Gedatolisib (Ged) vs Other Regimens Post-CDK4/6i and Endocrine Therapy (ET) in PIK3CA Wild-Type (WT) HR+/ HER2- Advanced Breast Cancer (aBC): A Network Meta-Analysis (NMA)

Sara M. Tolaney[1], VK Gadi[2], Rena D. Callahan[3], Barbara Pistilli[4], Evan Popoff[5], Michael J. Zoratti[5], Alana M. Stilla[5], Thelma Ubani[5], Anik R. Patel[5], Sidonie Niba[6], Chiemeka Ike[6], Seth A. Wander[7]

[1]Dana-Farber Cancer Institute, Boston, MA, USA; [2]University of Illinois Cancer Center, Chicago, IL, USA; [3]UCLA Health Jonsson Comprehensive Cancer Center, Los Angeles, CA, USA; [4]Gustave Roussy, Villejuif, France; [5]RainCity Analytics, Vancouver, BC, Canada; [6]Celcuity, Minneapolis, MN, USA; [7]Massachusetts General Hospital, Boston, MA, USA

FPN #1873P
Presented at the
2026 ESMO Annual Congress

BACKGROUND

- The treatment landscape of advanced breast cancer (aBC) is evolving rapidly. Notably, the introduction of cyclin-dependent kinase 4/6 inhibitors (CDK4/6i) has substantially improved clinical outcomes for patients with HR+/HER2− aBC.
- However, treatment options are limited for patients with *PIK3CA* wild-type (WT) HR+/HER2− aBC post-progression on CDK4/6i and endocrine therapy (ET).
- Gedatolisib, a kinase inhibitor of class I PI3K isoforms (α, β, δ, γ) and mTOR complexes mTORC1 and mTORC2, has emerged as an important investigational therapy with promising activity in combination with ET and CDK4/6i.[1]
- On July 14, 2026, gedatolisib received US FDA approval for use in combination with fulvestrant, with or without palbociclib, for the treatment of adult patients with HR+, HER2-negative locally advanced or metastatic breast cancer without a *PIK3CA* mutation detected following progression on or after treatment with at least one line of ET in the metastatic setting.
- In the absence of head-to-head trials to inform clinical decision-making, we conducted a systematic literature review (SLR) and NMA to assess the relative efficacy of gedatolisib vs alternate regimens.

OBJECTIVE

- The objective of this study was to estimate the relative efficacy of gedatolisib-based regimens vs approved/emerging treatments for patients with HR+/HER2− aBC and *PIK3CA* WT tumors who were previously treated with CDK4/6i.

METHODS

- Systematic searches were conducted in MEDLINE, Embase, and CENTRAL, supplemented by hand searches of pertinent conferences and US FDA CDER reports, for randomized controlled trials of systemic treatments evaluated among patients with HR+/HER2− aBC with CDK4/6i experience and *PIK3CA* WT tumors.
- Hazard ratios (HRs) for PFS were extracted or estimated from reconstructed Kaplan-Meier data.
- A Bayesian NMA was performed for time-to-event outcomes on the log-HR scale with a normal likelihood and vague priors. Fixed-effects models were selected.
- A sensitivity analysis was conducted where the assumption of equivalent imlunestrant and fulvestrant efficacy was relaxed by including the EMBER-3 fulvestrant arm, using the broader CDK4/6-experienced *PIK3CA*-pooled population due to data availability limitations.
- All analyses were performed using R (version 4.3.1) and JAGS.

METHODS (cont'd.)

Figure 1. PICOS and Study Selection

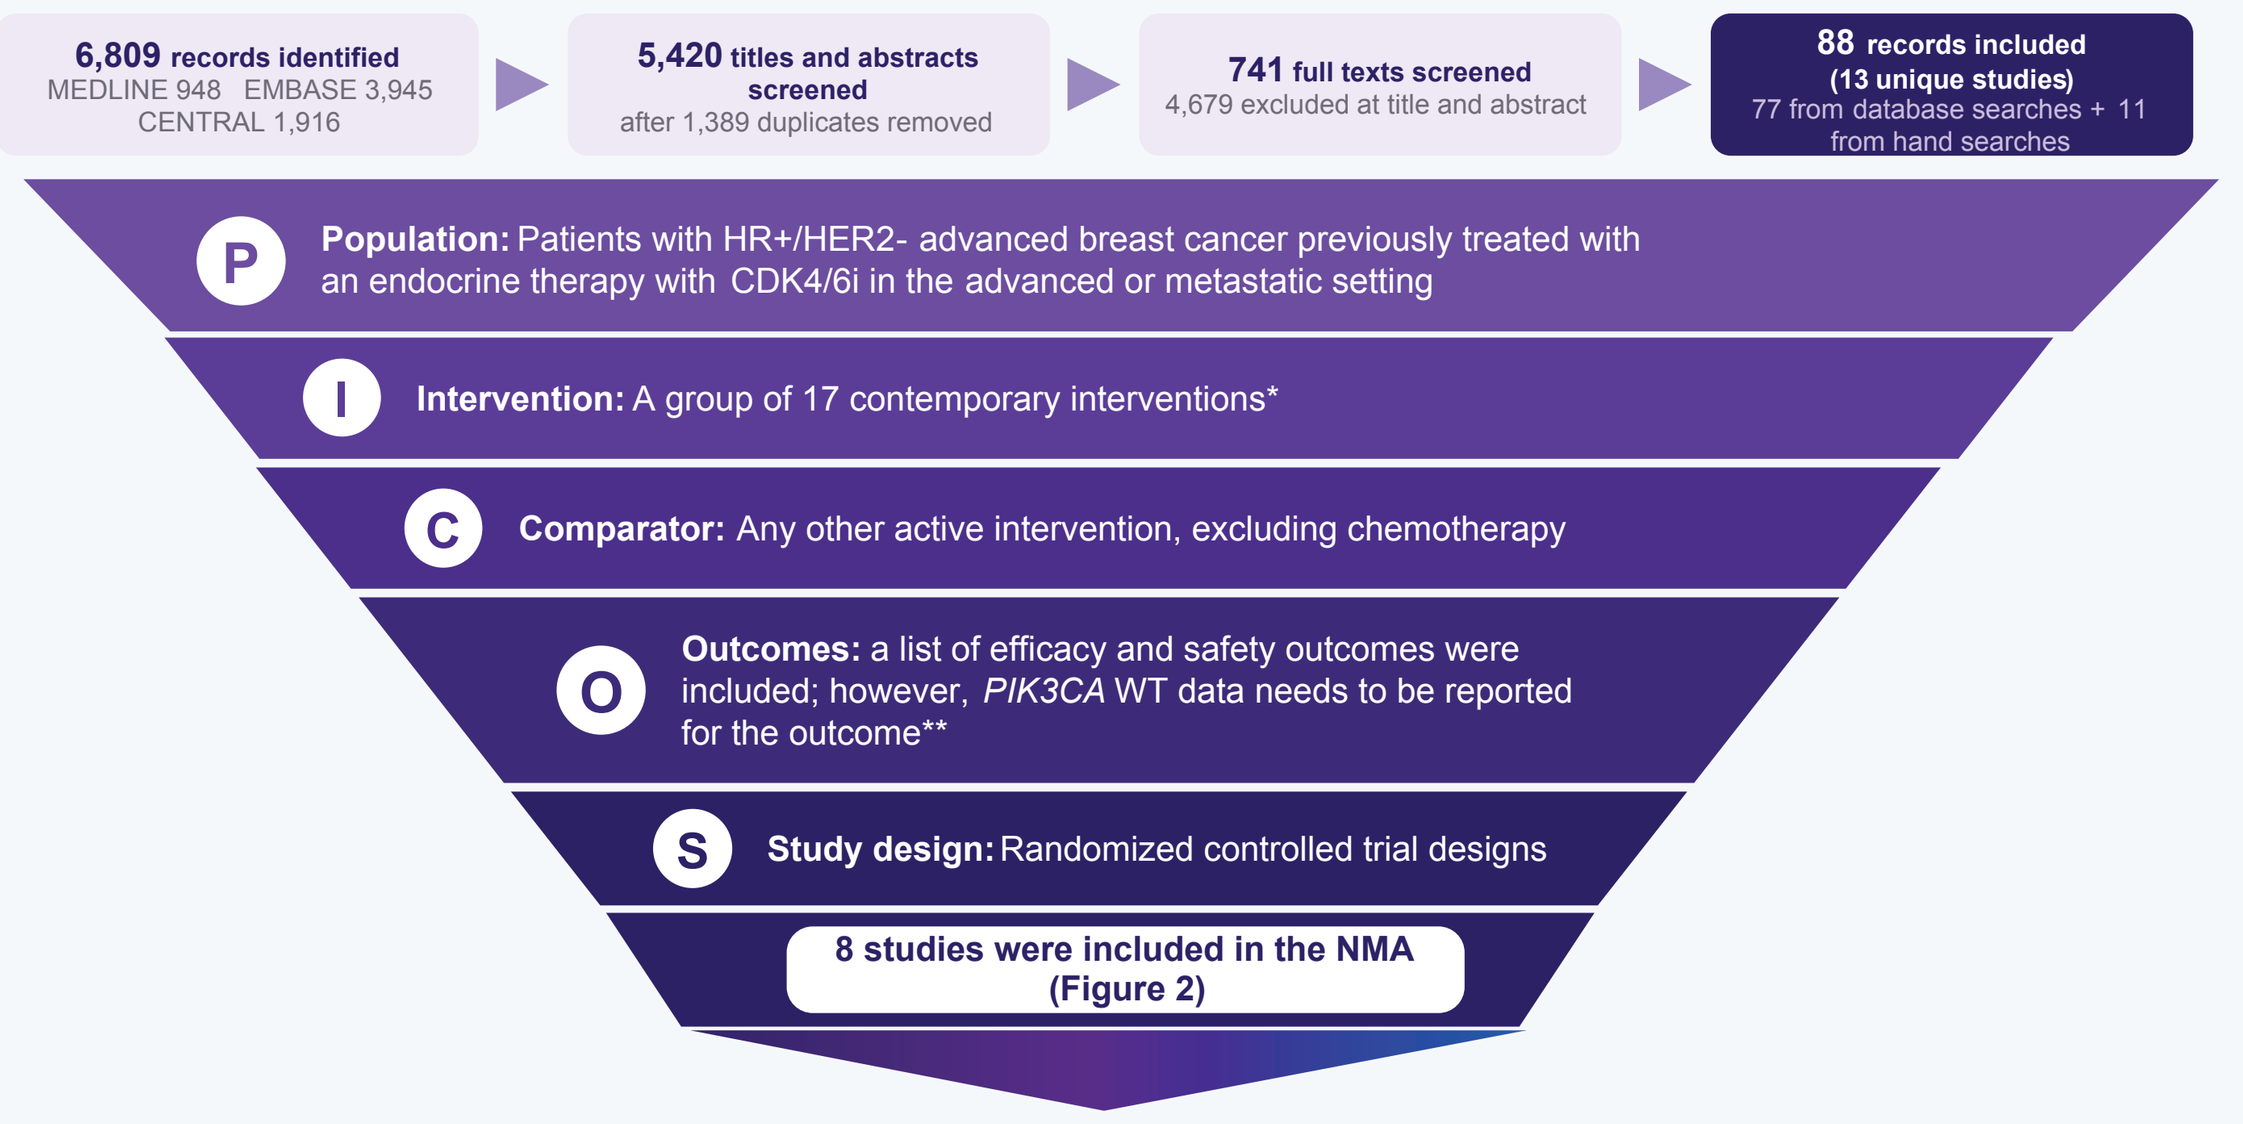
6,809 records identified
MEDLINE 948 EMBASE 3,945
CENTRAL 1,916
5,420 titles and abstracts screened
after 1,389 duplicates removed
741 full texts screened
4,679 excluded at title and abstract
88 records included
(13 unique studies)
77 from database searches + 11 from hand searches
P
Population: Patients with HR+/HER2- advanced breast cancer previously treated with an endocrine therapy with CDK4/6i in the advanced or metastatic setting
I
Intervention: A group of 17 contemporary interventions*
C
Comparator: Any other active intervention, excluding chemotherapy
O
Outcomes: a list of efficacy and safety outcomes were included; however, PIK3CA WT data needs to be reported for the outcome**
S
Study design: Randomized controlled trial designs
8 studies were included in the NMA
(Figure 2)

*Gedatolisib + palbociclib + fulvestrant; Gedatolisib + fulvestrant; Fulvestrant; Everolimus + exemestane; Fam-trastuzumab deruxtecan-nxki; Sacituzumab govitecan; Elacestrant; Imlunestrant monotherapy; Imlunestrant + abemaciclib; Giredestrant + everolimus; Vepdegestrant; Palbociclib + fulvestrant; Abemaciclib + fulvestrant; Ribociclib + fulvestrant; Olaparib; Talazoparib; Datopotamab deruxtecan
**Efficacy outcomes: Overall survival; Progression-free survival; Duration of response; Median duration of time on treatment; Time to next treatment; Time to discontinuation; Overall (or objective) response rate; Disease control rate; Complete response; Partial response; Stable disease; Clinical benefit rate.
**Safety outcomes: Any adverse event; Any serious adverse event; Treatment discontinuation due to adverse events; Stomatitis; Neutropenia; Nausea; Rash; Vomiting; Fatigue; Diarrhea; Hyperglycemia

Figure 2. Network of Evidence

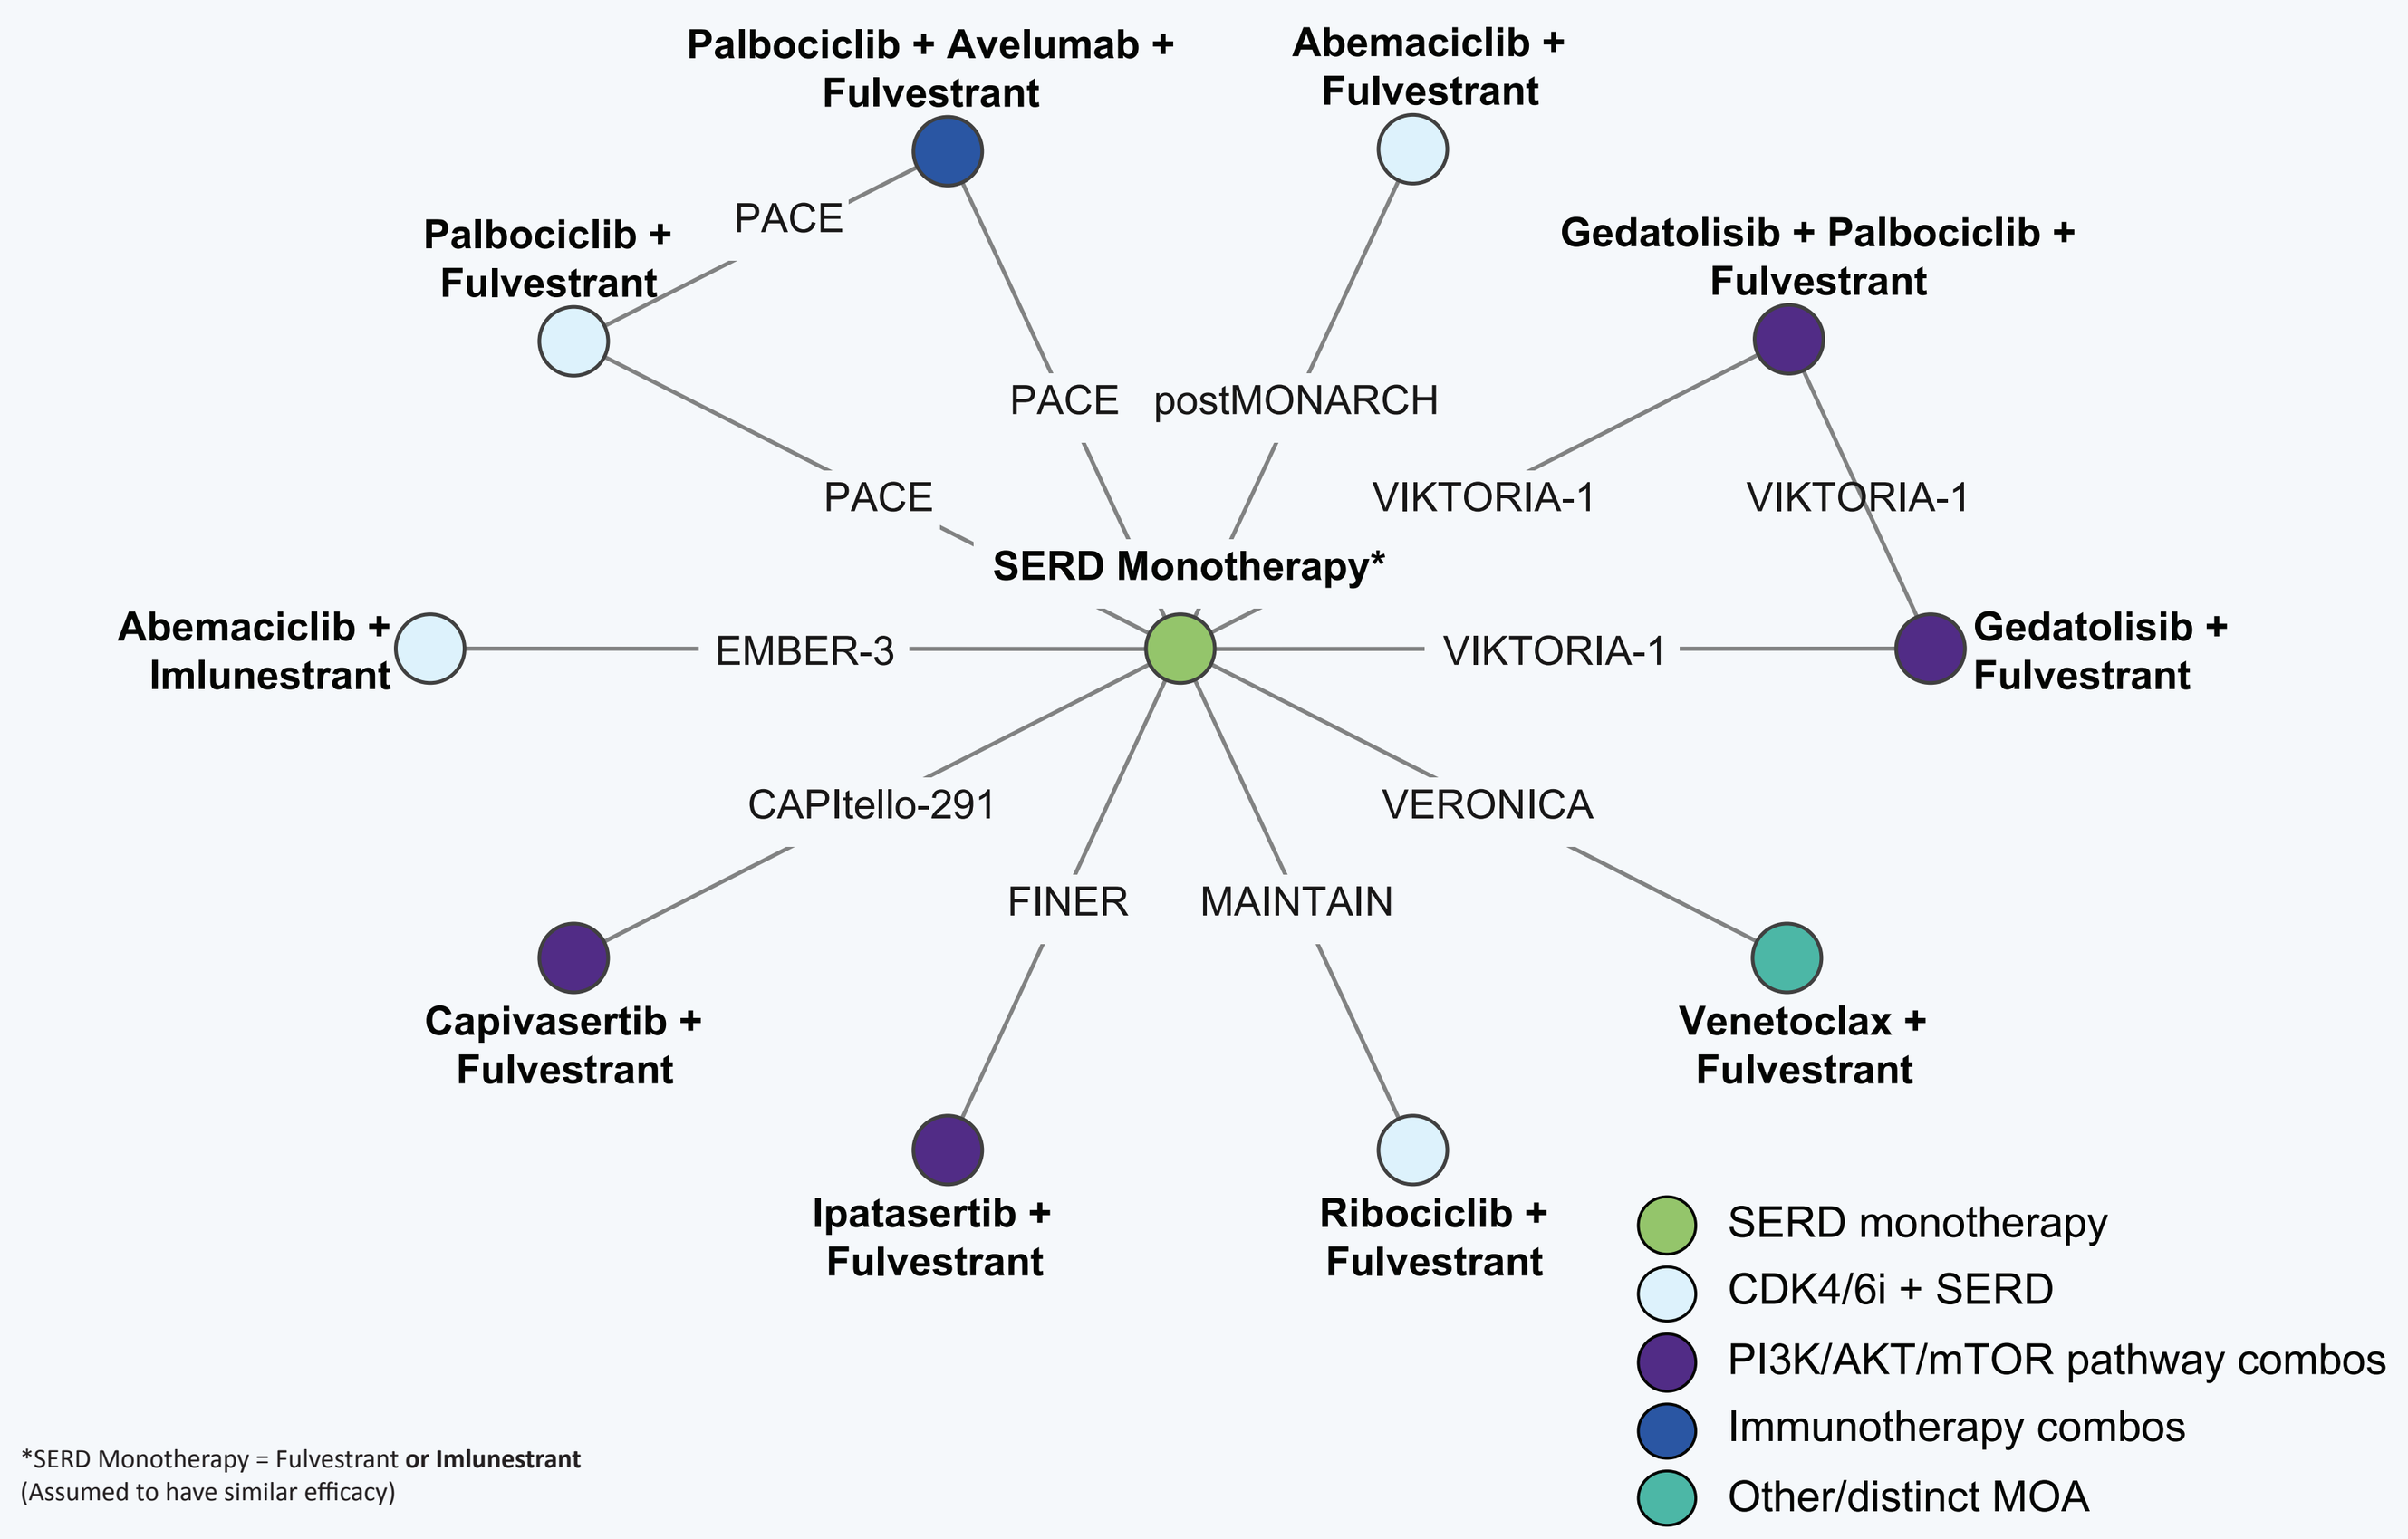
Palbociclib + Avelumab + Fulvestrant
Abemaciclib + Fulvestrant
Palbociclib + Fulvestrant
Gedatolisib + Palbociclib + Fulvestrant
PACE
PACE
postMONARCH
PACE
VIKTORIA-1
VIKTORIA-1
SERD Monotherapy*
Abemaciclib + Imlunestrant
EMBER-3
VIKTORIA-1
Gedatolisib + Fulvestrant
CAPItello-291
VERONICA
FINER
MAINTAIN
Capivasertib + Fulvestrant
Venetoclax + Fulvestrant
Ipatasertib + Fulvestrant
Ribociclib + Fulvestrant
SERD monotherapy
CDK4/6i + SERD
PI3K/AKT/mTOR pathway combos
Immunotherapy combos
Other/distinct MOA

*SERD Monotherapy = Fulvestrant **or Imlunestrant**
(Assumed to have similar efficacy)

RESULTS

- Database searches were conducted on February 13, 2026, and 13 studies were identified (**Figure 1**)
 - Five were not eligible for inclusion due to the absence of a shared comparator (DESTINY-Breast06, TROPiCS-02 and evERA) or availability of *PIK3CA* WT data only in patients with an *ESR1* mutation (EMERALD and VERITAC-2).
 - Finally, eight studies were eligible for inclusion in the analysis set.
- Progression-free survival (PFS) was selected as the primary endpoint for the NMA because response and safety outcomes were not consistently reported for the *PIK3CA* WT population across all studies.
- In EMBER-3, *PIK3CA* WT subgroup data were reported only for imlunestrant plus abemaciclib versus imlunestrant monotherapy.
 - Imlunestrant was assumed to have comparable efficacy as fulvestrant for the purposes of this analysis.
- Study and patient characteristics of included studies are presented in **Table 1** and **Table 2**. Key differences in inclusion criteria across the studies included prior CDK4/6i exposure, postmenopausal status and prior treatment with chemotherapy.
- The gedatolisib triplet demonstrated a statistically significant improvement in PFS relative to all non-gedatolisib regimens, except ribociclib + fulvestrant, which showed a non-significant trend in favor of the gedatolisib triplet (**Figure 3**).
- The percentage of patients progression-free at 6 months for gedatolisib triplet was 20% higher compared to imlunestrant+ abemaciclib and 38% higher vs capivasertib + fulvestrant.
- Similarly, statistically significant improvement in PFS was observed with gedatolisib doublet regimen compared to other therapies except for ribociclib + fulvestrant and imlunestrant + abemaciclib (**Figure 4**).
- Gedatolisib doublet also demonstrated a significant 31% PFS gain at 6 months compared to capivasertib + fulvestrant. PFS gain compared to imlunestrant + abemaciclib was also observed; however, it was not statistically significant.
- Results of the sensitivity analysis were consistent with the base-case analysis for both the gedatolisib triplet and doublet vs abemaciclib + imlunestrant, indicating that inclusion of the EMBER-3 fulvestrant arm using the broader CDK4/6-experienced, *PIK3CA*-pooled population did not materially alter the comparative findings.

RESULTS (cont'd.)

Table 1: Study Characteristics of Trials in the *PIK3CA* WT Network

Trial	Study design	Stratification factors	Intervention	n	% *PIK3CA* WT	*PIK3CA* WT and prior CDK4/6i progression (n)	Follow-up
CAPItello-291[2]	Double blind, Phase III; Placebo controlled	Liver metastases (yes/no), prior CDK4/6i (yes/no); Region	Capivasertib + fulvestrant	355	56%	92	13 months
			Placebo + fulvestrant	353	62%	121	12.7 months
EMBER-3[3]	Open label, Phase III; Active controlled	Prior CDK4/6i treatment (yes or no), the presence or absence of visceral metastases, and geographic region (East Asia, North America or Western Europe, and other regions)	Imlunestrant + abemaciclib	213	59%	75	28.5 months
			Imlunestrant	331	61%	69	
			Fulvestrant or exemestane	330	61%	--	
FINER[4]	Triple blind, Phase III; Placebo controlled	PIK3CA/PTEN AKT altered vs WT/unknown; Primary vs secondary endocrine resistance	Ipatasertib + fulvestrant	125	54%	68	15.2 months
			Placebo + fulvestrant	125	57%	71	
MAINTAIN[5]	Double blind, Phase II; Placebo controlled	--	Ribociclib + ET	60	55%	33	18.2 months
			Placebo + ET	59	41%	24	
PACE[6]	Open label, Phase II; Active controlled	Receipt or nonreceipt of chemotherapy between previous CDK4/6i and random assignment	Avelumab + palbociclib + fulvestrant	54	57%	31	23.6 months
			Palbociclib + fulvestrant	111	57%	63	
			Fulvestrant	55	65%	36	
postMONARCH[7]	Double blind, Phase III; Placebo controlled	Geography; Presence or absence of visceral metastasis (including lung, liver, brain, pleural, or peritoneal involvement); Duration of previous CDK4/6i therapy (>/< 12 months in the metastatic setting or recurrence during/after treatment in the adjuvant setting)	Abemaciclib + fulvestrant	182	57%	103	13 months
			Placebo + fulvestrant	186	49%	92	
VERONICA[8]	Open label, Phase II; Active controlled	Prior LoTs in the locally advanced or metastatic breast cancer settings (one versus two); BCL2 clinical status (high versus low)	Venetoclax + fulvestrant	51	57%	29	9.9 months
			Fulvestrant	52	62%	32	
VIKTORIA-1 (Study 1)[1]	Open label, Phase III; Active controlled	Lung/liver metastases (yes/no), TTP on immediate prior therapy (≤ or >6 months), region (US/Canada or ROW)	Gedatolisib + palbociclib + fulvestrant	131	100%	131	10.1 months
			Gedatolisib + fulvestrant	130	100%	130	
			Fulvestrant	131	100%	131	

Abbreviations; AKT, protein kinase B; BCL, B-cell lymphoma; CDK4/6i; cyclin-dependent kinase 4/6 inhibitor; ET, endocrine therapy; LoT, line of therapy; PIK3CA; phosphatidylinositol-4, 5-bisphosphate 3-kinase catalytic subunit α PTEN; phosphatase and tensin homolog; ROW, rest of world; TTP, time to progression; WT, wild-type

Table 2: Patient Baseline Characteristics of ITT Population in Included Trials

Characteristic[†]	CAPItello-291 *Cap+Ful vs Pbo+Ful N=708*	EMBER-3 *Iml+Abe vs Iml vs Ful/Exe N=874*	FINER *Ipa+Ful vs Pbo+Ful N=250*	MAINTAIN *Rib+Ful/Exe vs Pbo+Ful/Exe N=119*	PACE *Ave+Pal+Ful vs Pal+Ful vs Ful N=220*	postMONARCH *Abe+Ful vs Pbo+Ful N=368*	VERONICA *Ven+Ful vs Ful N=108*	VIKTORIA-1 (Study 1) *Ged+Pal+Ful vs Ged+Ful vs Ful N=392*
Median age	59; 58	62; 61; 62	60; 59	55, 59	58; 55; 58	58; 61	58; 59.5	57; 57; 54
Visceral metastasis	67.5%	55.5%	--	59.7%	60%	60%	87.4%	80.4%
Liver metastasis	43.2%	30%	66.4%	--	--	37.8%	61.2%	58.2%
Bone-only metastasis	14.5%	23.9%	16.8%	18.5%	13.6%	20.1%	1.9%	6.6%
*ESR1*m	--	37%	--	42.9%	54%	45.3%	42.6%	--
Post-menopausal	78%	86%	--	--	81.7%	100%	--	73.9%
Prior CDK4/6i	70%**	60%**	100%	100%	100%	99.7%	100%	100%
Prior chemo in aBC	18.2%	0%	--	9.2%*	16.4%*	0%*	0%	0%
Prior ET in aBC	86.9%	67%	--	83.2%*	100%*	--	100%*	97.7%

[†]Proportion of male patients was less than 2% across all studies.
*Metastatic setting
**ITT population distribution; the subgroup data with 100% prior CDK 4/6i use were used in this analysis
Abbreviations: aBC, advanced breast cancer; Abe, Abemaciclib; Ave, Avelumab; Cap, Capivasertib; CDK4/6i; cyclin-dependent kinase 4/6 inhibitor; ET, endocrine therapy; Exe, Exemestane; Ful, Fulvestrant; Ged, Gedatolisib; Iml, Imlunestrant; Ipa, Ipatasertib; Pal, Palbociclib; Pbo, Placebo; Rib, Ribociclib; Ven, Venetoclax

Figure 3: Relative Efficacy (PFS) of Ged Triplet vs Other Therapies

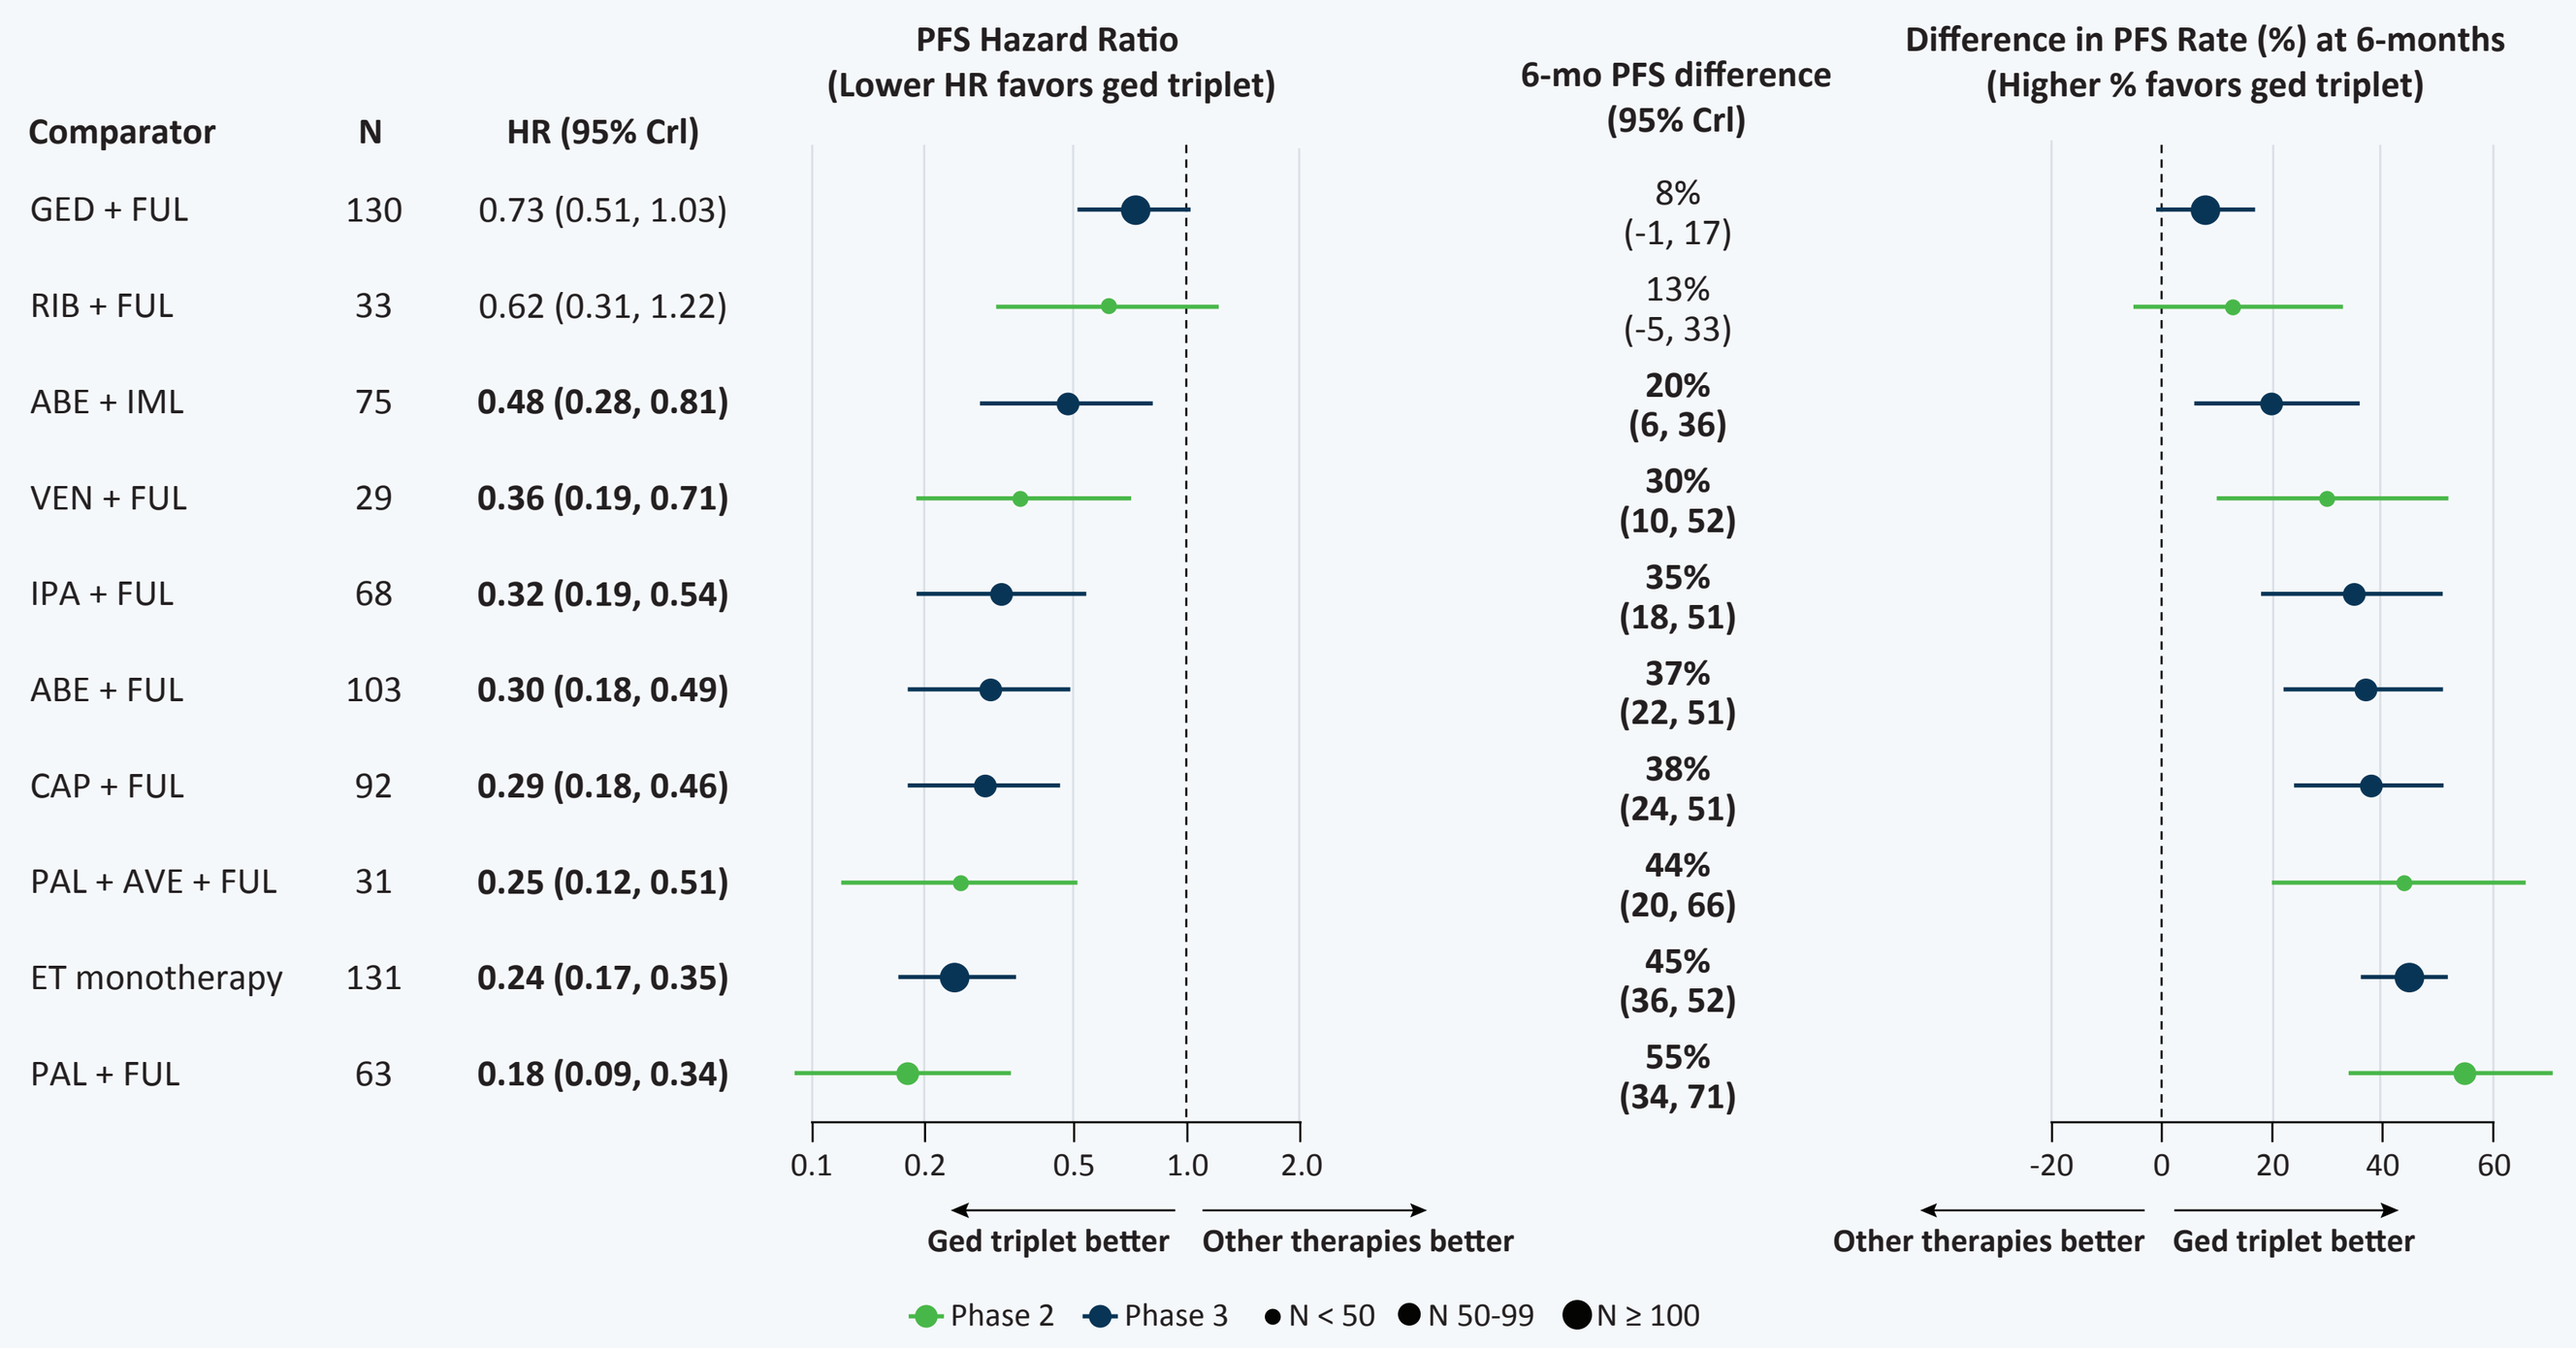
Comparator
N
HR (95% CrI)
PFS Hazard Ratio
(Lower HR favors ged triplet)
6-mo PFS difference
(95% CrI)
Difference in PFS Rate (%) at 6-months
(Higher % favors ged triplet)
GED + FUL 130 0.73 (0.51, 1.03) 8% (-1, 17)
RIB + FUL 33 0.62 (0.31, 1.22) 13% (-5, 33)
ABE + IML 75 0.48 (0.28, 0.81) 20% (6, 36)
VEN + FUL 29 0.36 (0.19, 0.71) 30% (10, 52)
IPA + FUL 68 0.32 (0.19, 0.54) 35% (18, 51)
ABE + FUL 103 0.30 (0.18, 0.49) 37% (22, 51)
CAP + FUL 92 0.29 (0.18, 0.46) 38% (24, 51)
PAL + AVE + FUL 31 0.25 (0.12, 0.51) 44% (20, 66)
ET monotherapy 131 0.24 (0.17, 0.35) 45% (36, 52)
PAL + FUL 63 0.18 (0.09, 0.34) 55% (34, 71)
0.1 0.2 0.5 1.0 2.0
Ged triplet better
Other therapies better
-20 0 20 40 60
Other therapies better
Ged triplet better
Phase 2
Phase 3
N < 50
N 50-99
N ≥ 100

Statistically significant values are **bolded**
ABE, Abemaciclib; AVE, Avelumab; CAP, Capivasertib; CrI, credible interval; ET, endocrine therapy; FUL, Fulvestrant; GED, Gedatolisib; HR, hazard ratio; IML, Imlunestrant; IPA, Ipatasertib; PAL, Palbociclib; PFS, progression-free survival; RIB, Ribociclib; VEN, Venetoclax

Figure 4: Relative Efficacy (PFS) of Ged Doublet vs Other Therapies

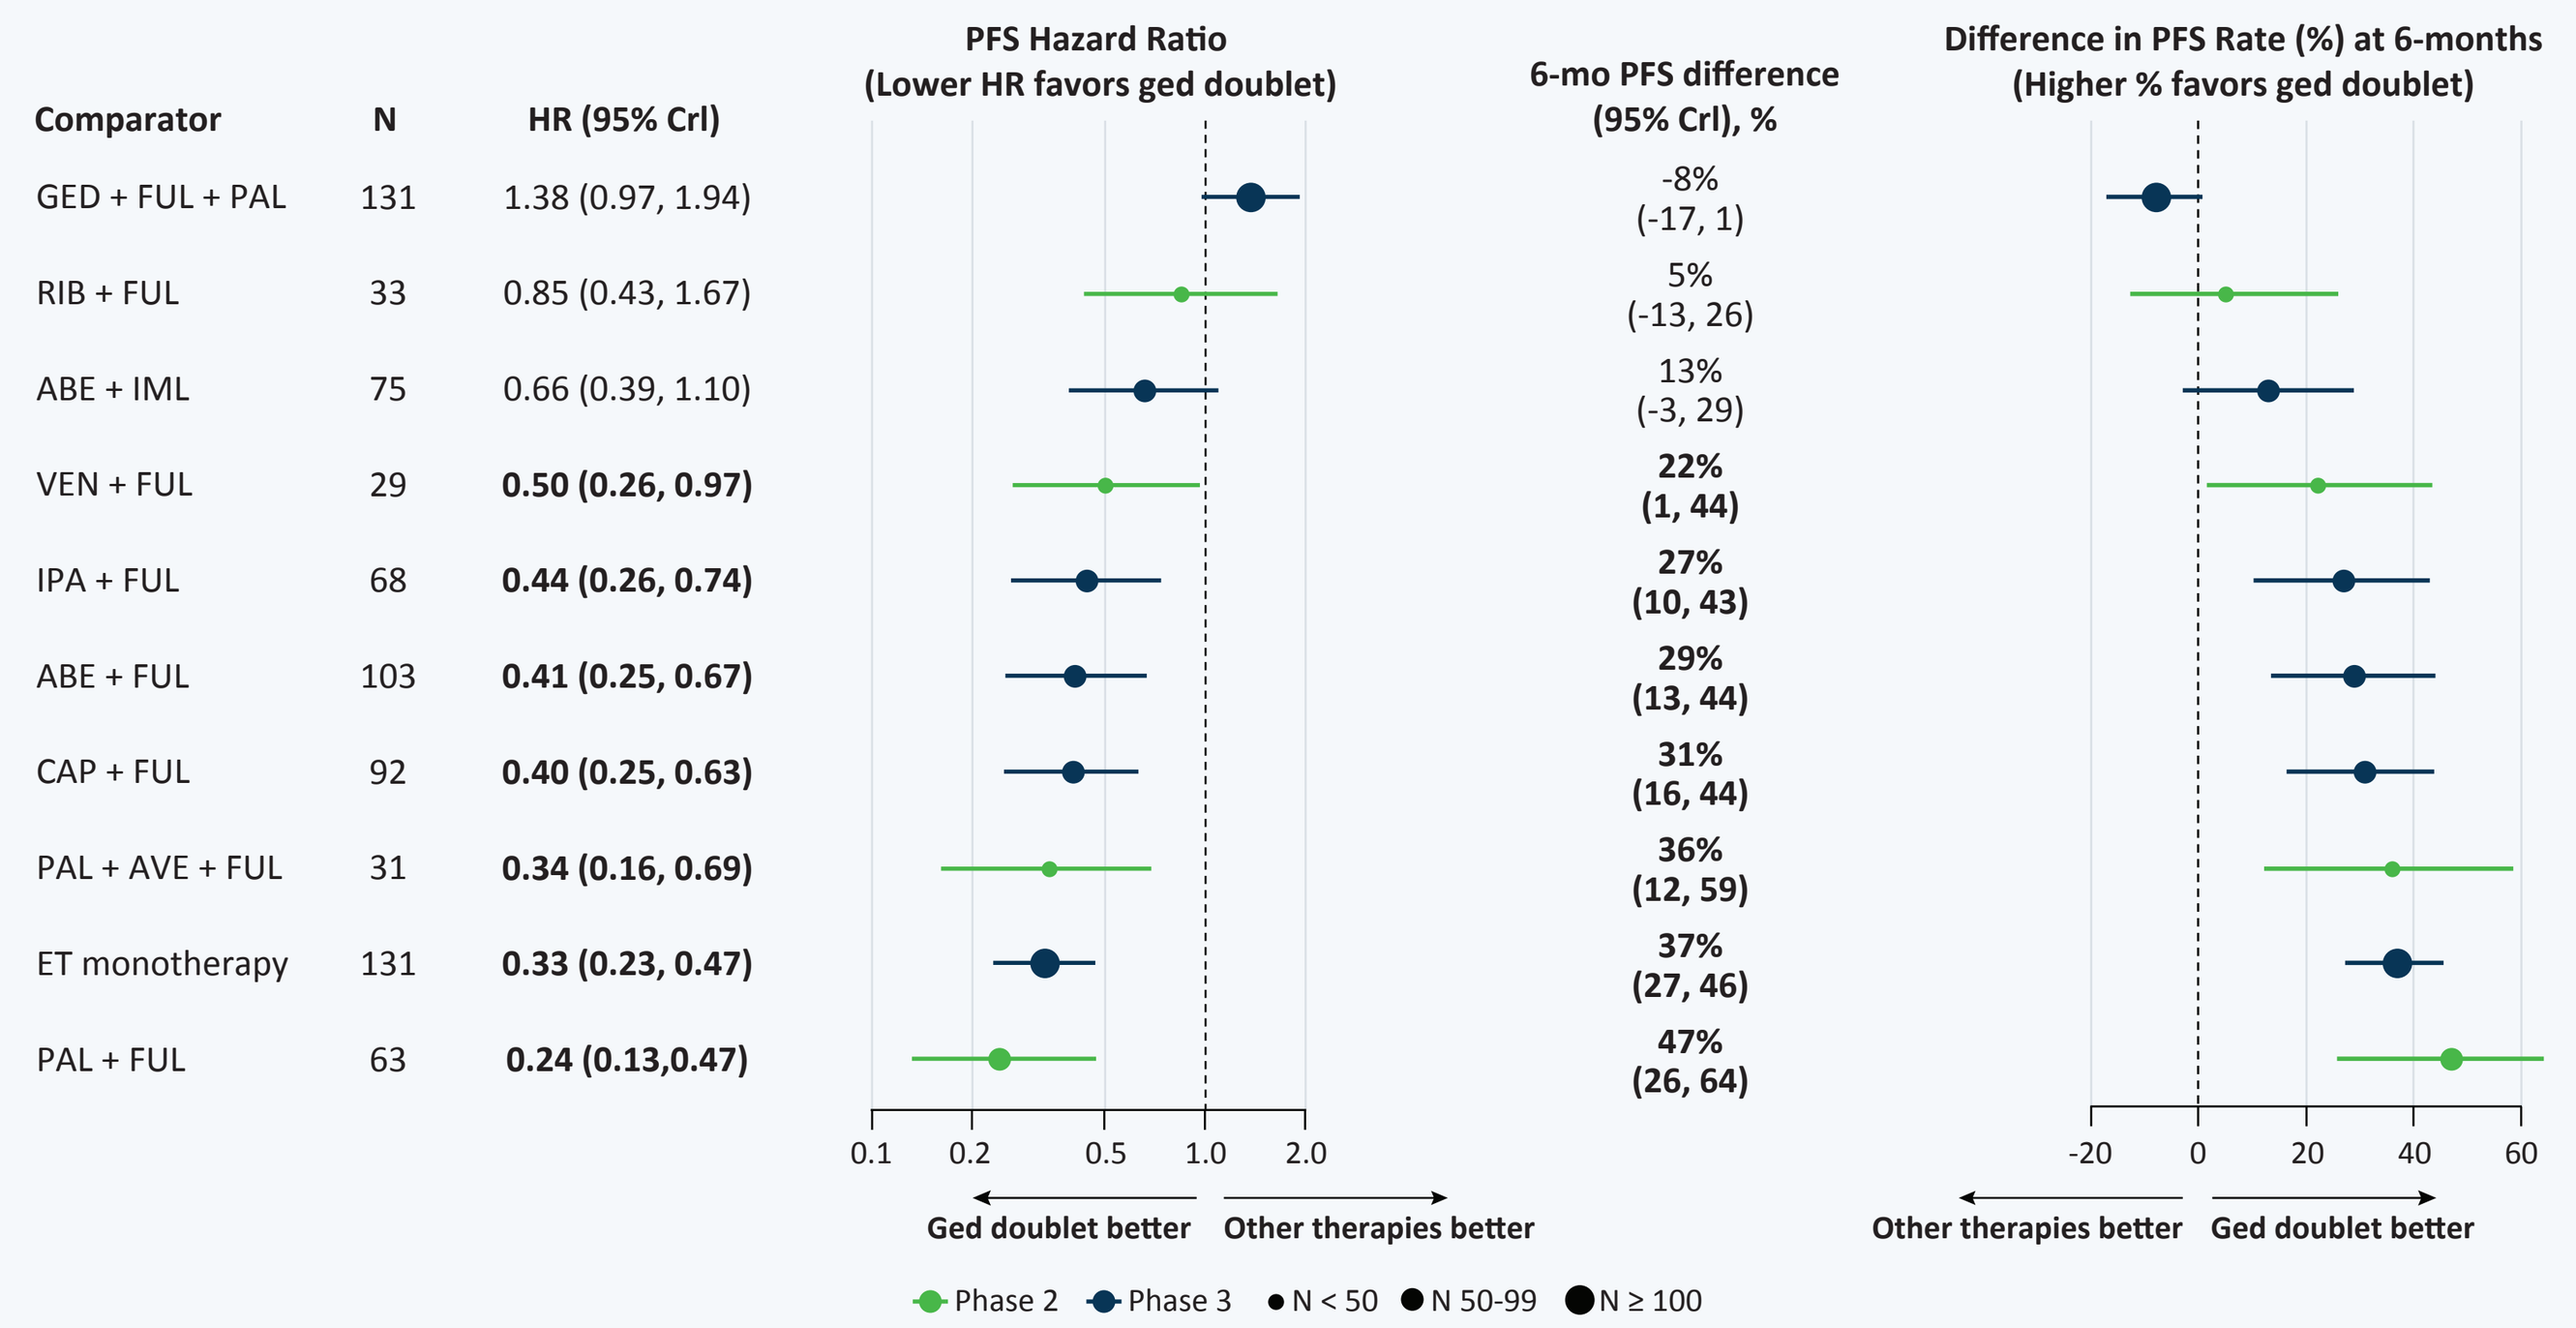
Comparator
N
HR (95% CrI)
PFS Hazard Ratio
(Lower HR favors ged doublet)
6-mo PFS difference
(95% CrI), %
Difference in PFS Rate (%) at 6-months
(Higher % favors ged doublet)
GED + FUL + PAL 131 1.38 (0.97, 1.94) -8% (-17, 1)
RIB + FUL 33 0.85 (0.43, 1.67) 5% (-13, 26)
ABE + IML 75 0.66 (0.39, 1.10) 13% (-3, 29)
VEN + FUL 29 0.50 (0.26, 0.97) 22% (1, 44)
IPA + FUL 68 0.44 (0.26, 0.74) 27% (10, 43)
ABE + FUL 103 0.41 (0.25, 0.67) 29% (13, 44)
CAP + FUL 92 0.40 (0.25, 0.63) 31% (16, 44)
PAL + AVE + FUL 31 0.34 (0.16, 0.69) 36% (12, 59)
ET monotherapy 131 0.33 (0.23, 0.47) 37% (27, 46)
PAL + FUL 63 0.24 (0.13,0.47) 47% (26, 64)
0.1 0.2 0.5 1.0 2.0
Ged doublet better
Other therapies better
-20 0 20 40 60
Other therapies better
Ged doublet better
Phase 2
Phase 3
N < 50
N 50-99
N ≥ 100

Statistically significant values are **bolded**
ABE, Abemaciclib; AVE, Avelumab; CAP, Capivasertib; CrI, credible interval; ET, endocrine therapy; FUL, Fulvestrant; GED, Gedatolisib; HR, hazard ratio; IML, Imlunestrant; IPA, Ipatasertib; PAL, Palbociclib; PFS, progression-free survival; RIB, Ribociclib; VEN, Venetoclax

CONCLUSION

- The NMA estimated favorable PFS outcomes with ged-based regimens compared to several comparator treatments, supporting their potential role in patients with *PIK3CA*-WT progression on CDK4/6i.
- Separately reported positive results from the VIKTORIA-1 *PIK3CA*-mutant cohort, which were not included in the present analysis, provide important new evidence for future comparative analyses.
- This analysis used a robust methodology for indirect treatment comparisons, although findings should be interpreted in the context of cross-trial differences and assumptions inherent to NMA.

REFERENCES:
1. Hurvitz et al. *J Clin Oncol*. 2026;44(12):1108-1119. doi:10.1200/JCO-25-02643. **2.** Turner NC, et al. *N Engl J Med*. 2023;388:2058–2070. doi:10.1056/NEJMoa2214131. **3.** Jhaveri KL, et al. *N Engl J Med*. 2025;392:1189–1202. doi:10.1056/NEJMoa2410858. **4.** Chia SKL, et al. *J Clin Oncol*. 2025;43:LBA1005. doi:10.1200/JCO.2025.43.17_suppl.LBA1005. **5.** Kalinsky K, et al. *J Clin Oncol*. 2023;41:4004–4013. doi:10.1200/JCO.22.02392. **6.** Mayer EL, et al. *J Clin Oncol*. 2024;42:2050–2060. doi:10.1200/JCO.23.01940. **7.** Kalinsky K, et al. *J Clin Oncol*. 2025;43:1101–1112. doi:10.1200/JCO-24-02086. **8.** Lindeman GJ, et al. *Clin Cancer Res*. 2022;28:3256–3267. doi:10.1158/1078-0432.CCR-21-3811.

DISCLOSURES:
- Presenting author disclosures: Sara Tolaney declares institutional research support from Genentech/Roche, Merck, Exelixis, Pfizer/SeaGen, Lilly, Novartis, Bristol Myers Squibb, AstraZeneca, NanoString Technologies, Gilead, OncoPep, Daiichi Sankyo, Menarini/Stemline, and Jazz Pharma ; and consulting fees from Novartis, Pfizer/SeaGen, Merck, Eli Lilly, AstraZeneca, Genentech/Roche, Eisai, Bristol Myers Squibb/Systimmune, Daiichi Sankyo, Gilead, Blueprint Medicines, Reveal Genomics, Sumitovant Biopharma, Artios Pharma, Menarini/Stemline, Bayer, Jazz Pharmaceuticals, Cullinan Oncology, Circle Pharma, Arvinas, BioNTech, Launch Therapeutics, Zuellig Pharma, Johnson & Johnson/Ambrx, Bicycle Therapeutics, BeOne Therapeutics/BeiGene, Mersana, Summit Therapeutics, Avenzo Therapeutics, Atkis Oncology, Boehringer Ingelheim, Celcuity, Samsung Bioepis, Olema Pharmaceuticals, Tempus, Boundless Bio, and Denali Therapeutics.
- Email address for presenting author: sara_tolaney@dfci.harvard.edu
- Study sponsored by Celcuity

Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission of the authors.