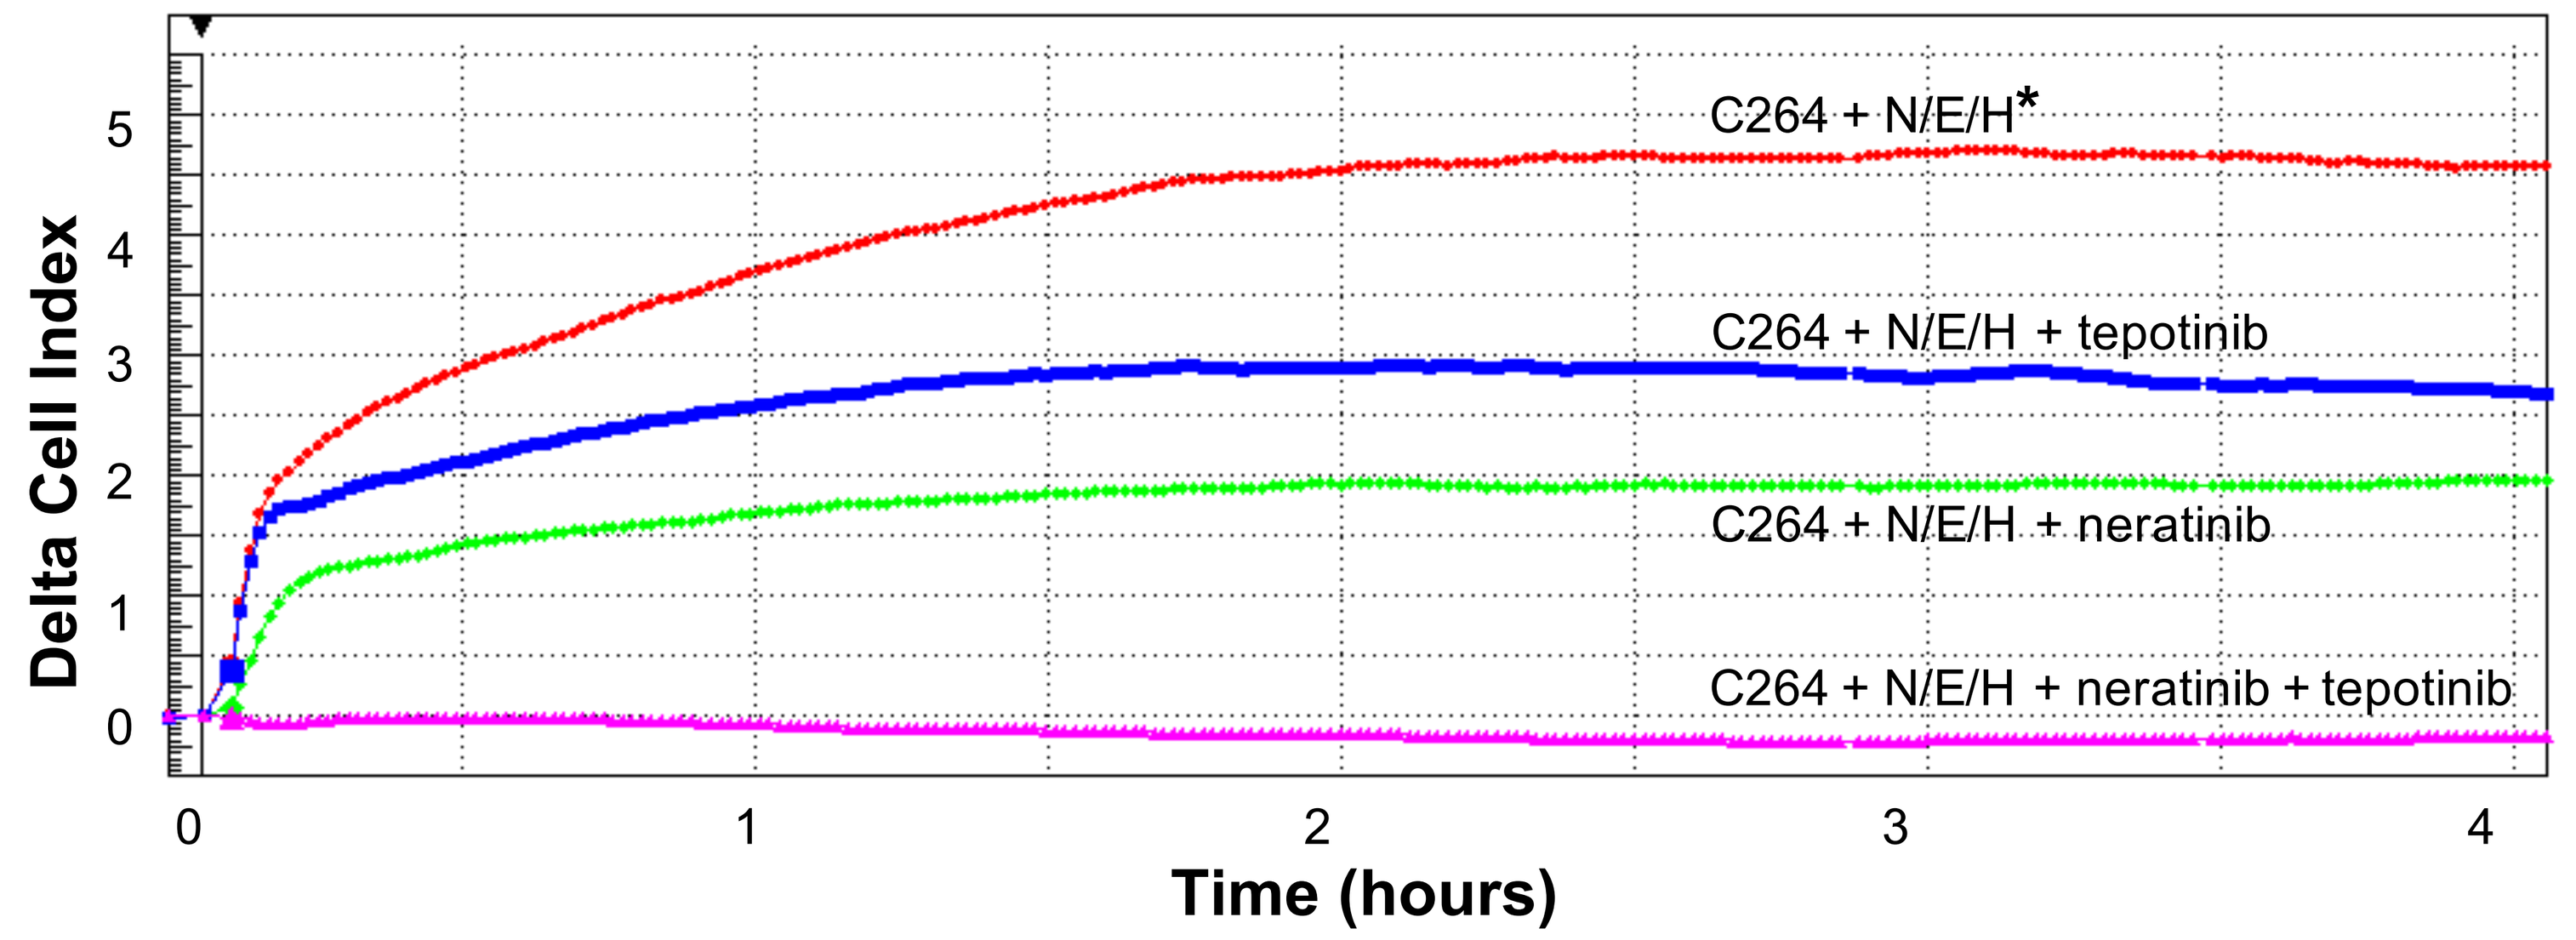
C264 + N/E/H*
C264 + N/E/H + tepotinib
C264 + N/E/H + neratinib
C264 + N/E/H + neratinib + tepotinib
Delta Cell Index
Time (hours)

B

Pan-HER Inhibitors	Average Inhibition (%) with Different c-Meti's
Poziotinib	100
Neratinib	100
Ibrutinib	99
Dacomitinib	100
Sapitinib	98
Lapatinib	80

c-Met Inhibitors	Average Inhibition (%) with Different HERi's
Capmatinib	94
Savolitinib	98
Tepotinib	96
Cabozantinib	99
Crizotinib	100

(A) CELx MP Test data showing activation by a NRG1b/EGF/HGF cocktail (N/E/H, red line) with c-Met (tepotinib) or pan-HER (neratinib) inhibitors alone or in combination. Antagonists were added 6 hours after initial seeding followed by an incubation period prior to growth factor addition.
(B) (Table 1, left) Average percent inhibition of the N/E/H signal with five different c-Met inhibitors against each pan-HER inhibitor. (Table 2, right) Average percent inhibition of the N/E/H signal with six different pan-HER inhibitors against each c-Met inhibitor.
Targeting c-Met and HER signaling pathways effectively eliminates signaling in the CELx signaling function assay.

Summary of Results

- The average IC50 values for the individual pan-HER inhibitors ranged from 1.23 nM – 137.27 nM.
- The average IC50 values for the individual c-Met inhibitors ranged from 3.10 nM – 28 nM.
- In drug efficacy studies, an average of at least 80% of the ErbB and c-MET signaling activated by NRG1b, EGF, and HGF co-stimulation was inhibited by each combination of c-Met and pan-HER inhibitors.

Conclusions

- The CELx MP test using live cells measures IC50 values comparable to those derived using cell-free methods.
- Every combination of pan-HER and c-Met inhibitors provided comparably high (at least 80%) levels of inhibitory activity effect *ex vivo*.
- These results suggest the sub-group of HER2-negative breast cancer patients diagnosed with coincident hyperactive c-Met and ErbB signaling by the CELx MP test may respond to virtually any pan-HER and c-MET inhibitor combination.
- Studying combinations designed to minimize drug toxicities without sacrificing efficacy should thus be possible.

References

1. Al-Kuraya K, et al. Prognostic relevance of gene amplifications and coamplifications in breast cancer. *Cancer Res.* 2004;64:8534-8540.
2. Dawood S, et al. Prognosis of women with metastatic breast cancer by HER2 status and trastuzumab treatment: an institutional-based review. *J Clin Oncol.* 2010;28:92-98.
3. Ross JS, et al. The HER-2 receptor and breast cancer: ten years of targeted anti-HER-2 therapy and personalized medicine. *Oncologist.* 2009;14:320-368.
4. Slamon DJ, et al. Human breast cancer: correlation of relapse and survival with amplification of the HER-2/neu oncogene. *Science.* 1987;235:177-182.
5. Perez EA, et al. HER2 testing: current status and future directions. *Cancer Treat Rev.* 2014;40:276-284.
6. Paik S, et al. HER2 status and benefit from adjuvant trastuzumab in breast cancer. *N Engl J Med.* 2008;358:1409-1411.
7. Perez EA, et al. HER2 and chromosome 17 effect on patient outcome in the N9831 adjuvant trastuzumab trial. *J Clin Oncol.* 2010;28:4307-4315.
8. Ko B, et al. MET/HGF pathway activation as a paradigm of resistance to targeted therapies. *Ann Transl Med.* 2017;5(1):4.
9. Engelman JA, et al. MET amplification leads to gefitinib resistance in lung cancer by activating ERBB3 signaling. *Science.* 2007;316(5827):1039-1043.
10. Breindel J, et al. EGF receptor activates MET through MAPK to enhance non-small cell lung carcinoma invasion and brain metastasis. *Cancer Res.* 2013;73(16):5053.
11. Mo HN and Liu P. Targeting MET in cancer therapy. *Chronic Diseases and Translational Medicine.* 2017;3:148-153.
12. Huang Y, et al. A functional signal profiling test for identifying a subset of HER2-negative breast cancers with abnormally amplified HER2 signaling activity. *Oncotarget.* 2016; in press.
13. Huang Y, et al. Development of a test that measures real-time HER2 signaling activity in live breast cancer cell lines and primary cells. *BMC Cancer.* 2016; in revision.
14. Stampfer MR, et al. Culture of human mammary epithelial cells. In: Freshney RI, Freshney MG, eds. *Culture of Epithelial Cells.* 2002.
15. Proia DA, et al. Reconstruction of human mammary tissues in a mouse model. *Nat Protoc.* 2006;1:206-214.

P2-05-05

This presentation is the intellectual property of Celcuity Inc. Contact Brian Sullivan at BSullivan@Celcuity.com for permission to reprint and/or distribute.