Mechanism of action of gedatolisib in combination with fulvestrant and/or palbociclib in estrogen receptor-positive breast cancer models

Stefano Rossetti, Aaron Broege, Megan Seibel, Jhomary Molden, Igor Gorbatchevsky, Brian Sullivan, Lance Laing
Celcuity Inc., Minneapolis, MN, USA

P2-02-05
Presented at the
San Antonio Breast Cancer Symposium
December 10-13, 2024

BACKGROUND

- The PAM (PI3K-AKT-mTOR) pathway is one of the most commonly activated oncogenic pathways in breast cancer (BC). PAM pathway dysregulation is frequently associated with mutations of PAM pathway genes, e.g., PIK3CA activating mutations and loss of PTEN function [1].
- Adaptive activation of the PAM pathway has also been associated with resistance to hormonal therapy (HT) and CDK4/6 inhibitors [2,3].
- Current standard-of-care therapy options for patients with advanced BC (ABC) include treatment with HT (e.g., letrozole, fulvestrant) in combination with CDK4/6 inhibitors (e.g., palbociclib) or PAM inhibitors (everolimus, alpelisib, capivasertib).
- Due to the crosstalk between the PAM pathway and the estrogen receptor (ER) and CDK pathways (**Figure 1**), resistance can arise through compensatory mechanisms when only one of these pathways is inhibited [4,5].
- In addition, due to feedback loops between PI3K isoforms, AKT, and mTOR that cross-activate uninhibited sub-units, PAM inhibitors that target single PAM pathway nodes cannot achieve optimal therapeutic effect, even when combined with HT [6,7].
- Nonclinical studies have shown that concomitant inhibition of PAM, ER, and CDK pathways can induce more effective and durable tumor growth inhibition by preventing or counteracting resistance mechanisms associated with inhibition of only one of these pathways [4].
- Gedatolisib is a PAM inhibitor exerting potent anti-proliferative and cytotoxic effects in BC cells. The comprehensive inhibition of the PAM pathway induced by gedatolisib affects PAM-controlled functions critical for BC progression, such as cell cycle, cell survival, protein synthesis, and glucose metabolism.
- Comparative studies have shown that gedatolisib inhibits the PAM pathway and PAM-controlled functions more effectively than single node PAM inhibitors, resulting in greater anti-proliferative and cytotoxic effects [7,8].
- In a phase 1b clinical trial in patients with HR+/HER2- ABC, the combination of gedatolisib with endocrine therapy and palbociclib showed promising efficacy and safety compared with the published results for standard-of-care therapies [9].
- The present study aimed to investigate the mechanism of action of the gedatolisib + fulvestrant + palbociclib combination and compare the effects of gedatolisib and single node PAM inhibitors in combination with fulvestrant and/or palbociclib in BC cells.

Figure 1. Crosstalk Between PAM, ER, and CDK Pathways

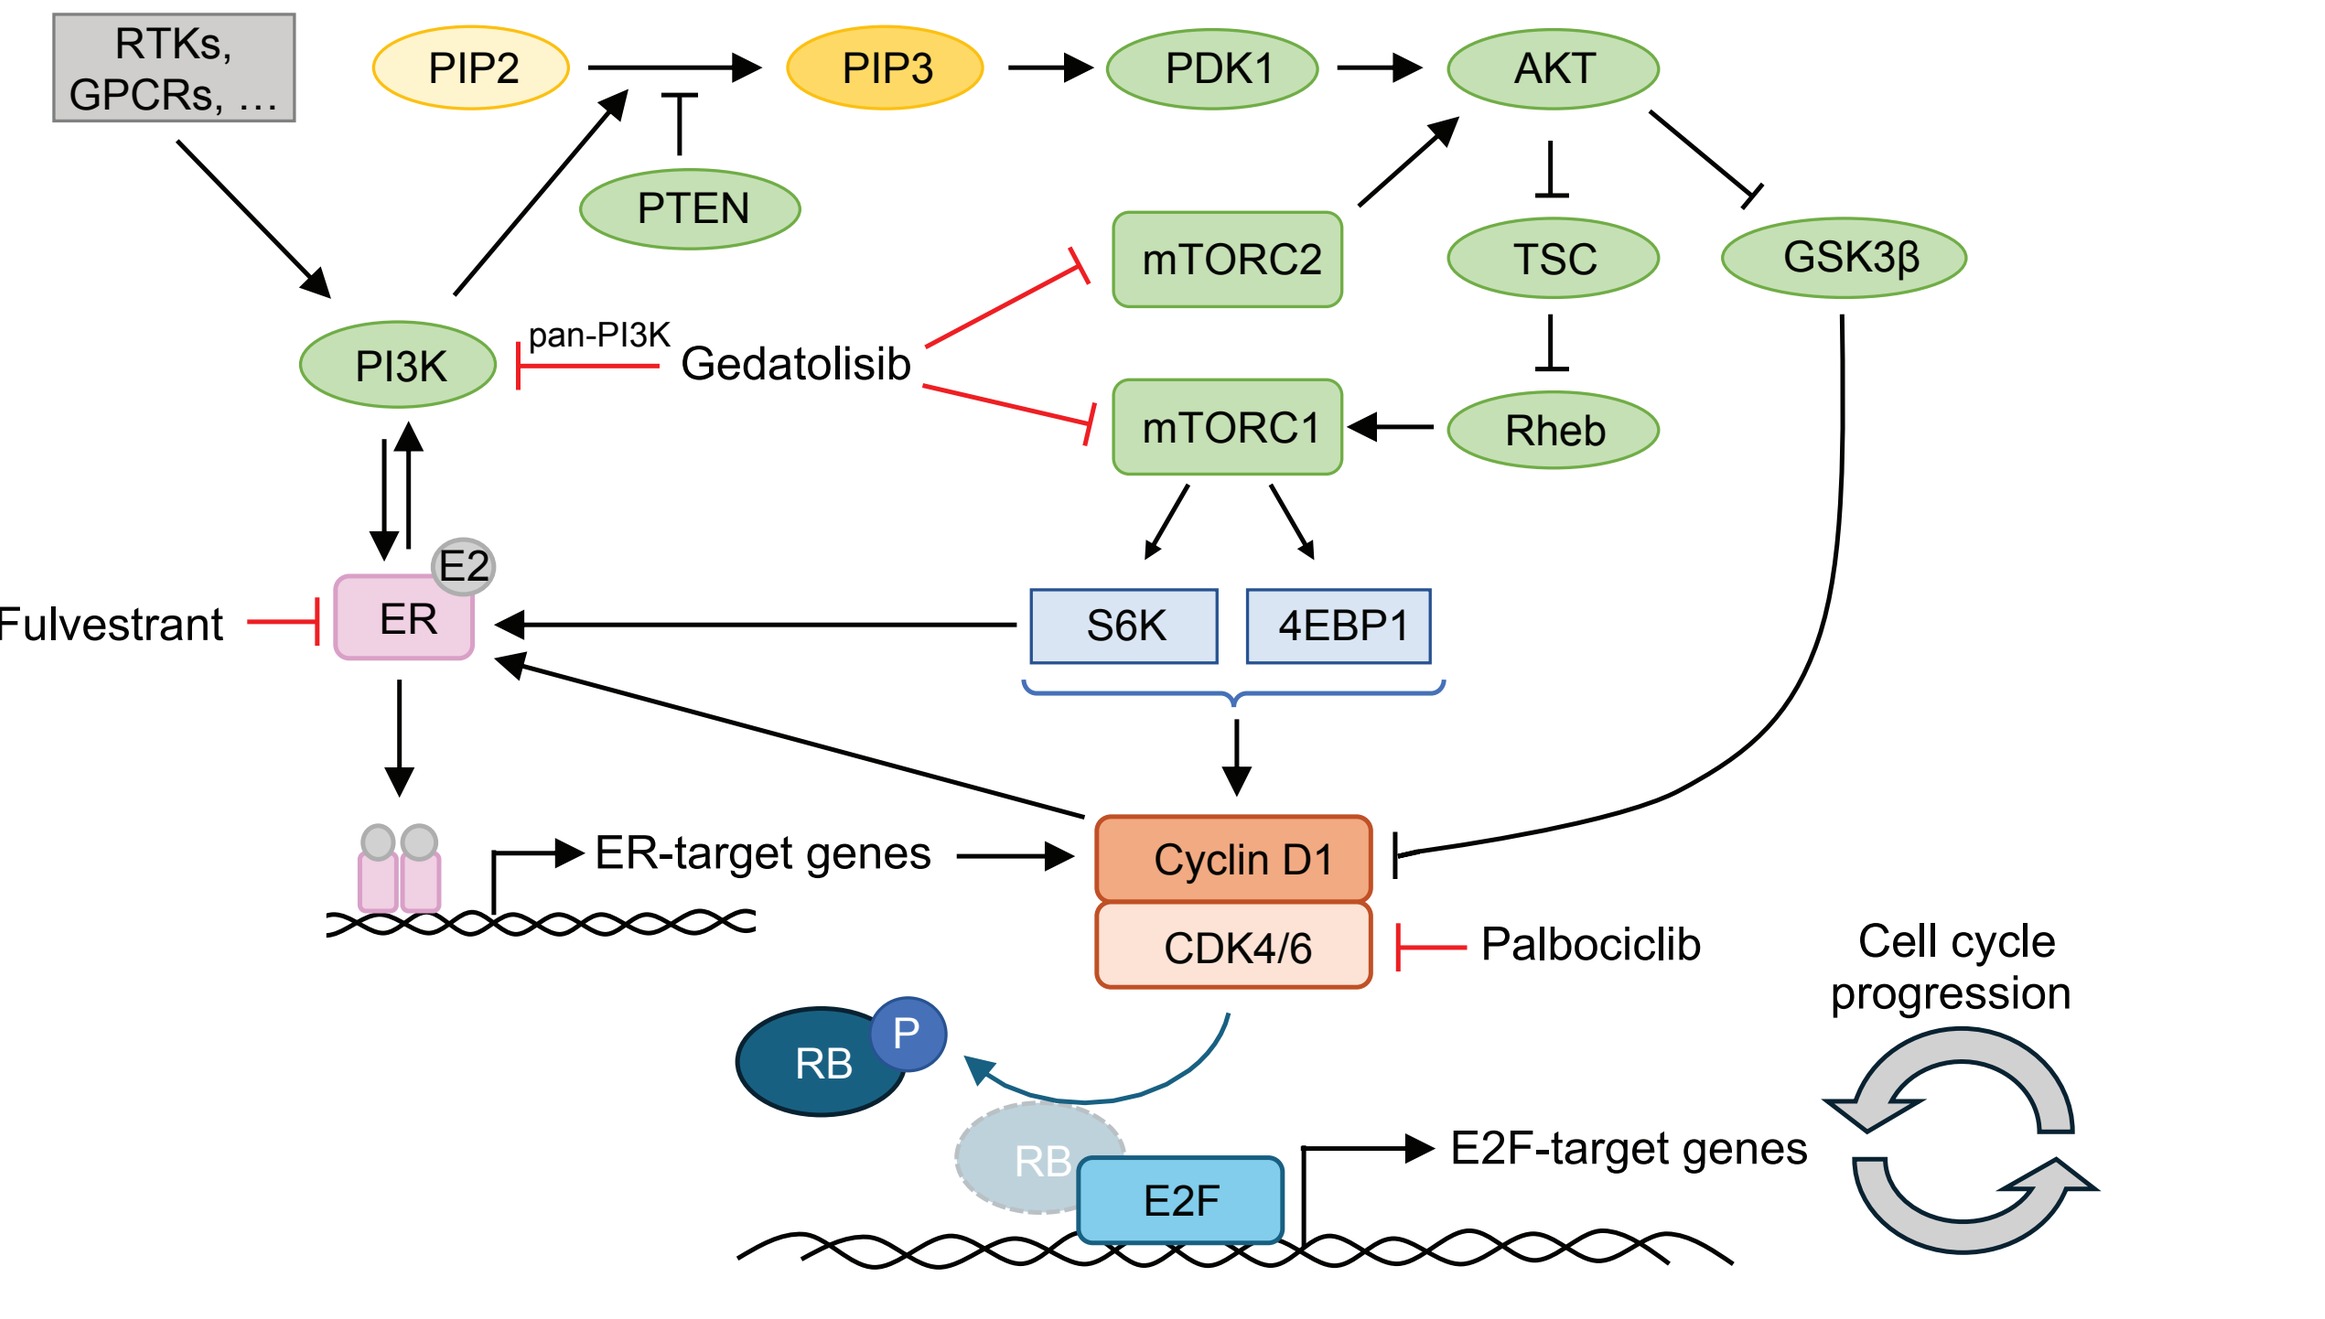
RTKs, GPCRs, …
PIP2
PIP3
PDK1
AKT
PTEN
mTORC2
TSC
GSK3β
PI3K
pan-PI3K
Gedatolisib
mTORC1
Rheb
E2
Fulvestrant
ER
S6K
4EBP1
ER-target genes
Cyclin D1
CDK4/6
Palbociclib
Cell cycle progression
RB
P
E2F
E2F-target genes

METHODS

Cell Lines. A panel of 9 ER+ BC cell lines was used in this study (see **Figure 2**). Cells were maintained according to American Type Culture Collection (ATCC) recommendations and authenticated by short tandem repeat (STR) profiling. Genetic alterations in PAM pathway genes were identified by cBioPortal (https://www.cbioportal.org) analysis of the Cancer Cell Line Encyclopedia (CCLE). Only driver alterations are shown.

Growth Rate (GR) Assays. Cells were seeded in 96-well plates were analyzed for cell viability before and after drug treatment by RT-Glo MT luciferase assay (Promega). Cell viability assessments were used to calculate GR values as described [10]. The GR approach was used to rule out confounding effects of traditional IC50 metrics, such as the number of cell divisions occurring during the assay [10].

Flow Cytometry. After treatment in 96-well plates, cells were harvested, stained with a viability dye (Zombie), fixed in 1.6% paraformaldehyde, permeabilized with methanol, stained with antibodies, and analyzed by flow cytometry on the Agilent Novocyte 3005. PAM pathway activity was assessed using an antibody against p4EBP1, a marker that integrates PAM signaling pathway outputs from PI3K/mTORC1 and mTORC2/AKT. CDK signaling was assessed by using an antibody against pRB, a key effector of the cyclinD1/CDK4/6 complex. Cell cycle phases were assessed by combining DNA staining with FxCycle with 5-ethynyl-2'-deoxyuridine (EdU) incorporation Click-iT assay (Invitrogen). Cell death was assessed by staining with Zombie dye (identifying dead cells with compromised membrane permeability). Protein synthesis was assessed by O-propargyl-puromycin (OPP) incorporation followed by Click-iT reaction (Invitrogen).

Metabolic Studies. Cells were seeded in 96-well plates and treated with the indicated drugs for 24 hours and analyzed for glucose uptake by using the Glucose Uptake-Glo assay (Promega). Lactate levels in the conditioned medium were measured using the Biosen R-line instrument (EKF Diagnostic Holdings) and used to calculate lactate consumption. Glucose uptake and lactate production values were normalized to cell number.

Colony Formation Assay. Cells were seeded at low density (1000 cells/plate) in 6-well plates, allowed to attach, and treated for 3 or 6 days. After treatment, the drug was removed by media exchange, and colonies were allowed to grow for a total of 2-3 weeks and stained by crystal violet. After imaging, crystal violet was eluted with 33% acetic acid to assess colony growth by spectrophotometric analysis.

Animal Studies. Estrogenized female SCID mice were inoculated in the mammary fat pad with $5x10^6$ MCF7 cells. When tumors reached ~230 mm^3, mice (N=10/arm) were randomly assigned to either a control vehicle group; or treatment groups that received gedatolisib, fulvestrant, palbociclib, or various combinations of the three drugs (see **Figure 9**). Fulvestrant was dosed subcutaneously (s.q.) once a day (QD) for 3 days, then once every 3 days (Q3D). Palbociclib was dosed orally (PO) QD. Gedatolisib was dosed intravenously (i.v.) once every 4 days (Q4D). Tumor volumes were measured twice a week with a digital caliper.

REFERENCES:
[1] Millis SZ, et al. *JAMA Oncol.* 2016;2:1565-1573.
[2] Vasan N, et al. *Ann Oncol.* 2019;30:x3-x11.
[3] Goel S, et al. *Nat Rev Cancer.* 2022;22:356-372.
[4] Alves CL, et al. *Int J Mol Sci.* 2023;24.
[5] Clark AS, et al. *Clin Cancer Res.* 2021;27:371-373.
[6] Thorpe LM, et al. *Nat Rev Cancer.* 2015;15:7-24.
[7] Rossetti S, et al. *NPJ Breast Cancer.* 2024;10:40.
[8] Sen A, et al. *Mol Oncol.* 2024.
[9] Layman RM, et al. *Lancet Oncol.* 2024;25:474-487.
[10] Hafner M, et al. *Nat Methods.* 2016;13:521-527.
[11] Mallon R, et al. *Clin Cancer Res.* 2011;17:3193-3203.
[12] Fritsch C, et al. *Mol Cancer Ther.* 2014;13:1117-1129.
[13] Addie M, et al. *J Med Chem.* 2013;56:2059-2073.
[14] Sedrani R, et al. *Transplant Proc.* 1998;30:2192-2194.

RESULTS

Figure 2. Effects of Gedatolisib in Combination with Fulvestrant and Palbociclib on Growth Rate (GR) Metrics

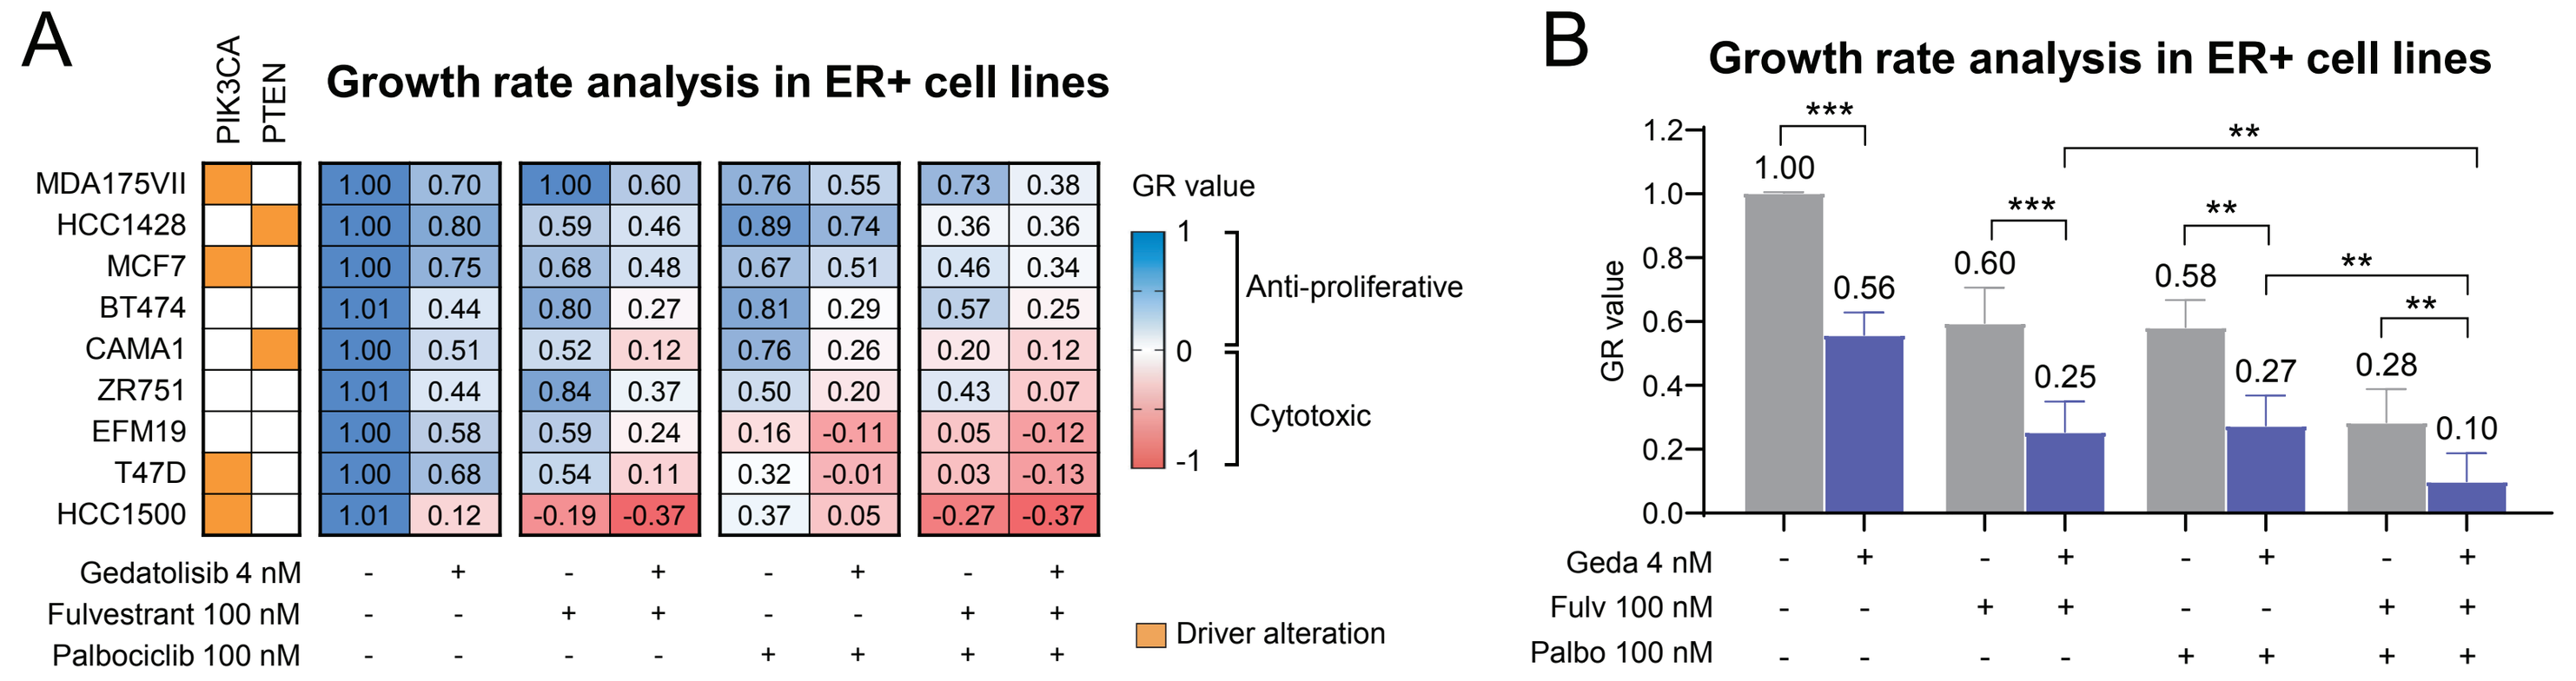
A
Growth rate analysis in ER+ cell lines
PIK3CA
PTEN
MDA175VII
HCC1428
MCF7
BT474
CAMA1
ZR751
EFM19
T47D
HCC1500
Gedatolisib 4 nM
Fulvestrant 100 nM
Palbociclib 100 nM
GR value
Anti-proliferative
Cytotoxic
Driver alteration
B
Growth rate analysis in ER+ cell lines
GR value
Geda 4 nM
Fulv 100 nM
Palbo 100 nM

GR metrics were used to assess the anti-proliferative and cytotoxic effects of gedatolisib, fulvestrant, and palbociclib in various combinations. A panel of nine ER+ BC cell lines was treated for 6 days and analyzed by RTGlo MT viability assay to assess anti-proliferative and cytotoxic effects of the drug combinations. GR values between 0 and 1 indicate anti-proliferative effects; GR = 0 indicates complete cytostasis; GR values < 0 indicate cytotoxic effects, where -1 indicates complete cell killing. GR values for each cell line are shown in **(A)**, while the mean GR values of all nine cell lines +/- standard error are shown in **(B)**. ** $P < 0.01$, *** $P < 0.001$

- **The triplet combination of gedatolisib + fulvestrant + palbociclib exerted greater anti-proliferative and cytotoxic effects than the single agents or doublet combinations in ER+ BC cells lines.**
- **The triplet combination was effective regardless of *PIK3CA/PTEN* genetic alterations.**

Figure 3. Effects of Gedatolisib/Fulvestrant/Palbociclib on PAM and CDK Signaling Pathways

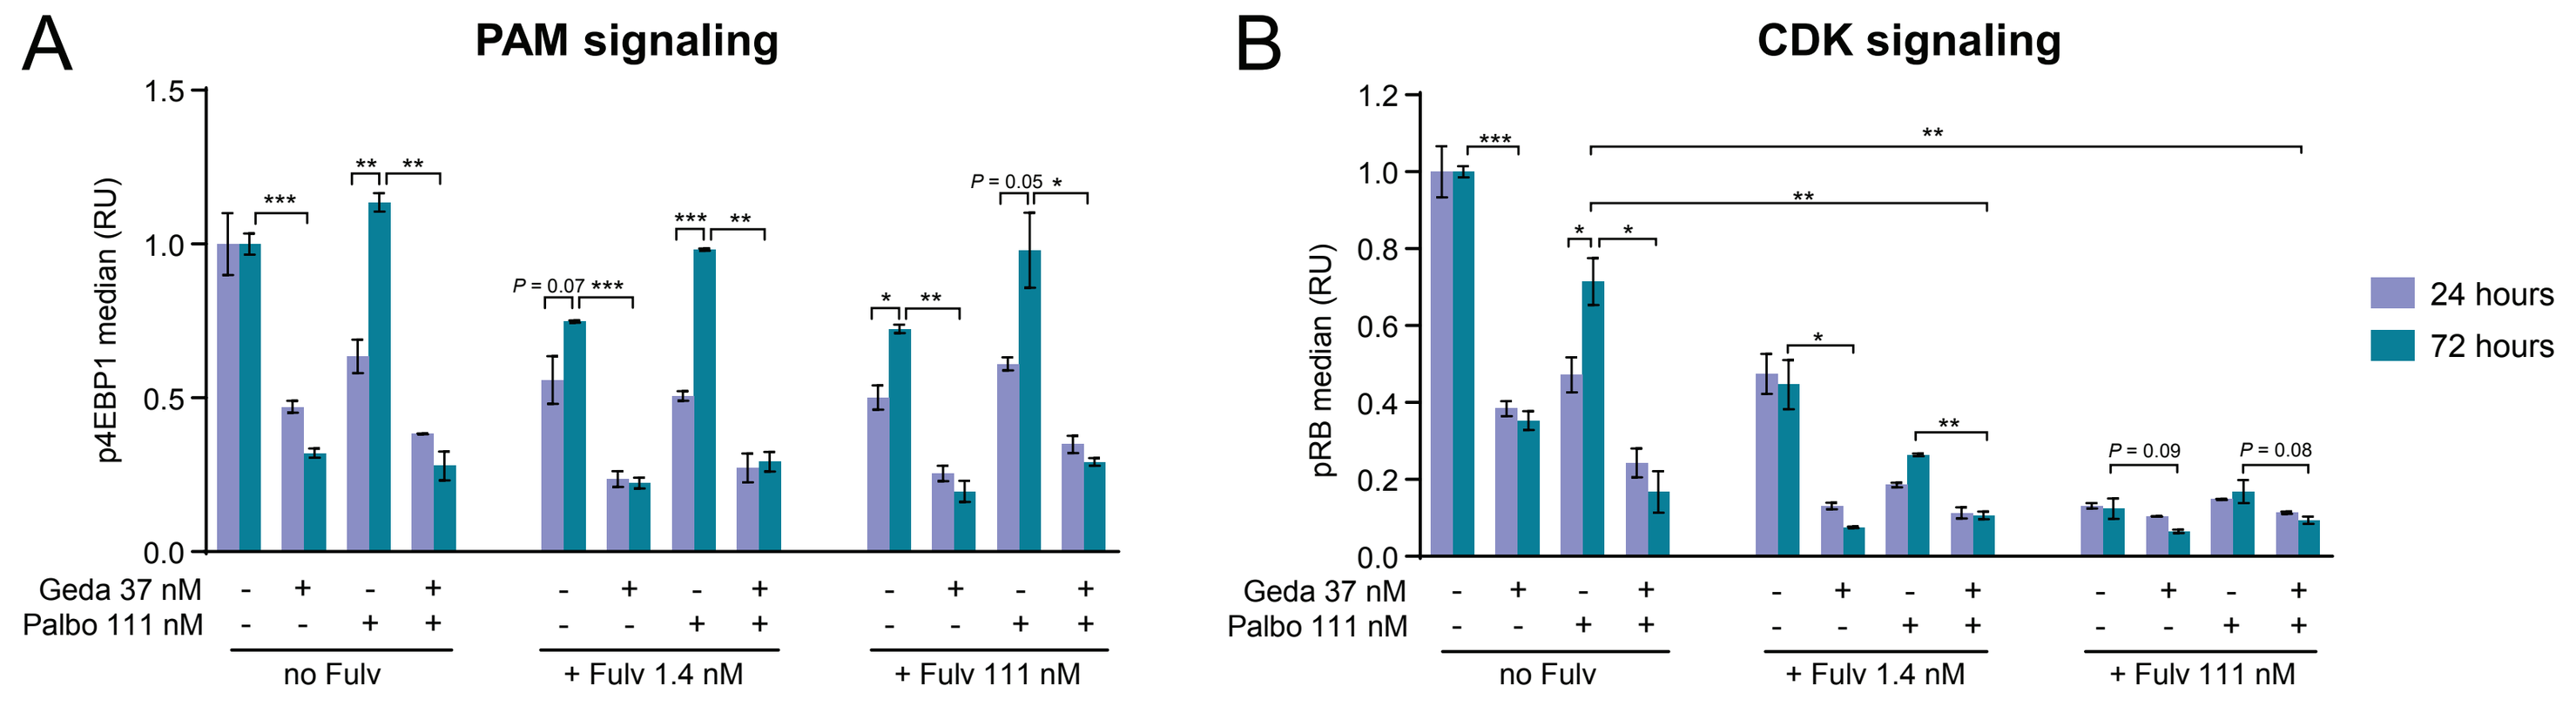
A
PAM signaling
p4EBP1 median (RU)
B
CDK signaling
pRB median (RU)
24 hours
72 hours
Geda 37 nM
Palbo 111 nM
no Fulv
+ Fulv 1.4 nM
+ Fulv 111 nM

MCF7 cells were treated with various combinations of gedatolisib, fulvestrant, and palbociclib for 24 or 72 hours and tested for **(A)** PAM pathway activity by flow cytometry analysis of p4EBP1; and **(B)** CDK pathway activity by flow cytometry analysis of phosphorylated RB (pRB). Data are relative to DMSO-treated cells (set as 1) and represent mean +/- SD (n=2-4). * $P < 0.05$, ** $P < 0.01$, *** $P < 0.001$

- **Treatment with palbociclib and/or fulvestrant triggered the following early adaptations:**
 - ✓ **Recovery of PAM signaling after 72-hour treatment with palbociclib and/or fulvestrant**
 - ✓ **Recovery of CDK signaling after 72-hour treatment with palbociclib**
- **Gedatolisib prevented PAM and/or CDK signaling recovery in response to palbociclib or fulvestrant.**
- **The triplet combination induced effective and durable inhibition of PAM and CDK pathways.**

Figure 4. Effects of Gedatolisib/Fulvestrant/Palbociclib on Cell Cycle

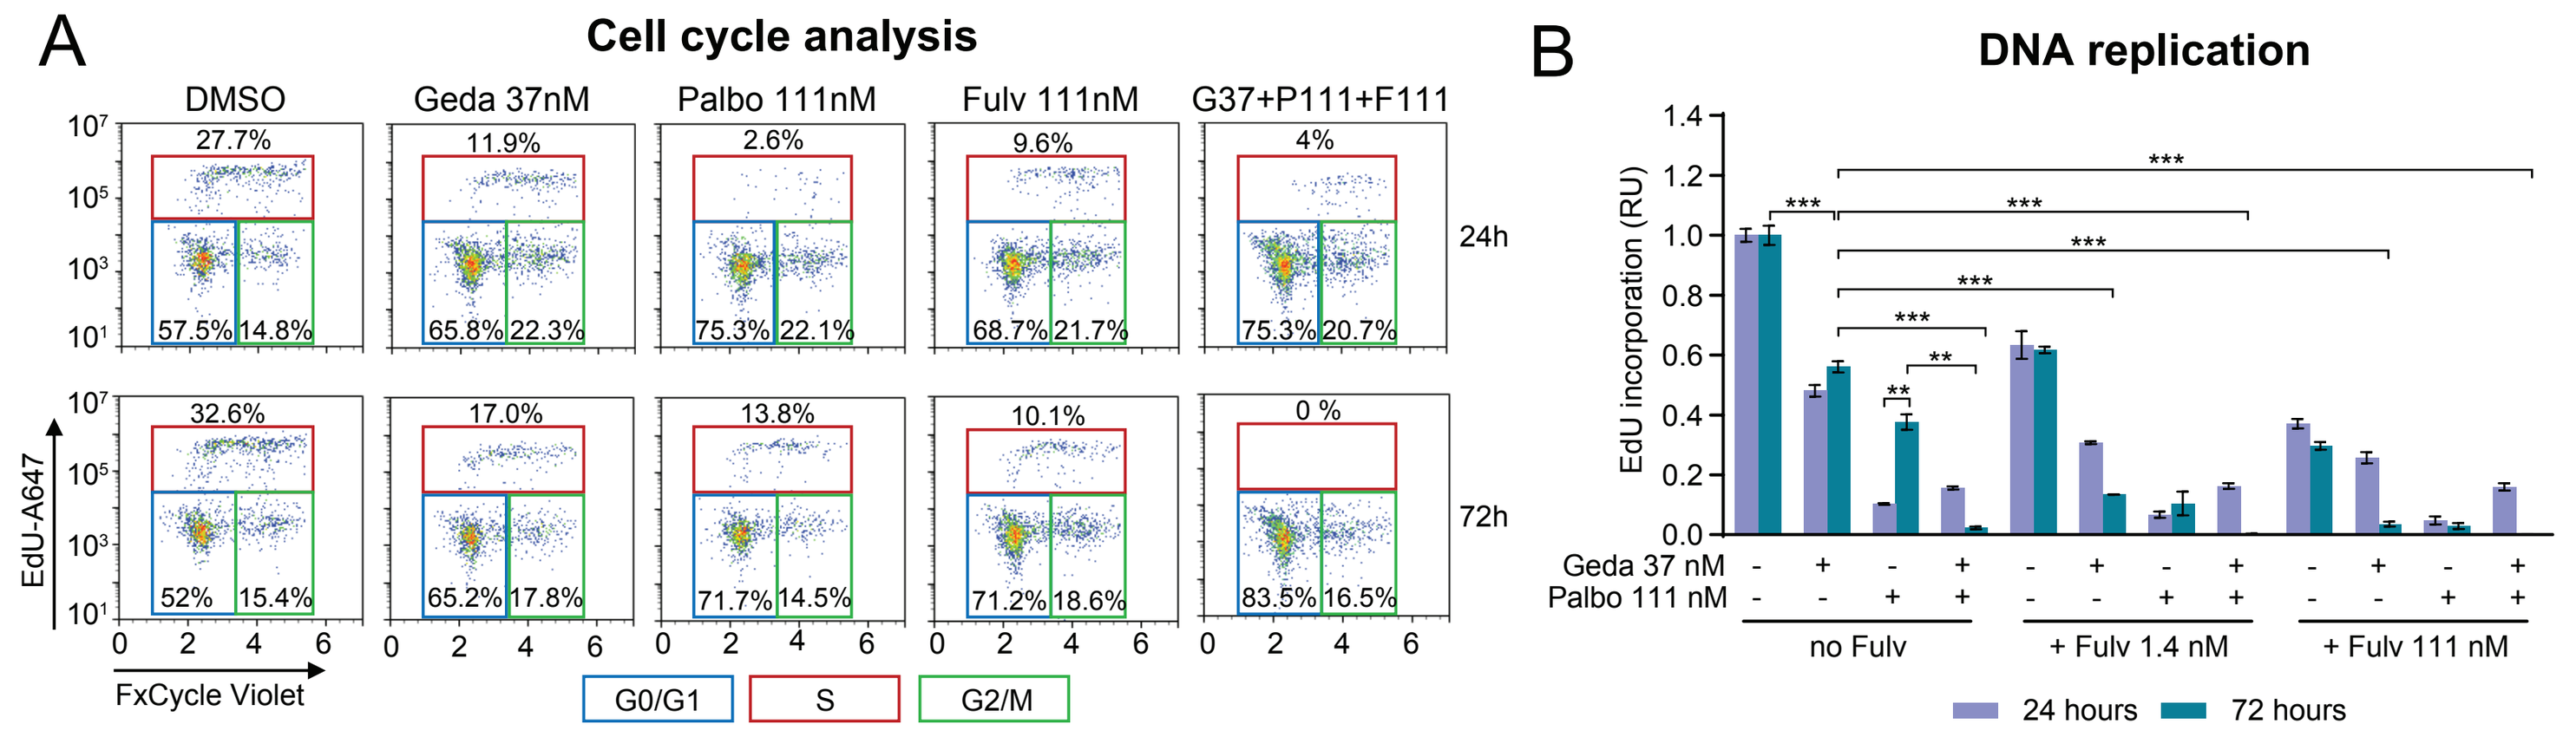
A
Cell cycle analysis
DMSO
Geda 37nM
Palbo 111nM
Fulv 111nM
G37+P111+F111
24h
72h
EdU-A647
FxCycle Violet
G0/G1
S
G2/M
B
DNA replication
EdU incorporation (RU)
Geda 37 nM
Palbo 111 nM
no Fulv
+ Fulv 1.4 nM
+ Fulv 111 nM
24 hours
72 hours

MCF7 cells were treated with various combinations of gedatolisib, fulvestrant, and palbociclib for 24 or 72 hours and analyzed for cell cycle phases and DNA replication by flow cytometry analysis of DNA content (assessed by FxCycle staining) and incorporation of the nucleoside analog EdU into newly synthesized DNA. **(A)** Examples of cell cycle analysis identifying S, G0/G1, and G2/M phases by FxCycle/EdU gating of live cells. **(B)** Quantification of EdU incorporation under different treatment conditions. Data are relative to DMSO-treated cells (set as 1) and represent mean +/- SD (n=2-4). ** $P < 0.01$, *** $P < 0.001$

- **Palbociclib induced transient cell cycle inhibition at 24 hours, followed by partial rebound at 72 hours while the drug was still present.**
- **Gedatolisib and fulvestrant prevented cell cycle recovery after palbociclib treatment rebound.**
- **The triplet combination gedatolisib + fulvestrant + palbociclib induced effective and durable inhibition of cell cycle progression without rebound.**

Figure 5. Effects of Gedatolisib/Fulvestrant/Palbociclib on Cell Survival

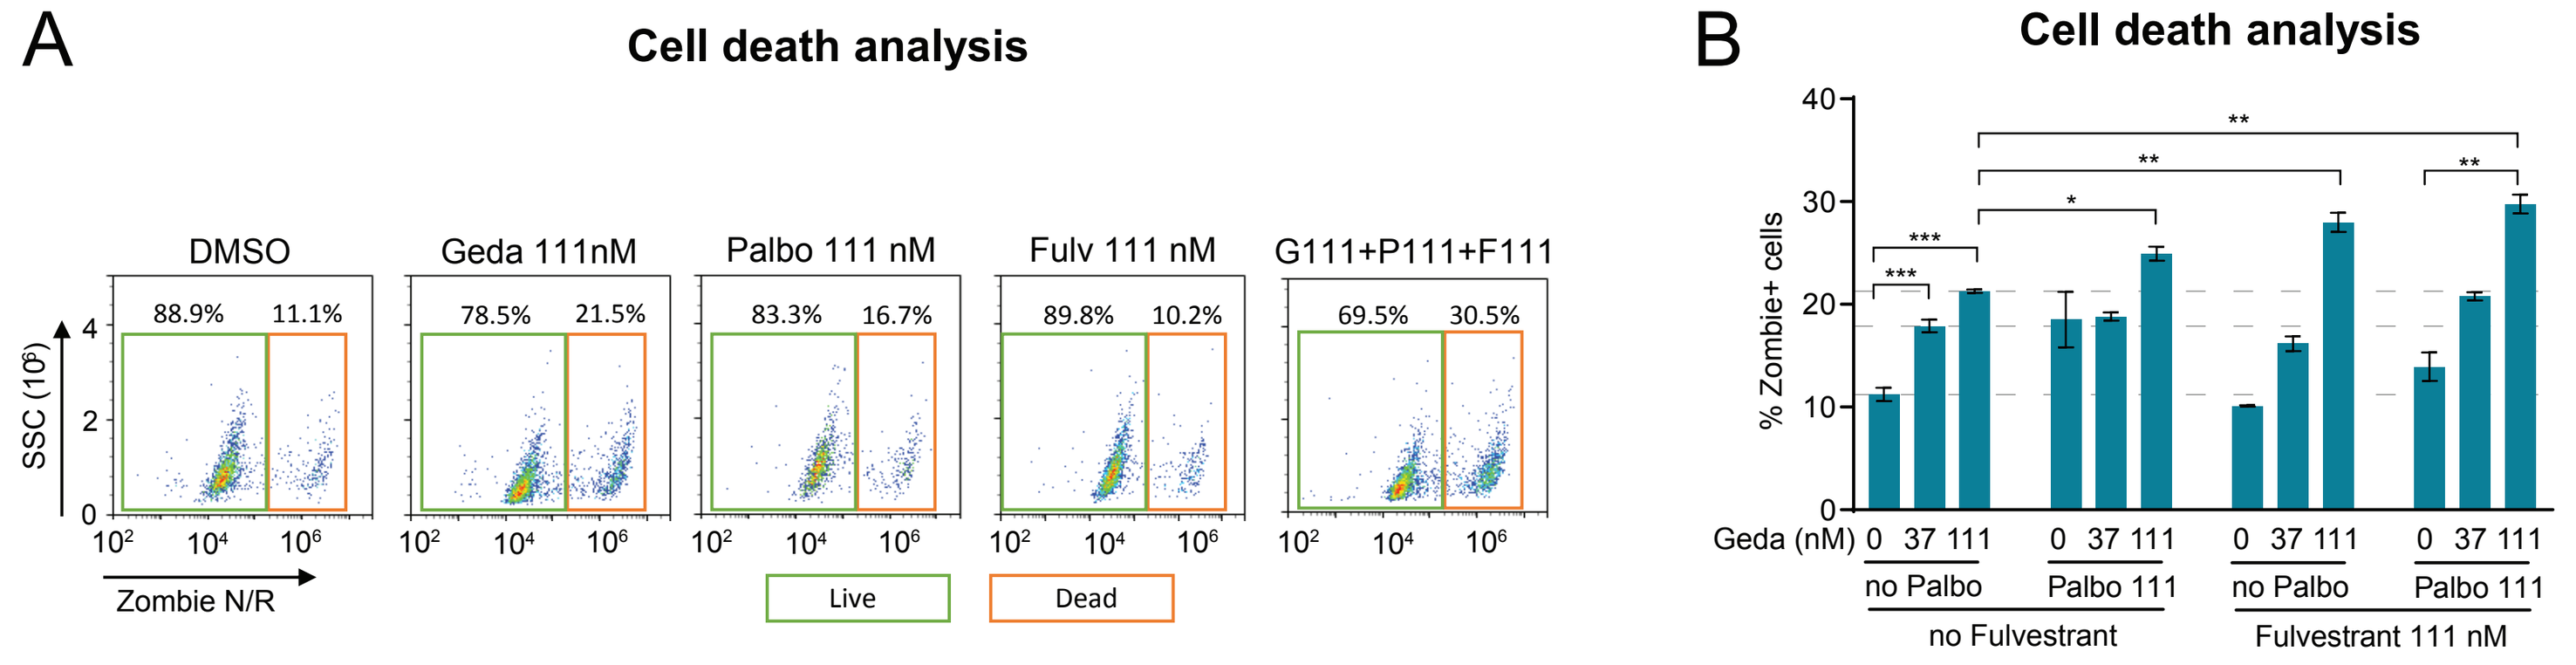
A
Cell death analysis
DMSO
Geda 111nM
Palbo 111 nM
Fulv 111 nM
G111+P111+F111
SSC
Zombie N/R
Live
Dead
B
Cell death analysis
% Zombie+ cells
Geda (nM)
no Palbo
Palbo 111
no Fulvestrant
Fulvestrant 111 nM

MCF7 cells were treated with various combinations of gedatolisib, fulvestrant, and palbociclib for 72 hours and analyzed by flow cytometry for cell death using Zombie staining. **(A)** Examples of flow cytometry plots gating live and dead cells. **(B)** Quantification of Zombie+ cells (dead cells) under different treatment conditions. Data represent mean +/- SD (n=2-4). * $P < 0.05$, ** $P < 0.01$, *** $P < 0.001$

- **Gedatolisib induced dose-dependent cell death as single agent.**
- **The triplet combination gedatolisib + fulvestrant + palbociclib induced more cell death than gedatolisib alone.**
- **The gedatolisib + palbociclib and gedatolisib + fulvestrant doublets induced more cell death than the palbociclib + fulvestrant doublet.**

Figure 6. Effects of Gedatolisib/Fulvestrant/Palbociclib on Protein Synthesis

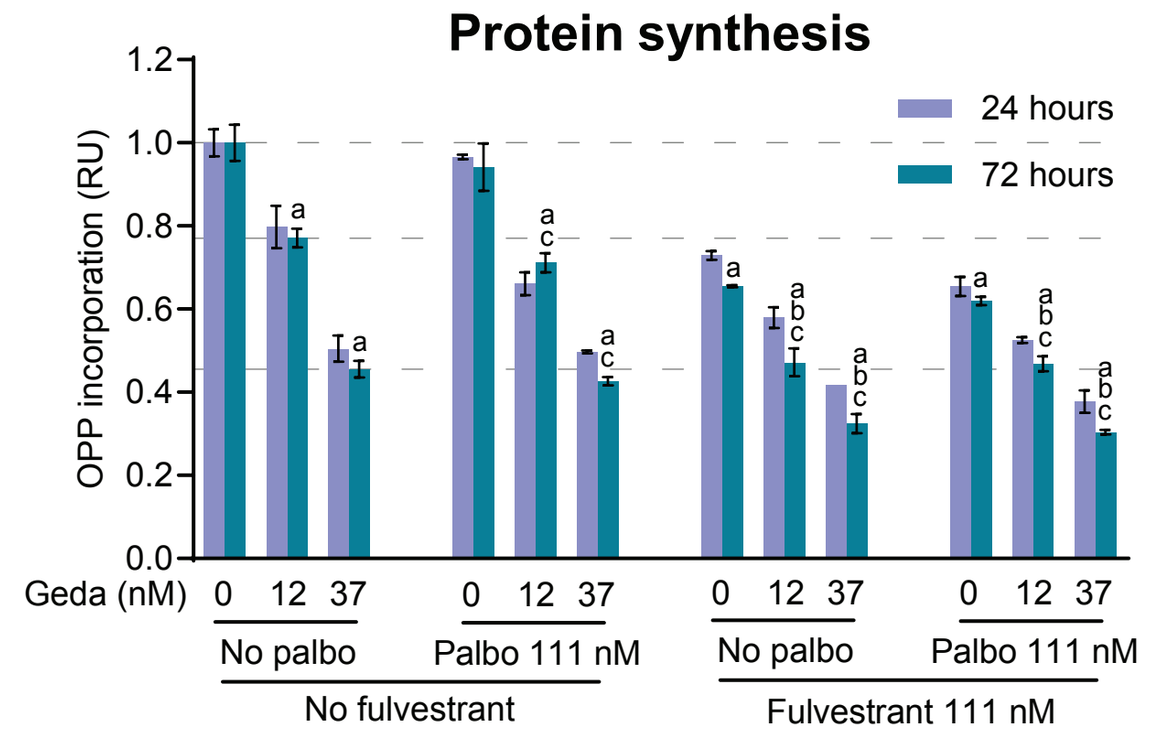
Protein synthesis
OPP incorporation (RU)
24 hours
72 hours
Geda (nM)
No palbo
Palbo 111 nM
No fulvestrant
Fulvestrant 111 nM

MCF7 cells were treated with various combinations of gedatolisib, fulvestrant, and palbociclib for 24 or 72 hours and analyzed for protein synthesis by flow cytometry analysis of OPP incorporation into newly synthesized proteins. Data are relative to DMSO-treated cells (set as 1) and represent mean +/- SD (n=2-4). a = $P < 0.05$ versus DMSO, b = $P < 0.05$ versus gedatolisib only, c = $P < 0.05$ versus no gedatolisib within group.

- **The triplet combination gedatolisib + fulvestrant + palbociclib reduced protein synthesis significantly more than gedatolisib alone or the palbociclib + fulvestrant doublet.**
- **The gedatolisib + palbociclib and gedatolisib + fulvestrant doublets each reduced protein synthesis more effectively than the palbociclib + fulvestrant doublet.**

Figure 7. Effects of Gedatolisib/Fulvestrant/Palbociclib on Glucose Metabolism

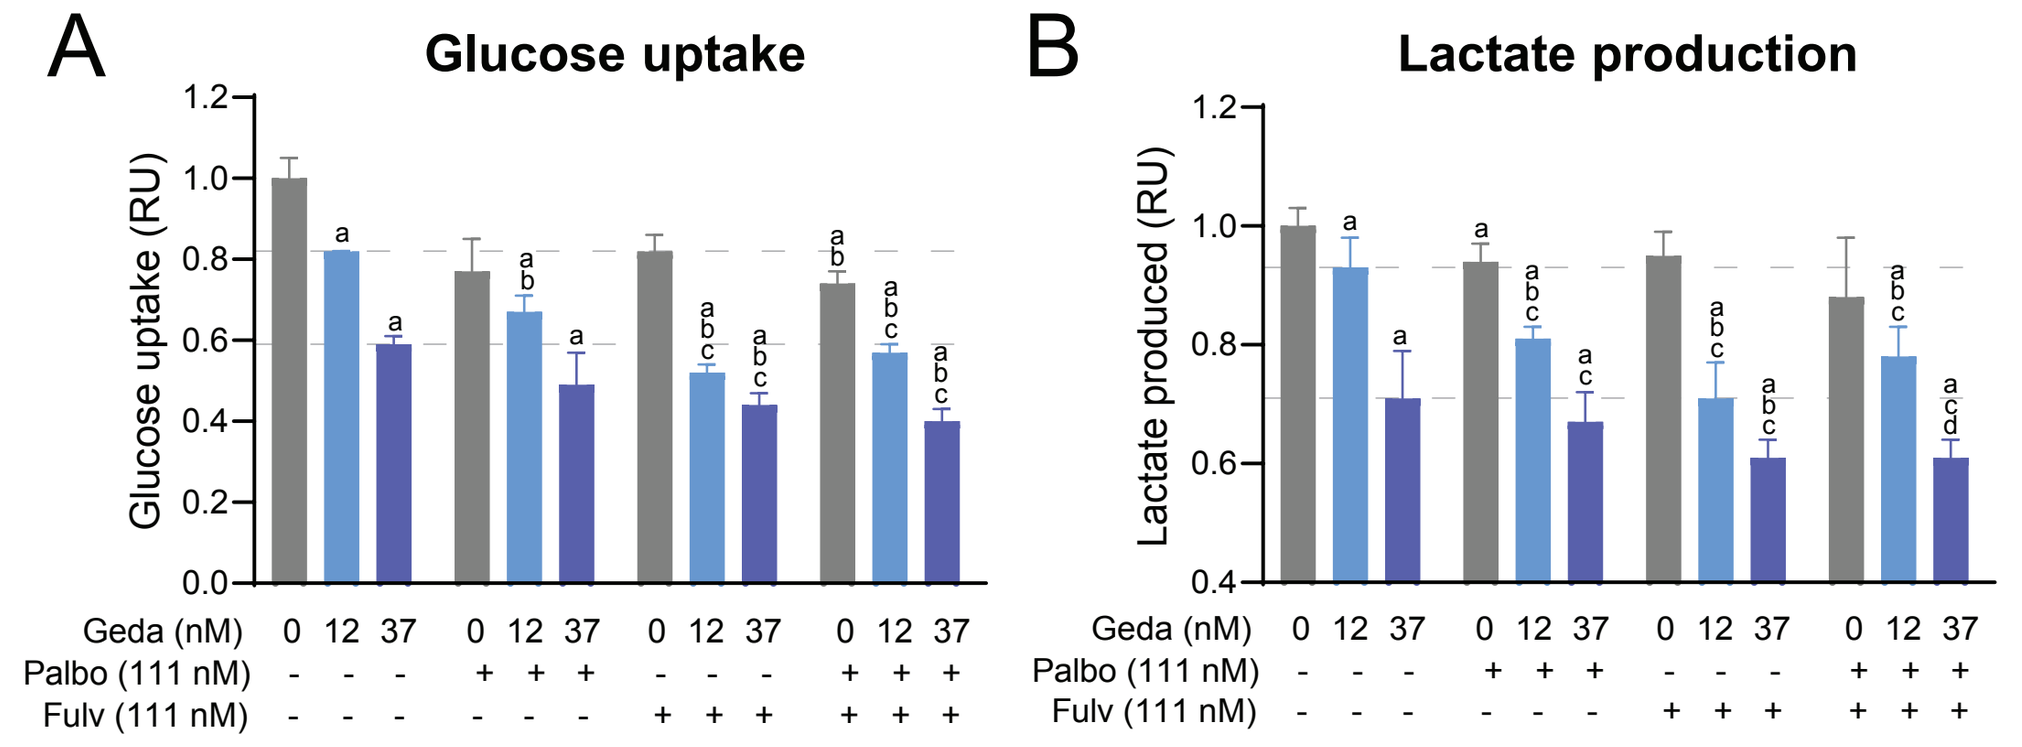
A
Glucose uptake
Glucose uptake (RU)
B
Lactate production
Lactate produced (RU)
Geda (nM)
Palbo (111 nM)
Fulv (111 nM)

MCF7 cells were treated with various combinations of gedatolisib, fulvestrant, and palbociclib for 24 hours and analyzed for **(A)** glucose uptake and **(B)** lactate production, the end product of glycolysis. Data are relative to DMSO-treated cells (set as 1) and represent mean +/- SD (n=2-4). a = $P < 0.05$ versus DMSO, b = $P < 0.05$ versus gedatolisib only, c = $P < 0.05$ versus no gedatolisib within group; d = $P = 0.052$ versus gedatolisib only.

- **The triplet combination gedatolisib + fulvestrant + palbociclib reduced glucose uptake and glycolysis significantly more than gedatolisib alone or the palbociclib + fulvestrant doublet.**
- **The gedatolisib + palbociclib and gedatolisib + fulvestrant doublets each reduced glucose uptake and glycolysis more effectively than the palbociclib + fulvestrant doublet.**

Figure 8. Long-term Effects of Gedatolisib/Fulvestrant/Palbociclib on Cell Growth

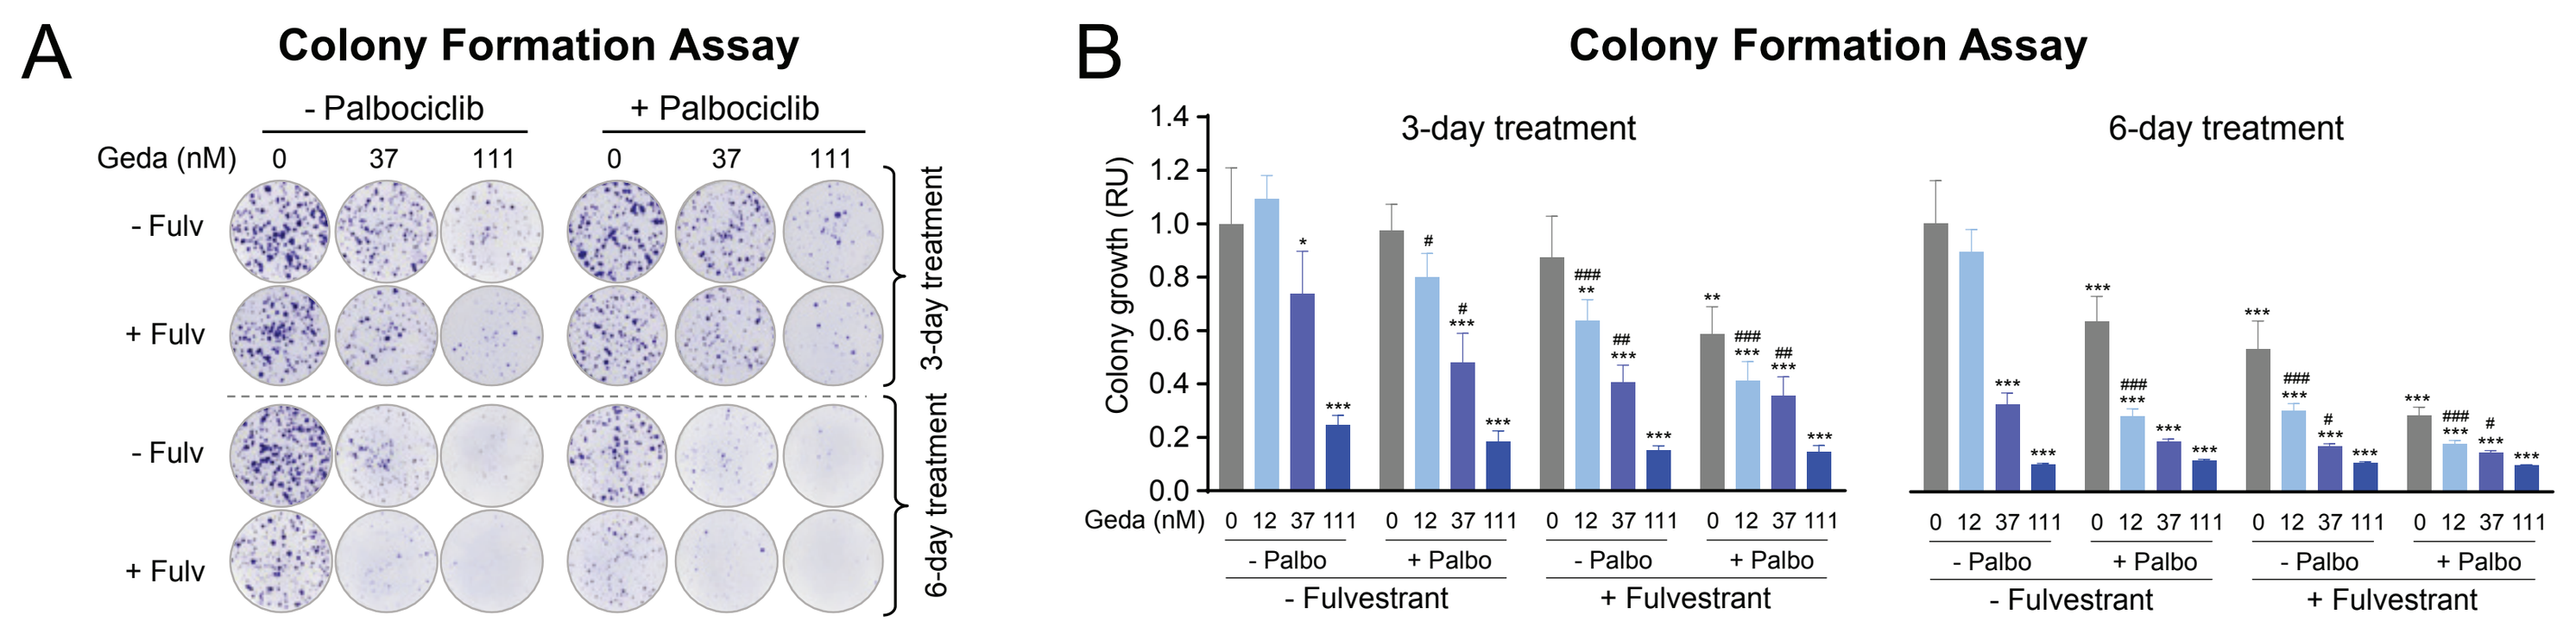
A
Colony Formation Assay
- Palbociclib
+ Palbociclib
Geda (nM)
- Fulv
+ Fulv
3-day treatment
6-day treatment
B
Colony Formation Assay
3-day treatment
6-day treatment
Colony growth (RU)
- Palbo
+ Palbo
- Fulvestrant
+ Fulvestrant

MCF7 cells were treated with 37-111 nM gedatolisib, 1.4 nM fulvestrant, and/or 111 nM palbociclib for 3 or 6 days and let grow for 2-3 weeks until discrete colonies were visible. Colonies were stained by crystal violet **(A)** and colony growth was quantified by spectrophotometric analysis of eluted crystal violet **(B)**. Data in B are relative to DMSO-treated cells (set as 1) and represent mean +/- SD (n=3). *,**,*** = $P < 0.05$, 0.01, 0.001 versus DMSO; #, ##, ### = $P < 0.05$, 0.01, 0.001 versus gedatolisib.

- **The triplet combination gedatolisib + fulvestrant + palbociclib reduced colony formation more effectively than the single drugs or the palbociclib + fulvestrant doublet. These long-term effects could prevent expansion of drug-resistant BC cells.**
- **The gedatolisib + palbociclib and gedatolisib + fulvestrant doublets each reduced colony formation more effectively than the palbociclib + fulvestrant doublet.**

Figure 9. In vivo Efficacy of the Gedatolisib/Fulvestrant/Palbociclib Combination

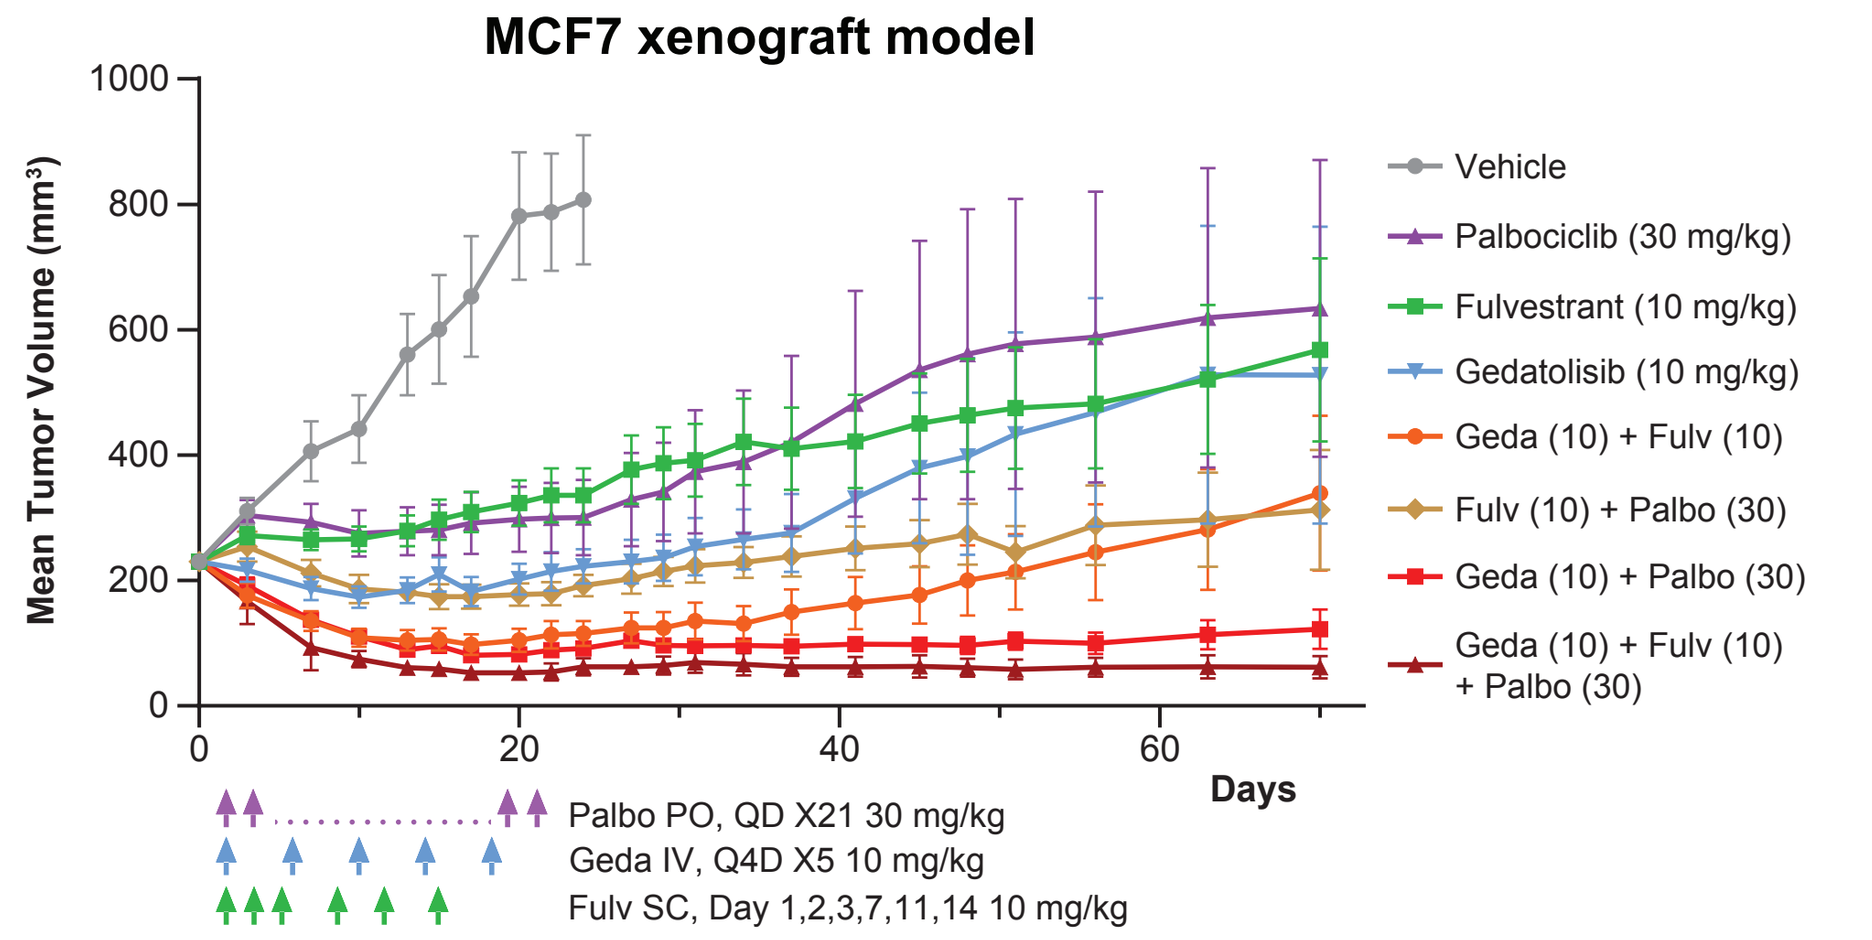
MCF7 xenograft model
Mean Tumor Volume (mm³)
Days
Vehicle
Palbociclib (30 mg/kg)
Fulvestrant (10 mg/kg)
Gedatolisib (10 mg/kg)
Geda (10) + Fulv (10)
Fulv (10) + Palbo (30)
Geda (10) + Palbo (30)
Geda (10) + Fulv (10) + Palbo (30)
Palbo PO, QD X21 30 mg/kg
Geda IV, Q4D X5 10 mg/kg
Fulv SC, Day 1,2,3,7,11,14 10 mg/kg

Treatment	TGI % on Day 20	p values: Vehicle	Geda	Geda + Palbo
Vehicle	NA	NA		
Fulvestrant	83	<0.00001		
Palbociclib	88	<0.00001		
Gedatolisib	105	<0.00001	NA	
Palbociclib Fulvestrant	109	<0.00001	>0.05	
Gedatolisib Fulvestrant	123	<0.00001	<0.0001	
Gedatolisib Palbociclib	127	<0.00001	<0.00001	NA
Gedatolisib Palbociclib Fulvestrant	132	<0.00001	<0.00001	<0.01

SCID female mice were xenografted with MCF7 cells in the mammary fat pad and treated with gedatolisib +/- fulvestrant +/- palbociclib in the indicated combination for up to 21 days. Tumor volumes were assessed during and after treatment up to a total of 70 days. Tumor growth inhibition and statistical significance at day 20 are reported in the table on the right.

- **The triplet combination gedatolisib + fulvestrant + palbociclib induced greater and more durable tumor growth inhibition (TGI) and regression than single drugs or the doublet combinations.**

Figure 10. Comparison of Gedatolisib and Single Node PAM Inhibitors Combined with Fulvestrant and Palbociclib

A

Drug	PAM specificity	Cell-free Assay Ki (nM): PI3Kα	PI3Kβ	PI3Kγ	PI3Kδ	mTOR	AKT1/2/3	Ref
Gedatolisib	PanPI3K/mTOR	0.4	6	8	6	1	-	[11]
Alpelisib	PI3Kα	5	>1000	250	290	-	-	[12]
Capivasertib	AKT	-	-	-	-	-	3/8/8	[13]
Everolimus	mTOR	-	-	-	-	1.6	-	[14]

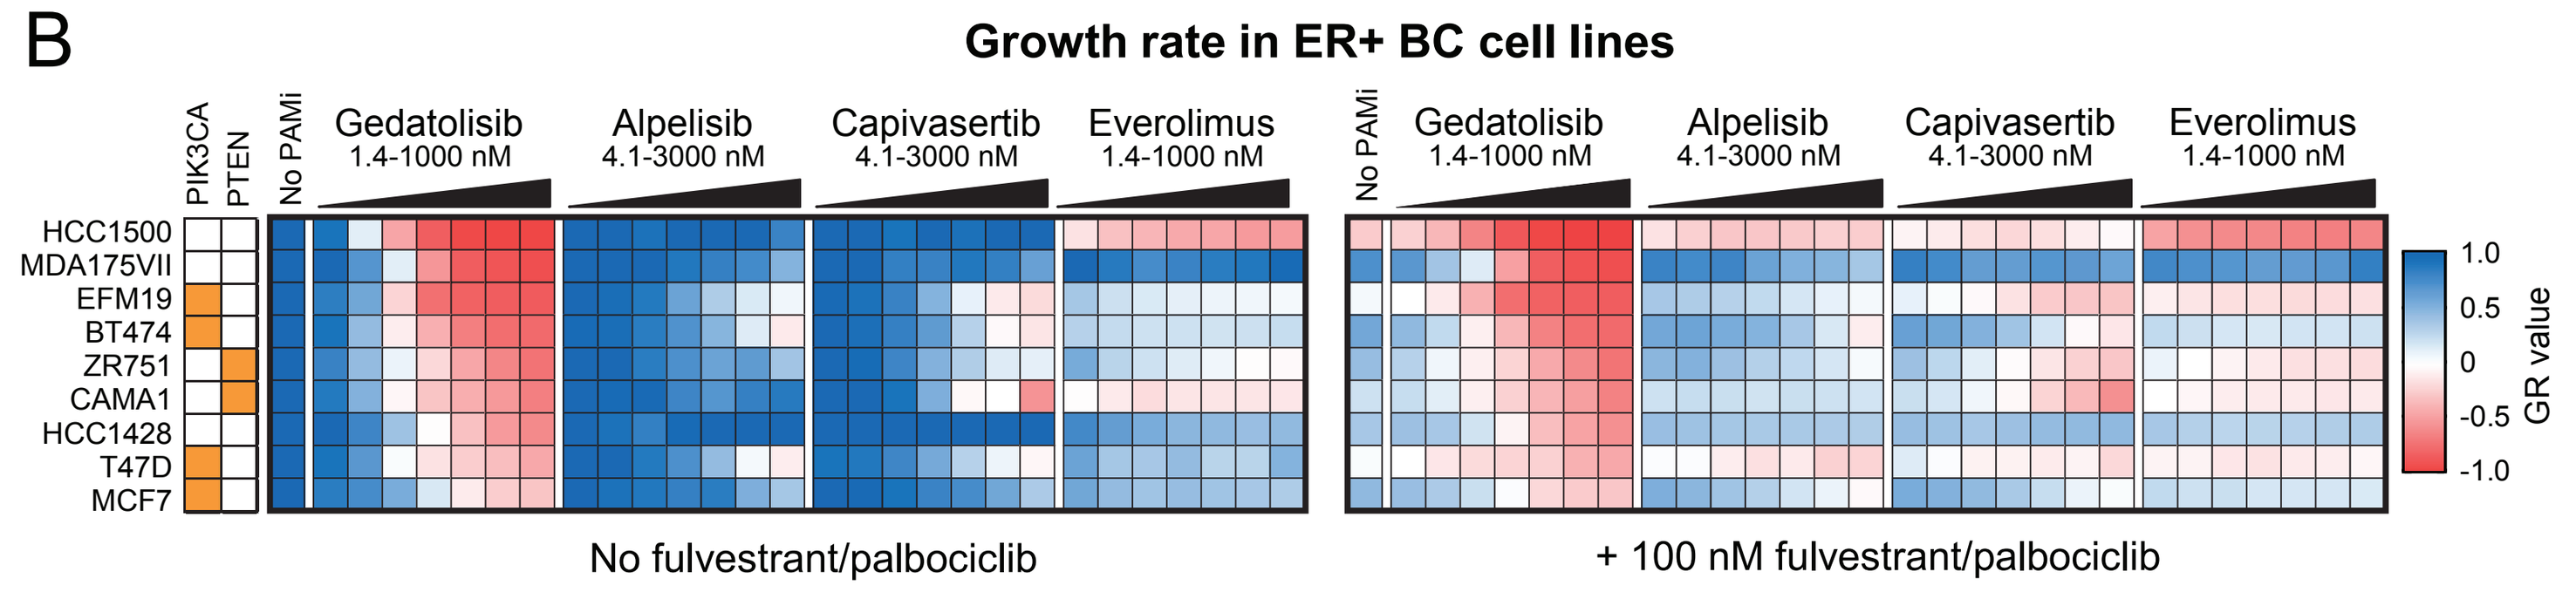
B
Growth rate in ER+ BC cell lines
PIK3CA
PTEN
No PAMi
Gedatolisib 1.4-1000 nM
Alpelisib 4.1-3000 nM
Capivasertib 4.1-3000 nM
Everolimus 1.4-1000 nM
HCC1500
MDA175VII
EFM19
BT474
ZR751
CAMA1
HCC1428
T47D
MCF7
No fulvestrant/palbociclib
+ 100 nM fulvestrant/palbociclib
GR value

A panel of ER+ BC cells with various PAM pathway mutational status was treated with escalating doses of PAM inhibitors in combination with 100 nM fulvestrant and 100 nM palbociclib for 6 days and analyzed by GR metrics. **(A)** Potency and specificity of the PAM inhibitors tested. **(B)** Heatmaps showing average GR values (n=2) in the cell lines treated. PAMi = PAM inhibitor

- **Gedatolisib exerted greater anti-proliferative and cytotoxic effects than single-node PAM inhibitors both as single agent and in combination with fulvestrant and palbociclib in BC cell lines, regardless of PIK3CA/PTEN mutations.**

SUMMARY AND CONCLUSIONS

- **The gedatolisib + fulvestrant + palbociclib triplet exerted greater anti-proliferative and cytotoxic effects relative to single agents or doublet combinations in ER+ BC cells lines, regardless of PIK3CA/PTEN genetic alterations.**
- **Treatment with palbociclib and/or fulvestrant as single agents can trigger early adaptations leading to reactivation of PAM pathway and cell cycling, which can be prevented by gedatolisib.**
- **The triplet combination induced a more effective and durable inhibition of functions controlled by the PAM, ER, and CDK pathways (e.g., cell cycle, cell survival, protein synthesis, glucose metabolism) relative to single agent treatments.**
- **The triplet combination induced greater and more durable tumor growth inhibition than single agents or doublet combinations in an ER+/HER2- BC xenograft model.**
- **Compared to FDA-approved single-node PAM inhibitors, gedatolisib exerted greater anti-proliferative and cytotoxic effects both as monotherapy and in combination with fulvestrant and palbociclib.**
- **Gedatolisib has previously demonstrated promising preliminary clinical efficacy and safety data in patients with ER+/HER2- ABC in combination with hormonal therapy and palbociclib [9].**
- **A Phase 3 study (VIKTORIA-1, NCT05501886) evaluating gedatolisib plus fulvestrant with and without palbociclib is underway in patients with ER+/HER2- ABC whose disease progressed while on treatment with a CDK4/6 inhibitor.**
- **Another Phase 3 trial (VIKTORIA-2, EU CT #2024-518933-28-00) is planned to evaluate gedatolisib in combination with fulvestrant and a CDK4/6 inhibitor as first line treatment for patients with HR+/HER2- ABC.**

This presentation is the intellectual property of the authors/presenter. Contact srossetti@celcuity.com for more information.